ALX2004: A Novel Anti-EGFR Topoisomerase I Inhibitor Antibody-Drug Conjugate for the Treatment of EGFR-Expressing Solid Tumors

Janica Wong, Ana Monegal Granados, Hanako Daino Laizure, Jia-Xu Li, Jessica L. Morgan, Sarah Bolton, Andrew Olson, Yin Labson, Connie Lu, Sen Xu, Venita De Almeida, Hadi Falahatpisheh, Peter Strop, Jaume Pons, **Marija Vrljic**

ALX Oncology Inc., South San Francisco, CA, USA

2025 AACR-NCI-EORTC

Introduction

- EGFR remains one of the most validated and clinically actionable targets in oncology. However, attempts to develop EGFR-targeted ADCs have resulted in failures and no FDA approvals. Toxicity associated with the previously utilized payloads (e.g., tubulin inhibitors and cross-linking DNA agents) and the off-tumor skin toxicity associated with this target have significantly limited the therapeutic window and clinically meaningful dose
- ALX2004 is a differentiated anti-EGFR TOP1i ADC with a lysosomal protease-cleavable linker and payload linked to native IgG1 interchain cysteines with DAR 8, resulting in a homogenous product (**Figure 1**)
- ALX2004 demonstrated potent antitumor activity in CDX mouse models with differing levels of EGFR expression and varied mutational status across the EGFR signaling pathway, including EGFR with wild-type and mutant kinase domain and wild-type and mutant KRAS and BRAF

ALX2004 was meticulously designed to maximize the therapeutic window and has the potential to establish proof-of-concept early in the development cycle

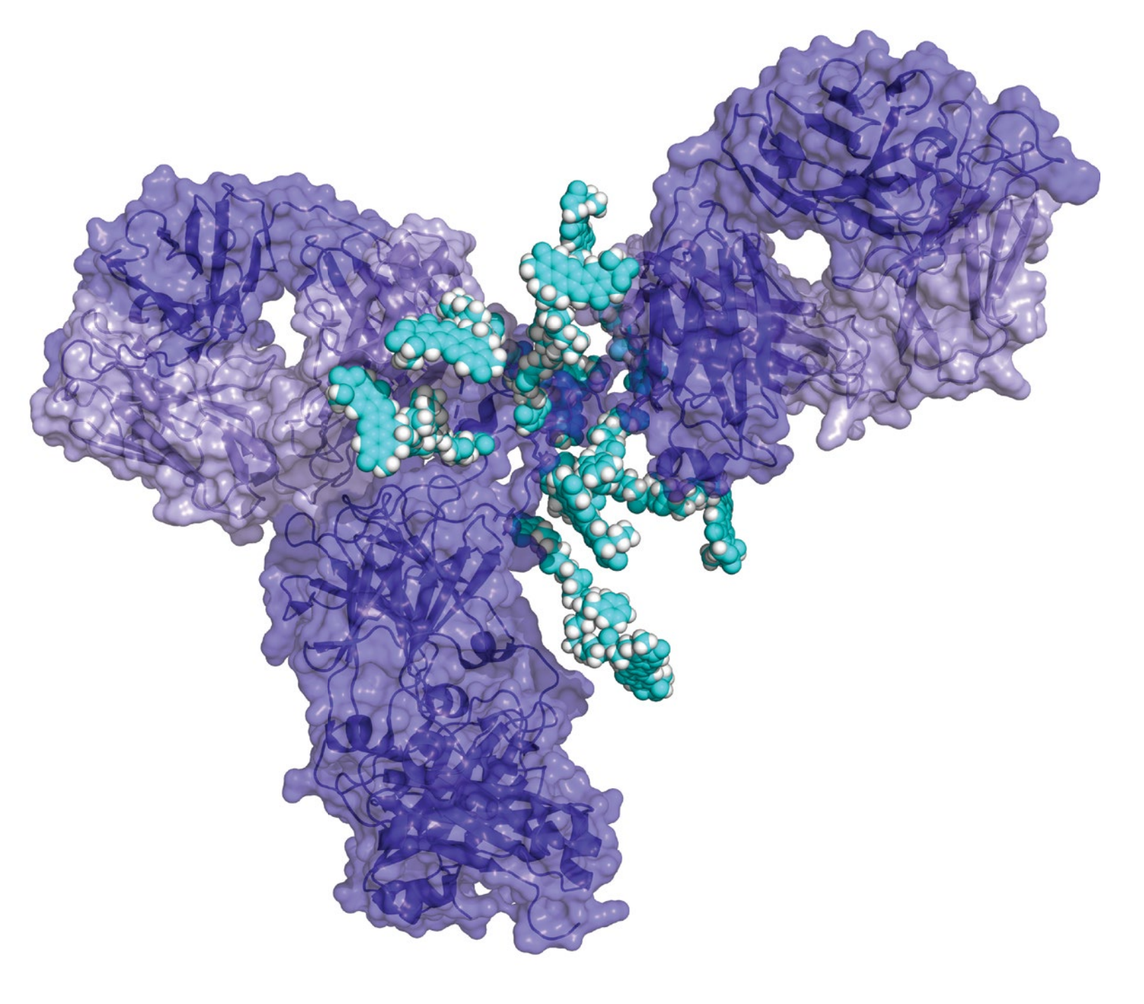

Figure 1. ALX2004
EGFR-targeted ADC DAR 8
TOP1i payload

EGFR Antibody
Matuzumab-derived EGFR antibody selected to minimize off-tumor skin toxicity and to maximize therapeutic window
Epitope distinct from that of FDA-approved EGFR antibodies

Proprietary Linker-Payload
Lysosomal cleavage like deruxtecan ADCs with improved linker-antibody stability to minimize off-tumor payload release

Proprietary Top1i Payload, DAR 8
TOP1i with similar direct cytotoxic potency and enhanced bystander activity compared to deruxtecan

ALX2004 Mechanism of Action

ALX2004_payload selected based on robust cytotoxicity in eight cancer cell lines

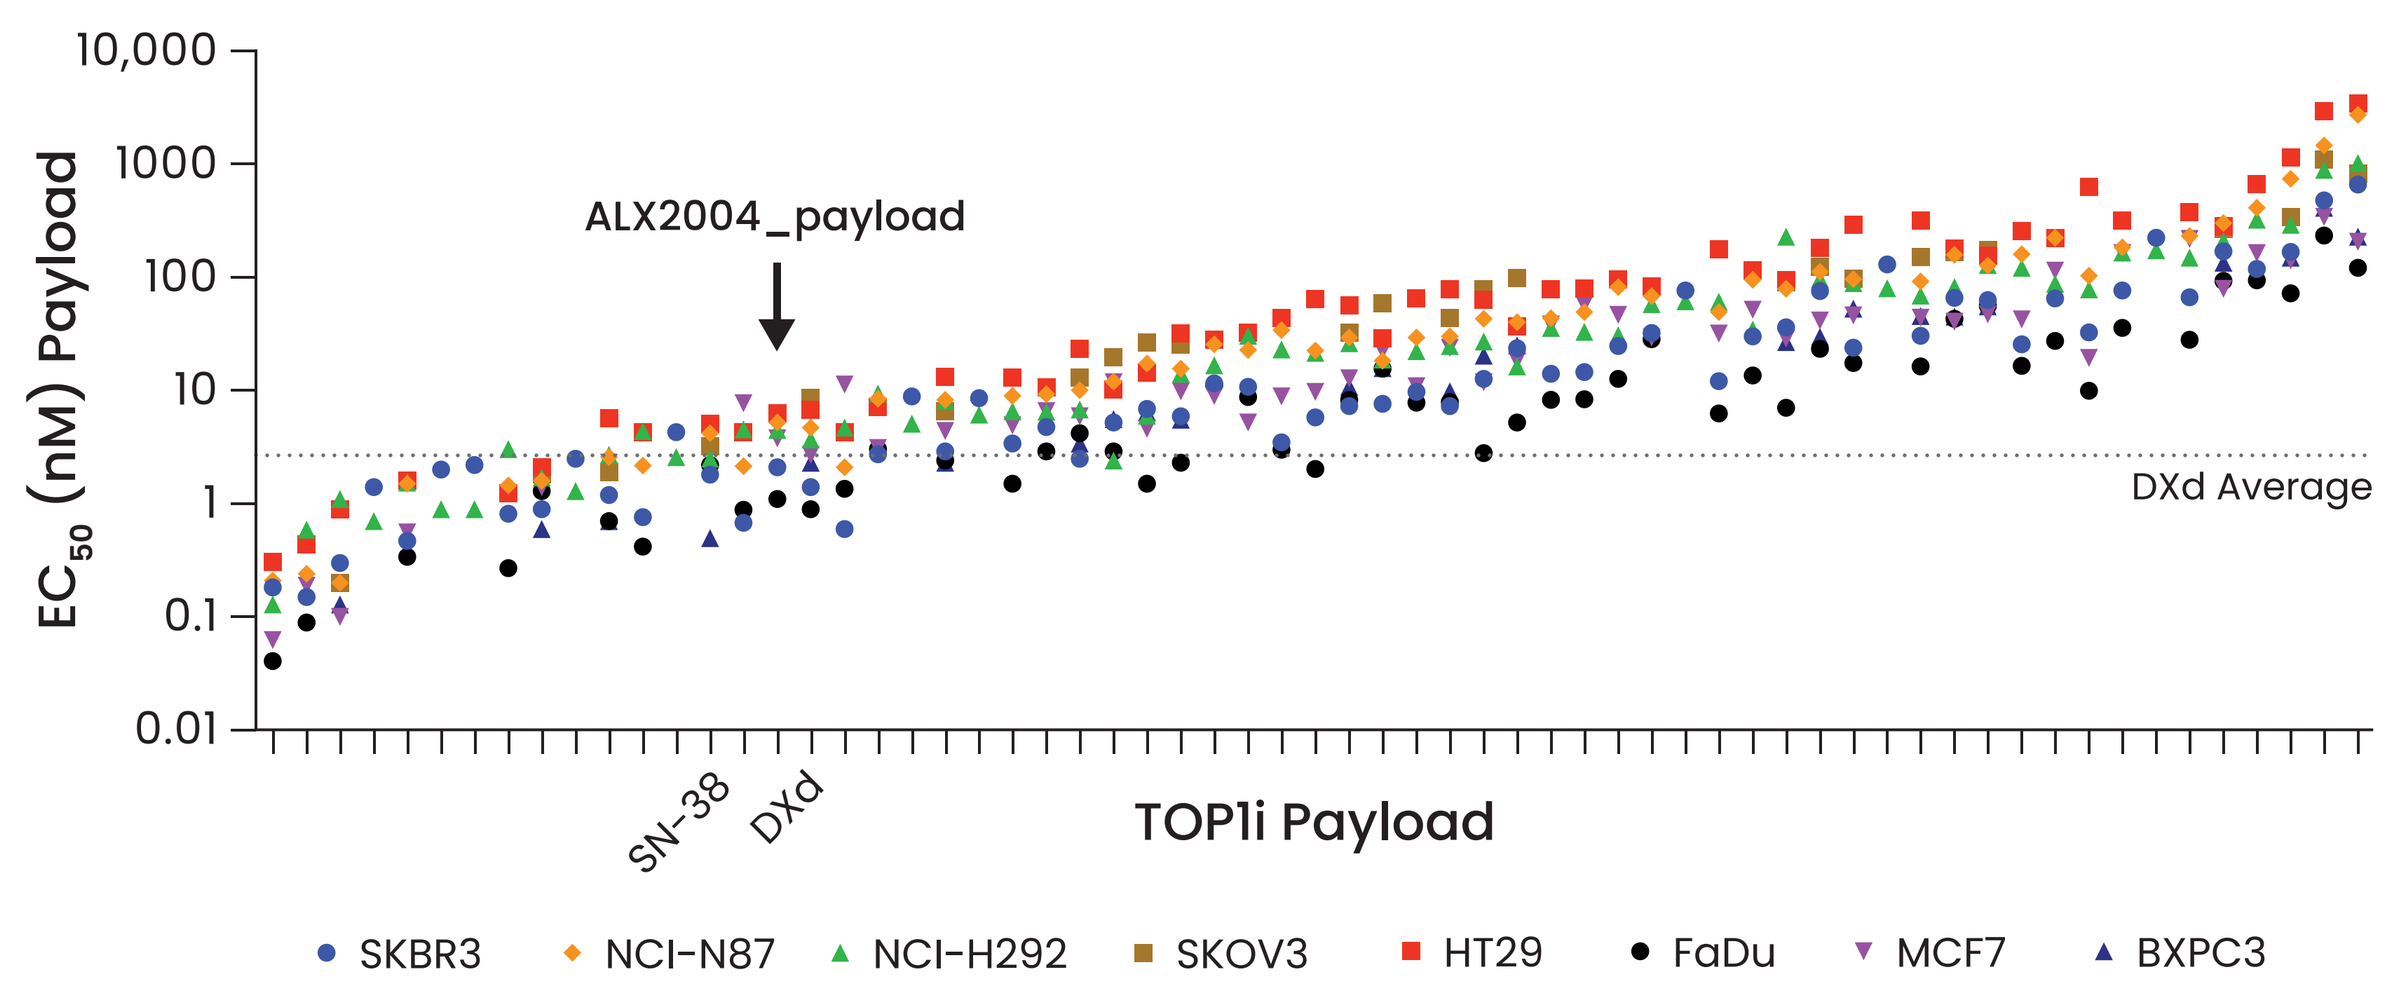
EC50 (nM) Payload
ALX2004_payload
DXd Average
SN-38
DXd
TOP1i Payload
SKBR3
NCI-N87
NCI-H292
SKOV3
HT29
FaDu
MCF7
BXPC3

Figure 2. Cytotoxicity EC_{50} (nM) of synthesized TOP1i payloads robustly tested in eight cancer cell lines and compared to SN-38 (govitecan payload) and DXd (deruxtecan payload). Each x-axis tick corresponds to a payload. The arrow marks ALX2004_payload. The dashed lines show the average EC_{50} (nM) of DXd across cell lines.

ALX2004's linker-payload shows superior bystander effect compared to deruxtecan in a cell-based bystander assay

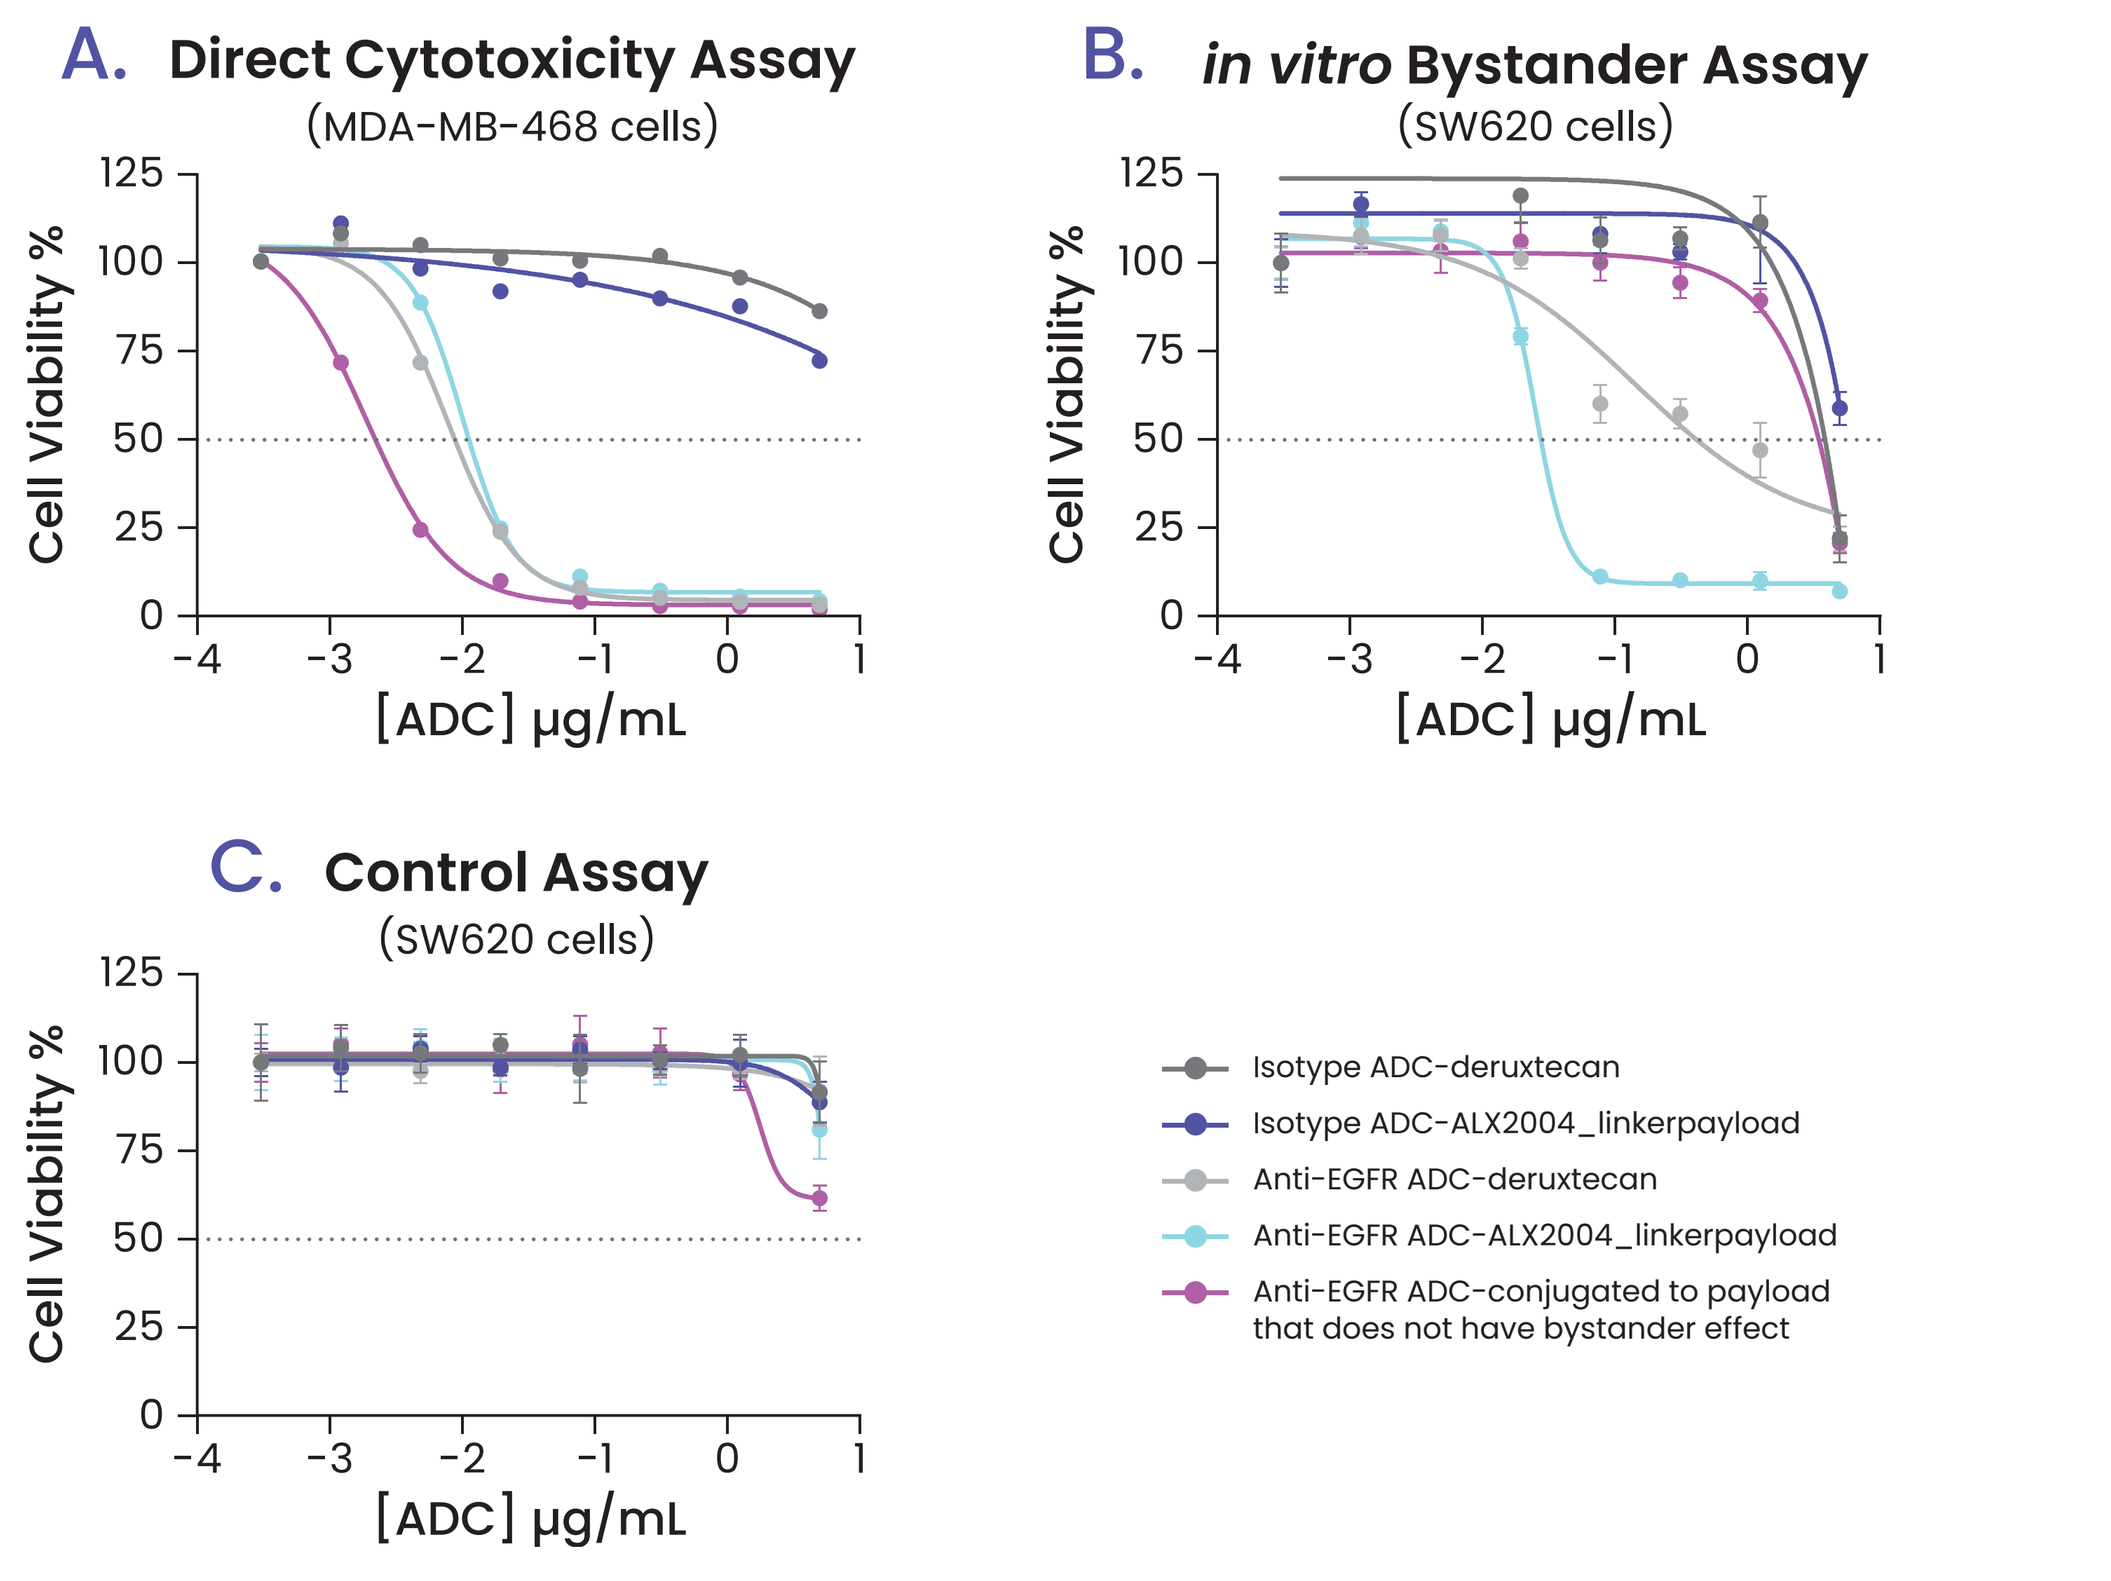
A. Direct Cytotoxicity Assay
(MDA-MB-468 cells)
B. in vitro Bystander Assay
(SW620 cells)
C. Control Assay
(SW620 cells)
Cell Viability %
[ADC] μg/mL
Isotype ADC-deruxtecan
Isotype ADC-ALX2004_linkerpayload
Anti-EGFR ADC-deruxtecan
Anti-EGFR ADC-ALX2004_linkerpayload
Anti-EGFR ADC-conjugated to payload that does not have bystander effect

Figure 3. In a cell-based bystander assay, the ADC with ALX2004's linker-payload showed a superior bystander effect compared to ADC with deruxtecan linker-payload (Figure **4A-C**). The *in vitro* bystander activity of ALX2004_linkerpayload vs deruxtecan was compared by generating two anti-EGFR ADCs, one conjugated to ALX2004_linkerpayload and the other to deruxtecan (both with DAR ~8). To evaluate the specificity and potency of the bystander effect, (**A**) EGFR positive MDA-MB-468 cells (~441,000 EGFR per cell surface) were first treated with anti-EGFR ADCs conjugated to ALX2004_linkerpayload (DAR ~8) or deruxtecan (DAR ~8) or respective isotype control ADCs (DAR ~8) for 6 days. (**B**) Then, cell supernatants from treated MDA-MB-468 cells in (**A**) were harvested and used to treat EGFR ultra-low SW620 cells (~2000 EGFR/cell surface) for 6 days. (**C**) As control, SW620 ultra-low EGFR cells (~2000 EGFR per cell surface) were treated with anti-EGFR ADCs and respective isotype control ADCs for 6 days. An anti-EGFR ADC conjugated to payload that has no bystander effect is shown as control in (**A-C**).

ALX2004 shows superior anti-tumor activity compared to anti-EGFR ADC conjugated to deruxtecan in CDX and *in vivo* bystander effect CDX mouse models

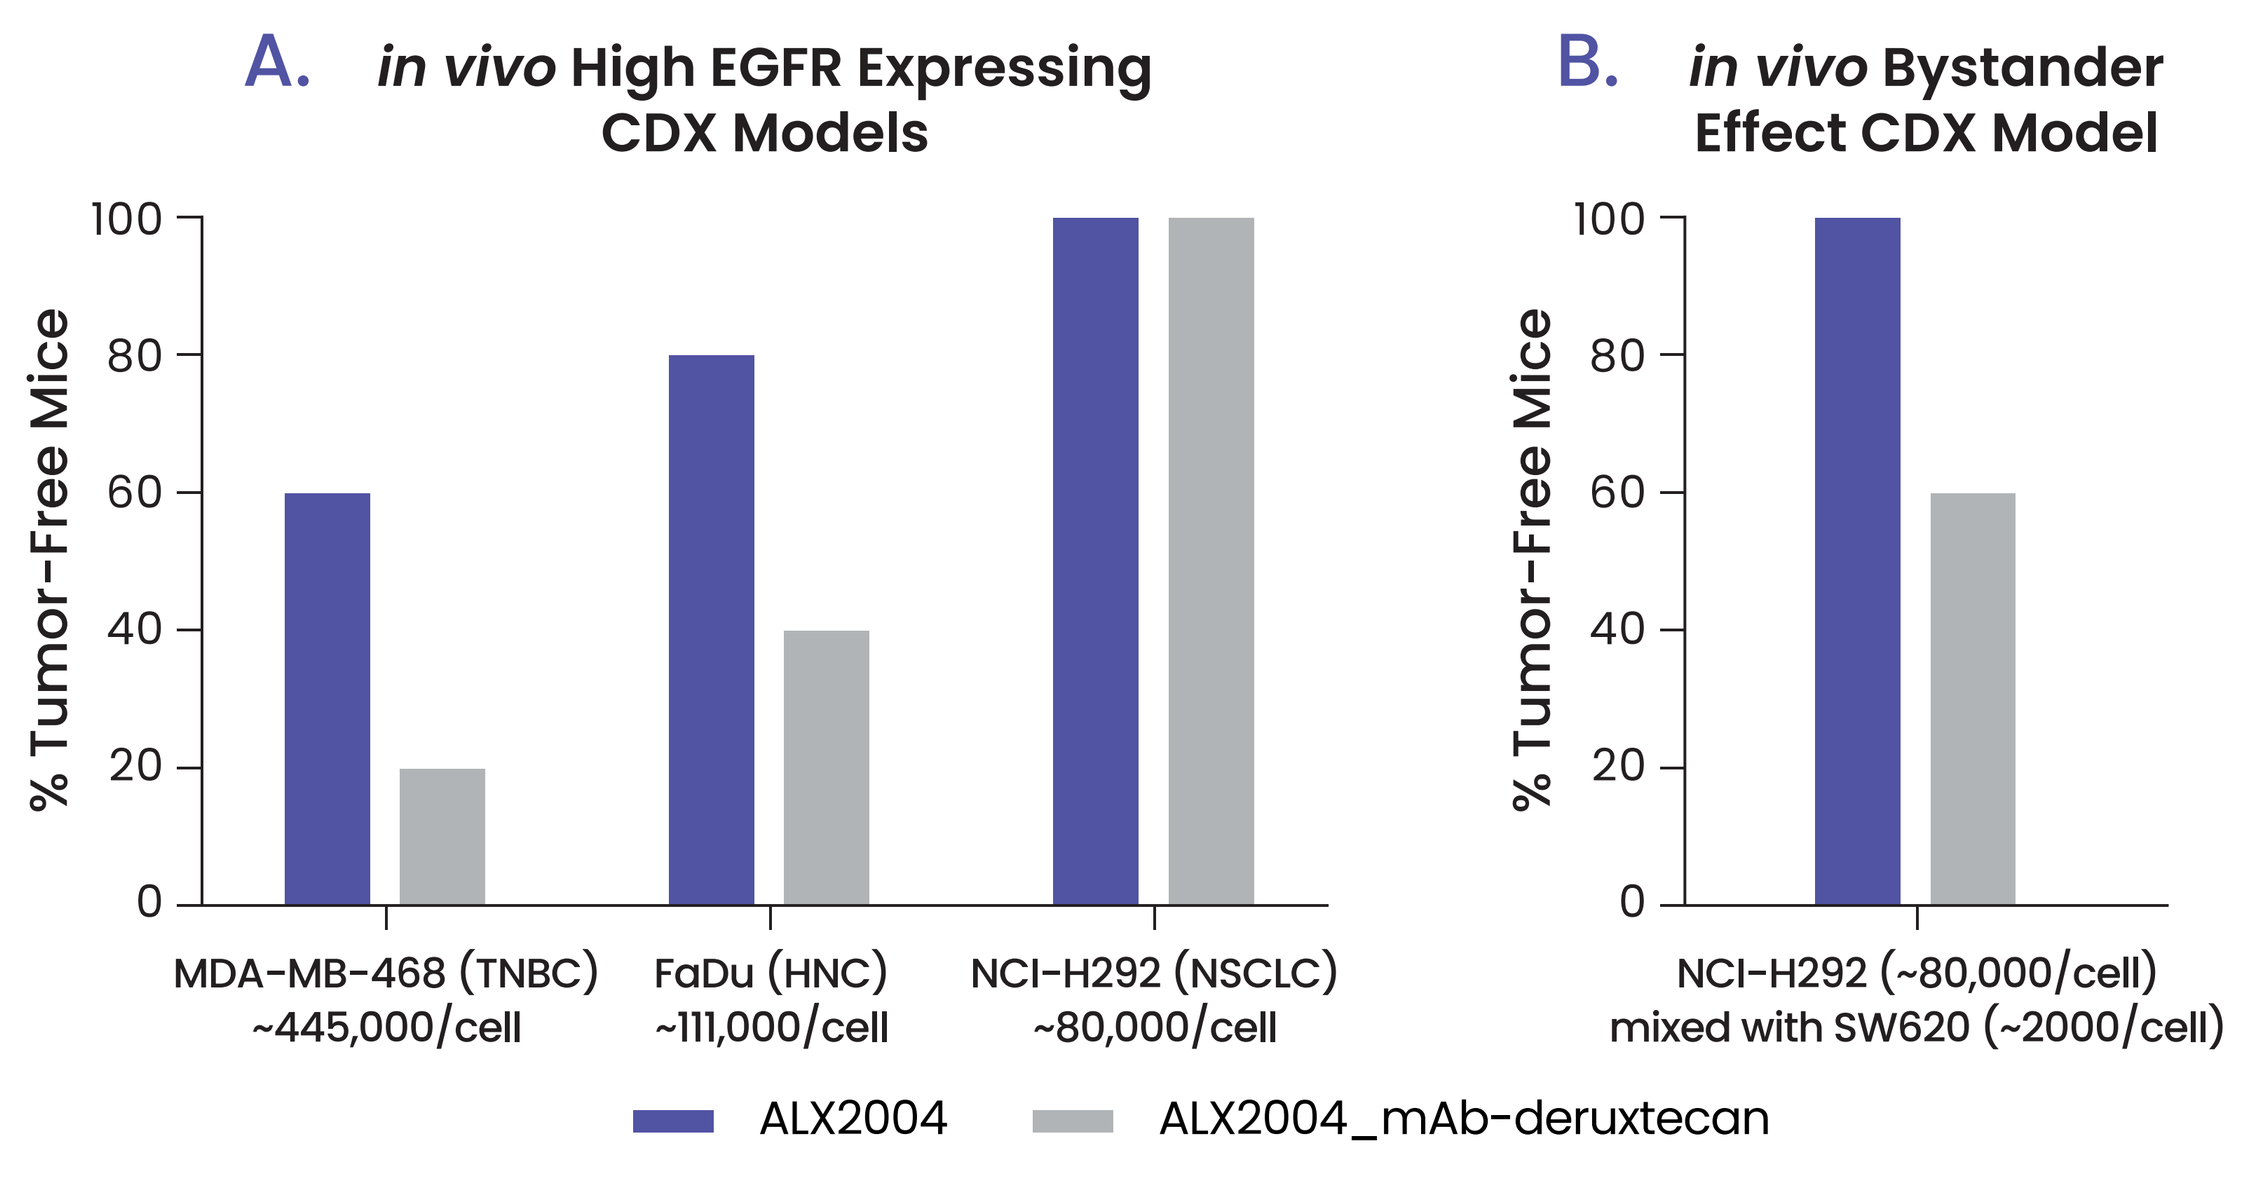
A. in vivo High EGFR Expressing CDX Models
B. in vivo Bystander Effect CDX Model
% Tumor-Free Mice
MDA-MB-468 (TNBC) ~445,000/cell
FaDu (HNC) ~111,000/cell
NCI-H292 (NSCLC) ~80,000/cell
NCI-H292 (~80,000/cell) mixed with SW620 (~2000/cell)
ALX2004
ALX2004_mAb-deruxtecan

Figure 4. Percent of tumor-free mice in CDX models dosed with ALX2004 or ALX2004_mAb-deruxtecan (ADC composed of ALX2004's antibody conjugated to deruxtecan) (both ADCs, DAR ~8). (**A**) MDA-MB-468 (3 mg/kg, 1 dose), FaDu (1 mg/kg, 3 doses, Q1W), NCI-H292 (3 mg/kg, 3 doses, Q1W), (**B**) Bystander effect CDX model composed of 1:1 NCI-H292 cells (~80,000 EGFR/cell surface) and SW620 cells (~2000 EGFR/cell surface) dosed 3 mg/kg, 3 doses, Q1W. NOD SCID mice, n=5 per group.

ALX2004's linker-payload designed to deliver payloads to tumors, not the periphery

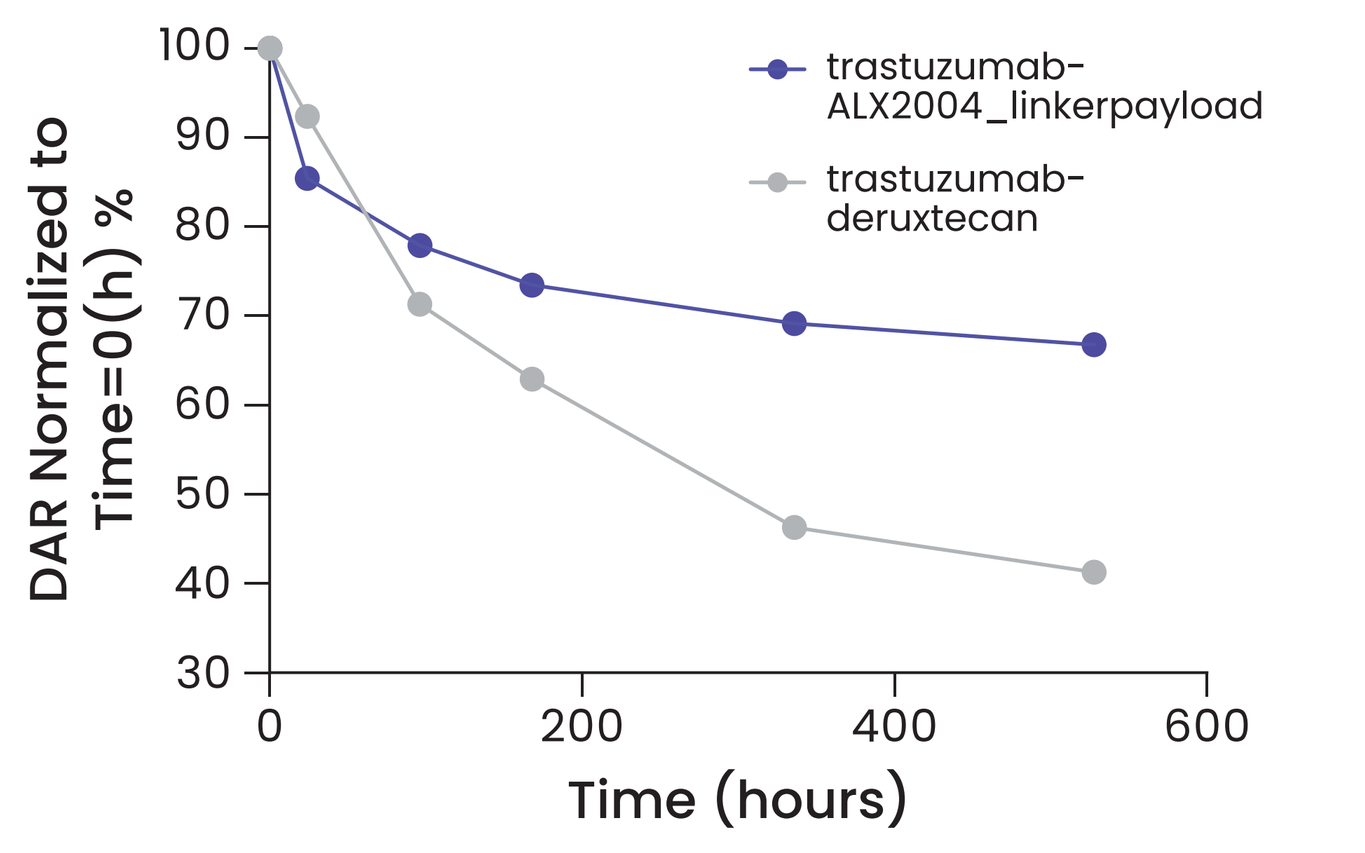
DAR Normalized to Time=0(h) %
Time (hours)
trastuzumab-ALX2004_linkerpayload
trastuzumab-deruxtecan

Figure 5. Improved DAR stability of ALX2004_linkerpayload compared to deruxtecan. Graph shows DAR of two ADCs: trastuzumab conjugated to ALX2004_linkerpayload (DAR ~8) and trastuzumab conjugated to deruxtecan (DAR ~8), as a function of time in non-human primate (NHP) dosed at 30 mg/kg (n=2 per group). DAR was analyzed using reduced middle-down RP-LCMS.

ALX2004 induces cell markers of immunogenic cell death *in vitro*

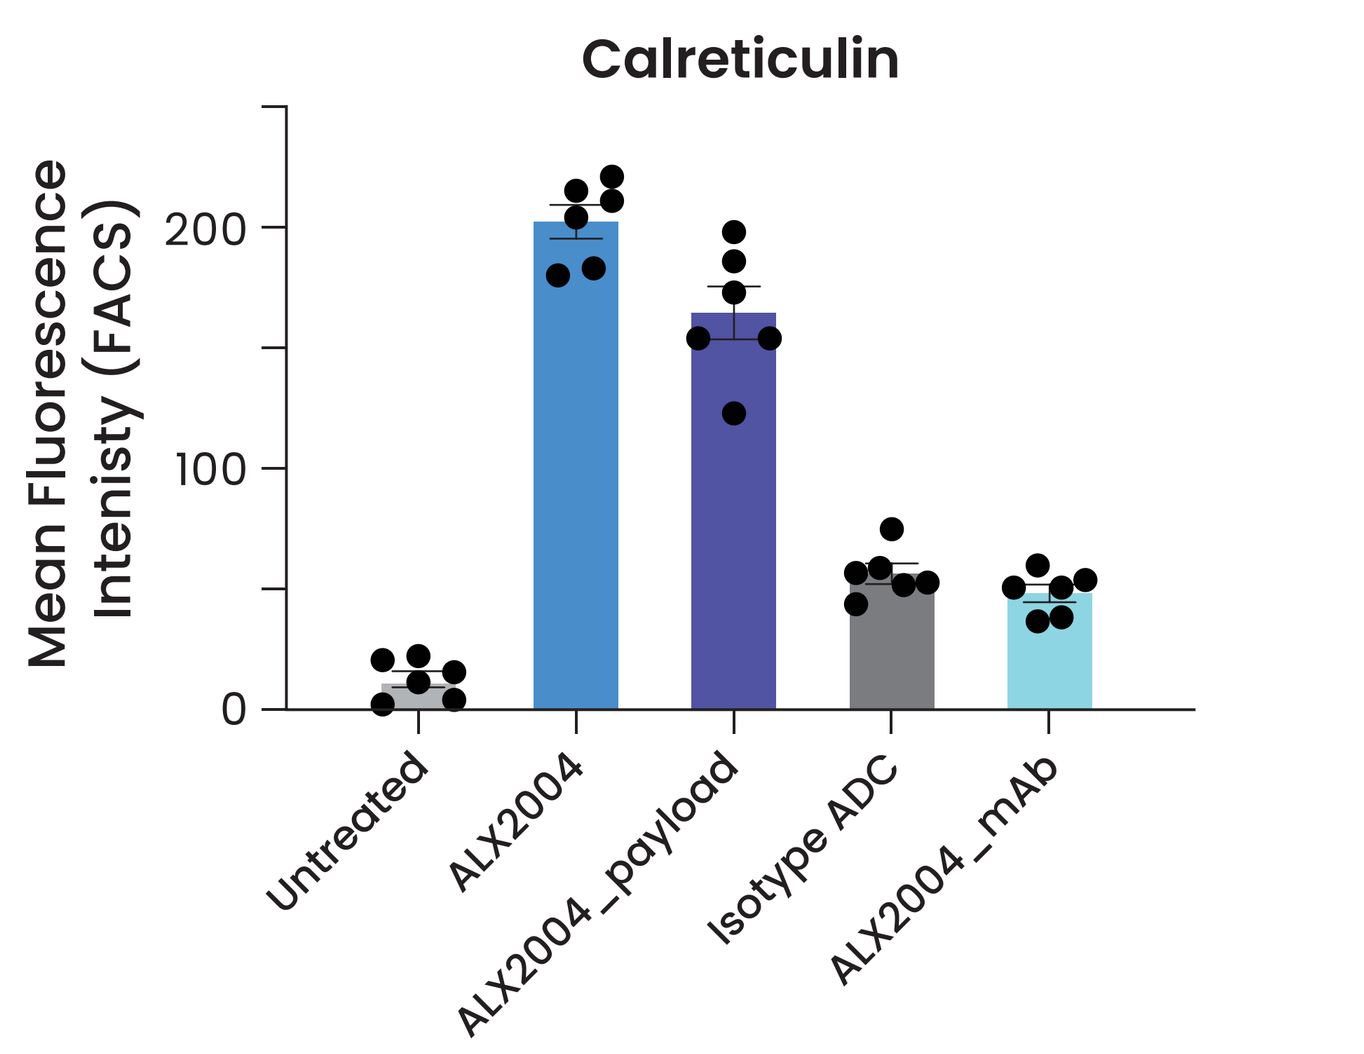
Calreticulin
Mean Fluorescence Intenisty (FACS)
Untreated
ALX2004
ALX2004_payload
Isotype ADC
ALX2004_mAb

Figure 6. Cell surface levels of calreticulin in MDA-MB-468 cell line following 72-hour incubation with indicated test agents. Similar trends were observed for induction of HMGB1 and ATP cell markers (data not shown). Isotype ADC was conjugated to ALX2004_linker payload (DAR ~8).

ALX2004 induces ADCC and ADCP

Target Cell Line	EGFR Number Expressed on Cell Surface	ALX2004 ADCC EC_{50} (nM)	Isotype Control ADC ADCC EC_{50} (nM)
MDA-MB-468	441,000	0.1	N/A
		ADCP EC_{50} (nM)	ADCP EC_{50} (nM)
MDA-MB-468	441,000	1.0	N/A

Table 1. ALX2004 induced ADCC and ADCP *in vitro*. ADCC was assayed using Jurkat-Lucia™ NFAT-CD16 effector reporter cells and MDA-MB-468 target cells. ADCP was assayed using Jurkat-Lucia™ NFAT-CD32 effector reporter cells and MDA-MB-468 target cells. Isotype ADC was conjugated to A2004_linkerpayload (DAR ~ 8).

ALX2004 maintains anti-EGFR based antitumor activity by blocking EGFR tyrosine kinase domain phosphorylation

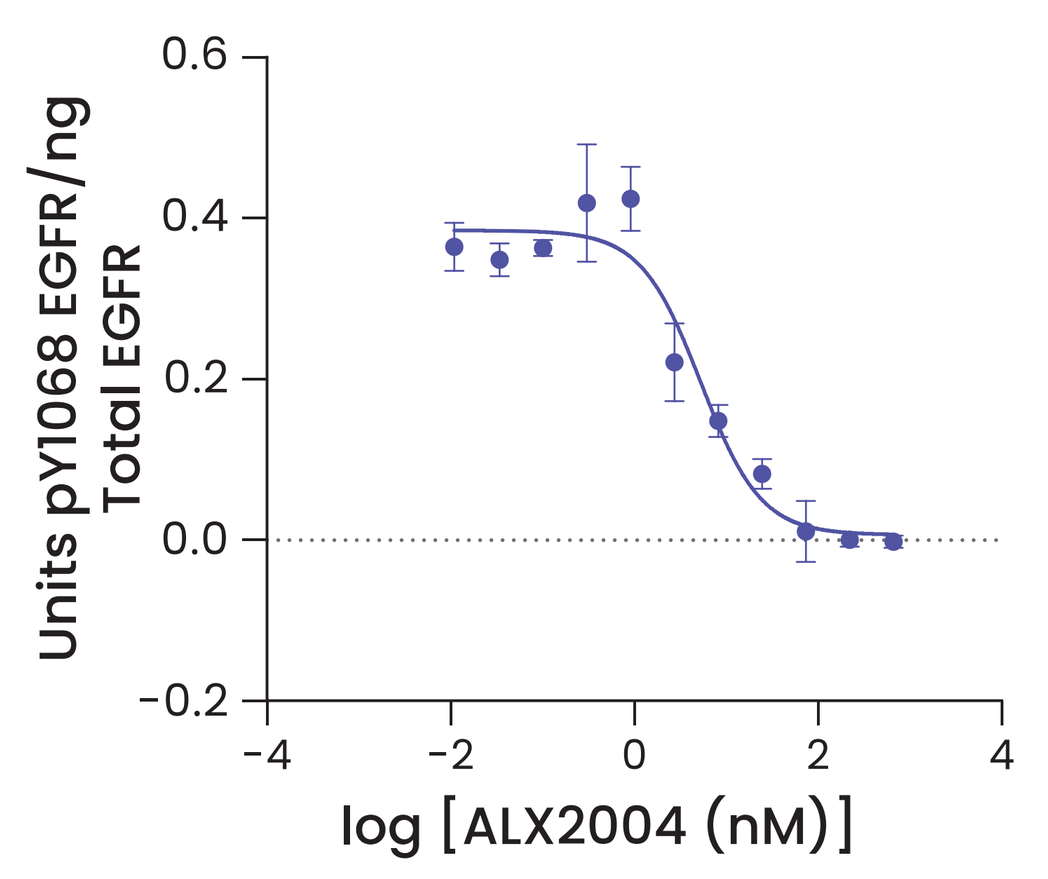
Units pY1068 EGFR/ng Total EGFR
log [ALX2004 (nM)]

Figure 7. Dose-response curve of inhibitory activity of ALX2004 against EGF-induced phosphorylation of EGFR intracellular tyrosine kinase domain in MDA-MB-468. Phosphorylation of EGFR at site Y1068 was quantified by ELISA, total levels of full-length EGFR independent of the phosphorylation status were quantified by ELISA. Phosphorylated EGFR (units/mL extracted protein) was normalized to 0 ng/mL EGF treated background control and to ng total EGFR protein amount (total EGFR ng/mL extracted protein).

ALX2004 Demonstrated Potent *in vivo* Anti-Tumor Activity

ALX2004 demonstrated potent antitumor activity in *in vivo* models with differing levels of EGFR expression and varied mutational status across the EGFR signaling pathway, including EGFR with wild-type and mutant kinase domain and wild-type and mutant KRAS and BRAF in NSCLC, HNC, CRC, TNBC, and PDAC CDX or PDOX models

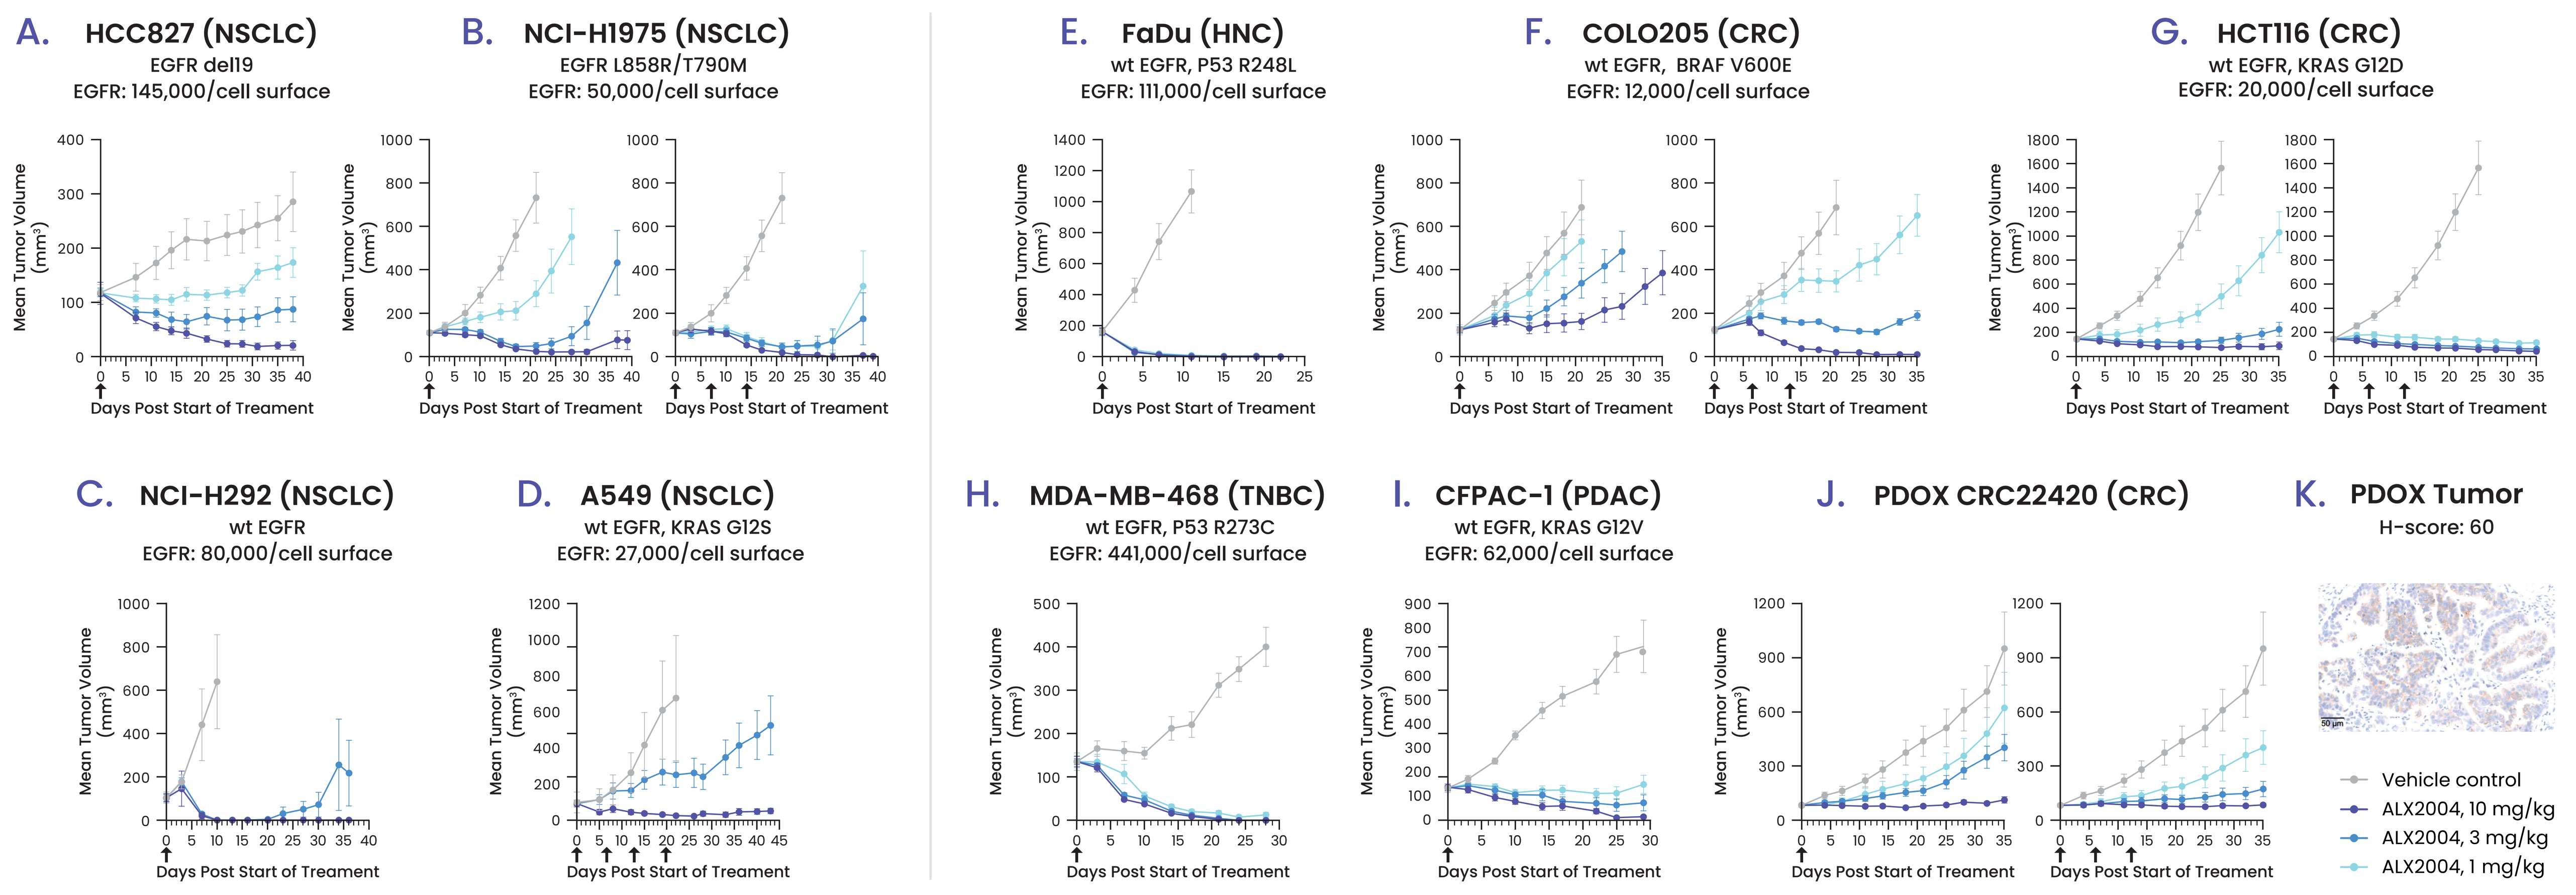
A. HCC827 (NSCLC)
EGFR del19
EGFR: 145,000/cell surface
B. NCI-H1975 (NSCLC)
EGFR L858R/T790M
EGFR: 50,000/cell surface
E. FaDu (HNC)
wt EGFR, P53 R248L
EGFR: 111,000/cell surface
F. COLO205 (CRC)
wt EGFR, BRAF V600E
EGFR: 12,000/cell surface
G. HCT116 (CRC)
wt EGFR, KRAS G12D
EGFR: 20,000/cell surface
C. NCI-H292 (NSCLC)
wt EGFR
EGFR: 80,000/cell surface
D. A549 (NSCLC)
wt EGFR, KRAS G12S
EGFR: 27,000/cell surface
H. MDA-MB-468 (TNBC)
wt EGFR, P53 R273C
EGFR: 441,000/cell surface
I. CFPAC-1 (PDAC)
wt EGFR, KRAS G12V
EGFR: 62,000/cell surface
J. PDOX CRC22420 (CRC)
K. PDOX Tumor
H-score: 60
Mean Tumor Volume (mm³)
Days Post Start of Treament
Vehicle control
ALX2004, 10 mg/kg
ALX2004, 3 mg/kg
ALX2004, 1 mg/kg

Figure 8. ALX2004 has inhibited tumor growth *in vivo*. Tumor volume (mm^3) after IV administration of a single dose or three doses of ALX2004 at indicated dose levels (mg/kg) and dosing frequencies in CDX mouse models of NSCLC (**A-D**) and other EGFR-expressing tumors (**E-J**): (**A**) HCC827, (**B**) NCI-H1975, (**C**) NCI-H292, (**D**) A549, (**E**) FaDu, (**F**) COLO205, (**G**) HCT116, (**H**) MDA-MB-468, (**I**) CFPAC-1, (**J**) PDOX model CRC22420. Dosing frequency indicated by black arrows. Data represented as mean ± SEM, (**A-B**, **E-J**) n=12 per group, NU/NU mice, (**C-D**) n=5 per group, NOD SCID mice. (**K**) Representative IHC staining of EGFR in PDOX tissue. wt refers to EGFR wild-type tyrosine kinase domain.

Safety Profile Findings in Non-Human Primates Toxicity Study

- The toxicity and toxicokinetic profile of ALX2004 was evaluated in a 6-week repeat-dose (Q3W dosing) GLP toxicity study in monkeys, at doses of 5, 10, and 20 mg/kg
- No dose-limiting major target organ toxicity, including on-target toxicity (i.e., skin or other EGFR-expressing cells), was observed
- There was no evidence of ILD
- NOAEL was 10 mg/kg; HNSTD was 20 mg/kg
- All findings were minimal to moderate and fully recoverable

Summary

- ALX2004 carries a proprietary TOP1i payload designed and selected to offer similar direct cytotoxic potency with enhanced bystander effect compared to the clinical standard, deruxtecan
- The linker-payload of ALX2004 has been designed with improved stability in circulation to minimize off-tumor linker-payload release
- ALX2004 has antibody-dependent antitumor activities (blocks EGFR signaling, ADCC, and ADCP), as demonstrated by *in vitro* assays
- As shown in *in vitro* assays, ALX2004 could activate the immune system for longer-term tumor suppression (ICD, ADCC, and ADCP)
- ALX2004 has shown potent antitumor activity in EGFR-expressing CDX models across a range of EGFR expression levels and mutation status, e.g., with wild-type and mutated tyrosine kinase domain and mutations in BRAF and KRAS, as well as several tumor types (NSCLC, HNC, CRC, TNBC, and PDAC)
- A favorable preclinical NHP safety profile (e.g., no skin toxicity or ILD) in combination with potent antitumor activity across multiple CDX tumor types in mouse models support the clinical evaluation of ALX2004
- A phase 1, first-in-human study evaluating ALX2004 in patients with advanced or metastatic solid tumors is enrolling (NCT07085091), and trial details are presented at this meeting by Spira A, et al., on the poster "A Phase 1, First-in-Human, Open-Label, Multicenter Study to Evaluate ALX2004, an Antibody–Drug Conjugate Targeting EGFR, in Patients with Advanced or Metastatic Select Solid Tumors (ALX2004-01)," Abstract #LB-A004

Abbreviations: **ADC** – Antibody drug conjugate; **ADCC** – Antibody-dependent cellular cytotoxicity; **ADCP** – Antibody-dependent cellular phagocytosis; **ALX2004** – ADC; **ALX2004_linkerpayload** – Linker-payload used in ALX2004; **ALX2004_mAb** – Antibody used in ALX2004; **ALX2004_mAb-deruxtecan** – Antibody used in ALX2004 conjugated to deruxtecan; **ALX2004_payload** – Payload used in ALX2004; **CDX** – Cell line derived xenograft; **CRC** – Colorectal cancer; **DAR** – Drug to antibody ratio; **EGFR** – Epidermal growth factor receptor; **ELISA** – Enzyme-linked immunosorbent assay; **FACS** – Fluorescence-activated cell sorting; **FDA** – U.S. Food and Drug Administration; **GLP** – Good laboratory practice; **HNC** – Head and neck cancer; **HNSTD** – Highest non-severely toxic dose; **ICD** – Immunogenic cell death; **IgG** – Immunoglobulin G; **IHC** – Immunohistochemistry; **ILD** – Interstitial lung disease; **IV** – Intravenous; **mAb** – Monoclonal antibody; **NHP** – Non-human primate; **NOAEL** – No observed adverse effect level; **NSCLC** – Non-small cell lung cancer; **PDAC** – Pancreatic cancer; **PDOX** – Patient-derived organoid xenograft; **RP-LCMS** – Reversed-phase liquid chromatography mass spectrometry; **SEM** – Standard error of the mean; **TNBC** – Triple-negative breast cancer; **TOP1i** – Topoisomerase 1 inhibitor; **Trastuzumab-ALX2004_linker payload** – Trastuzumab conjugated to linker-payload used in ALX2004; **Q1W** – Dosing frequency of once every week; **Wt** – Wild-type EGFR tyrosine kinase domain.

Disclosures: Marija Vrljic is an employee of ALX Oncology. **Acknowledgments:** We thank all the participating patients, their families and site research staff. **Contact Email:** ALX2004trial@alxoncology.com **Trial Registration:** ClinicalTrials.gov identifier, NCT07085091 **Presented at:** The AACR-NCI-EORTC Annual Meeting 2025, October 17-21, 2025. Abstract #A119.