A Phase 1, First-in-Human, Open-Label, Multicenter Study to Evaluate ALX2004, an Antibody–Drug Conjugate Targeting EGFR, in Patients with Advanced or Metastatic Select Solid Tumors (ALX2004-01)

Alexander I. Spira[1], William B. McKean[2], Arvind Chaudhry[3], Siqing Fu[4], Cherry Mao[5], Max Ma[5], Athanasios C. Tsiatis[5], Alan Sandler[5], Nehal J. Lakhani[6]

[1]Virginia Cancer Specialists, Fairfax, VA, USA; [2]START Center for Cancer Research, West Valley City, UT, USA; [3]Summit Cancer Centers, North Spokane, WA, USA; [4]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [5]ALX Oncology Inc., South San Francisco, CA, USA; [6]START Center for Cancer Research, Grand Rapids, MI, USA

2025 AACR-NCI-EORTC

Background

- ALX2004 is a uniquely designed EGFR-targeted TOP1i-based ADC (DAR 8; **Figure 1**)[1]
- ALX2004 has demonstrated antitumor activity *in vitro* and *in vivo* across ten cell line-derived xenograft models and five tumor types with varying levels of EGFR expression and mutation status[1]
- In non-human primate studies, ALX2004 showed a favorable safety profile with no major organ toxicity (eg, skin toxicity or interstitial lung disease) at clinically relevant doses[1]
- **ALX2004-01** is a first-in-human study to evaluate the safety, tolerability, and preliminary efficacy of ALX2004 in participants with advanced or metastatic select solid tumors that are known to express EGFR
- Preclinical data for ALX2004 are presented at this meeting by Marija Vrljic, et al., on the poster titled "ALX2004: a novel topoisomerase I inhibitor antibody–drug conjugate for the treatment of EGFR-expressing solid tumors"[1]

Figure 1. ALX2004 was Meticulously Designed to Maximize the Therapeutic Window and has the Potential to Establish Proof-of-Concept Early in the Development Cycle

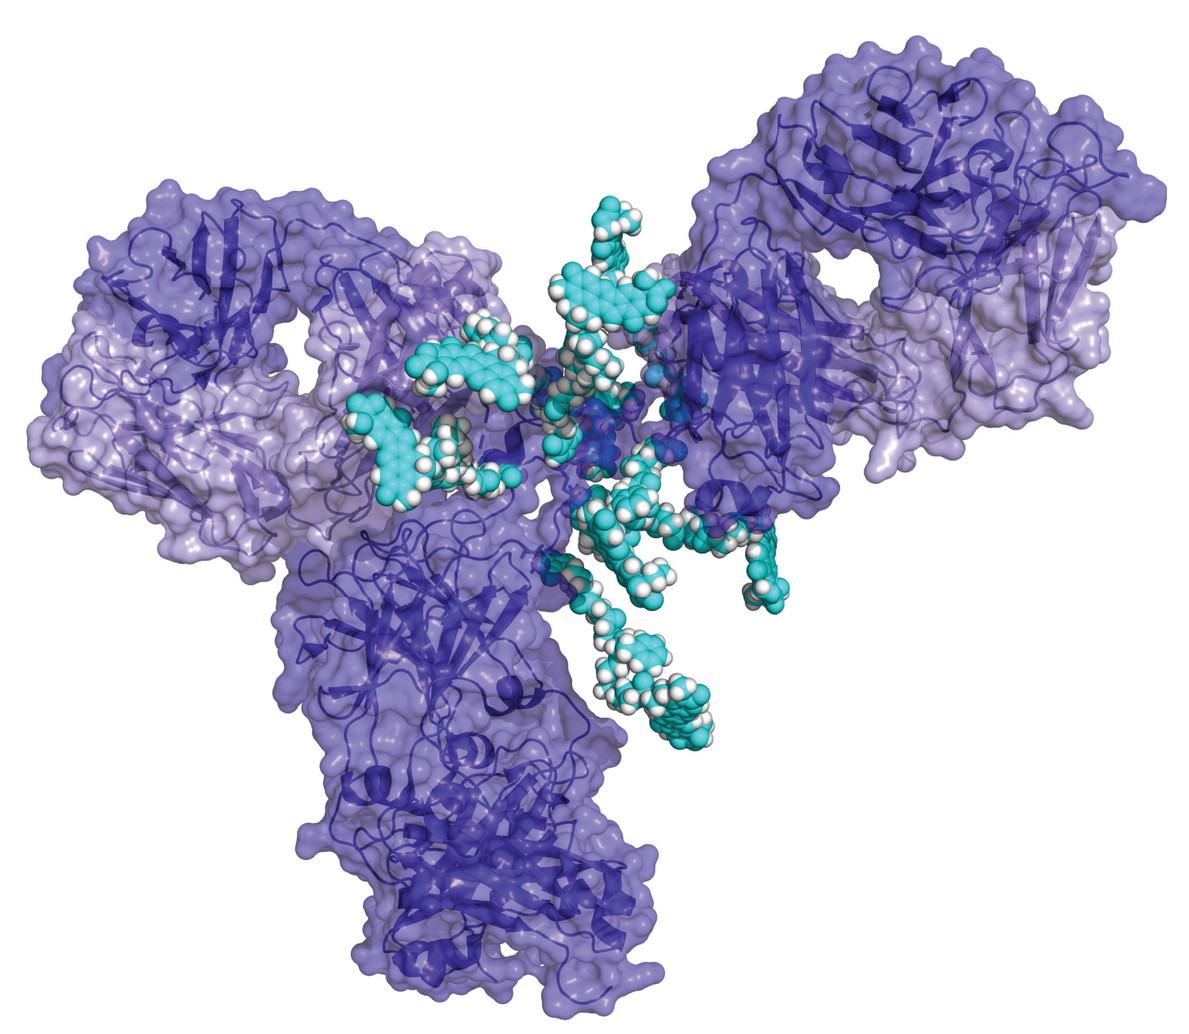

ALX2004
EGFR-targeted ADC DAR 8
TOP1i payload

EGFR Antibody
Matuzumab-derived EGFR antibody selected to minimize off-tumor skin toxicity and to maximize therapeutic window
Epitope distinct from that of FDA-approved EGFR antibodies

Proprietary Linker-Payload
Lysosomal cleavage like deruxtecan ADCs with improved linker-antibody stability to minimize off-tumor payload release

Proprietary TOP1i Payload, DAR 8
TOP1i with similar direct cytotoxic potency and enhanced bystander activity compared to deruxtecan

ALX2004 Mechanism of Action

- Preclincal *in vitro* and/or *in vivo* models demonstrated that multiple mechanisms of action may contribute to the antitumor activity of ALX2004 (**Figure 2**)

1. Direct tumor-cell killing by its cytotoxic payload via inhibition of TOP1
2. Payload-mediated bystander killing of neighboring cells
3. Payload-mediated immunogenic cell death
4. Antibody-dependent cellular cytotoxicity and antibody-dependent cellular phagocytosis mediated by immunoglobulin G1 fragment crystallizable domain
5. Inhibition of EGFR signaling pathway

- The clinical potential of ALX2004 is further supported by tumor regression in animal models
- In non-human primate studies, ALX2004 showed a favorable safety profile with no major organ toxicity (eg, skin toxicity or interstitial lung disease) at clinically relevant doses

Figure 2. ALX2004 is Designed to Optimize All Mechanisms of ADC Cancer Killing While Maximizing the Therapeutic Window

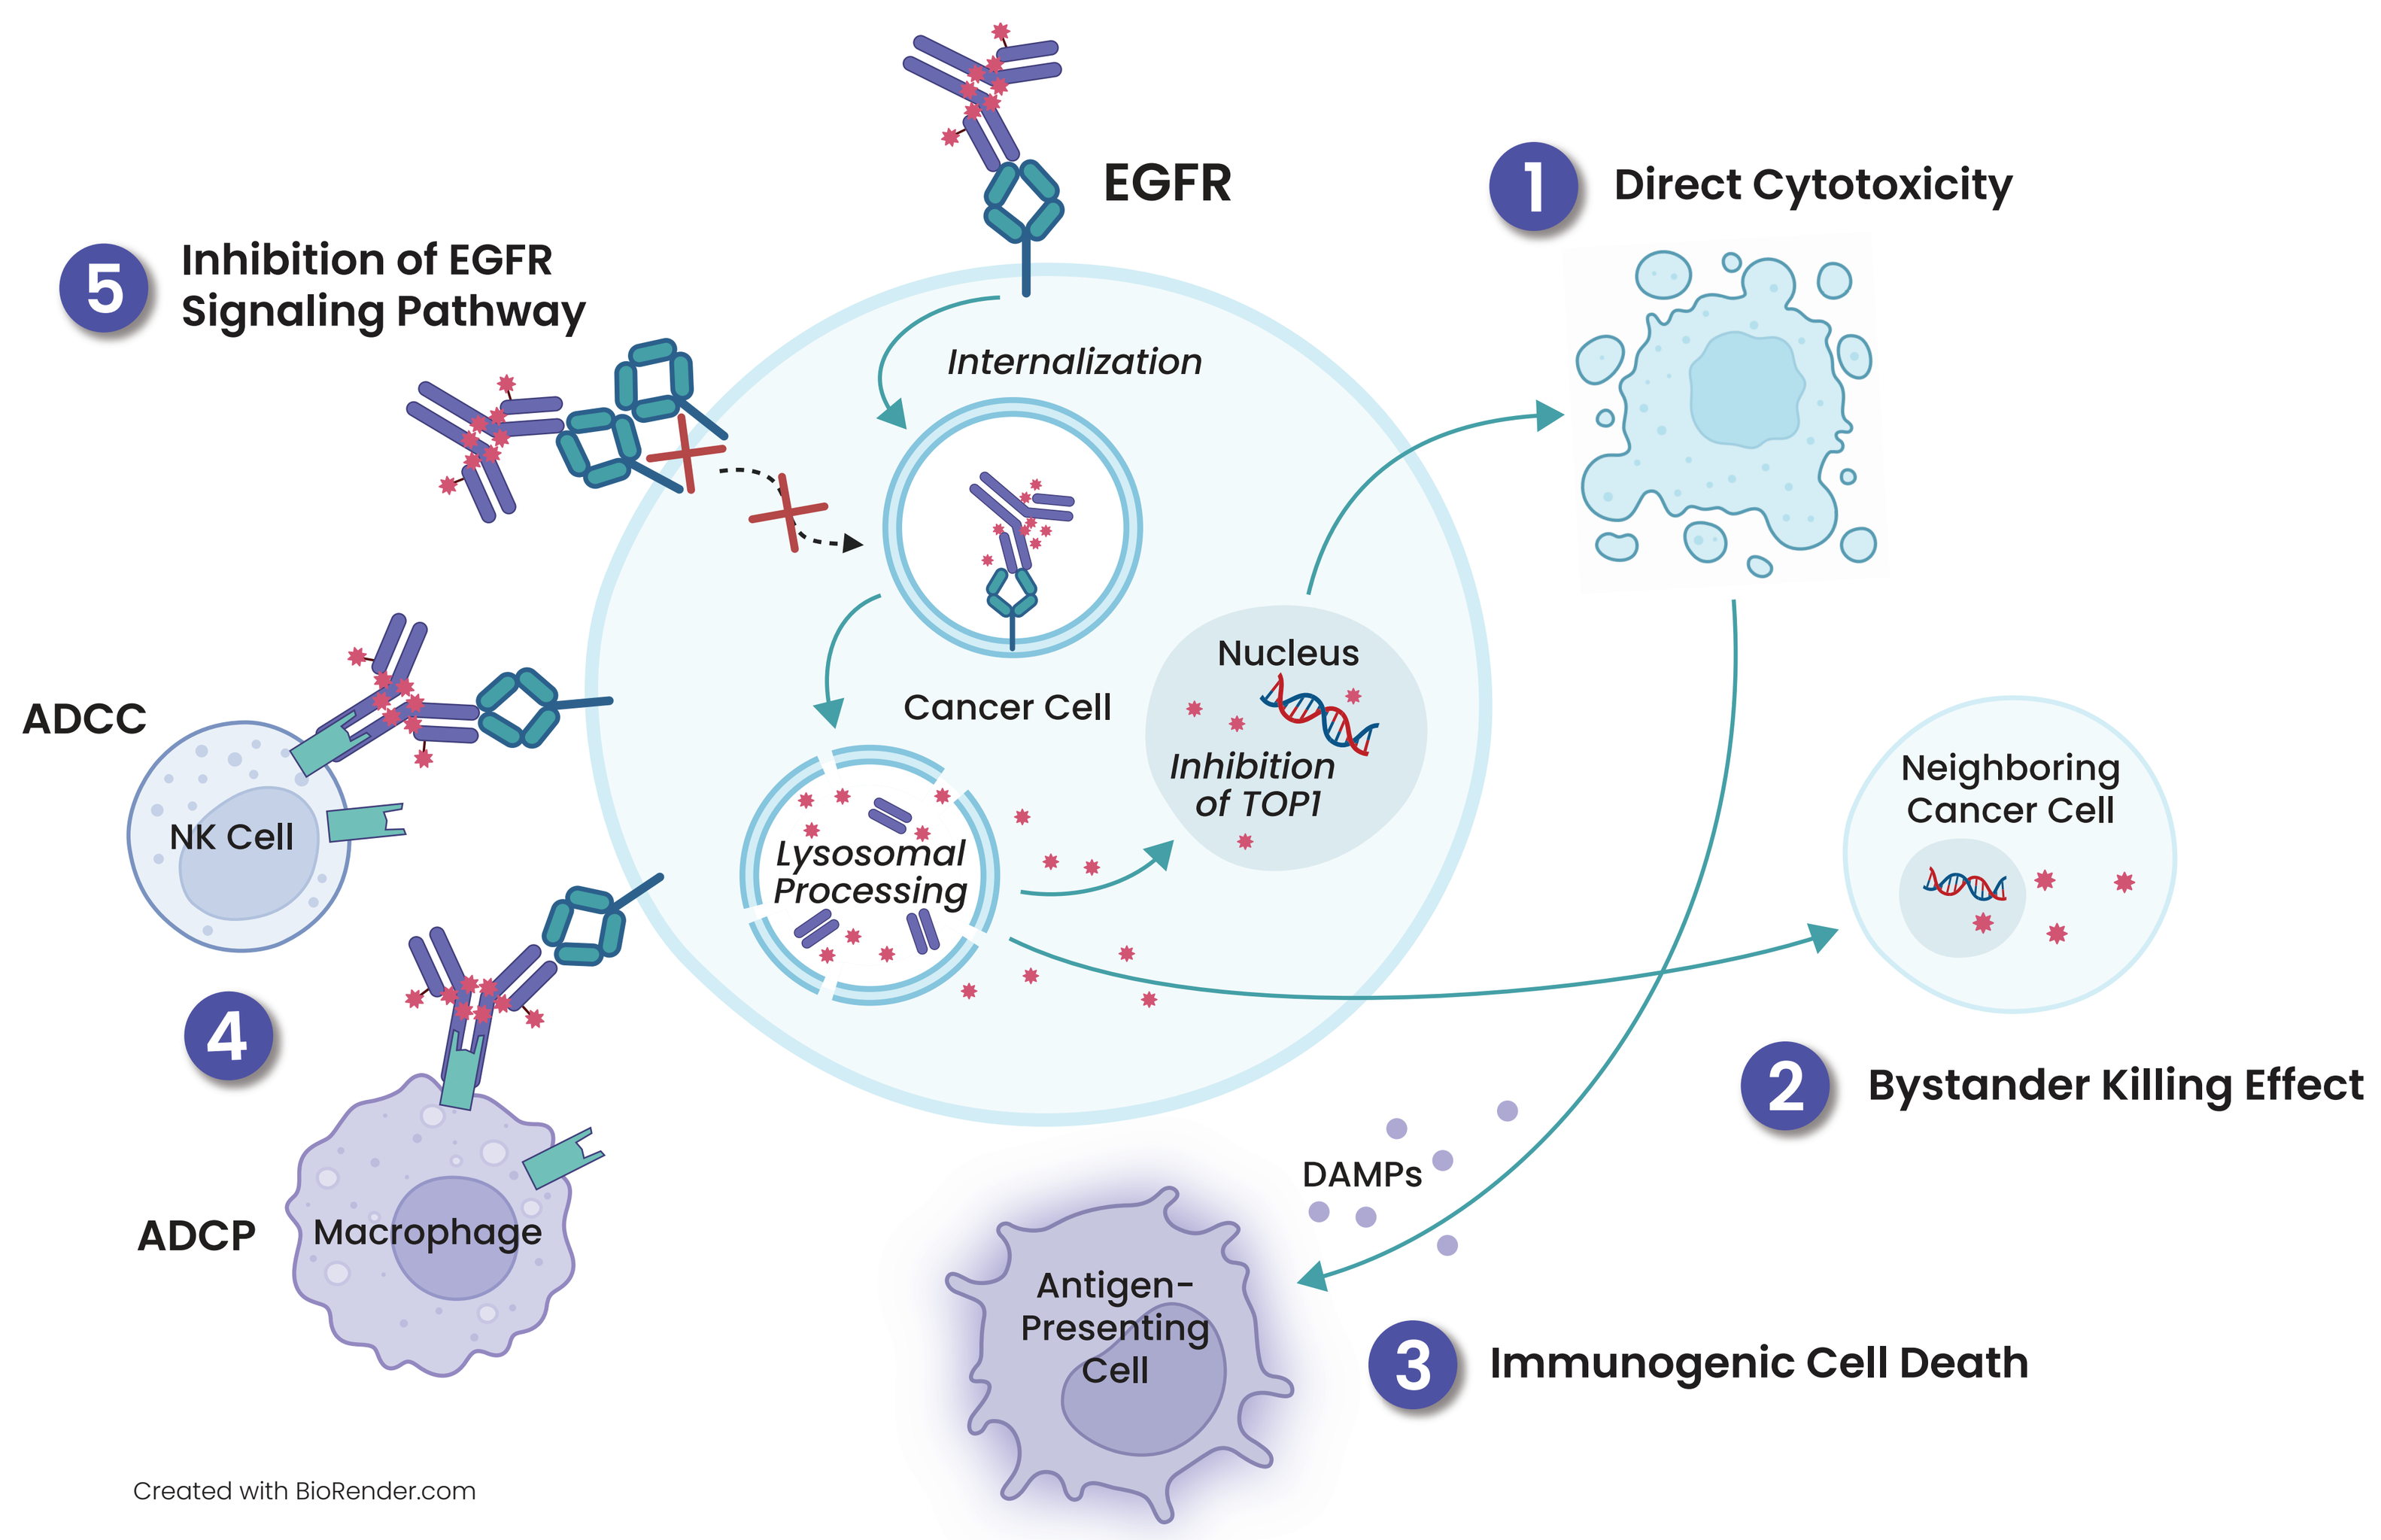

EGFR
1 Direct Cytotoxicity
5 Inhibition of EGFR Signaling Pathway
Internalization
Cancer Cell
Nucleus
Inhibition of TOP1
Lysosomal Processing
ADCC
NK Cell
4
ADCP
Macrophage
Neighboring Cancer Cell
2 Bystander Killing Effect
DAMPs
Antigen-Presenting Cell
3 Immunogenic Cell Death
Created with BioRender.com

Best-in-Class Design Elements

- Payload and stable linker conjugation
 - Potent direct killing of targeted tumor cells
 - Bystander indirect killing of neighboring target-negative and ultra-low tumor cells
 - Cleaves in the tumor
 - Maintains stability elsewhere to minimize systemic off-target toxicity inherent to cytotoxic payloads

Immunomodulatory

- Activation of immune system through recruiting APCs, for longer-term tumor suppression

Antibody

- Retain antibody-dependent anticancer activities
- Minimize off-tumor effects of payload delivery

Study Methods

- **ALX2004-01** is a Phase 1, first-in-human, open-label study to evaluate the safety, tolerability, and preliminary efficacy of ALX2004 in participants with advanced or metastatic select solid tumors that are known to express EGFR
- Phase 1a is composed of dose escalation and optional dose exploration to identify the Randomized Dose(s) for Expansion. Up to 80 patients may be enrolled in the Dose Expansion. (Phase 1b) (**Figure 3**)
- This protocol will investigate the safety and tolerability, MTD, RDE(s), single- and multiple-dose PK profiles, and PD markers of ALX2004, as well as evaluate the preliminary efficacy of ALX2004 given as monotherapy in select tumor types known to express EGFR
 - Tumor types for Dose Escalation are NSCLC, HNSCC, CRC, ESCC
 - Tumor types for Dose Exploration and Dose Expansion will be a subset of these original four
- During Dose Escalation, ALX2004 will be administered IV in escalating DL cohorts beginning at the first DL of Q3W 1 mg/kg
- Dose Escalation will proceed using the BOIN design

Figure 3. ALX2004-01 Trial Design

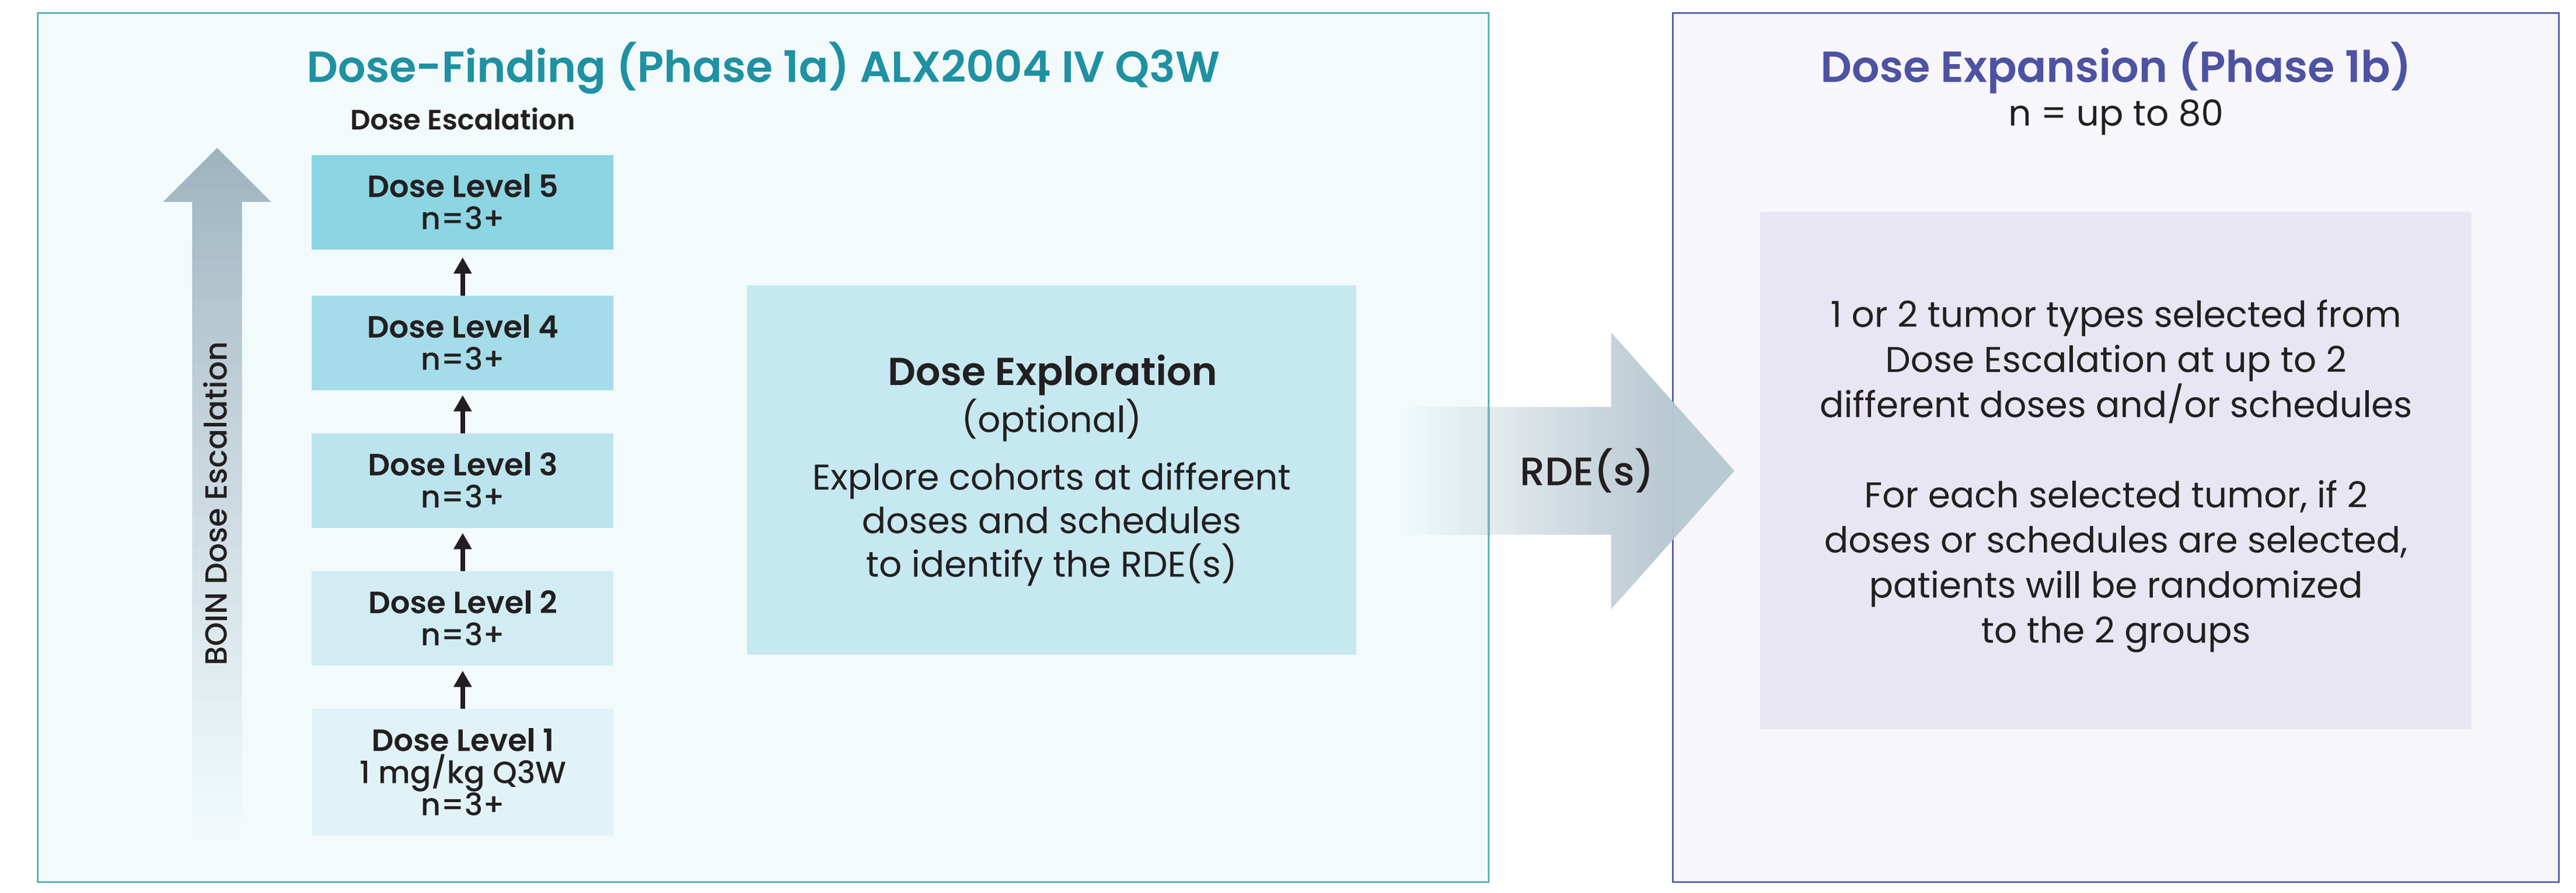

Dose-Finding (Phase 1a) ALX2004 IV Q3W
Dose Escalation
BOIN Dose Escalation
Dose Level 5 n=3+
Dose Level 4 n=3+
Dose Level 3 n=3+
Dose Level 2 n=3+
Dose Level 1 1 mg/kg Q3W n=3+
Dose Exploration (optional)
Explore cohorts at different doses and schedules to identify the RDE(s)
RDE(s)
Dose Expansion (Phase 1b)
n = up to 80
1 or 2 tumor types selected from Dose Escalation at up to 2 different doses and/or schedules
For each selected tumor, if 2 doses or schedules are selected, patients will be randomized to the 2 groups

- This clinical trial is registered with ClinicalTrials.gov and is now enrolling (NCT07085091)

Eligibility Criteria

Key Inclusion Criteria

- Histologically confirmed solid tumor types known to express EGFR: NSCLC, HNSCC, CRC, or ESCC
 - **Dose Escalation:** Participants who have relapsed or progressed following prior anticancer therapy in the advanced/metastatic setting and for whom no approved or standard therapy is available
 - **Dose Exploration:** All or a subset of tumors tested in Dose Escalation including NSCLC (≤2 prior lines of therapy), HNSCC (≤3 prior lines of therapy), CRC (≤4 prior lines of therapy), and ESCC (≤3 prior lines of therapy)
 - **Dose Expansion:** Subset of tumors tested in Dose Escalation
- ECOG performance status must be 0 or 1
- Participants must have measurable disease according to RECIST v1.1

Key Exclusion Criteria

- Disease suitable for local therapy with curative intent
- Prior treatment with any ADCs that have a TOP1i-based component
- Participants receiving systemic anticancer therapy within 28 days or five half-lives (whichever is shorter) of C1D1
- Any major surgery, non-healing wound, ulcer, or bone fracture requiring hospitalization within 2 months prior to the first dose of study drug
- History of symptomatic uncontrolled ascites, pleural effusion, or pericardial effusion requiring therapeutic intervention within 1 month prior to the first dose of study drug

Study Objectives

Primary Objective

- To evaluate the safety and tolerability of ALX2004 as a single agent in previously treated participants with known EGFR-expressing advanced or metastatic select solid tumors
- To determine the MTD at 3-weekly intervals and the RDE(s) of ALX2004 as a single agent

Key Secondary Objectives

- To characterize the single- and multiple-dose PK profiles of ALX2004 total antibody, total ADC, and TOP1i payload
- To evaluate the preliminary antitumor activity of ALX2004

Key Exploratory Objectives

- To explore the PD effects of ALX2004 and their association with PK, safety, and efficacy endpoints

References: 1. Vrljic M, et al. Presented at the AACR-NCI-EORTC 2025, Abstract #119.
Abbreviations: **ADC** – Antibody–drug conjugate; **ADCC** – Antibody-dependent cell mediated cytotoxicity; **ADCP** – Antibody-dependent cellular phagocytosis; **APC** – Antigen-presenting cell; **BOIN** – Bayesian optimal interval; **C1D1** – Cycle 1, day 1; **CRC** – Colorectal carcinoma; **DAMP** – Damage-associated molecular pattern; **DAR** – Drug-to-antibody ratio; **DL** – Dose level; **ECOG** – Eastern Cooperative Oncology Group; **EGFR** – Epidermal growth factor receptor; **ESCC** – Esophageal squamous cell carcinoma; **FDA** – Food and Drug Administration; **HNSCC** – Head and neck squamous cell carcinoma; **IV** – Intravenously; **MTD** – Maximum tolerated dose; **NK** – Natural killer; **NSCLC** – Non-small cell lung cancer; **PD** – Pharmacodynamic; **PK** – Pharmacokinetic; **Q2W** – Every 2 weeks; **Q3W** – Every 3 weeks; **RDE** – Recommended dose for expansion; **TOP1i** – Topoisomerase I inhibitor.
Disclosures: **Leadership:** NEXT Oncology Virginia; **Honoraria:** AbbVie, Amgen, Astellas Pharma, Bayer, Bristol Myers Squibb, CytomX Therapeutics, Janssen Oncology, Merck, Novartis, Prelude Therapeutics, Takeda; **Consulting or Advisory Roles:** Amgen, ArriVent BioPharma, AstraZeneca/MedImmune, Black Diamond Therapeutics, Blueprint Medicines, Bristol Myers Squibb, Crispr Therapeutics, Daiichi Sankyo/AstraZeneca, Gritstone Bio, Gritstone Oncology, GSK, Incyte, Janssen Research and Development, Jazz Pharmaceuticals, Lilly, Merck, Mersana, Mirati Therapeutics, Novartis, Regeneron, Revolution Medicines, Sanofi, Synthekine, Takeda; **Research Funding:** AbbVie, ADC Therapeutics, Alkermes, Amgen, ArriVent Biopharma, Astellas Pharma, Astex Pharmaceuticals, AstraZeneca, Black Diamond Therapeutics, Blueprint Medicines, BluPrint Oncology, Boehringer Ingelheim, Bristol Myers Squibb, CytomX Therapeutics, Daiichi Sankyo, Gritstone, Ignyta, Incyte, Janssen Oncology, LAM Therapeutics, Lilly, Loxo, MacroGenics, Medikine, MedImmune, Mersana, Nalo Therapeutics, Novartis, Plexxikon, Prelude Therapeutics, Regeneron, Revolution Medicines, Roche, Rubius, Scorpion Therapeutics, Synthekine, Takeda. **Acknowledgments:** We thank all the participating patients, their families and site research staff. **Contact Email:** ALX2004trial@alxoncology.com **Presented at:** AACR-NCI-EORTC 2025, Abstract #LB-A004. **Funding:** Study sponsored by ALX Oncology.