#780

ALTA-002, a SIRPα-Directed TLR9 Agonist Antibody Conjugate, Activates Myeloid Cells And Promotes Anti-Tumor Immunity

Ons Harrabi[1], Amy Chen[1], Emma Sangalang[1], Danielle Fontaine[1], Min Li [1], Jaume Pons[2], Hong I. Wan[1], Janet Sim[1], Tracy C. Kuo[1]

[1]Tallac Therapeutics, Inc., Burlingame, CA; [2]ALX Oncology, South San Francisco, CA

TALLAC THERAPEUTICS

ALX ONCOLOGY

Introduction

- Novel therapies engaging both innate and adaptive immune responses may engender more robust and durable anti-cancer immunity [1].
- Activation of toll-like receptor 9 (TLR9) by unmethylated CpG oligonucleotides (ODNs) promotes innate inflammatory responses and the induction of adaptive immunity [2]. Several CpG oligodeoxynucleotides have demonstrated clinical response in patients with melanoma by intra-tumoral injection [3].
- We developed a novel Toll-like Receptor Agonist Antibody Conjugate (TRAAC) molecule comprised of a differentiated TLR9 agonist (T-CpG) conjugated to an antibody against SIRPα termed SIRPα TRAAC.
- Signal regulatory protein α (SIRPα) is a myeloid inhibitory receptor that suppresses immune activation following binding of its ligand CD47 [4].
- Blockade of CD47-SIRPα myeloid checkpoint pathway has demonstrated clinical response in patients with solid tumors [5].
- We present preclinical data demonstrating that SIRPα TRAAC delivers T-CpG to myeloid cells via SIRPα and FcγR engagement, triggering TLR9 signaling, cell activation and immune modulation, resulting in robust anti-tumor activity.

Fig 1: SIRPα TRAAC, is a Toll-Like Receptor Agonist Antibody Conjugate, designed for systemic delivery of T-CpG, a potent TLR9 agonist

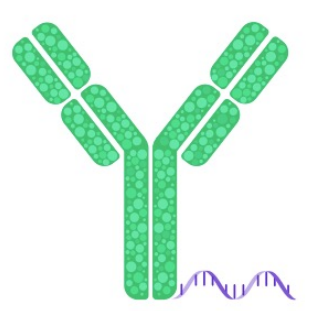

Antibody: anti-SIRPα
- Targets myeloid cells expressing SIRPα
- Localizes to tumor microenvironment of SIRPα⁺ tumors
- Engages Fc gamma receptors
- Blocks CD47-SIRPα myeloid checkpoint pathway

Immune Activator: T-CpG
- Potent TLR9 agonist
- Linear, monomeric, and non-aggregated
- Sequence optimized for potency and stability
- Innovative design enables efficient site-specific conjugation

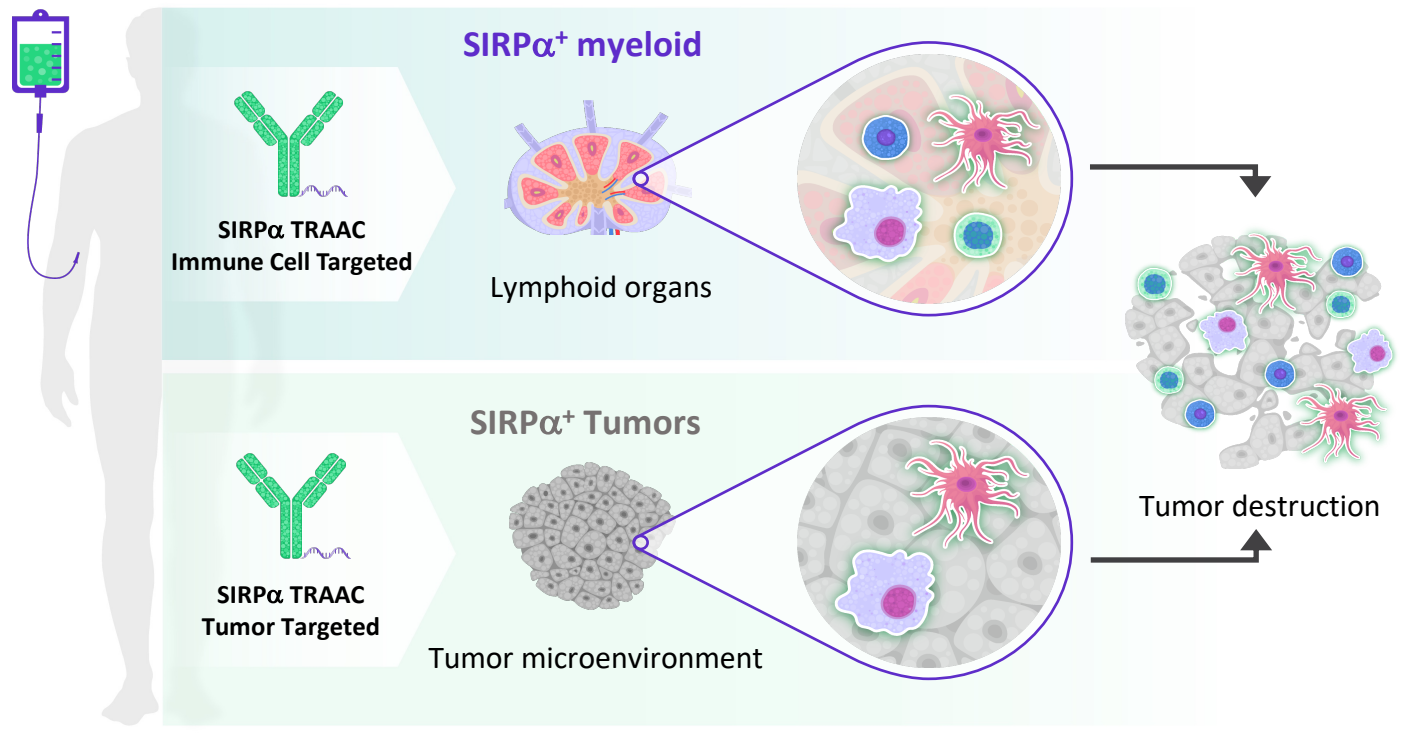
SIRPα⁺ myeloid
SIRPα TRAAC Immune Cell Targeted
Lymphoid organs
SIRPα⁺ Tumors
SIRPα TRAAC Tumor Targeted
Tumor microenvironment
Tumor destruction

Cell type	SIRPα+	TLR9
p(DC)	✓	✓
Myeloid	✓	✓
B cells	—	✓

Experimental Results

Fig 2: T-CpG is a potent TLR9 agonist with targeted activation across species

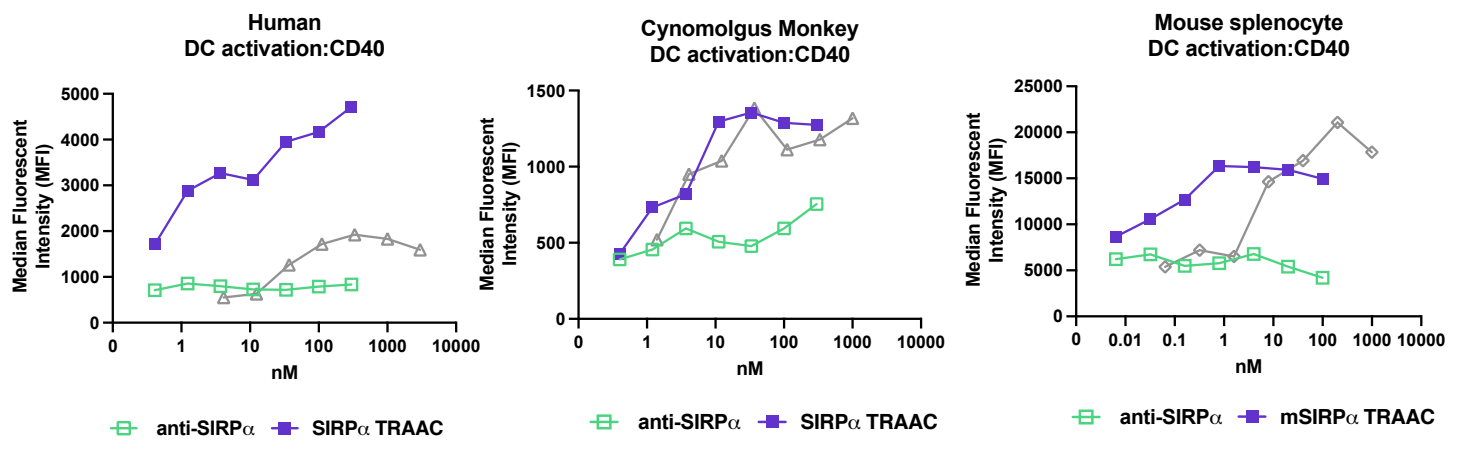
Human DC activation:CD40
Cynomolgus Monkey DC activation:CD40
Mouse splenocyte DC activation:CD40
Median Fluorescent Intensity (MFI)
nM
anti-SIRPα
SIRPα TRAAC
mSIRPα TRAAC

Figure 2: Human, cynomolgus PBMCs, and mouse splenocytes were stimulated with either anti-SIRPα, SIRPα TRAAC, T-CpG, or SIRPα TRAAC conjugated to murine reactive mT-CpG (mSIRPα TRAAC) for 24hr or 48hrs and surface marker expression was assayed by flow cytometry. DC: dendritic cells

Fig 3: SIRPα TRAAC specifically targets and activates SIRPα⁺ immune cells in human PBMC cultures

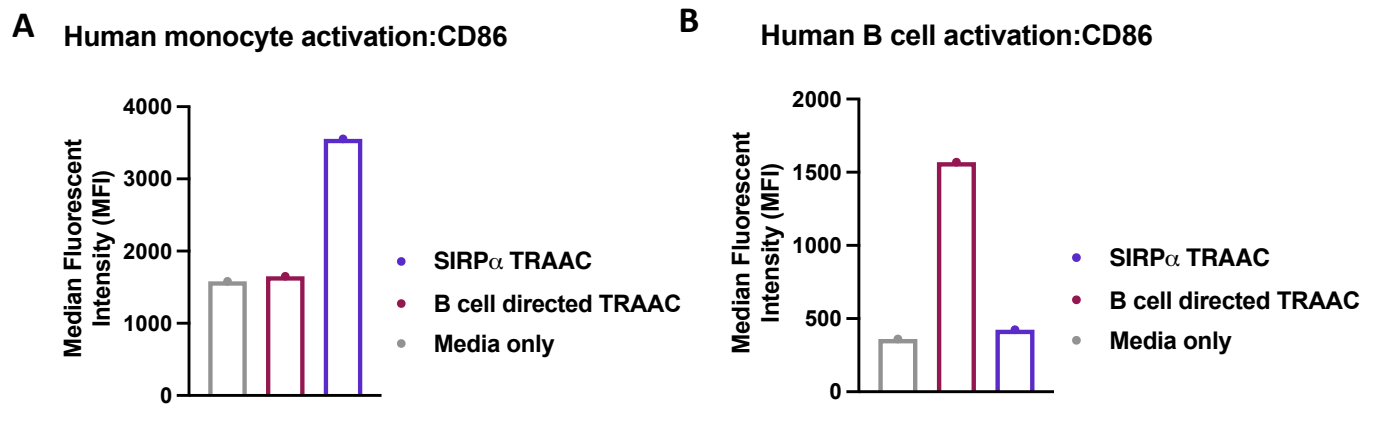
A
Human monocyte activation:CD86
B
Human B cell activation:CD86
Median Fluorescent Intensity (MFI)
SIRPα TRAAC
B cell directed TRAAC
Media only

Figure 3: Human PBMCs were stimulated with SIRPα TRAAC, B cell directed TRAAC or media only for 24hr and surface marker expression on monocytes (A) and B cells (B) was assayed by flow cytometry.

Fig 4: SIRPα TRAAC triggers TLR9 signaling in SIRPα⁺ DC and monocytes, eliciting pro-inflammatory and cytotoxic cytokine production in human PBMC cultures

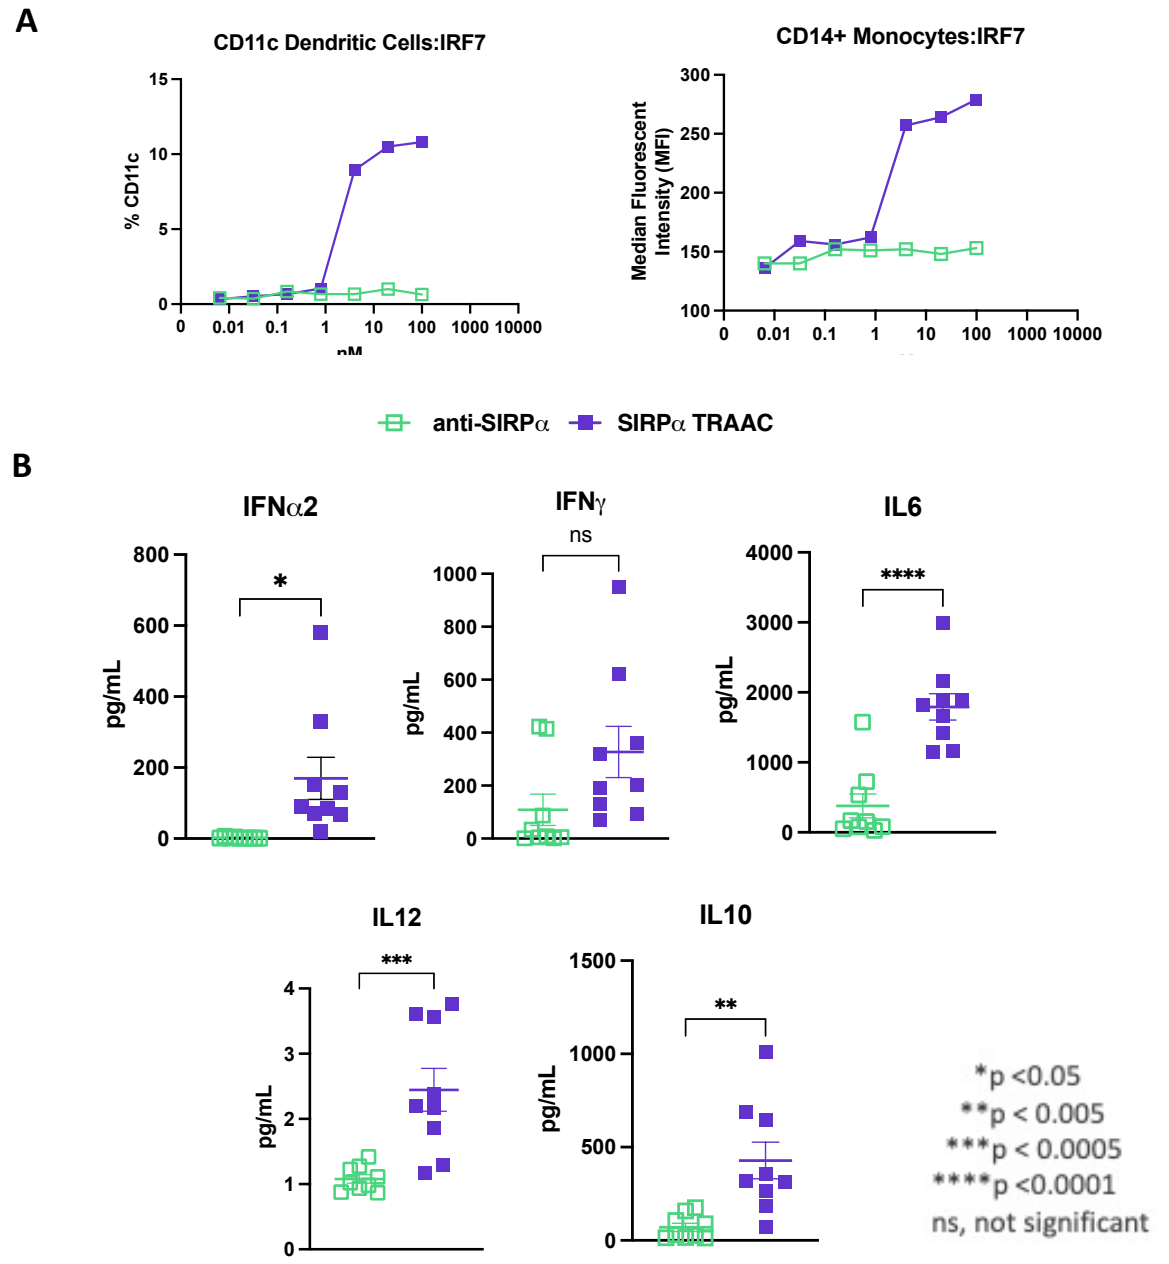
A
CD11c Dendritic Cells:IRF7
CD14+ Monocytes:IRF7
% CD11c
Median Fluorescent Intensity (MFI)
anti-SIRPα
SIRPα TRAAC
B
IFNα2
IFNγ
IL6
IL12
IL10
pg/mL
*
ns

**
*p <0.05
**p < 0.005
***p < 0.0005
****p <0.0001
ns, not significant

Figure 4. Human PBMCs were stimulated with anti-SIRPα or SIRPα TRAAC for 24hr and assayed for IRF7 induction (A) and cytokine production (B) from 9 healthy donors following 72hr stimulation by flow cytometry. P-values were calculated using unpaired t-test.

Fig 5: SIRPα expression on tumors potentiates myeloid activation by SIRPα TRAAC

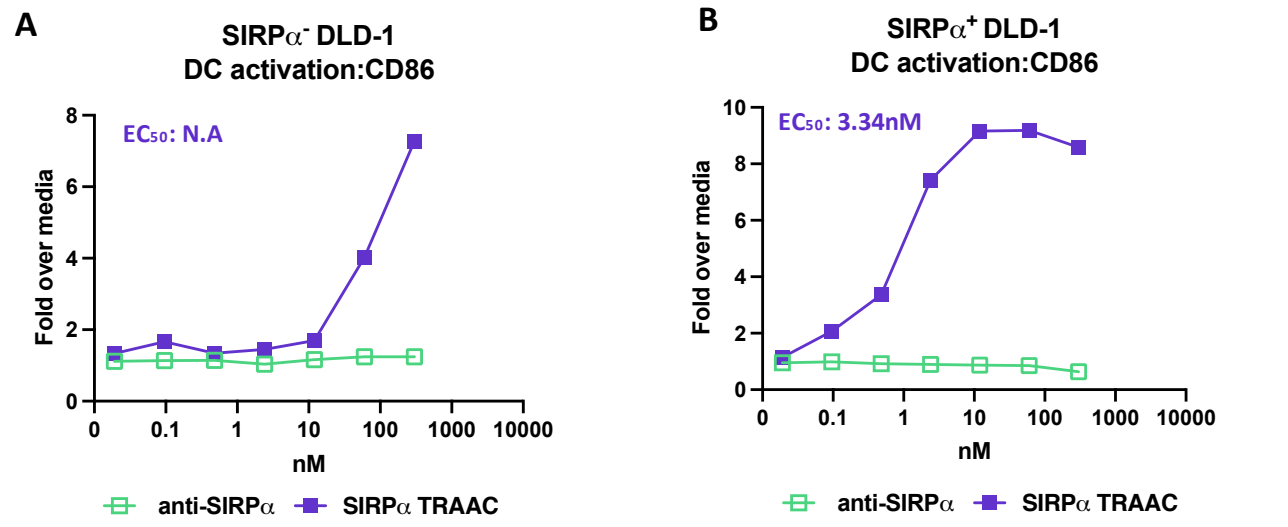
A
SIRPα⁻ DLD-1 DC activation:CD86
EC₅₀: N.A
B
SIRPα⁺ DLD-1 DC activation:CD86
EC₅₀: 3.34nM
Fold over media
nM
anti-SIRPα
SIRPα TRAAC

Figure 5. Human PBMCs were co-cultured either in presence of parental DLD-1 (A) or DLD-1 expressing SIRPα (B) and stimulated with anti-SIRPα, SIRPα TRAAC, or media only for 48hr. Surface marker expression was assayed by flow cytometry. Data is presented as fold over media only control.

Fig 6: SIRPα TRAAC promotes phagocytosis of SIRPα⁻ and SIRPα⁺ tumors by macrophages

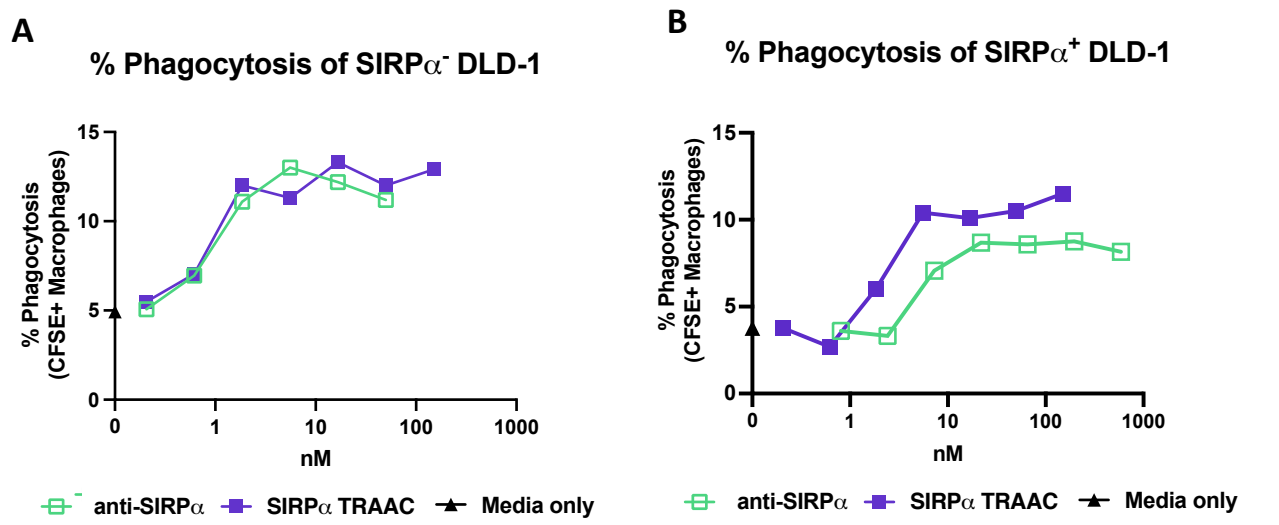
A
% Phagocytosis of SIRPα⁻ DLD-1
B
% Phagocytosis of SIRPα⁺ DLD-1
% Phagocytosis (CFSE+ Macrophages)
nM
anti-SIRPα
SIRPα TRAAC
Media only

Figure 6. Human monocyte derived macrophages were co-incubated for 2hrs with either parental DLD-1 (A) or DLD-1 expressing SIRPα (B) in presence of anti-SIRPα, SIRPα TRAAC or media only. % Phagocytosis was determined by flow cytometry.

Fig 7: Localizing mSIRPα TRAAC to SIRPα⁺ tumor demonstrates superior anti-tumor activity in syngeneic model when administrated systemically

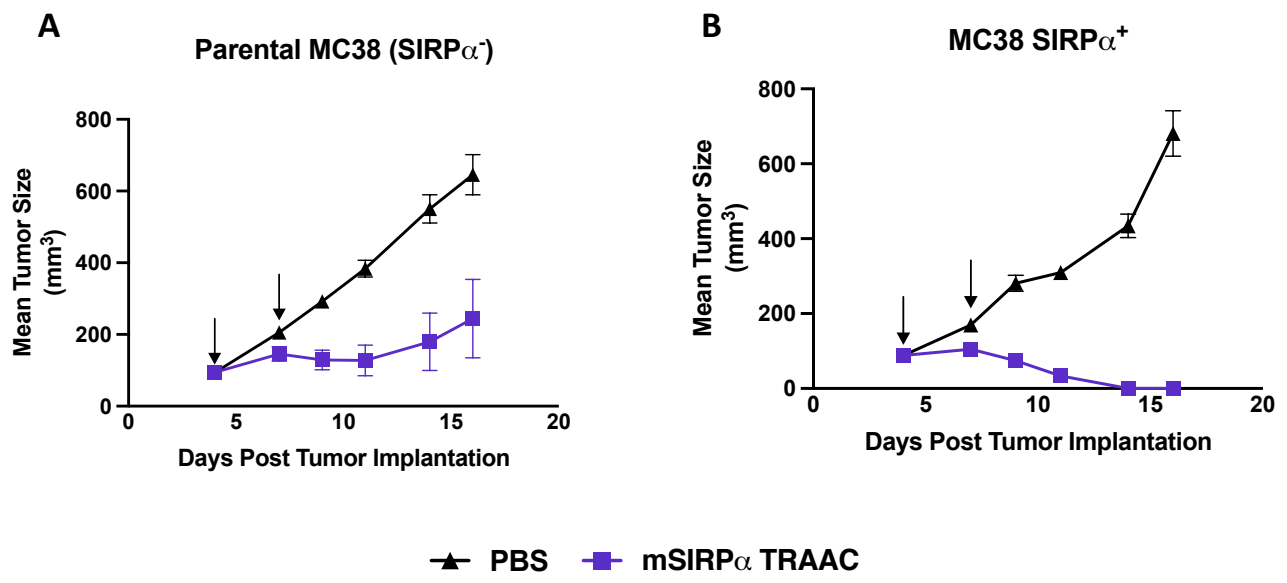
A
Parental MC38 (SIRPα⁻)
B
MC38 SIRPα⁺
Mean Tumor Size (mm³)
Days Post Tumor Implantation
PBS
mSIRPα TRAAC

Figure 7. Mice bearing either parental SIRPα⁻ MC38 tumors (A) or expressing SIRPα (B) were dosed intraperitoneally (i.p.) twice, three days apart with mSIRPα TRAAC conjugated with murine reactive mT-CpG at 1mg/kg or PBS control. Arrows indicate doses administered.

Fig 8: mSIRPα TRAAC elicits potent single agent anti-tumor response in RENCA, a SIRPα⁺ model refractory to anti-PD-1

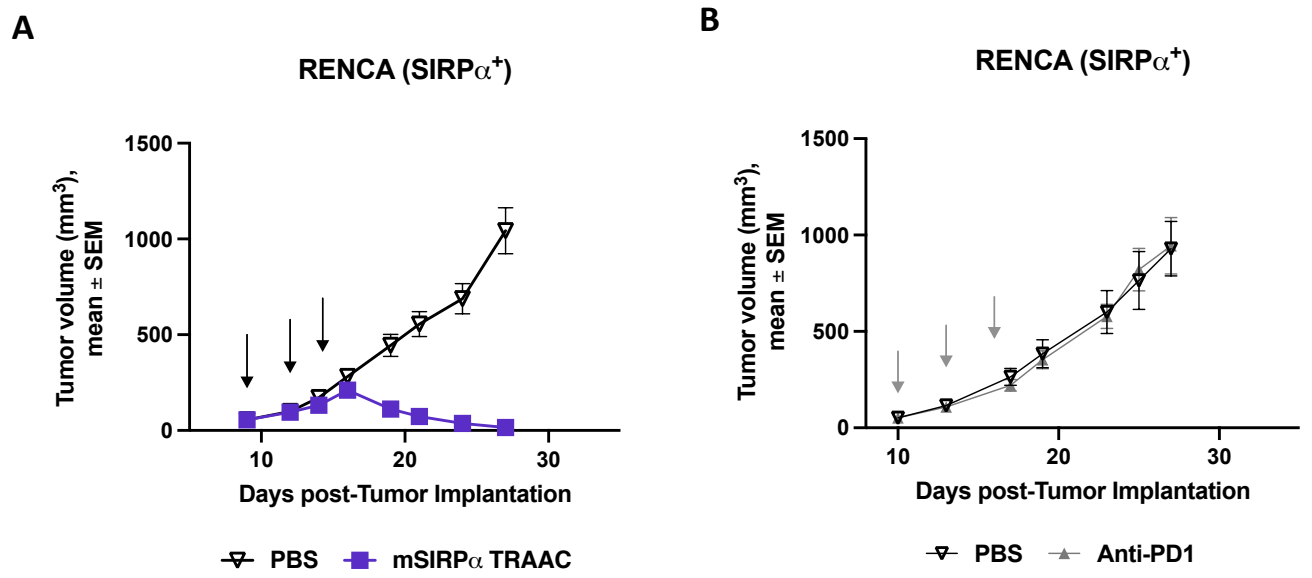
A
RENCA (SIRPα⁺)
B
RENCA (SIRPα⁺)
Tumor volume (mm³), mean ± SEM
Days post-Tumor Implantation
PBS
mSIRPα TRAAC
Anti-PD1

Figure 8. Mice bearing RENCA tumors were dosed intraperitoneally (i.p.) three times, three days apart with mSIRPα TRAAC at 10mg/kg or PBS (A). A separate cohort was dosed with anti-PD1 at 10mg/kg three times, three days apart or PBS (B). Arrows indicate doses administrated.

Fig 9: The combination of suboptimal dose of mSIRPα TRAAC and anti-PD-1 elicits enhanced anti-tumor response in CT26, a SIRPα negative tumor model

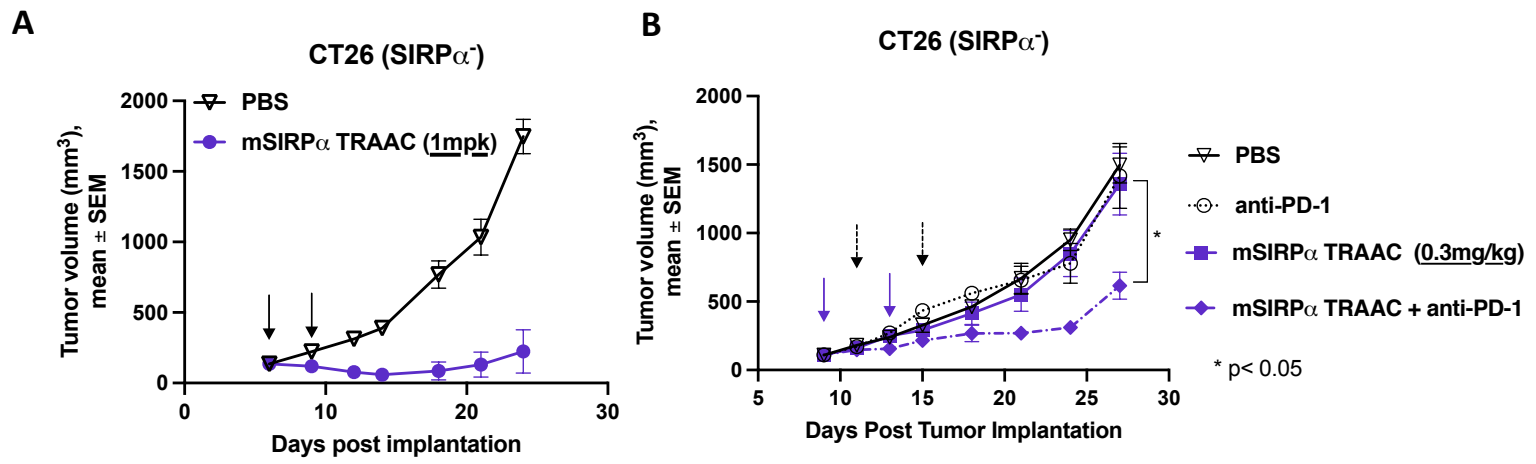
A
CT26 (SIRPα⁻)
PBS
mSIRPα TRAAC (1mpk)
Tumor volume (mm³), mean ± SEM
Days post implantation
B
CT26 (SIRPα⁻)
PBS
anti-PD-1
mSIRPα TRAAC (0.3mg/kg)
mSIRPα TRAAC + anti-PD-1
* p< 0.05
Days Post Tumor Implantation

Figure 9. Mice bearing CT26 tumors were intraperitoneally (i.p.) treated with mSIRPα TRAAC at 1mg/kg or PBS (A) or mSIRPα TRAAC at 0.3mg/kg, anti-PD-1 at 10mg/kg, in combination, or PBS control (B). Arrows indicate doses administrated. P-values comparing combination group vs. anti-PD-1 in were calculated using unpaired t-test.

Conclusions

- SIRPα TRAAC is a SIRPα-directed TLR9 receptor agonist antibody conjugate designed for systemic administration.
- SIRPα TRAAC specifically targets myeloid cells triggering TLR9 signaling via SIRPα and FcγR engagement leading to robust cellular activation and cytokine induction in cultured immune cells.
- SIRPα expression on tumors potentiates the activation of myeloid cells by SIRPα TRAAC.
- SIRPα TRAAC promotes phagocytosis of tumors independent of SIRPα expression.
- Localization of mSIRPα TRAAC to SIRPα+ tumors demonstrates robust and curative single agent activity in multiple models including anti-PD-1 refractory tumors.
- mSIRPα TRAAC enhances tumor regression in combination with anti-PD-1 in syngeneic tumor model.
- **ALTA-002, our SIRPα TRAAC candidate, is in preclinical development for the treatment of patients with various types of malignancies**

References:

1. Kobold et al., Innate and adaptive immunity combined for cancer treatment. *PNAS* 2019, 116 (4) 1087-1088.
2. Dowling et al., Toll-like receptors: the swiss army knife of immunity and vaccine development. *Clin. Transl. Immunology*. 2016 *5*(5), e85–10.
3. Hamid et al., Intratumoral Immunotherapy—Update 2019. *The Oncol*. 2019; 25:343–359.
4. Kuo, T et al., Targeting the myeloid checkpoint receptor SIRPα potentiates innate and adaptive immune responses to promote anti-tumor activity. 2020. J Hematol *Oncol* 13, 160.
5. Lee KW et al., ALX148, a CD47 blocker in combination with standard chemotherapy and antibody regimens in patients with gastric/gastroesophageal junction (GC) cancer and head and neck squamous cell carcinoma (HNSCC); ASPEN-01. SITC 2020.