A Phase 2 Study of Evorpacept (ALX148) in Combination with Pembrolizumab and Chemotherapy in Patients with Advanced Head and Neck Squamous Cell Carcinoma (HNSCC); ASPEN-04

Ezra Cohen[1], Jean-Pascal Machiels[2], Sjoukje Oosting[3], Bhumsuk Keam[4], Beatriz Cirauqui[5], Annette M Lim[6], Keun-Wook Lee[7], Haiying Liu[8], Tim Welliver[8], Christine Ju[8], Feng Jin[8], Alison Forgie[8], Jaume Pons[8], Sophia S Randolph[8], Athanasios C Tsiatis[8], Kevin Harrington[9]

[1]University of California San Diego, San Diego, United States; [2]Cliniques Universitaires Saint-Luc, Brussels, Belgium; [3]University of Groningen, University Medical Center Groningen, Netherlands; [4]Seoul National University Hospital, Seoul, Korea; [5]Institut Catala d'Oncologia Badalona, Barcelona, Spain; [6]Peter MacCallum Cancer Centre, Melbourne, Australia; [7]Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, Korea; [8]ALX Oncology Inc., South San Francisco, United States; [9]The Royal Marsden Hospital / The Institute of Cancer Research, London, United Kingdom

Background

- Anticancer immunity relies on the release of tumor antigens and subsequent activation of both the innate and adaptive immune systems. Myeloid checkpoint inhibitors have been shown to help potentiate innate immune cell activity including antigen presentation after neoantigen release induced by chemotherapy.
- CD47, a marker of self, interacts with SIRPα on myeloid immune cells and can be upregulated by cancer cells to evade immune responses.
- Evorpacept is a high affinity CD47-blocking fusion protein with an inactive Fc region (**Figure 1**) designed to enhance standard anticancer therapeutics and not exacerbate their toxicities.
- The combination of evorpacept + pembrolizumab + platinum/5FU has been evaluated in the Phase 1 ASPEN-01 study in previously untreated, PD-L1-unselected recurrent/metastatic (R/M) HNSCC patients[1], demonstrating initial clinical activity and tolerability.
- The **ASPEN-04** study will assess the efficacy and safety of evorpacept in combination with pembrolizumab and chemotherapy in previously untreated patients with PD-L1-unselected R/M HNSCC.

Figure 1. Evorpacept Selectively and Potently Binds CD47 to Block Interaction with SIRPα

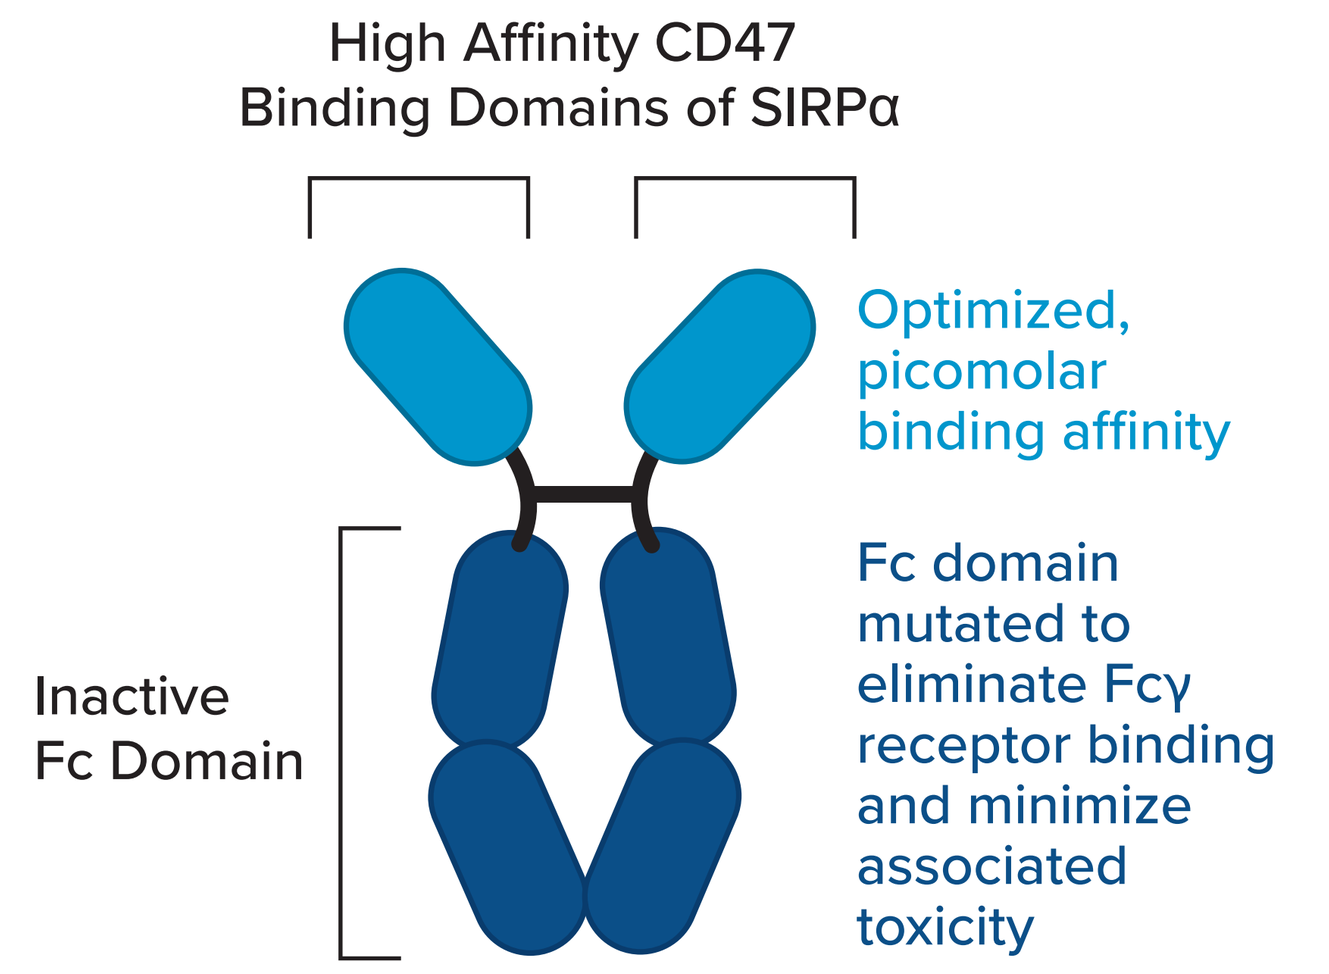
High Affinity CD47
Binding Domains of SIRPα
Optimized, picomolar binding affinity
Inactive Fc Domain
Fc domain mutated to eliminate Fcγ receptor binding and minimize associated toxicity

- Fc domain enables antibody-like PK.
- Molecular weight half the size of typical antibody.

Evorpacept, Pembrolizumab, and Chemotherapy Have Complementary Mechanisms of Action

- Pembrolizumab, a T cell checkpoint inhibitor, represents a standard treatment option for patients with previously untreated recurrent/metastatic (R/M) HNSCC, both as a monotherapy and in combination with platinum + 5FU. Through increased activation of the immune system, the combination of evorpacept + pembrolizumab + platinum/5FU might have greater anti-tumor activity in R/M HNSCC than standard therapeutic approaches (**Figure 2**)[2].
- This combination approach could be especially beneficial to R/M HNSCC patients with low PD-L1 expression, where anti-PD-(L)1 therapy does not provide benefit over cetuximab + platinum/5FU.

Figure 2. Evorpacept, Pembrolizumab, and Chemotherapy Combine to Activate Both the Innate and Adaptive Immune Responses

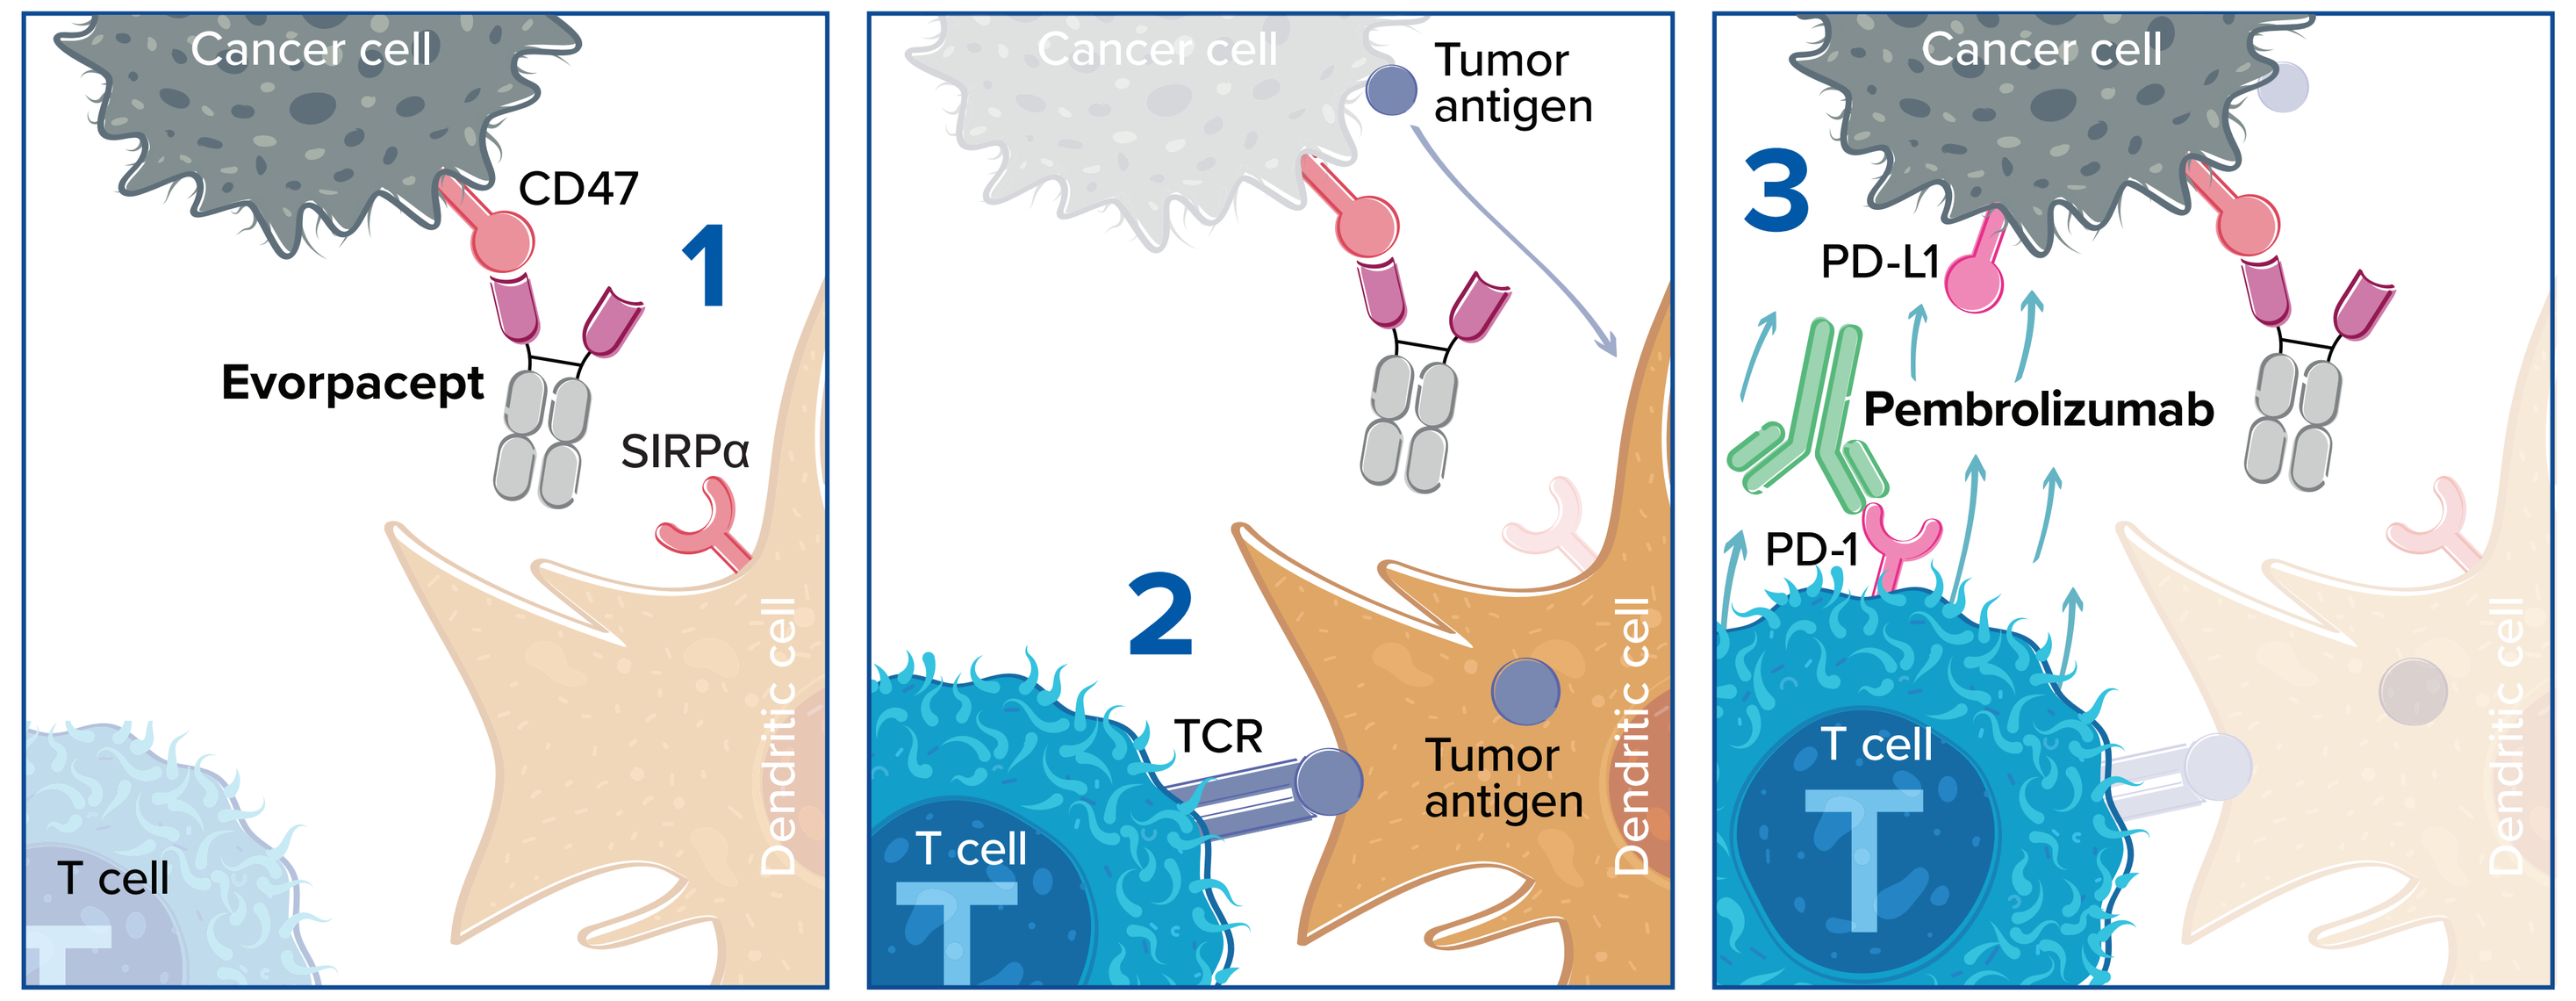
Cancer cell
CD47
1
Evorpacept
SIRPα
T cell
Dendritic cell
Cancer cell
Tumor antigen
2
TCR
T cell
Tumor antigen
Dendritic cell
Cancer cell
3
PD-L1
Pembrolizumab
PD-1
T cell
Dendritic cell

1 **Evorpacept binds to CD47 expressed by cancer cells to block the CD47-SIRPα myeloid checkpoint leading to activation of dendritic cells and enhanced phagocytosis**

2 **Activated dendritic cells cross-present tumor antigens (including tumor antigens released in response to treatment with chemotherapy) to cytotoxic T cells**

3 **Pembrolizumab binds to PD-1, relieving PD-1/PD-L1-mediated inhibition of activated T cells leading to enhanced T cell cytotoxic activity against the cancer cell**

Acknowledgments

We would like to thank all the participating patients and their families as well as site research staff.
This trial is being conducted in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.
Contact email: info@alxoncology.com
Trial Registration: ClinicalTrials.gov identifier, NCT04675333
Presented at the 2022 Society for Immunotherapy of Cancer (SITC) Annual Meeting; November 8-12, 2022. Abstract #676.

Study Methods

- **ASPEN-04** (**Figure 3**) is an ongoing, non-comparative, open-label, randomized Phase 2 global study of evorpacept + pembrolizumab + chemotherapy (5FU + either carboplatin or cisplatin) or pembrolizumab + chemotherapy in patients with PD-L1-unselected metastatic or unresectable recurrent HNSCC who have not yet been treated for their advanced disease.
- **ASPEN-04** will randomize approximately 162 patients to receive evorpacept + pembrolizumab + chemotherapy or pembrolizumab + chemotherapy using a 2:1 allocation.
- Geography, CPS, HPV (p16) status, tobacco habits and ECOG will be used as minimization factors across both arms.
- Patients in the evorpacept treatment arm will receive evorpacept 45 mg/kg IV Q3W.
- All patients will receive pembrolizumab 200 mg IV Q3W (for a maximum of 35 cycles), 5FU (1000 mg/m^2/day continuous infusion Days 1, 2, 3, 4 Q3W x 6 cycles) and either carboplatin (AUC 5 mg/ml/min as a 60 min infusion Day 1 Q3W x 6 cycles) or cisplatin (100 mg/m^2 as a 60 min infusion Day 1 Q3W x 6 cycles).

Figure 3. ASPEN-04 Trial Design

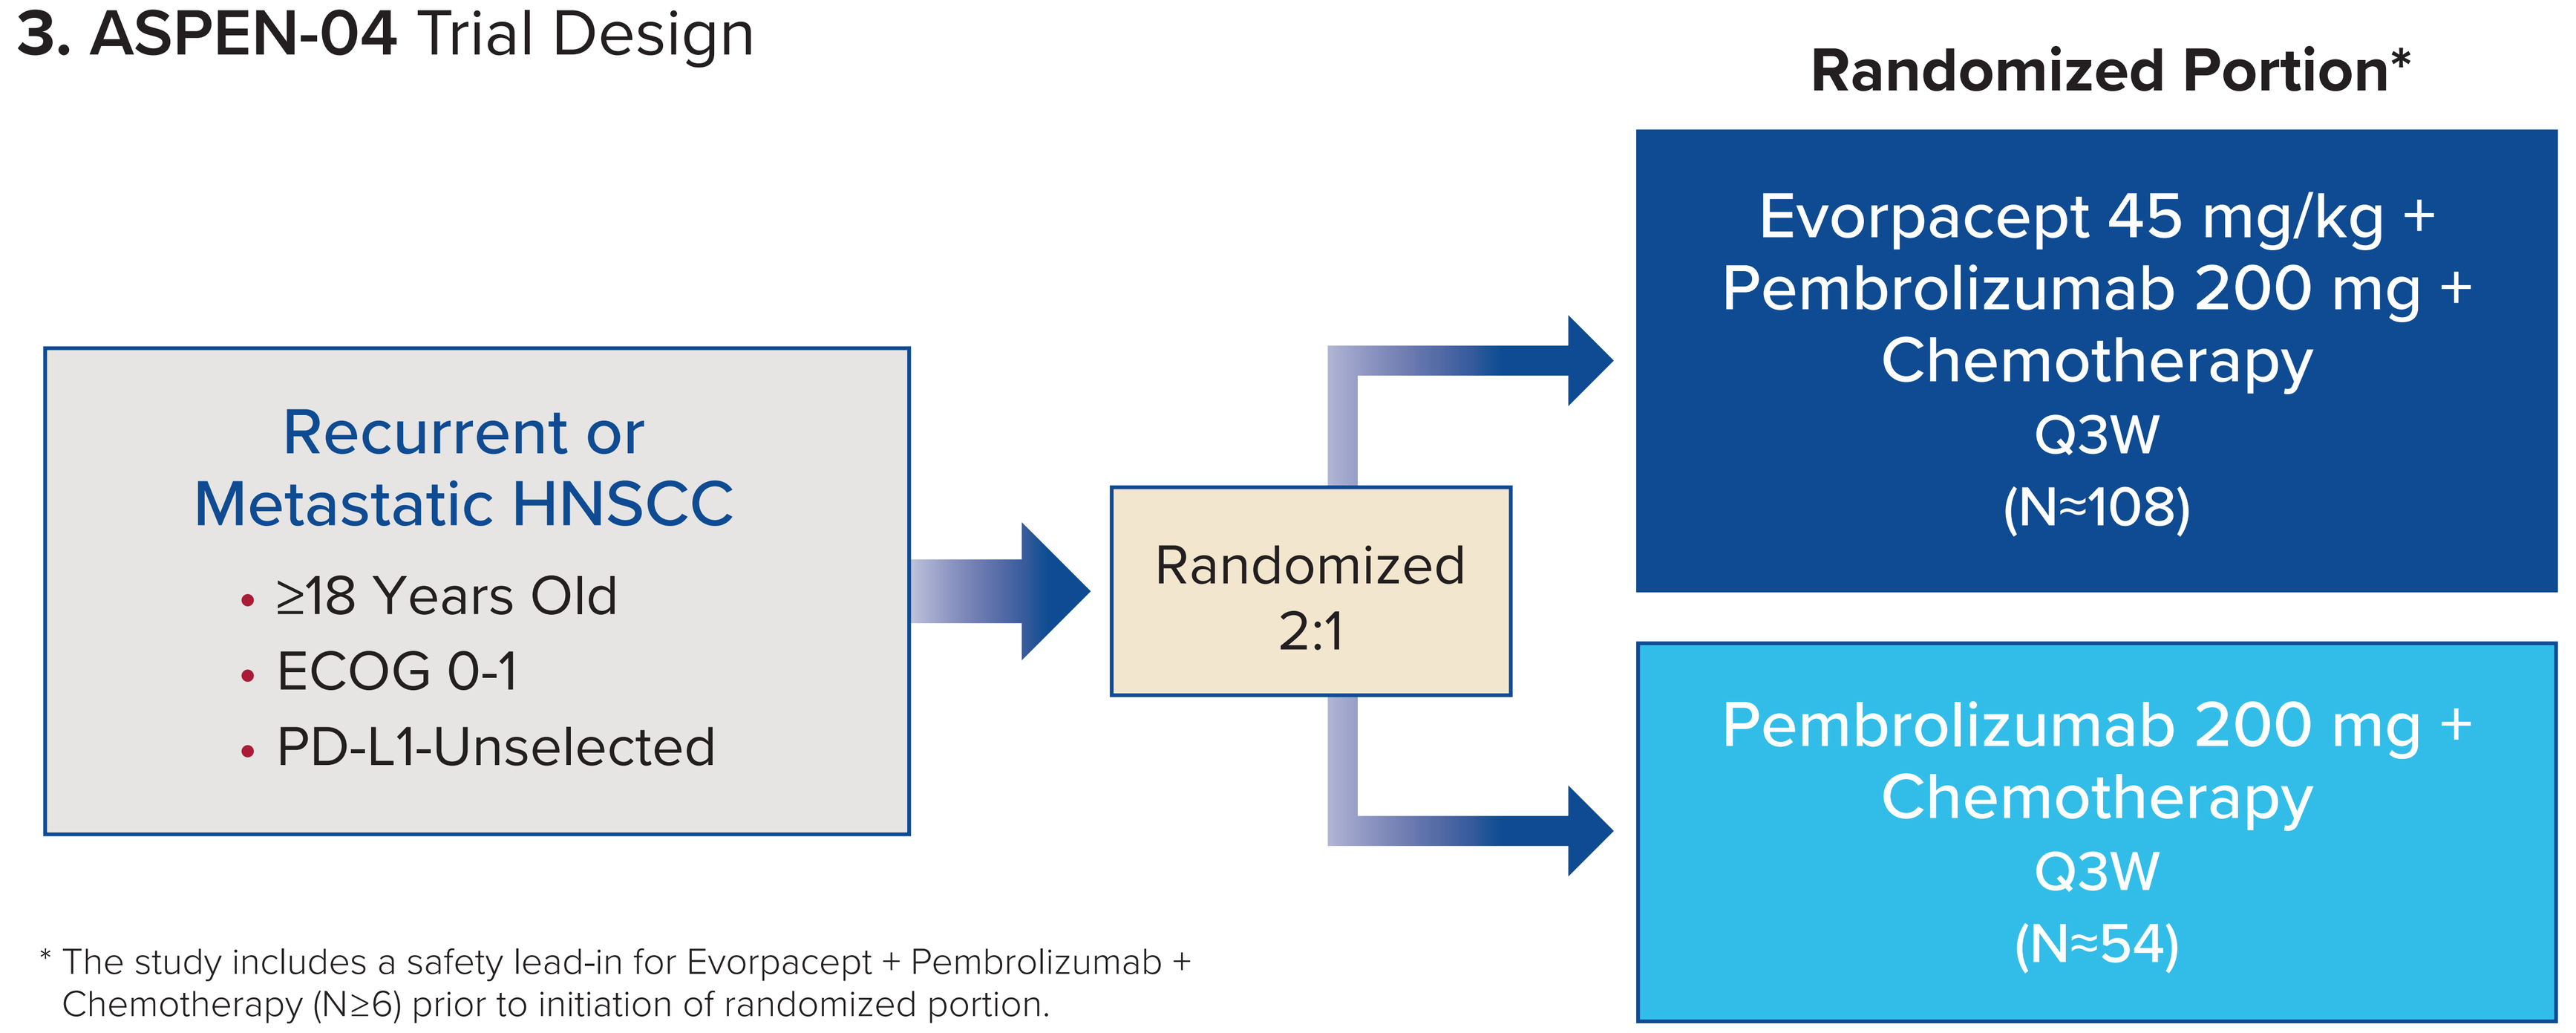
Randomized Portion*
Recurrent or Metastatic HNSCC
≥18 Years Old
ECOG 0-1
PD-L1-Unselected
Randomized 2:1
Evorpacept 45 mg/kg + Pembrolizumab 200 mg + Chemotherapy
Q3W
(N≈108)
Pembrolizumab 200 mg + Chemotherapy
Q3W
(N≈54)

* The study includes a safety lead-in for Evorpacept + Pembrolizumab + Chemotherapy (N≥6) prior to initiation of randomized portion.

Eligibility Criteria

Key Inclusion Criteria

- Patients with metastatic or unresectable, recurrent HNSCC who have not received prior systemic therapy for their advanced disease.
 - Prior systemic therapy for the treatment of locoregionally advanced disease is permitted if it was completed more than 6 months prior to enrollment.
- Measurable disease (RECIST v1.1).
- Adequate liver, renal, and bone marrow function.
- Age ≥18 years.
- ECOG performance status ≤1.

Key Exclusion Criteria

- HNSCC suitable for local therapy.
- Progressive disease within 6 months of completion of systemic therapy for the treatment of locoregionally advanced HNSCC.
- Nasopharyngeal carcinoma (NPC).
- Prior treatment with any anti-CD47 or anti-SIRPα agent.
- Prior treatment with an anti-PD-1 or anti-PD-L1, or anti-PD-L2 agent or with an agent directed to another stimulatory or co-inhibitory T cell receptor (e.g., CTLA-4, CD137, OX40).

Study Endpoints

Co-Primary Endpoints

- 12-month overall survival rate
- Objective response rate

Key Secondary Endpoints

- Duration of response
- Progression-free survival
- Overall survival
- Safety

Exploratory endpoints will characterize pharmacodynamic properties.

Sample Size Determination

- Statistical assumptions for the sample size determination:
 - 1-sided α = 2.5% and 80% power for 12-month OS rate.
 - Rule out a 12-month OS rate of 53%[2], assuming a 12-month OS rate of 66.3% for the Evorpacept + Pembrolizumab + Chemotherapy arm.
- Testing strategy: Fixed sequence procedure will be used for the testing of co-primary endpoints.

References

1. Lee KW, Chung H, Kim TM, et al. 498 Evorpacept (ALX148), a CD47 myeloid checkpoint inhibitor, in patients with head and neck squamous cell carcinoma (HNSCC) and with gastric/gastroesophageal cancer (GC); ASPEN-01. *Journal for ImmunoTherapy of Cancer* 2021;9:doi: 10.1136/jitc-2021-SITC2021.498.
2. Burtness B, Rischin D, Greil R, et al. Pembrolizumab Alone or With Chemotherapy for Recurrent/Metastatic Head and Neck Squamous Cell Carcinoma in KEYNOTE-048: Subgroup Analysis by Programmed Death Ligand-1 Combined Positive Score. *J Clin Oncol*. 2022 Jul 20;40(21):2321-2332.