CD47 Expression as a Predictive Biomarker for Evorpacept in HER2-positive Gastric/Gastroesophageal Cancer from the Phase 2 Randomized ASPEN-06 Trial

Zev Wainberg,[1] Keun-Wook Lee,[2] Josep Tabernero,[3] Eric Van Cutsem,[4] Clelia Coutzac,[5] Christelle De La Fouchardière,[6] Jeeyun Lee,[7] Sun Young Rha,[8] Yoon-Koo Kang,[9] Charlie Zhang,[10] Daniel Brickman,[10] Philip Fanning,[10] Alison Forgie,[10] Athanasios C. Tsiatis,[10] Alan Sandler,[10] Kohei Shitara[11]

[1]Ronald Reagan UCLA Medical Center, Los Angeles, CA, USA; [2]Seoul National University Bundang Hospital, Seoul National University College of Medicine, Seongnam, Korea; [3]Vall d'Hebrón University Hospital, Barcelona, Spain; [4]University Hospitals Gasthuisberg & KU Leuven, Leuven, Belgium; [5]Centre Léon Bérard, Lyon, France; [6]Institut Paoli-Calmettes, Marseille, France; [7]Samsung Medical Center, Seoul, Korea; [8]Severance Hospital, Seoul, Korea; [9]Asan Medical Center, Seoul, Korea; [10]ALX Oncology, South San Francisco, CA, USA; [11]National Cancer Center Hospital East, Kashiwa-shi, Japan

Background

- CD47 is a transmembrane protein that acts as a major checkpoint in the innate immune system; it is widely expressed on the surface of healthy cells, including hematopoietic cells[1–3]
- Cancer cells often overexpress CD47 to evade the innate immune system[1–4]
- Evorpacept blocks the CD47-SIRPα interaction, enhancing ADCP of cancer cells, and it has an inactive Fc domain to minimize impact on healthy cells (**Figure 1**)[5]
- ASPEN-06 is a global, randomized study evaluating evorpacept plus trastuzumab, ramucirumab, and paclitaxel (TRP) vs TRP in the 2L/3L treatment of patients with gastric/GEJ cancer who previously received HER2-targeted therapy[6]
 - In the Phase 2 portion of the study, evorpacept + TRP demonstrated an ORR of 41.3% and DOR of 15.7 months vs 26.6% and 9.1 months, respectively, with TRP alone, and safety data confirmed the combination of evorpacept and TRP is feasible. The full dataset was previously presented (N=127; data cut-off date December 2, 2024)[6]
- Maximum ADCP activity of evorpacept plus trastuzumab is expected to occur when their respective targets, CD47 and HER2, are sufficiently expressed[7–9]
 - There is evidence of variability in CD47 expression in gastric cancers[10]
 - Loss of HER2 expression after HER2-targeted treatment has been observed in some patients with gastric cancers[11]
- Here we present updated exploratory efficacy results to address the key question:
 - Is the benefit of evorpacept + TRP driven by patients with both CD47 tumor expression and retained HER2 expression?

FIGURE 1. Evorpacept Plus Trastuzumab Mechanism of Action

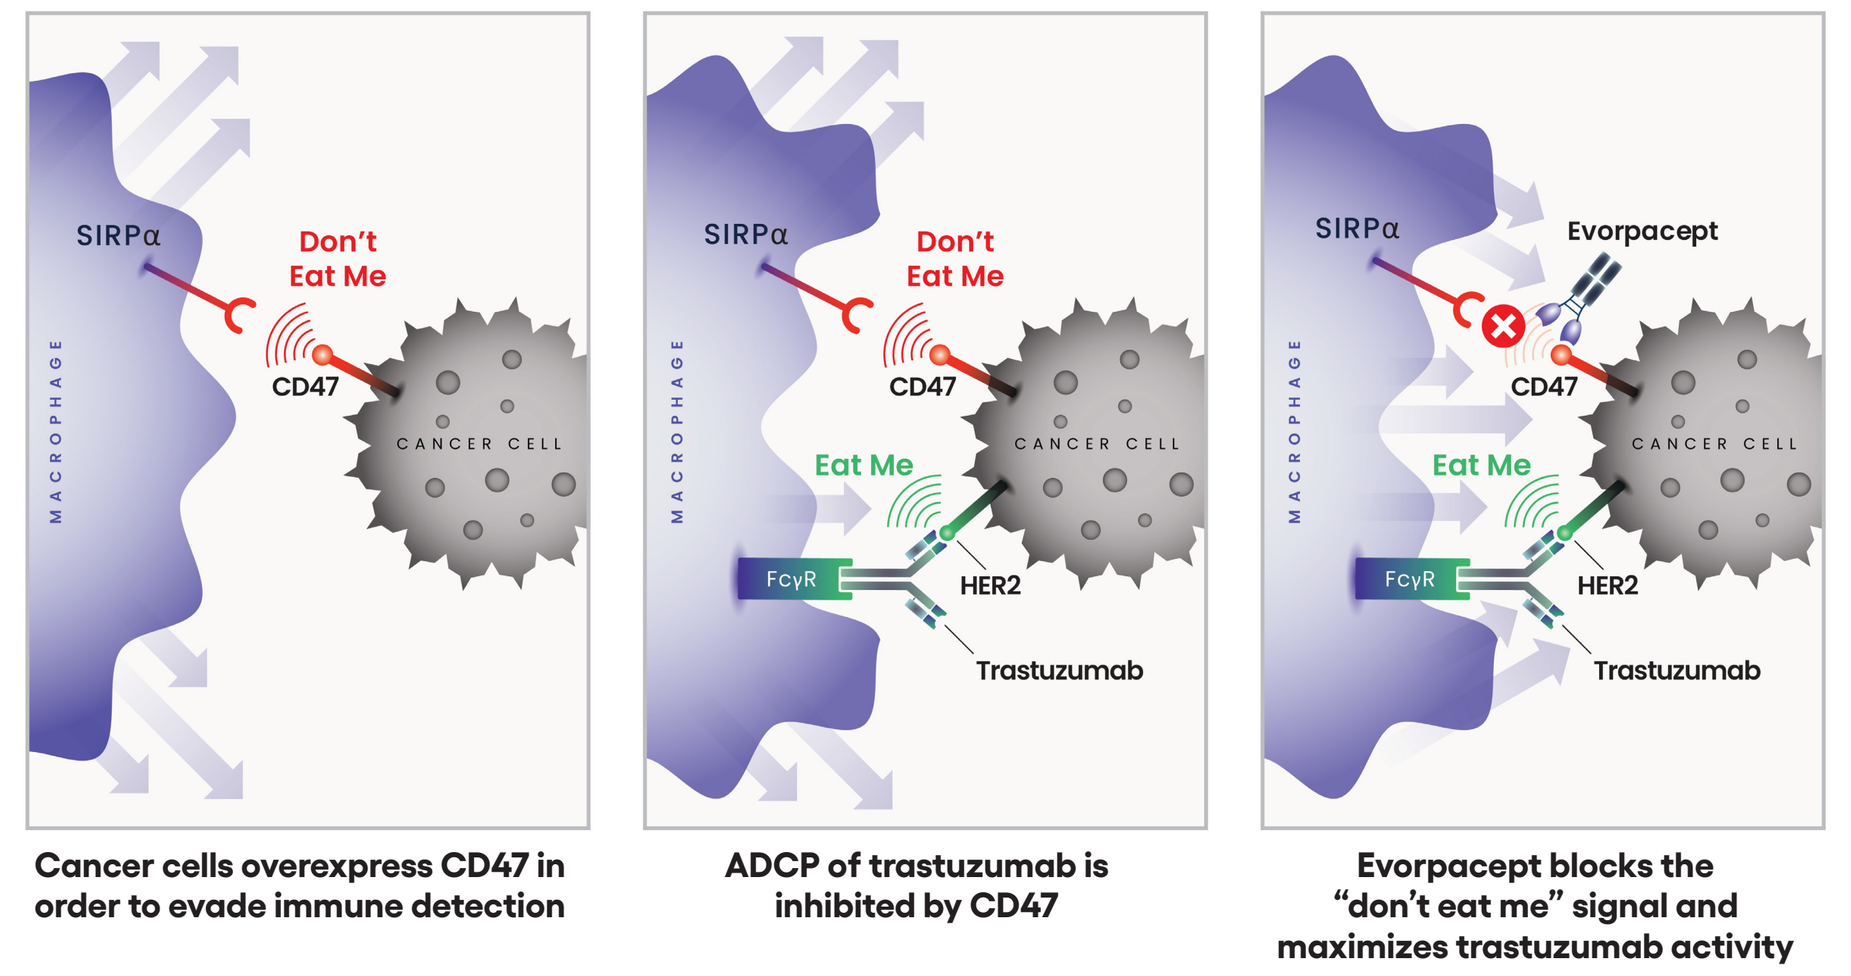
SIRPα
Don't Eat Me
CD47
CANCER CELL
MACROPHAGE
Eat Me
FcγR
HER2
Trastuzumab
Evorpacept
Cancer cells overexpress CD47 in order to evade immune detection
ADCP of trastuzumab is inhibited by CD47
Evorpacept blocks the "don't eat me" signal and maximizes trastuzumab activity

Methods

- **Objective:** To determine whether tumor CD47 expression is a predictive biomarker for evorpacept in the patient subgroup with HER2+ status in a fresh biopsy or HER2 amplification by ctDNA from the ASPEN-06 study (**Figure 2**)
- HER2+ status was determined locally (if available) or centrally, using an FDA-approved test for gastric/GEJ cancer, on the most recent tissue sample
- As fresh biopsies were only available for a subset of patients, HER2 (*ERBB2*) gene amplification in ctDNA extracted from plasma samples collected on day 1 of cycle 1 prior to dosing was also evaluated
- CD47 protein expression was evaluated by IHC and manually scored according to the percent of tumor cells with fractional membrane staining intensities of 0, 1+, 2+, and 3+. Tumors with ≥10% and <10% of cells with membrane CD47 IHC3+ staining were defined as CD47-High and CD47-Low, respectively
- All efficacy analyses (ORR and PFS per investigator and OS) were evaluated in the patient subgroup with HER2+ status in a fresh biopsy or HER2 amplification by ctDNA and by CD47-High vs CD47-Low expression
- OS by CD47-High vs CD47-Low expression was also evaluated in the TRP arm, to investigate whether CD47 expression is a negative prognostic factor

FIGURE 2. ASPEN-06 Study Design

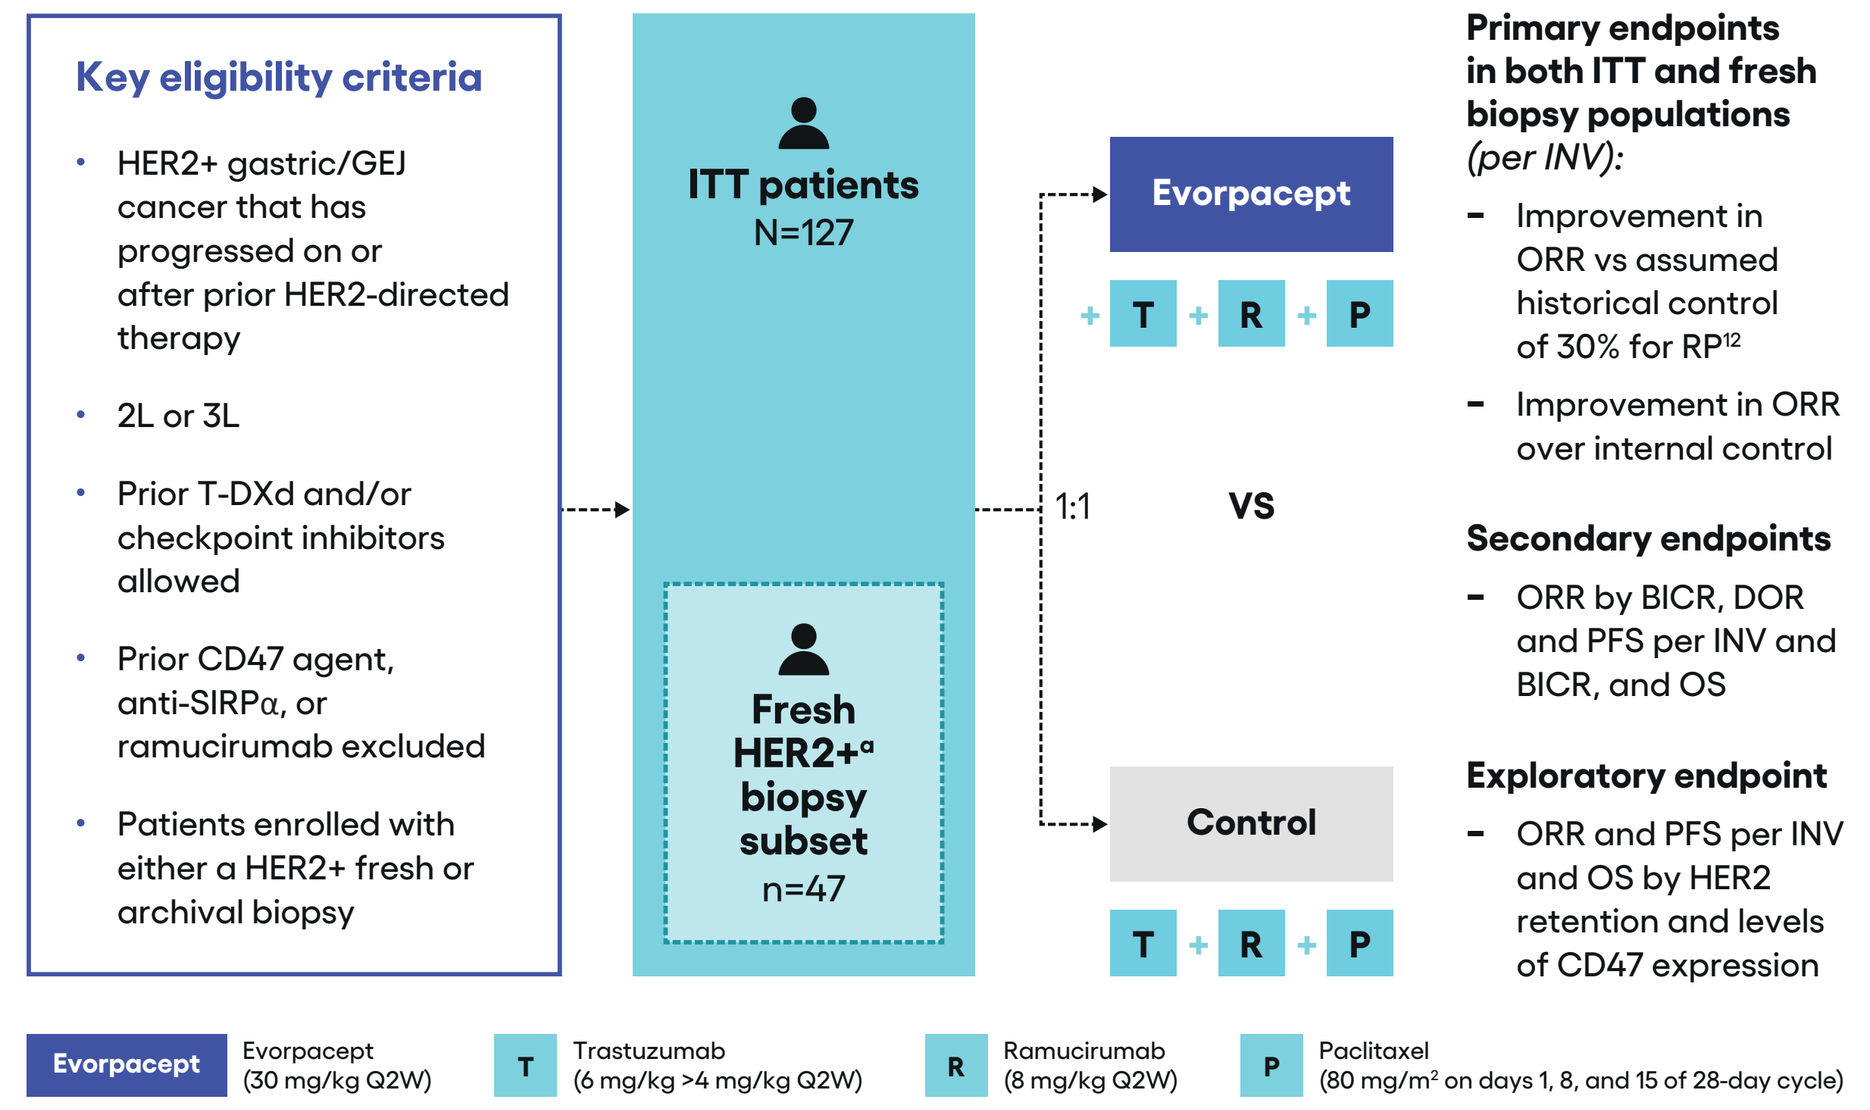
Key eligibility criteria
HER2+ gastric/GEJ cancer that has progressed on or after prior HER2-directed therapy
2L or 3L
Prior T-DXd and/or checkpoint inhibitors allowed
Prior CD47 agent, anti-SIRPα, or ramucirumab excluded
Patients enrolled with either a HER2+ fresh or archival biopsy
ITT patients N=127
Fresh HER2+ᵃ biopsy subset n=47
1:1
Evorpacept
T + R + P
VS
Control
T + R + P
Primary endpoints in both ITT and fresh biopsy populations (per INV):
Improvement in ORR vs assumed historical control of 30% for RP[12]
Improvement in ORR over internal control
Secondary endpoints
ORR by BICR, DOR and PFS per INV and BICR, and OS
Exploratory endpoint
ORR and PFS per INV and OS by HER2 retention and levels of CD47 expression
Evorpacept (30 mg/kg Q2W)
T Trastuzumab (6 mg/kg >4 mg/kg Q2W)
R Ramucirumab (8 mg/kg Q2W)
P Paclitaxel (80 mg/m² on days 1, 8, and 15 of 28-day cycle)

ᵃFresh HER2+ is defined as biopsies that were HER2+ after receiving prior HER2-targeted treatment

Results

- In total, 95 patients had HER2+ status in a fresh biopsy or HER2 amplification by ctDNA; 90 of these patients were also evaluable for CD47 expression (CD47-High, n=43; CD47-Low, n=47; data cut-off date May 15, 2025; **Figure 3**)
- In patients with retained HER2 expression and CD47-High expression, greater efficacy was observed in exploratory endpoints (per INV) for evorpacept + TRP vs TRP
 - ORR was 65.0% vs 26.1% and median DOR was 25.5 vs 8.4 months (**Figures 4** and **5**)
 - Median PFS was 18.4 vs 7.0 months (HR 0.39 [95% CI 0.17, 0.86]; **Figure 6A**)
 - Median OS was 17.0 vs 9.9 months (HR 0.63 [95% CI 0.30, 1.31]; **Figure 6B**)

FIGURE 3. Biomarker Consort

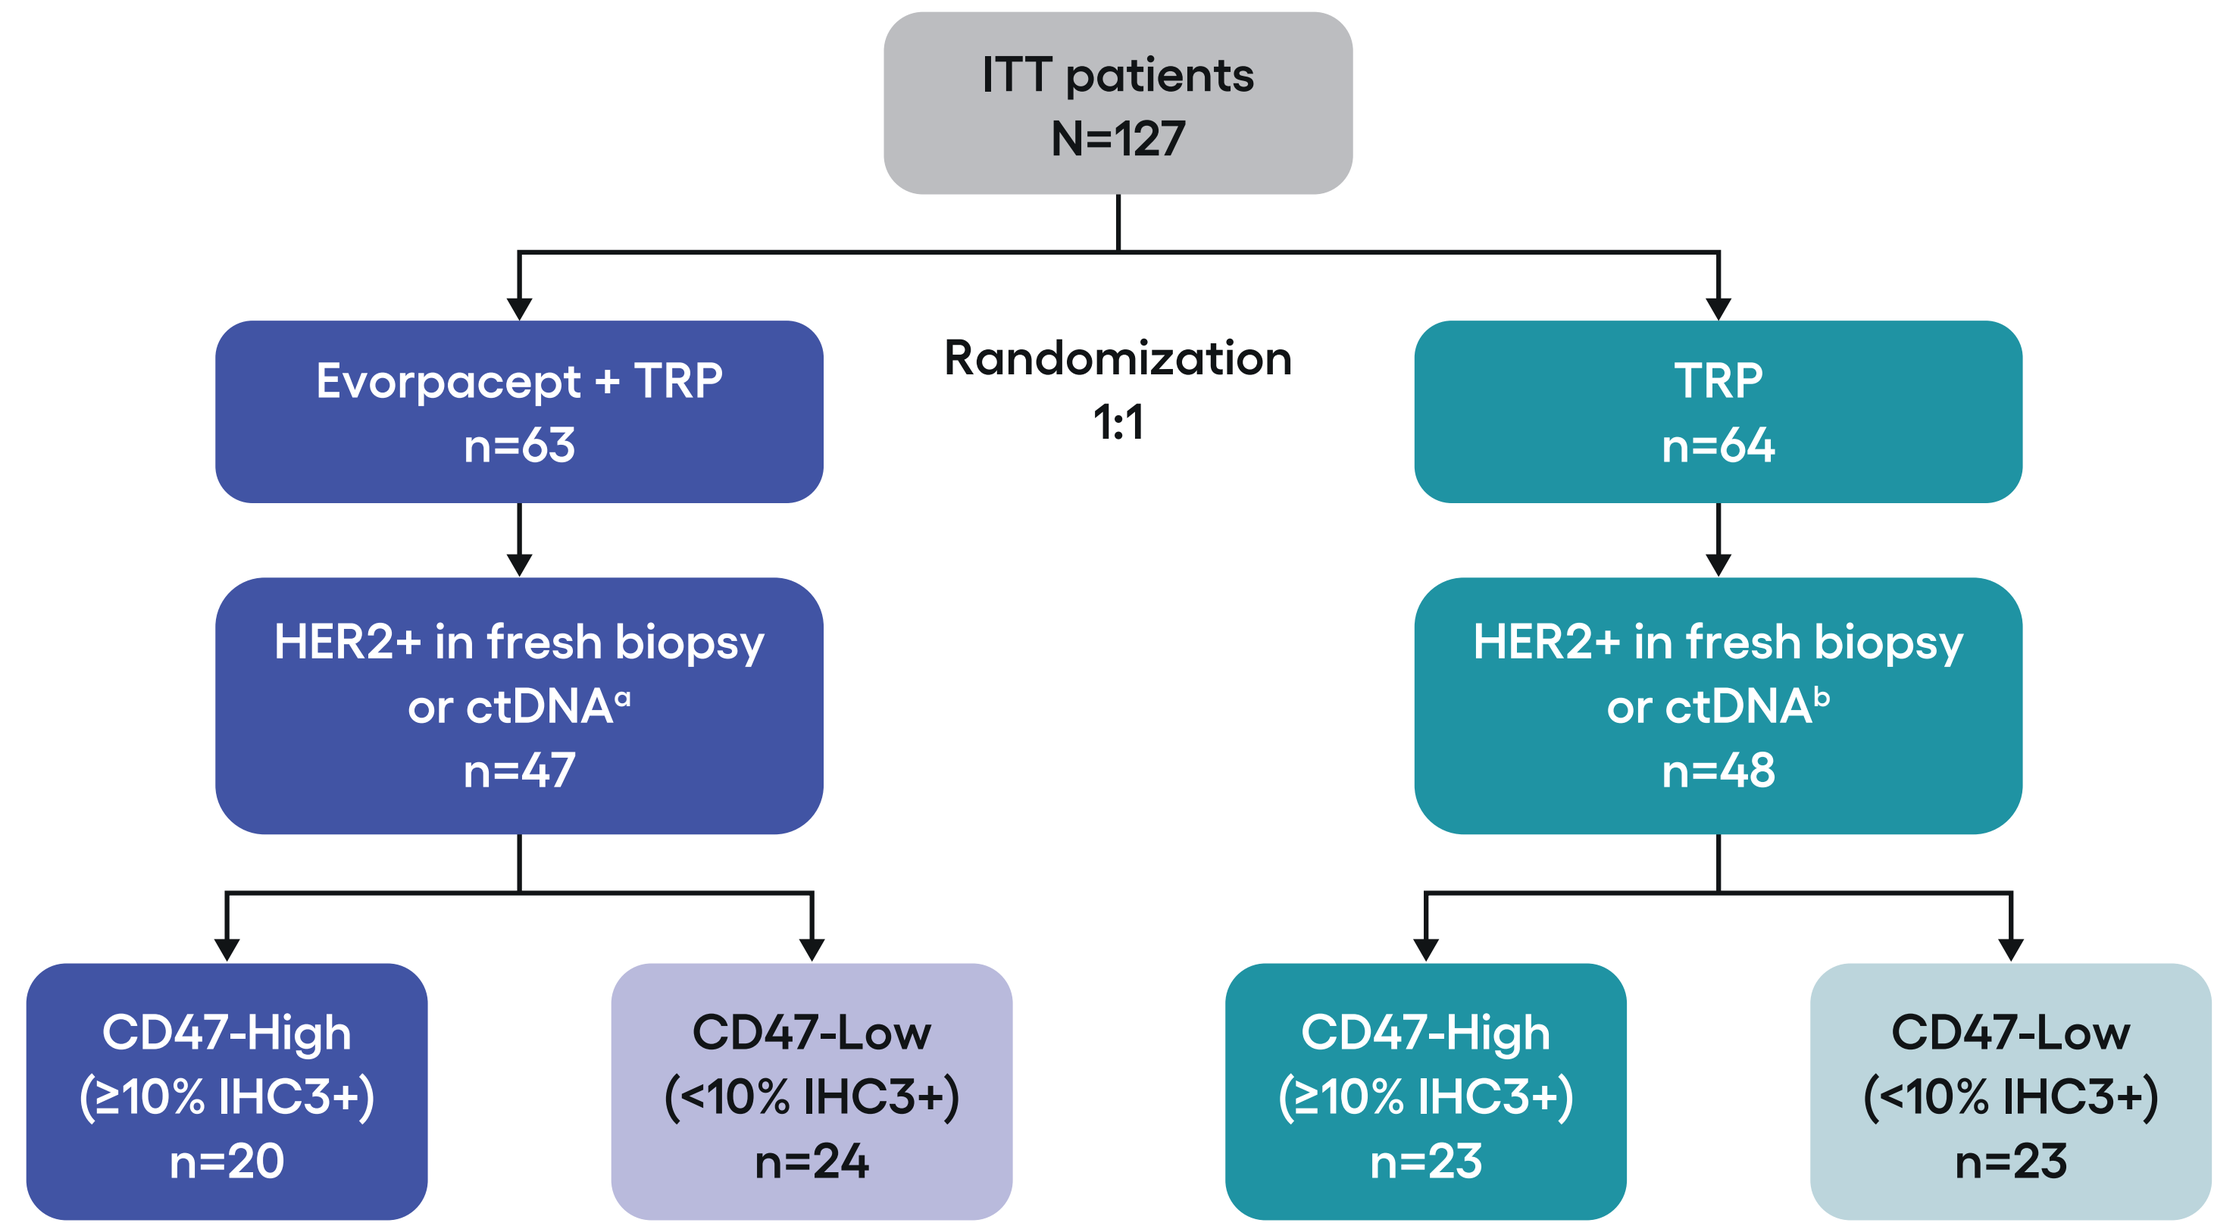
ITT patients N=127
Randomization 1:1
Evorpacept + TRP n=63
TRP n=64
HER2+ in fresh biopsy or ctDNAᵃ n=47
HER2+ in fresh biopsy or ctDNAᵇ n=48
CD47-High (≥10% IHC3+) n=20
CD47-Low (<10% IHC3+) n=24
CD47-High (≥10% IHC3+) n=23
CD47-Low (<10% IHC3+) n=23

ᵃ3 patients were not evaluable for CD47; ᵇ2 patients were not evaluable for CD47

FIGURE 4. Improved ORR with Evorpacept + TRP vs TRP in Patients with CD47-High Expression

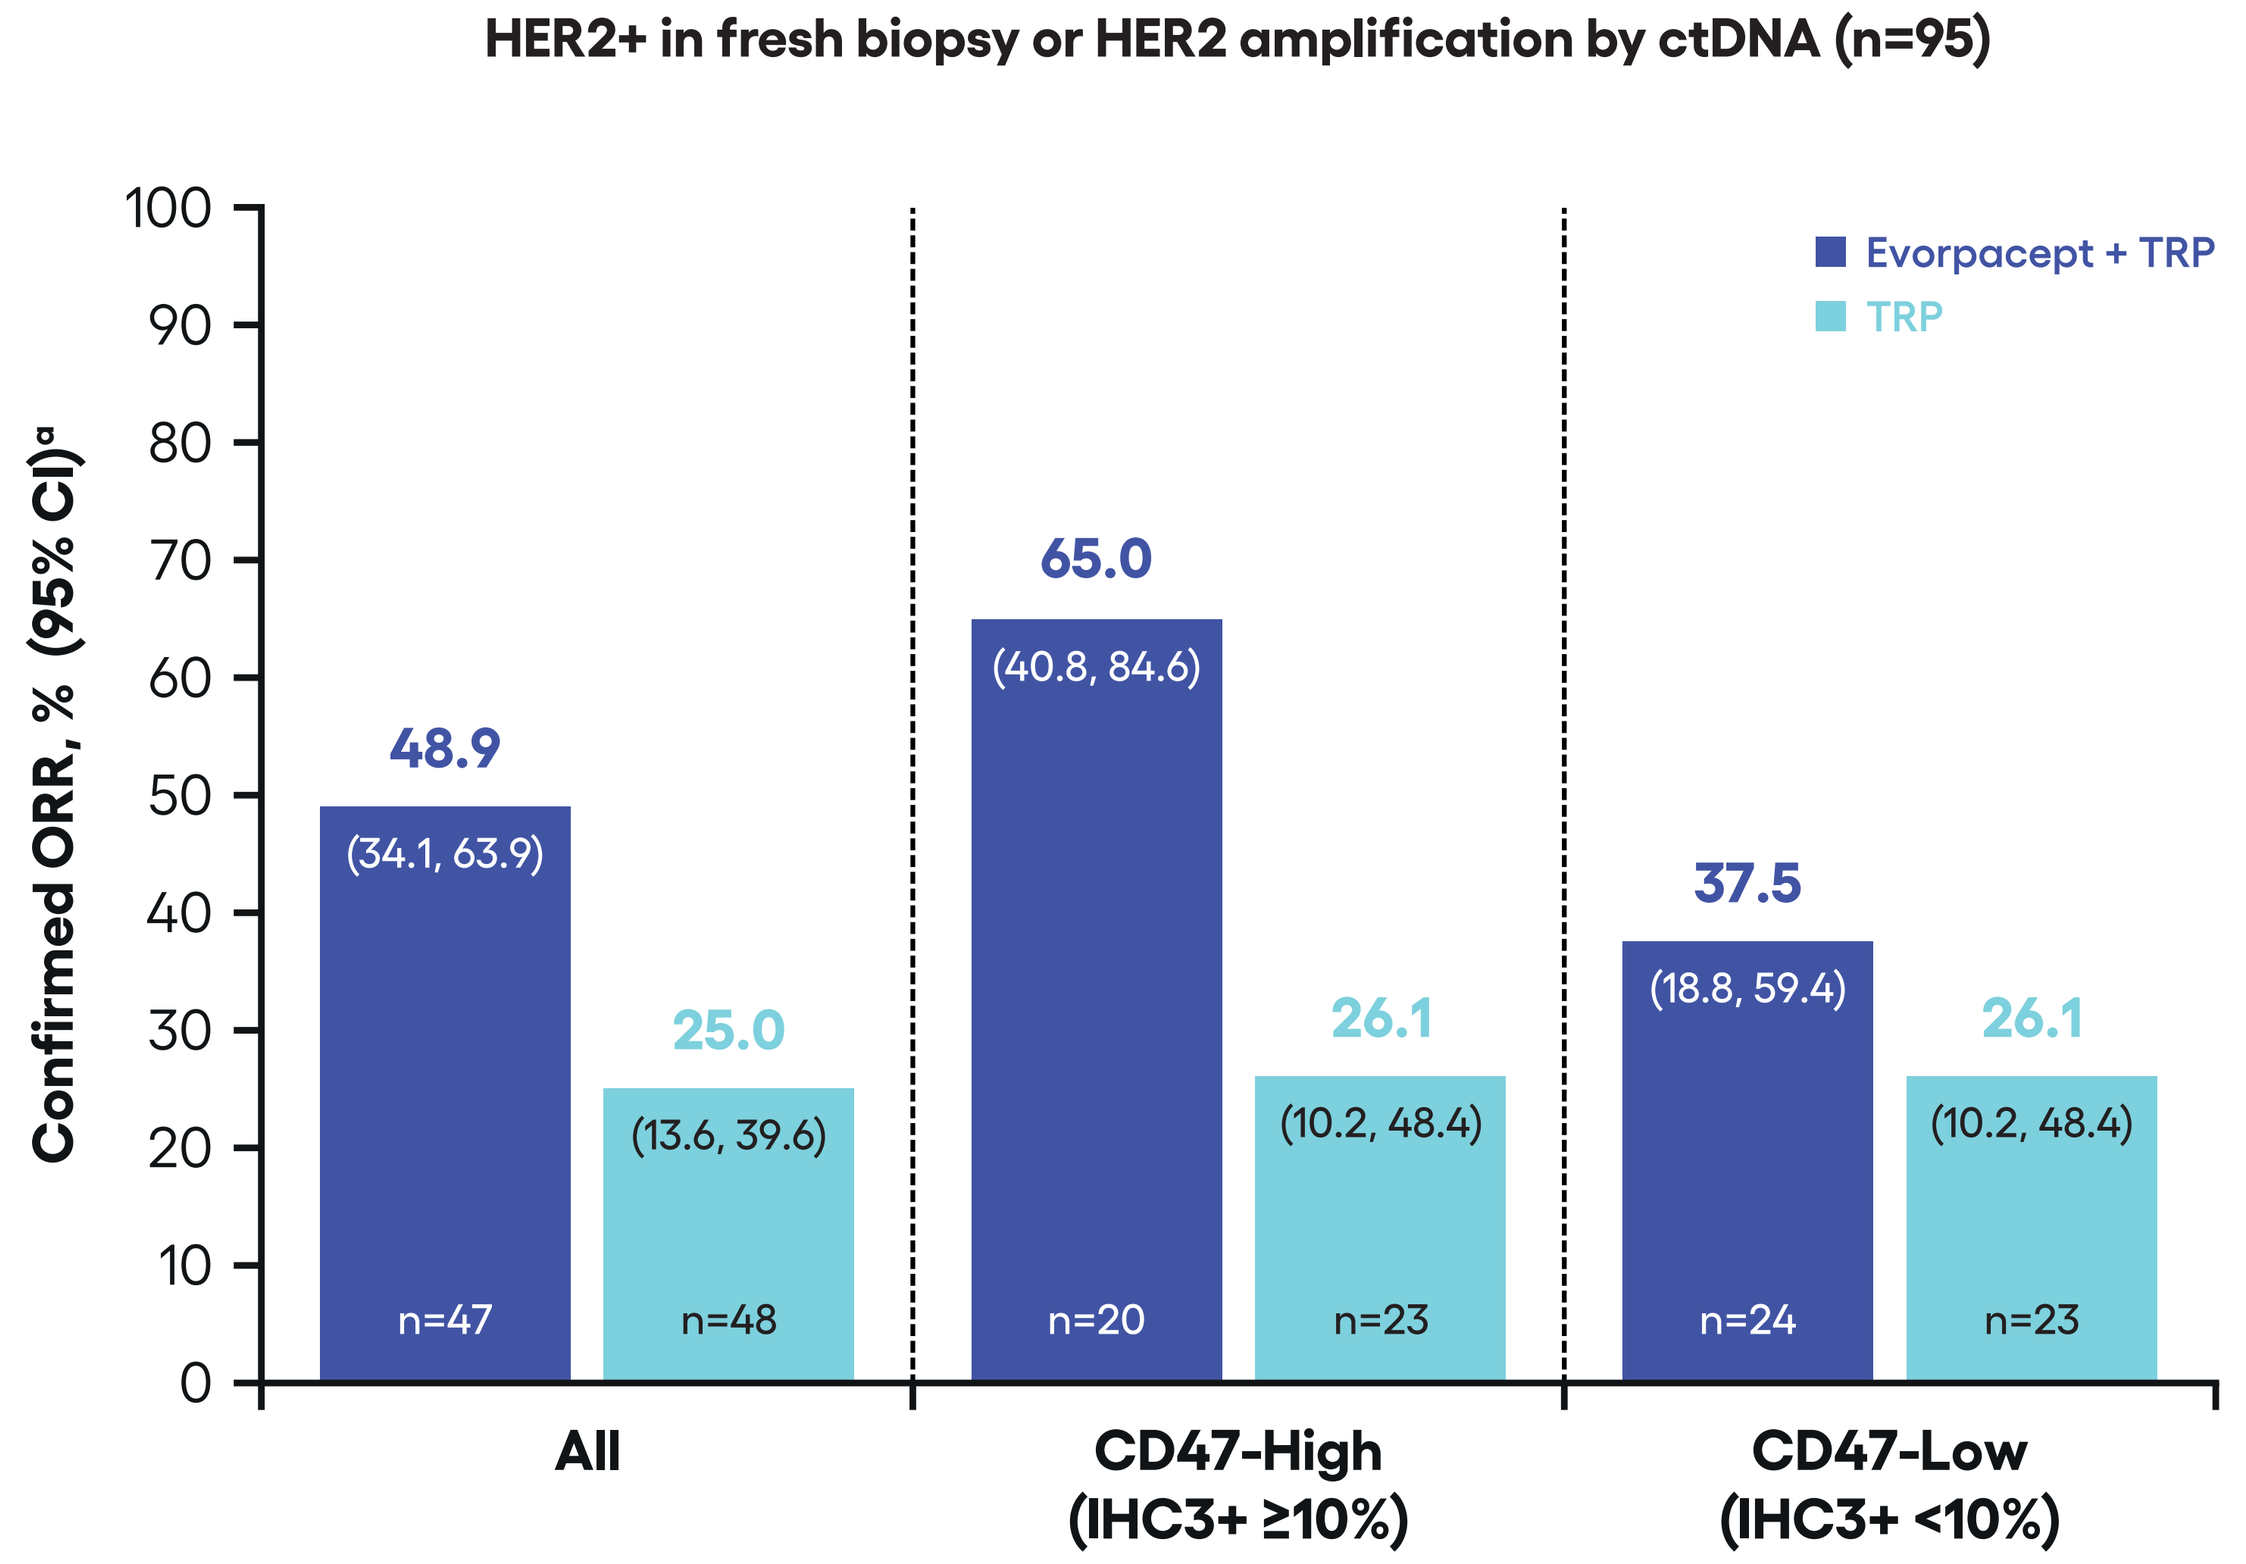
HER2+ in fresh biopsy or HER2 amplification by ctDNA (n=95)
Confirmed ORR, % (95% CI)ᵃ
Evorpacept + TRP
TRP
48.9
(34.1, 63.9)
n=47
25.0
(13.6, 39.6)
n=48
65.0
(40.8, 84.6)
n=20
26.1
(10.2, 48.4)
n=23
37.5
(18.8, 59.4)
n=24
26.1
(10.2, 48.4)
n=23
All
CD47-High (IHC3+ ≥10%)
CD47-Low (IHC3+ <10%)

ᵃInvestigator assessed

- In the CD47-High group, demographics were generally well-balanced except for line of therapy
 - Benefit with evorpacept in 2L only was similar to that seen in the full CD47-High subgroup (evorpacept + TRP vs TRP: ORR 69% vs 23%; median DOR 25.5 vs 14.5 months)
- Efficacy in patients with CD47-Low expression was similar between treatment arms
- Efficacy outcomes were improved with evorpacept + TRP vs TRP across multiple levels of high CD47 expression defined by CD47 membrane staining intensities of IHC2+ and/or IHC3+ (**Table 1**)

FIGURE 5. Median DOR in Patients with CD47-High Expression was Longer for Evorpacept + TRP vs TRP

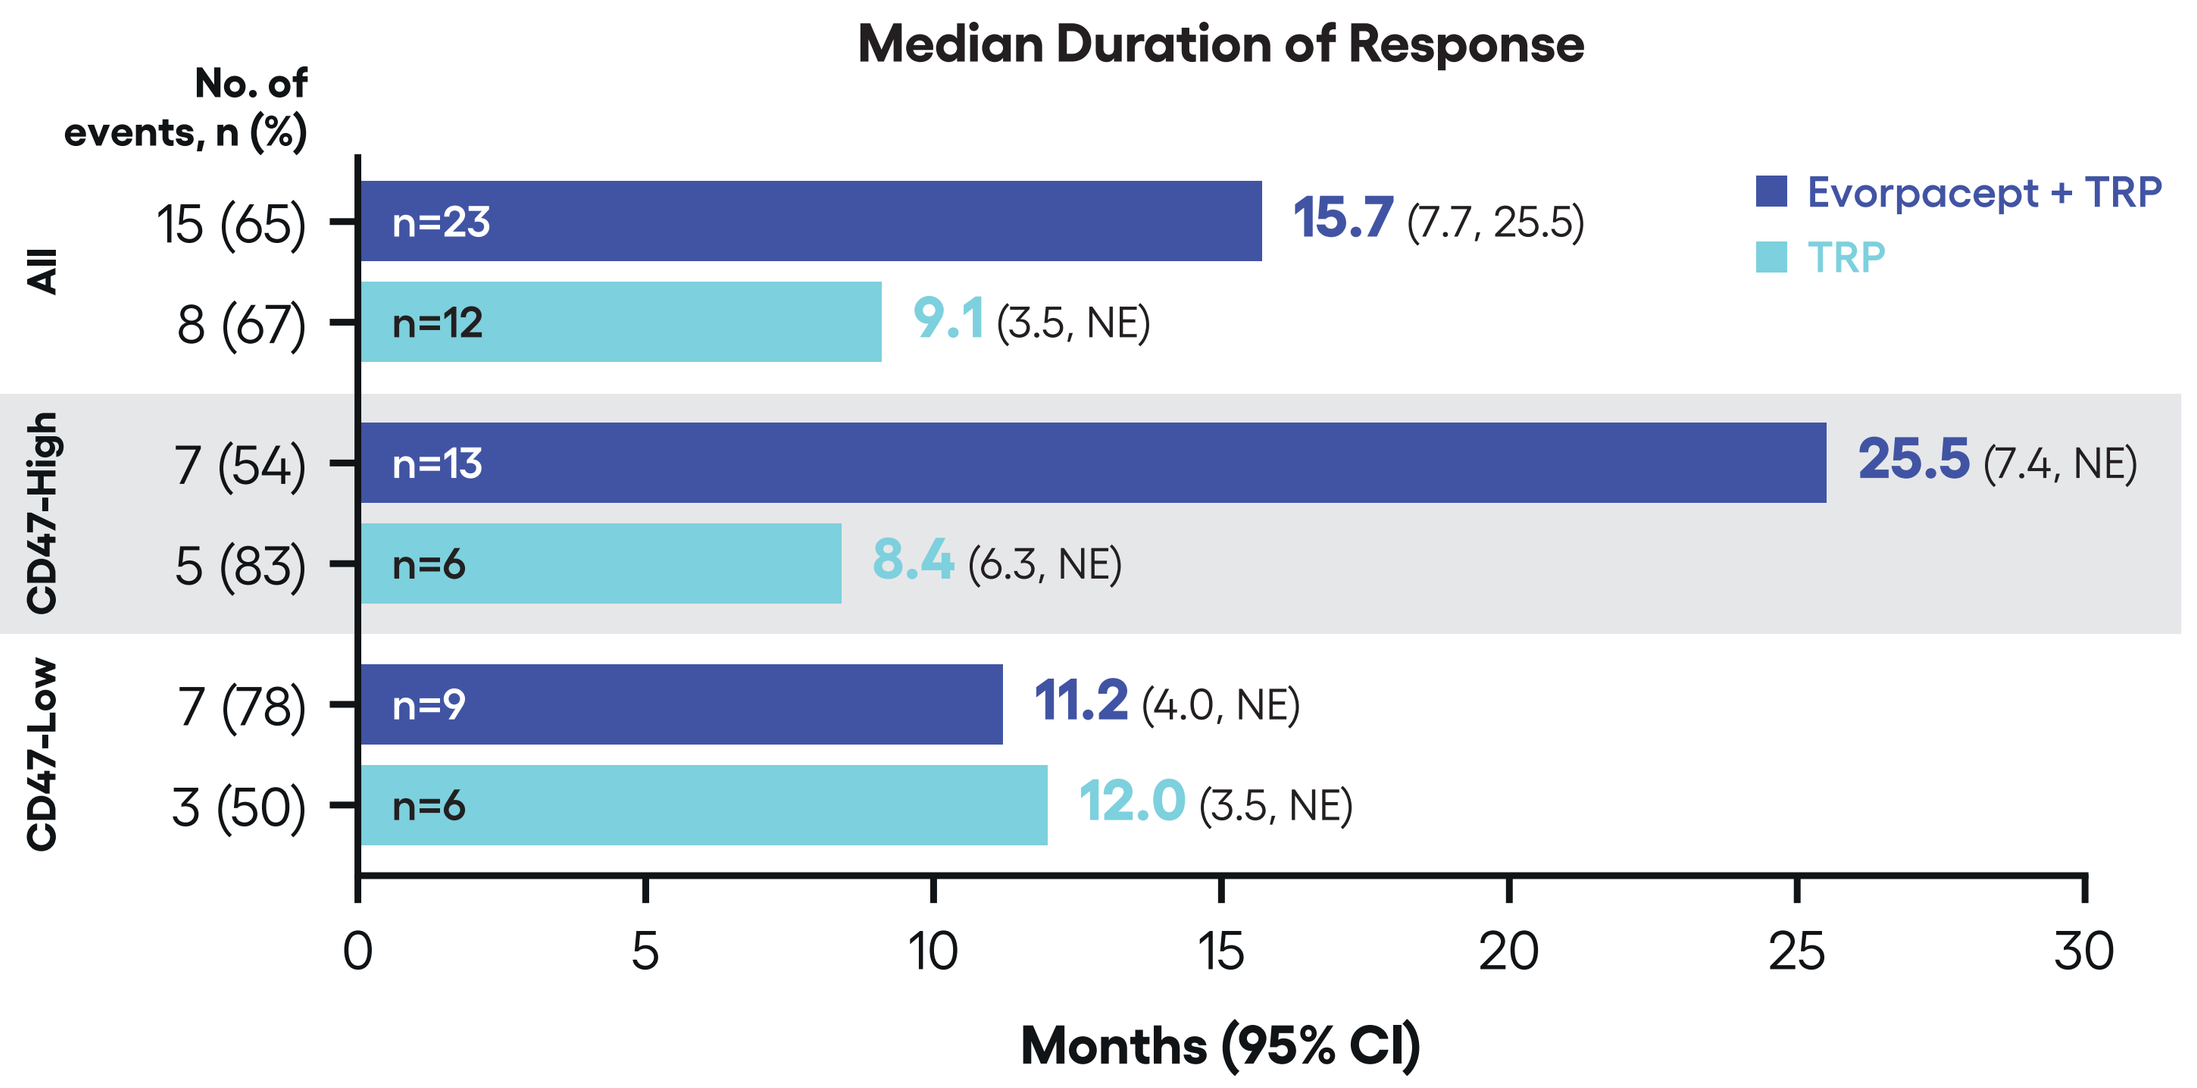
Median Duration of Response
No. of events, n (%)
Evorpacept + TRP
TRP
All
15 (65)
n=23
15.7 (7.7, 25.5)
8 (67)
n=12
9.1 (3.5, NE)
CD47-High
7 (54)
n=13
25.5 (7.4, NE)
5 (83)
n=6
8.4 (6.3, NE)
CD47-Low
7 (78)
n=9
11.2 (4.0, NE)
3 (50)
n=6
12.0 (3.5, NE)
Months (95% CI)

TABLE 1. Improved Efficacy Outcomes with Evorpacept + TRP vs TRP Across Multiple Levels of CD47 Expression

CD47 Cut-offᵃ	% of HER2+ Subgroup (n=95)	ORR		PFS HRᵇ (95% CI)	OS HRᵇ (95% CI)
		Evorpacept + TRP	TRP		
No cut-off	100%	**49%** (n=47)	**25%** (n=48)	**0.72** (0.44, 1.18)	**0.95** (0.58, 1.56)
≥10% Med/High	57%	**56%** (n=25)	**24%** (n=29)	**0.40** (0.19, 0.82)	**0.75** (0.39, 1.46)
≥25% Med/High	40%	**60%** (n=20)	**22%** (n=18)	**0.36** (0.15, 0.84)	**0.65** (0.30, 1.41)
≥5% High	51%	**64%** (n=22)	**23%** (n=26)	**0.38** (0.17, 0.84)	**0.66** (0.32, 1.37)
≥10% High	45%	**65%** (n=20)	**26%** (n=23)	**0.39** (0.17, 0.86)	**0.63** (0.30, 1.31)

ᵃCD47 cut-off defined by cell staining intensity: Medium = IHC2+; High = IHC3+.
ᵇThe HR is estimated from a Cox proportional hazards model with the treatment, region=Asia (Yes/No) and the use of prior T-DXd (Yes/No) as covariates

- CD47-High expression is a negative prognostic factor: in the TRP arm, OS was shorter in the CD47-High subgroup vs the CD47-Low subgroup (9.9 vs 19.1 months; HR 1.38 [95% CI 0.68, 2.83]; **Figure 7**)
- Efficacy by BICR was similar to INV (evorpacept + TRP vs TRP: ORR 60% vs 30%; median DOR 26.3 vs 9.2 months)

FIGURE 6. PFS per INV Assessment (A) and OS (B) was Improved with Evorpacept + TRP vs TRP in Patients with CD47-High Expression

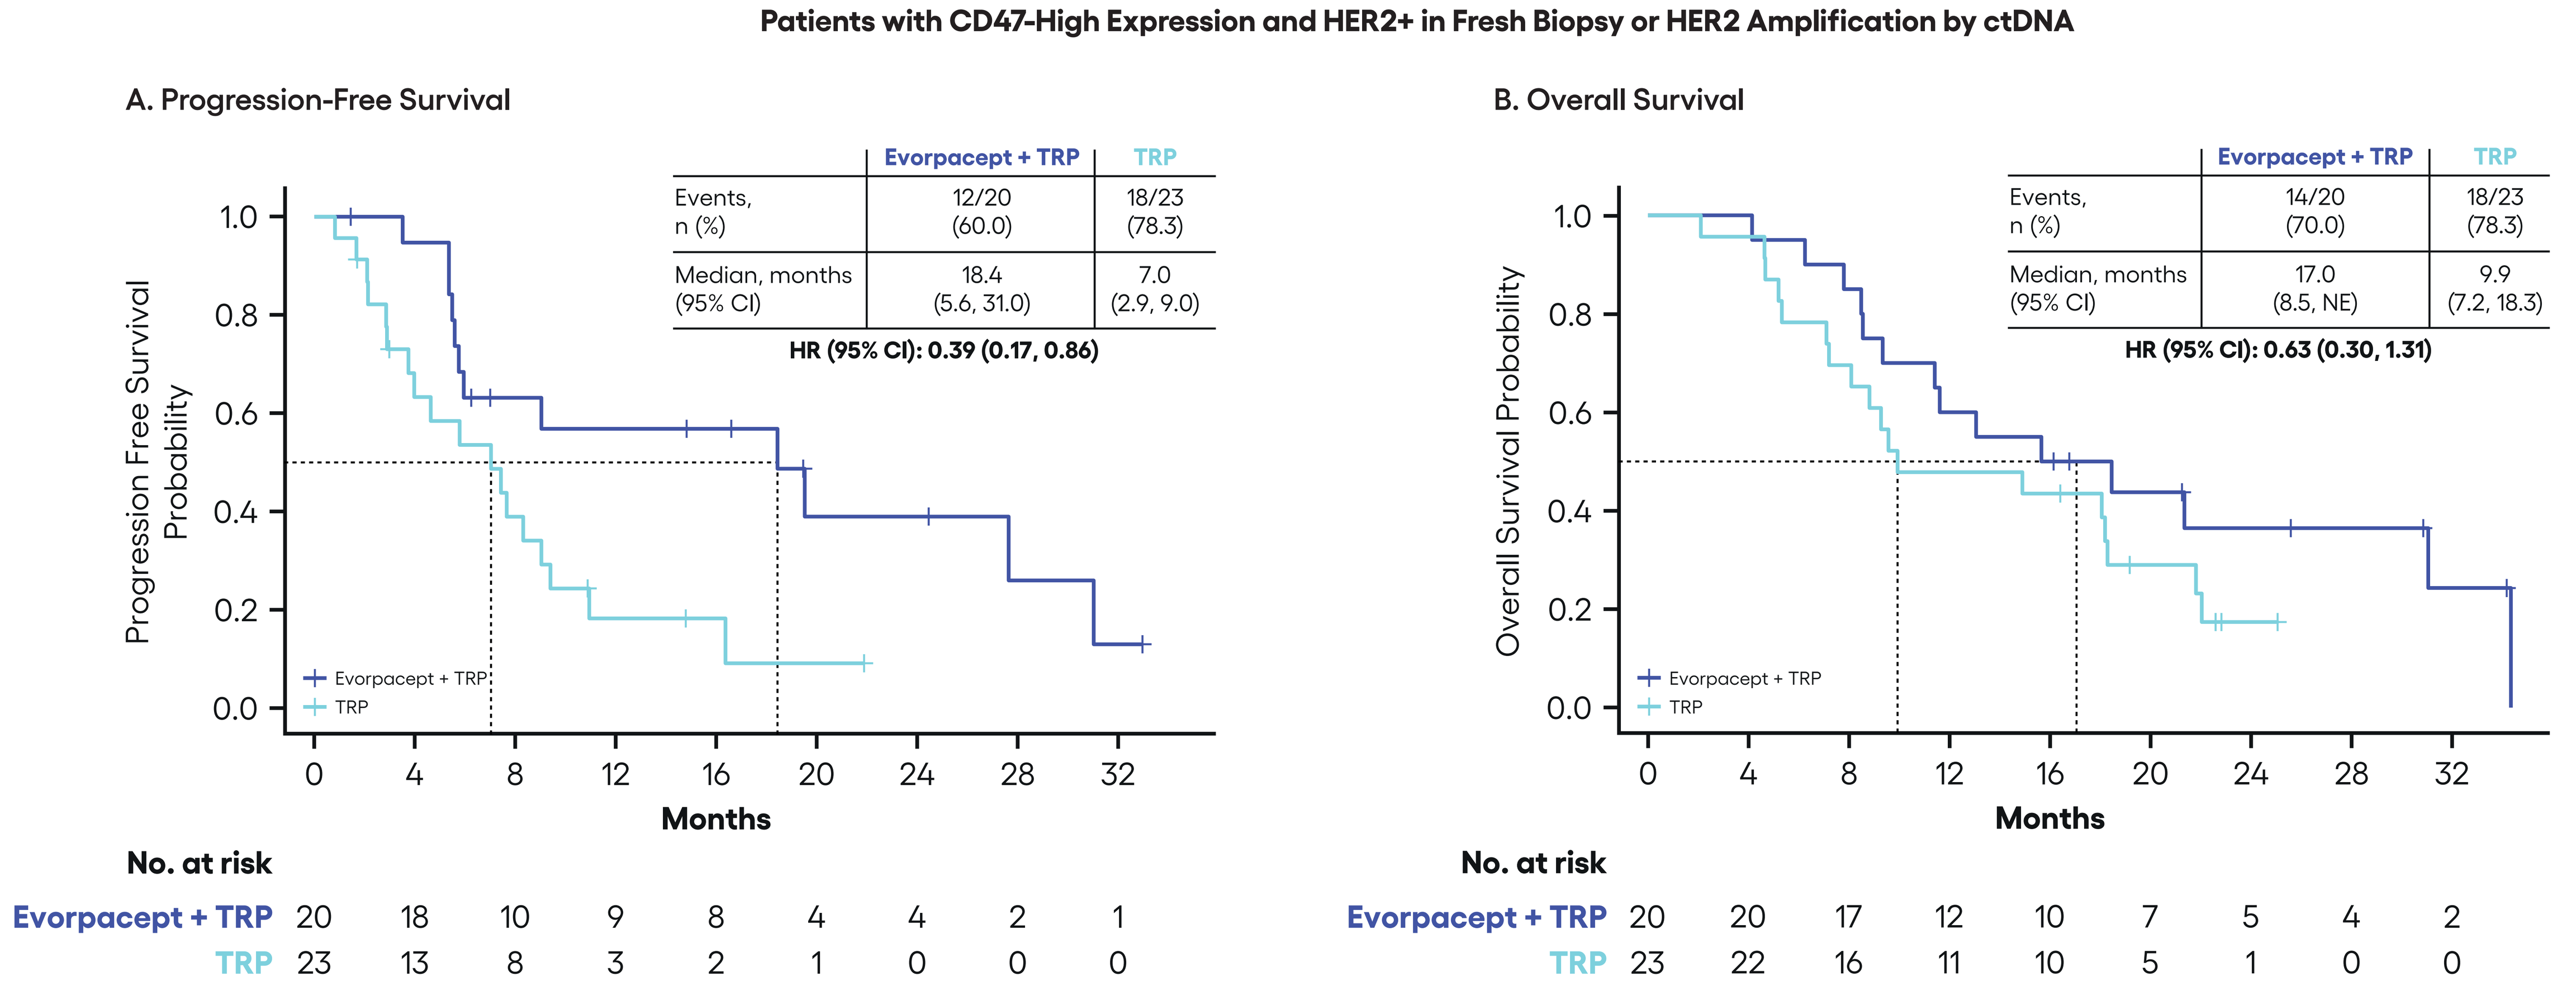
Patients with CD47-High Expression and HER2+ in Fresh Biopsy or HER2 Amplification by ctDNA
A. Progression-Free Survival
Progression Free Survival Probability
Evorpacept + TRP
TRP
Events, n (%) 12/20 (60.0) 18/23 (78.3)
Median, months (95% CI) 18.4 (5.6, 31.0) 7.0 (2.9, 9.0)
HR (95% CI): 0.39 (0.17, 0.86)
Months
No. at risk
Evorpacept + TRP 20 18 10 9 8 4 4 2 1
TRP 23 13 8 3 2 1 0 0 0
B. Overall Survival
Overall Survival Probability
Events, n (%) 14/20 (70.0) 18/23 (78.3)
Median, months (95% CI) 17.0 (8.5, NE) 9.9 (7.2, 18.3)
HR (95% CI): 0.63 (0.30, 1.31)
Months
No. at risk
Evorpacept + TRP 20 20 17 12 10 7 5 4 2
TRP 23 22 16 11 10 5 1 0 0

FIGURE 7. OS in TRP CD47-High vs CD47-Low Suggests that CD47 Expression is a Negative Prognostic Factor

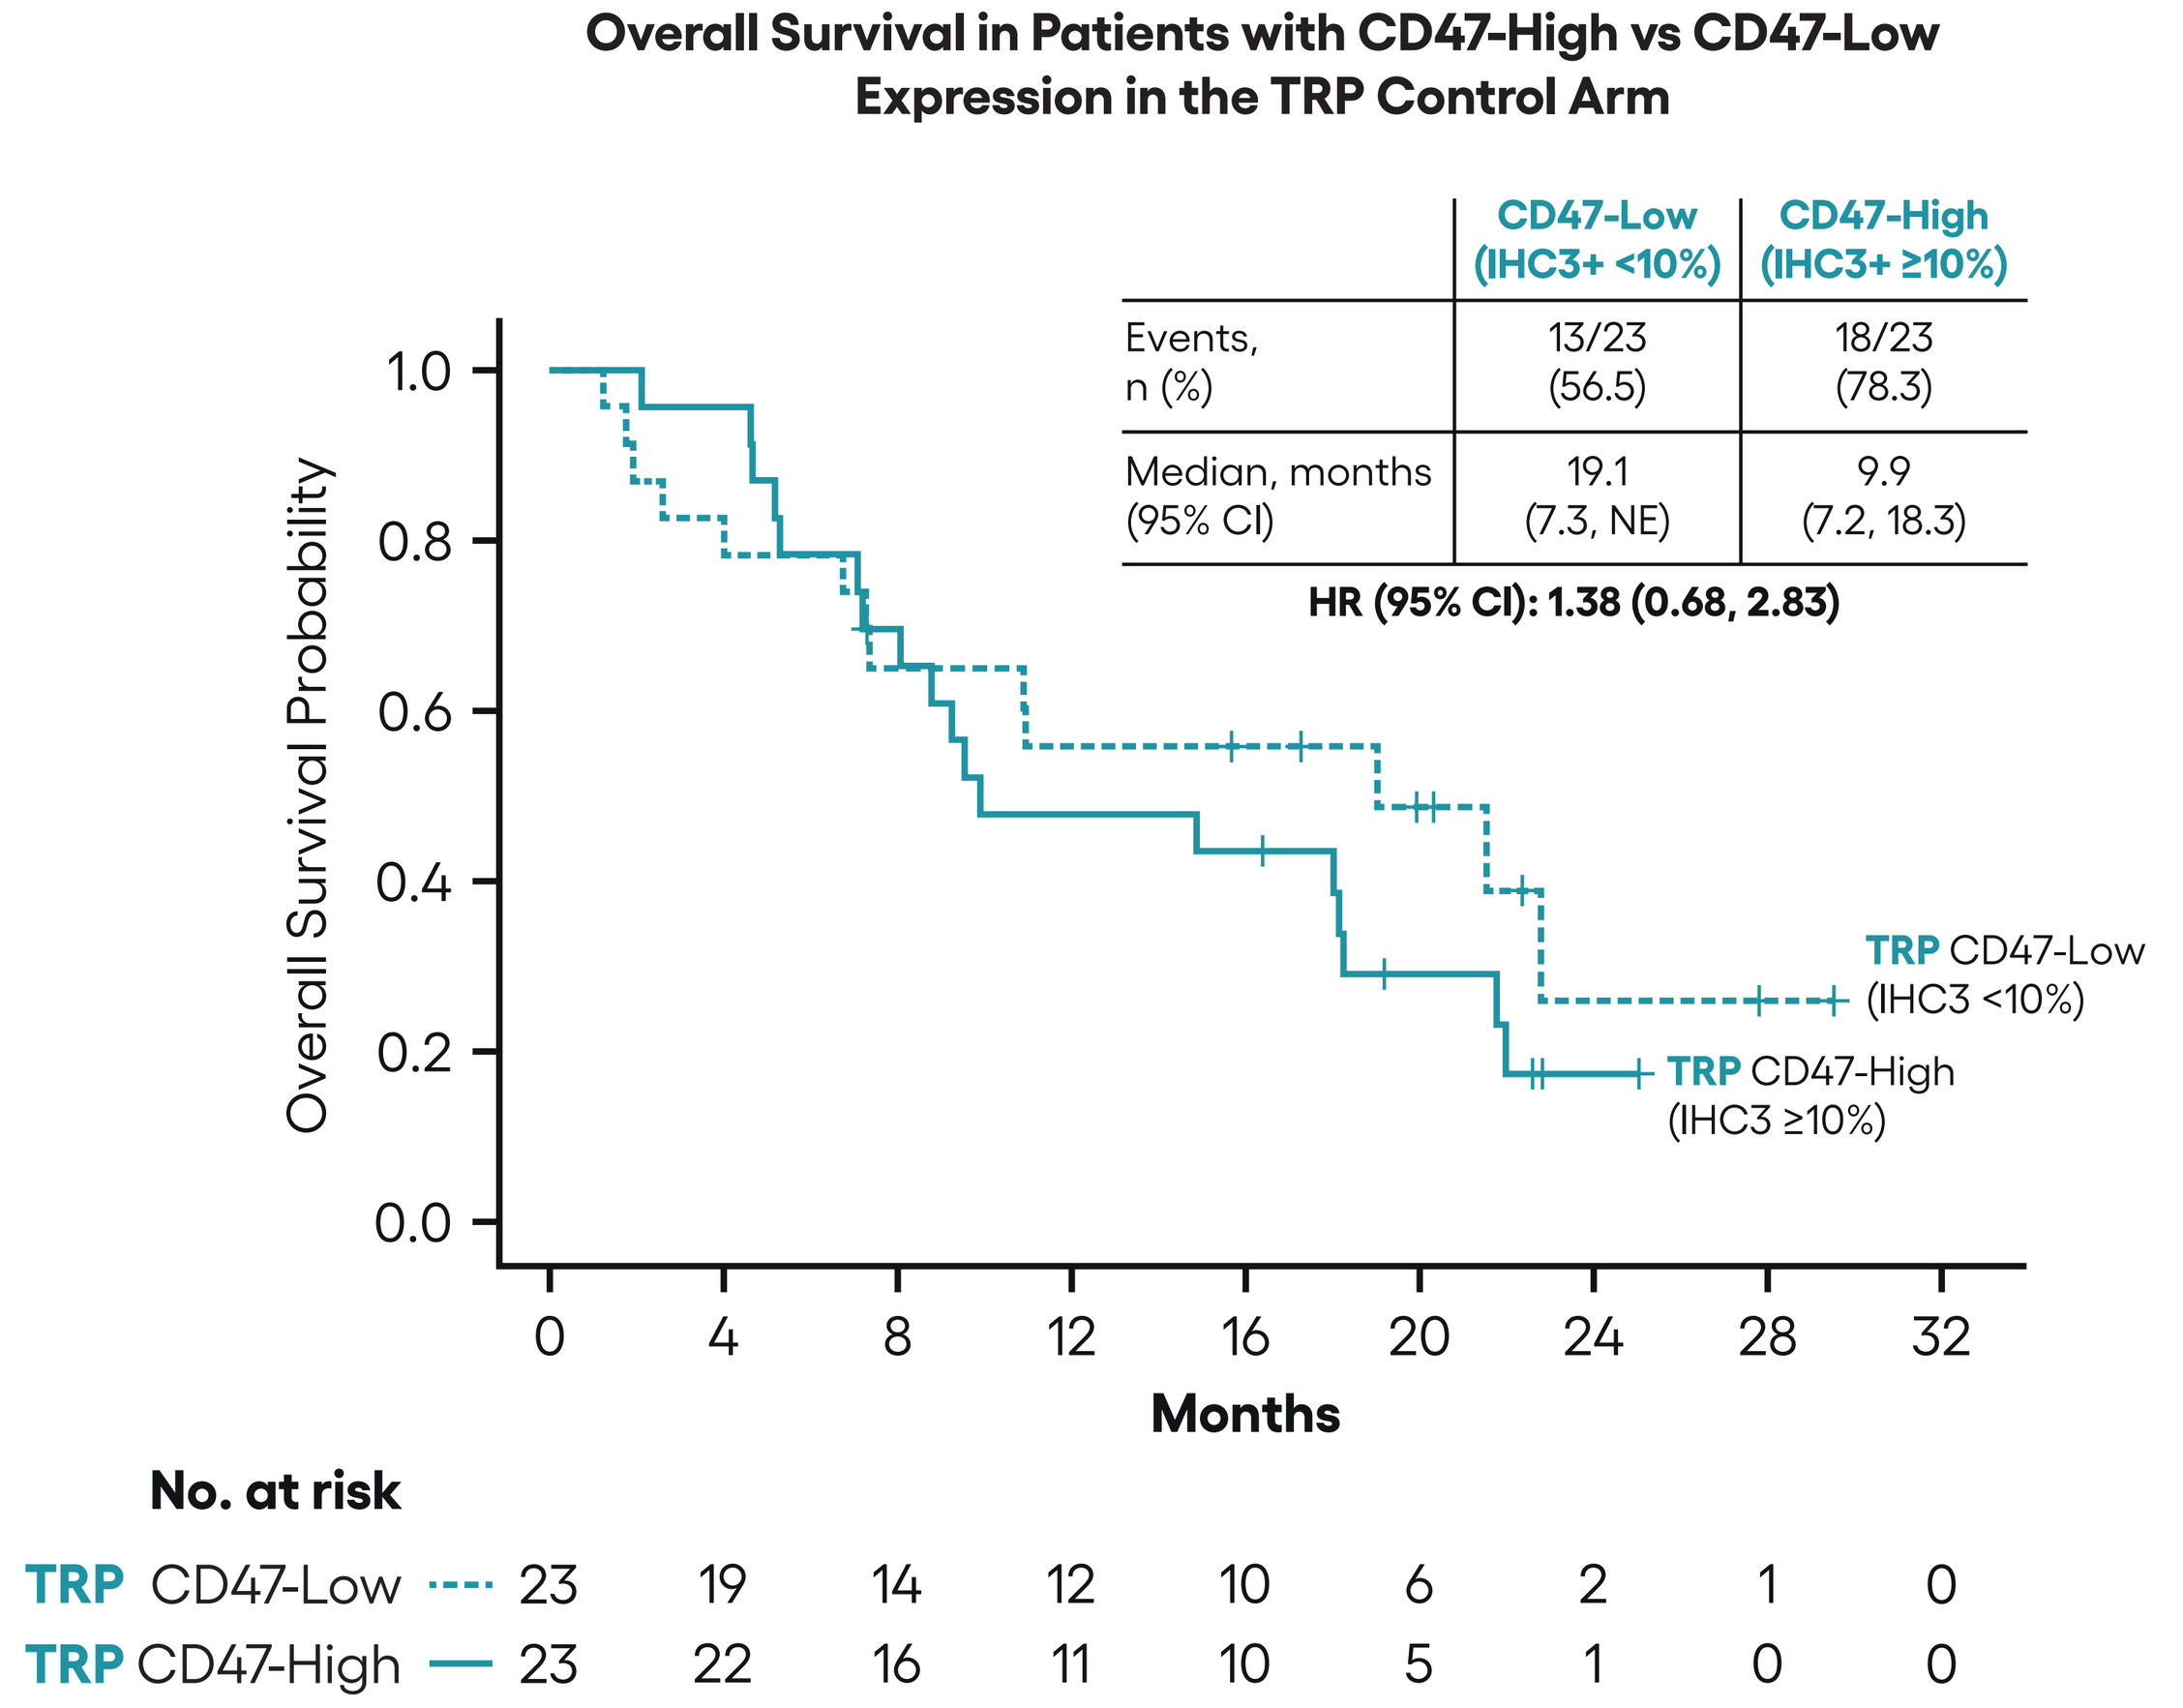
Overall Survival in Patients with CD47-High vs CD47-Low Expression in the TRP Control Arm
CD47-Low (IHC3+ <10%)
CD47-High (IHC3+ ≥10%)
Events, n (%) 13/23 (56.5) 18/23 (78.3)
Median, months (95% CI) 19.1 (7.3, NE) 9.9 (7.2, 18.3)
HR (95% CI): 1.38 (0.68, 2.83)
Overall Survival Probability
TRP CD47-Low (IHC3 <10%)
TRP CD47-High (IHC3 ≥10%)
Months
No. at risk
TRP CD47-Low 23 19 14 12 10 6 2 1 0
TRP CD47-High 23 22 16 11 10 5 1 0 0

Conclusions

- The results of this analysis suggest CD47 is a predictive biomarker for clinical benefit with evorpacept in 2L/3L gastric/GEJ cancer
 - Clinical activity was strongly enriched with evorpacept + TRP vs TRP in patients with both CD47 expression and retained HER2 expression (ORR: 65.0% vs 26.1%; median DOR: 25.5 vs 8.4 months; PFS HR: 0.39; OS HR: 0.63)
- These results are consistent with mechanistic rationale that both HER2 and CD47 expression are needed to drive maximal ADCP
- Numerically shorter OS in patients with CD47-High vs CD47-Low expression in the TRP control arm is consistent with the literature that CD47 expression is a negative prognostic factor[13]
- To our knowledge, ASPEN-06 is the first randomized study to demonstrate both efficacy and a manageable safety profile of CD47-SIRPα inhibition in solid tumors
- Confirmation in larger, controlled studies of evorpacept in combination with HER2-targeted and other anticancer antibodies is warranted, with prospective analysis of CD47 expression levels

References: 1. Lu Q, et al. *Onco Targets Ther.* 2020;13:9323–9331. **2.** Jiang Z, et al. *J Hematol Oncol.* 2021;14:180. **3.** Zhou X, et al. *Adv Funct Mater.* 2021;31:2006220. **4.** Liu Y, et al. *Signal Transduct Target Ther.* 2023;8:104. **5.** Kauder SE, et al. *PLoS One.* 2018;13:e0201832. **6.** Shitara K, et al. *J Clin Oncol.* 2025;43:A332. **7.** Bang YJ, et al. *Lancet.* 2010;376:687–697. **8.** Johnson LDS and Zhang Y. *Blood.* 2024;144:4604. **9.** Van Buuren N, et al. *J ImmunoTher Cancer.* 2024;12:A212. **10.** Chen C, et al. *iScience.* 2024;27:111478. **11.** Saeki H, et al. *Eur J Cancer.* 2018;105:41–49. **12.** Wilke H, et al. *Lancet Oncol.* 2014;15;1224–1235. **13.** Zhao H-J, et al. *Transl Cancer Res.* 2018;7:609–621. **Abbreviations: 2L**, second line; **3L**, third line; **ADCP**, antibody-dependent cellular phagocytosis; **BICR**, blinded independent central review; **CI**, confidence interval; **ctDNA**, circulating tumor DNA; **DOR**, duration of response; **FCγR**, FCγ receptor; **FDA**, US Food and Drug Administration; **GEJ**, gastroesophageal junction; **HER2(+)**, human epidermal growth factor receptor 2(-positive); **HR**, hazard ratio; **IHC**, immunohistochemistry; **INV**, investigator; **ITT**, intent to treat; **NE**, not estimable; **ORR**, objective response rate; **OS**, overall survival; **PFS**, progression-free survival; **Q2W**, every 2 weeks; **RP**, ramucirumab and paclitaxel; **SIRPα**, signal regulatory protein alpha, **T-DXd**, trastuzumab deruxtecan; **TRP**, trastuzumab, ramucirumab, and paclitaxel. **Disclosures:** Dr Wainberg reports consulting roles for Alligator Therapeutics, Amgen, AstraZeneca, Arcus, Boehringer Ingelheim, Bristol Myers Squibb, Daiichi Sankyo Company, Eli Lilly and Company, EMD Serono, Roche AG, Genentech, Ipsen, Johnson & Johnson, Merus N.V., Merck, Novartis, Novocure, Pfizer, Servier, Verastem. **Acknowledgments:** We thank all of the participating patients and their families as well as the site research staff. **Contact email:** info@alxoncology.com. **Presented at:** The 2025 Society for Immunotherapy of Cancer (SITC) Annual Meeting, November 5–9, 2025. Abstract #496. **Trial Registration:** ClinicalTrials.gov identifier, NCT05002127. This study was funded by ALX Oncology.