ALX148, a CD47 Blocker, in Combination with Standard Chemotherapy and Antibody Regimens in Patients with Gastric/Gastroesophageal Junction (GC) Cancer and Head and Neck Squamous Cell Carcinoma (HNSCC); ASPEN-01

Keun-Wook Lee[1], Hyun Cheol Chung[2], Won Seog Kim[3], Laura QM Chow[4*], Nehal Lakhani[5], Wells Messersmith[6], Yung-Jue Bang[7], Patricia LoRusso[8], Philip Fanning[9], Pierre Squifflet[10], Feng Jin[9], Alison Forgie[9], Hong Wan[9], Jaume Pons[9], Sophia Randolph[9], Justin Gainor[11]

[1]Seoul National University Bundang Hospital, Seoul National University College of Medicine, Seongnam, Korea; [2]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Korea; [3]Sungkyunkwan University School of Medicine, Samsung Medical Center, Seoul, Korea; [4]University of Washington, Seattle, WA, USA; [5]START Midwest, Grand Rapids, MI, USA; [6]University of Colorado Cancer Center, Aurora, CO, USA; [7]Seoul National University College of Medicine, Seoul, Korea; [8]Yale Cancer Center, New Haven, CT, USA; [9]ALX Oncology, Burlingame, CA, USA; [10]International Drug Development Institute, Brussels, Belgium; [11]Massachusetts General Hospital Cancer Center, Boston, MA, USA

Background

- CD47, a myeloid checkpoint and marker of self, signals the macrophage to ignore the cell on which CD47 is expressed by binding its receptor, SIRPα.[1] Tumors upregulate CD47 to evade the immune response.
- ALX148 is a high affinity CD47 blocker fusion protein with an inactive human immunoglobulin Fc region (Figure 1) designed to enhance the activity of anti-cancer targeted antibodies and checkpoint inhibitors with minimal hematologic toxicity.[2]
- ASPEN-01 (AT148001), a first-in-human Phase 1 study evaluates ALX148 administered as a single agent (Part 1) and in combination with established anti-cancer antibodies (Part 2).

Figure 1. ALX148 Potently and Selectively Binds CD47 to Block SIRPα Interaction

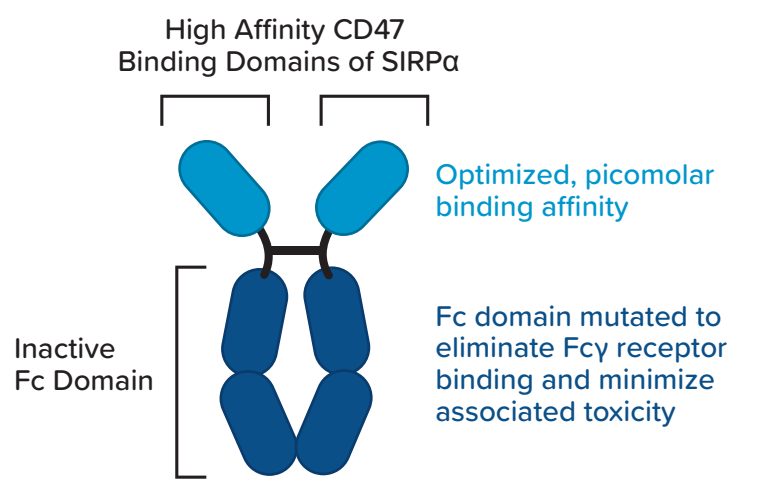
High Affinity CD47
Binding Domains of SIRPα
Optimized, picomolar binding affinity
Inactive Fc Domain
Fc domain mutated to eliminate Fcγ receptor binding and minimize associated toxicity

- Fc domain enables antibody-like PK.
- Molecular weight half the size of typical antibody.

Figure 2. ALX148 Bridges Innate and Adaptive Immunity[2]

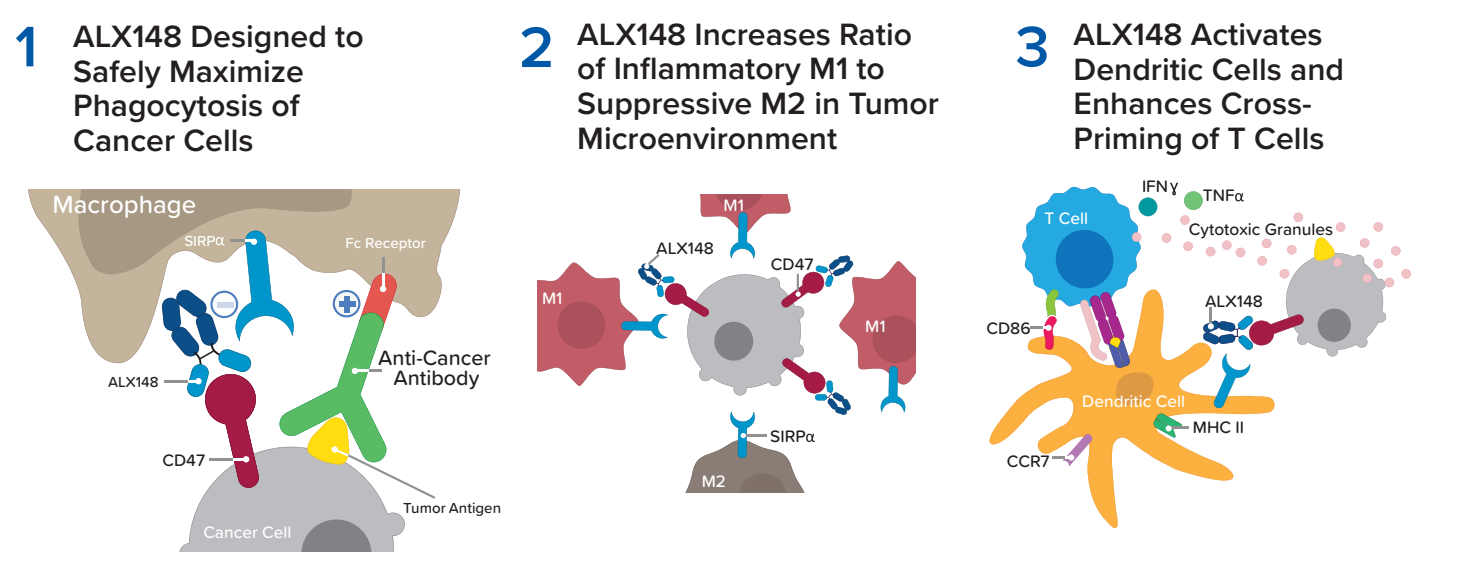
1 ALX148 Designed to Safely Maximize Phagocytosis of Cancer Cells
2 ALX148 Increases Ratio of Inflammatory M1 to Suppressive M2 in Tumor Microenvironment
3 ALX148 Activates Dendritic Cells and Enhances Cross-Priming of T Cells
Macrophage
SIRPα
Fc Receptor
Anti-Cancer Antibody
ALX148
CD47
Tumor Antigen
Cancer Cell
M1
M2
T Cell
IFNγ
TNFα
Cytotoxic Granules
CD86
Dendritic Cell
MHC II
CCR7

Methods

Study Design

- **Part 1 (single agent):** Patients were administered escalating doses of intravenous ALX148 (0.3 to 10 mg/kg QW; or 30 mg/kg QOW).[3]
- **Part 2 (combination):** Patients were administered ALX148 10 or 15 mg/kg QW in combination with pembrolizumab (200 mg IV Q3W), trastuzumab (8 mg/kg IV ➔ 6 mg/kg Q3W), ramucirumab (8 mg/kg Days 1, 15 Q4W), paclitaxel (80 mg/m^2 Days 1, 8, 15 Q4W), cisplatin (100 mg/m^2 Q3W x 6), carboplatin (AUC 5 mg/ml/min Day 1 Q3W x 6), and 5FU: 1,000 mg/m^2/day Days 1, 2, 3, 4 Q3W x 6)
 - Adequate organ function and hemoglobin ≥9 g/dL.
 - No prior treatment with an anti-CD47 or anti-SIRPα agent.
- CD47 target occupancy (TO) in peripheral blood T lymphocytes and erythrocytes was measured by flow cytometry.
- HER2, PD-L1, CD8, CD68 and CD163 expression on formalin fixed, paraffin embedded tumor tissue was measured by immunohistochemistry (IHC) assays. HER2 levels were determined by HercepTest™. PD-L1 (Clone 22C3) Combined Positive Score (CPS) was determined by pathologist scoring.

Table 1. ALX148 Combination Gastric/Gastroesophageal (GC) and HNSCC Tumor Cohorts

Combination Dose Expansion
≥2L HER2-Positive GC (N=14) ALX148 + Trastuzumab + Ramucirumab + Paclitaxel *Progressed on prior trastuzumab, fluoropyrimidine, or platinum*
≥2L HER2-Positive GC (N=20) ALX148 + Trastuzumab *Progressed on prior fluoropyrimidine (progression on trastuzumab and platinum allowed)*
1L HNSCC (N=5) ALX148 + Pembrolizumab + 5FU + Platinum *No prior treatment for advanced disease*
≥2L HNSCC (N=20) ALX148 + Pembrolizumab *Progressed on prior platinum*

- **Primary Study Objective:** Characterize ALX148 safety profile as a single agent (Part 1) and in combination with established anti-cancer antibodies with or without standard chemotherapy (Part2).
- Here we report preliminary data from the GC and HNSCC patient cohorts receiving ALX148 plus chemotherapy combinations, and updated data from the GC and HNSCC patient cohorts receiving ALX148 plus pembrolizumab and ALX148 plus trastuzumab, as of October 01, 2020.

Results

Patient Baseline Characteristics

- 59 solid tumor patients have been enrolled into Part 2 GC and HNSCC combination cohorts (Table 2).

Table 2. Baseline Characteristics

	ALX148 + Trastuzumab + Ram/Pac ≥2L GC (N=14)	ALX148 + Trastuzumab ≥2L GC (N=20)	ALX148 + Pembrolizumab + 5FU/Platinum 1L HNSCC (N=5)	ALX148 + Pembrolizumab ≥2L HNSCC (N=20)
Median Age				
Years (range)	63 (36-83)	58 (45-79)	61 (45-63)	62.5 (35-81)
Sex, n				
M	10	15	4	15
F	4	5	1	5
Race, n				
Asian	11	13	4	6
White	3	6	1	12
Other	—	1	—	2
ECOG PS, n				
0	5	7	4	7
1	9	13	1	13
Progressed Upon Prior Anti-HER2 Therapy, n (%)	13 (93)	19 (95)	N/A	N/A
Progressed Upon ≥2 Prior anti-HER2 Therapies n (%)	1 (7.1)	9 (45)	N/A	N/A
Progressed Upon Prior CPI Therapy, n (%)	1 (7.1)	9 (45)	0 (0)	10 (50)
Visceral Distant Metastasis, n (%)	13 (93)	17 (85)	1 (20)	12 (60)

N/A – not applicable; **CPI** – checkpoint inhibitor; **Ram/Pac** – ramucirumab + paclitaxel

Safety

- ALX148 in combination with trastuzumab + ramucirumab + paclitaxel (ram/pac) (N=14), and pembrolizumab + 5FU + platinum (N=5) was well tolerated, with most treatment related adverse events (TRAE) reported of low grade and frequency. Safety of ALX148 in combination with pembrolizumab and trastuzumab has been described elsewhere.[4]
- There were no dose limiting toxicities reported in patients treated with ALX148 + trastuzumab + ram/pac or ALX148 + pembrolizumab + 5FU + platinum.
- All patients in chemo-containing cohorts experienced at least 1 adverse event. Eight (57%) patients administered ALX148 + trastuzumab + ram/pac and no (0%) patient administered ALX148 + pembrolizumab + 5FU + platinum experienced a TRAE.
- The most common TRAEs of ALX148 in combination with trastuzumab + ram/pac (N=14) were low grade diarrhea, rash, urticaria (each 21%; Table 3). TRAEs ≥Grade 3 severity were of low frequency (Table 4).
- There were no treatment related SAEs reported in patients treated with ALX148 trastuzumab + ram/pac or ALX148 + pembrolizumab + 5FU + platinum.

Treatment Related Adverse Events

Table 3. ALX148 + Trastuzumab + Ram/Pac (N=14)

Adverse Event	Total N (%)
Diarrhea	3 (21)
RASH	3 (21)
Urticaria	3 (21)
Fatigue	2 (14)
Pruritus	2 (14)
Lymphocyte Count Decreased	1 (7)
Abdominal Pain	1 (7)

RASH – rash and dermatitis

No TRAEs were reported in patients receiving:

- ALX148 (10 and 15 mg/kg QW) + pembrolizumab + 5FU + platinum: N=5.

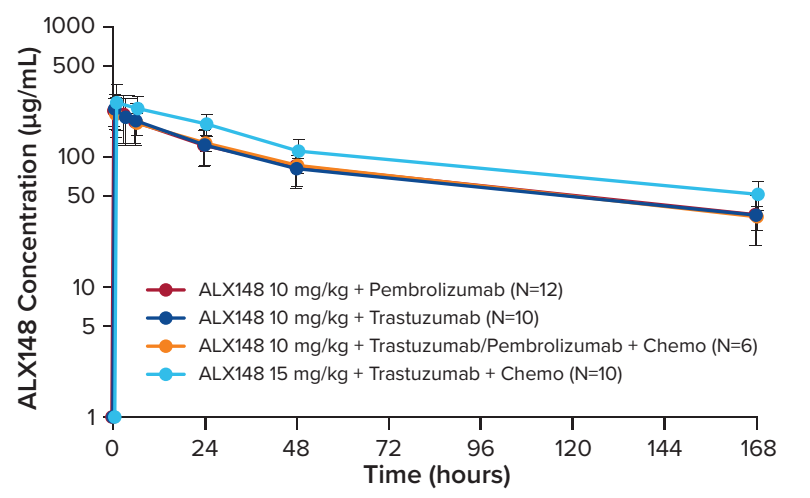
ALX148 Concentration (µg/mL)
1000
500
100
50
10
5
1
0
24
48
72
96
120
144
168
Time (hours)
ALX148 10 mg/kg + Pembrolizumab (N=12)
ALX148 10 mg/kg + Trastuzumab (N=10)
ALX148 10 mg/kg + Trastuzumab/Pembrolizumab + Chemo (N=6)
ALX148 15 mg/kg + Trastuzumab + Chemo (N=10)

Figure 3. ALX148 Concentration-Time Profiles by Dose Level and Combination Partner

Table 4. ≥Grade 3 Adverse Events

Adverse Event	ALX148 + Trastuzumab + Ramucirumab + Paclitaxel (N=14) All Causality n(%)		Related n (%)		ALX148 + Pembrolizumab + 5FU + Platinum (N=5) All Causality n(%)		Related n (%)	
Grade	3	4	3	4	3	4	3	4
Neutrophil Count Decreased	5 (36)	1 (7)	—	—	1 (20)	—	—	—
Hypertension	5 (36)	—	—	—	—	—	—	—
Anemia	1 (7)	—	—	—	1 (20)	—	—	—
Hypophosphatemia	1 (7)	—	—	—	—	—	—	—
Lymphocyte Count Decreased	1 (7)	—	1 (7)	—	—	—	—	—
Platelet Count Decreased	1 (7)	—	—	—	—	—	—	—
Urinary Tract Infection	1 (7)	—	—	—	—	—	—	—
Cardiac Tamponade	—	—	—	—	—	1 (20)*	—	—
Dysphagia	—	—	—	—	1 (20)	—	—	—
Pericarditis Constrictive	—	—	—	—	1 (20)*	—	—	—
Supraventricular Tachycardia	—	—	—	—	1 (20)*	—	—	—

*Events occurred in a single patient with malignant pericardial effusion.

Response

Table 5. Clinical Activity of ALX148 Chemotherapy Combinations in Response Evaluable Patients with ≥2L HER2 Positive GC Cancer and 1L HNSCC

Population ALX148 Dose	N	ORR (95% CI)	Median Follow-Up* (95% CI)
≥2L Gastric ALX148 + Trastuzumab + Ramucirumab + Paclitaxel	14	64.3% (38.8; 83.7)	5.3 mo (2.8; 6.7)
ALX148 (15 mg/kg)	11	63.6% (35.4; 84.8)	4.2 mo (2.4; 6.2)
ALX148 (10 mg/kg)	3	66.7% (20.8; 93.9)	8.9 mo (5.1; 9.6)
1L HNSCC ALX148 + Pembrolizumab + 5FU + Platinum	4	75% (30.0; 95.0)	5.0 mo (1.3; 8.8)
ALX148 (15 mg/kg)	1	100% (20.5; 100)	1.6 mo (1.3; 1.9)
ALX148 (10 mg/kg)	3	66.7% (20.8; 93.9)	5.3 mo (5.0; 8.8)

*Intent to treat population: HNSCC (10 + 15 mg/kg) N=5; HNSCC (15 mg/kg) N=2.

ALX148 Combination Expansion Cohorts – Clinical Activity in Response Evaluable Patients*

- **HER2 Positive GC Expansion**
 - **ALX148 (15 mg/kg QW) + trastuzumab + chemo, ≥2L GC:** N=11 [7 PR (5 confirmed), 3 SD, 1 PD]
 - **ALX148 (10 mg/kg QW) + trastuzumab + chemo, ≥2L GC:** N=3 [2 PR confirmed, 1 SD]
 - **ALX148 (10 mg/kg QW) + trastuzumab, ≥2L GC:** N=19 [4 PR (3 confirmed), 5 SD, 10 PD]
- **HNSCC Expansion**
 - **ALX148 (15 mg/kg QW) + pembrolizumab + chemo, CPI naive 1L HNSCC:** N=1 [1 PR unconfirmed]
 - **ALX148 (10 mg/kg QW) + pembrolizumab + chemo, CPI naive 1L HNSCC:** N=3 [1 CR unconfirmed, 1 PR confirmed, 1 PD]
 - **ALX148 (10 mg/kg QW) + pembrolizumab, CPI naive ≥2L HNSCC:** N=10 [4PR (2 confirmed), 2 SD, 4 PD]
 - **ALX148 (10 mg/kg QW) + pembrolizumab, progressed on prior CPI ≥2L HNSCC:** N=10 [3 SD, 7 PD]

* Based upon investigator assessed response using RECIST v1.1. All objective responses are unconfirmed as of the cut off date unless specified.

Table 6. Updated Clinical Activity in Response Evaluable Patients Receiving ALX148 + Trastuzumab and Pembrolizumab in ≥2L HER2 Positive GC and ≥2L HNSCC

Population ALX148 (10 mg/kg QW)	N	ORR (95% CI)	Median DOR (95% CI)	Median PFS (95% CI)	Median OS* (95% CI)	Median Follow-Up* (95% CI)
≥2L Gastric ALX148 + Trastuzumab	19	21.1% (8.5; 43.3)	8.7 mo (5.6; 9.4)	2.2 mo (1.9; 5.5)	8.1 mo (3.4; 12.6)	19.8 mo (11.7; 19.8)
≥2L HNSCC No Prior CPI ALX148 + Pembrolizumab	10	40% (16.8; 68.7)	4.3 mo (1.9; NC)	4.6 mo (0.5; 7.5)	22.1 mo (3.1; NC)	25.1 mo (15.3; 25.6)
≥2L HNSCC Progressed on Prior CPI ALX148 + Pembrolizumab	10	0% (0; 27.8)	N/A	2.0 mo (0.9; 3.6)	7.4 mo (3.1; 15.4)	24.4 mo (1.0; 24.4)

*Intent to treat population: Gastric N=20.

Figure 4. ALX148 + Trastuzumab + Ram/Pac ≥2L HER2 Positive GC Response Evaluable (n=14)

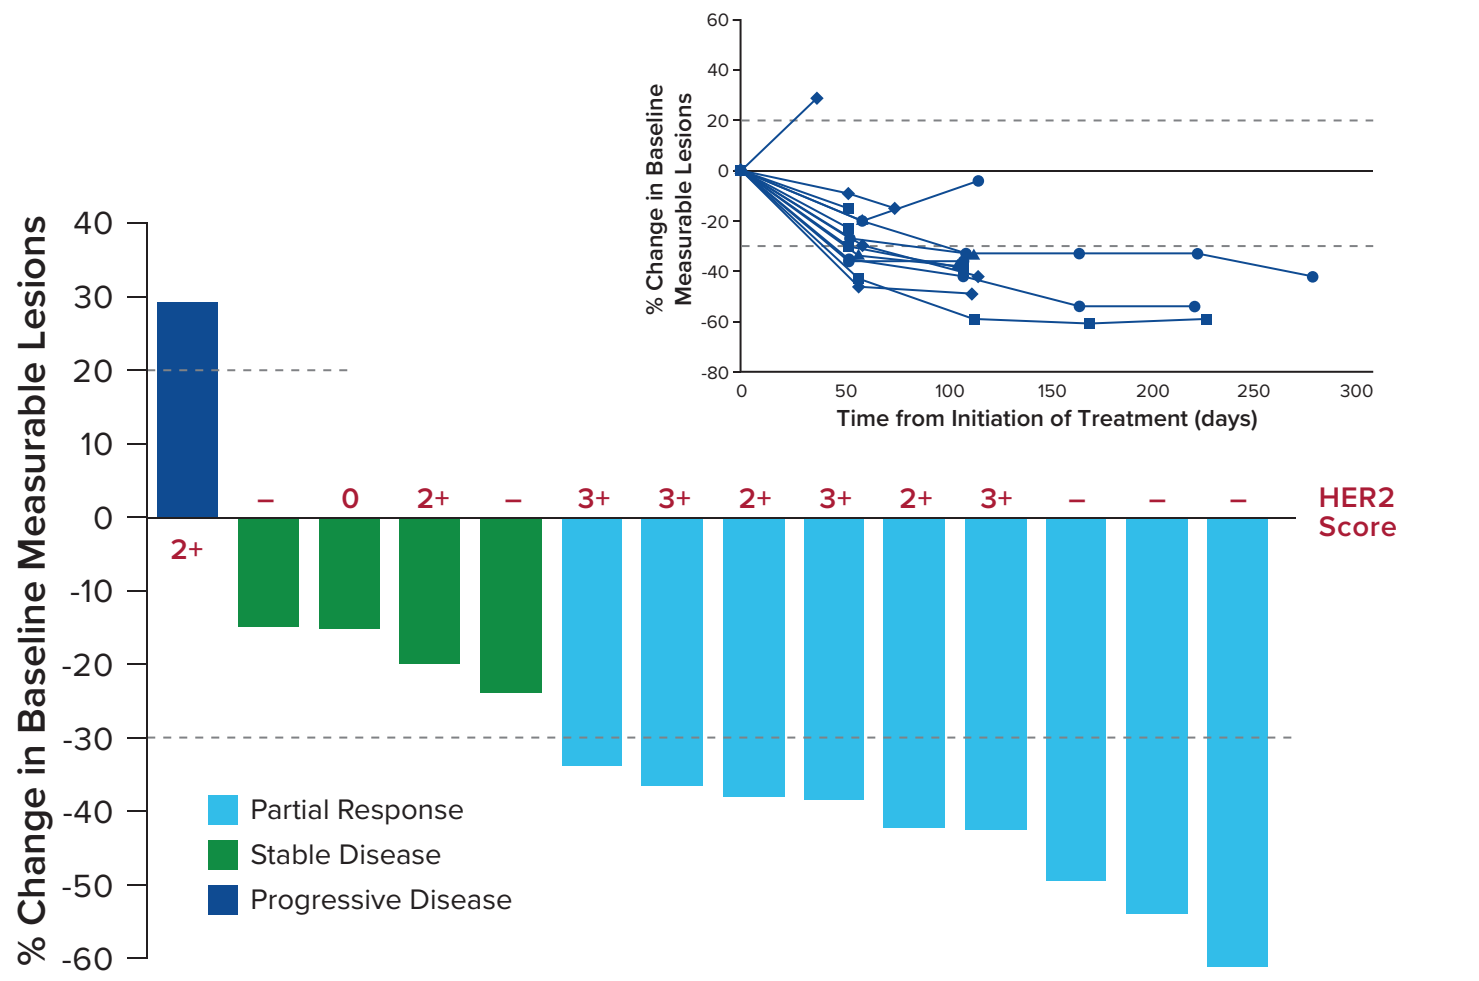
% Change in Baseline Measurable Lesions
2+
–
0
2+
–
3+
3+
2+
3+
2+
3+
–
–
–
HER2 Score
Partial Response
Stable Disease
Progressive Disease
Time from Initiation of Treatment (days)

Figure 5. Best Overall and Duration of Response in Patients While Receiving ALX148 + Trastuzumab + Ramucirumab + Paclitaxel

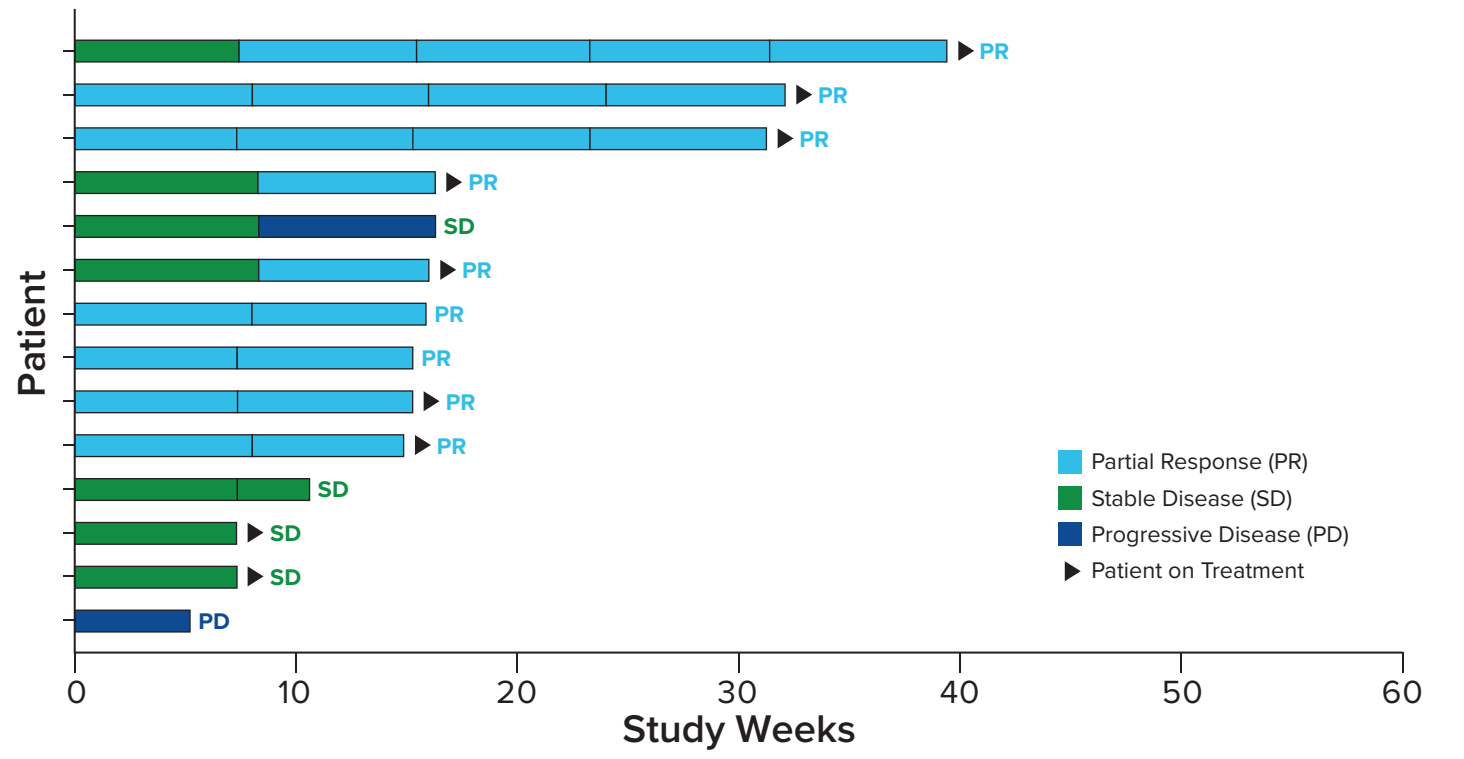
≥2L HER2 positive GC
Patient
Study Weeks
Partial Response (PR)
Stable Disease (SD)
Progressive Disease (PD)
Patient on Treatment

Figure 6. ALX148 + Pembrolizumab + 5FU/Platinum in 1L HNSCC CPI Naïve Patients Response Evaluable (n=4)

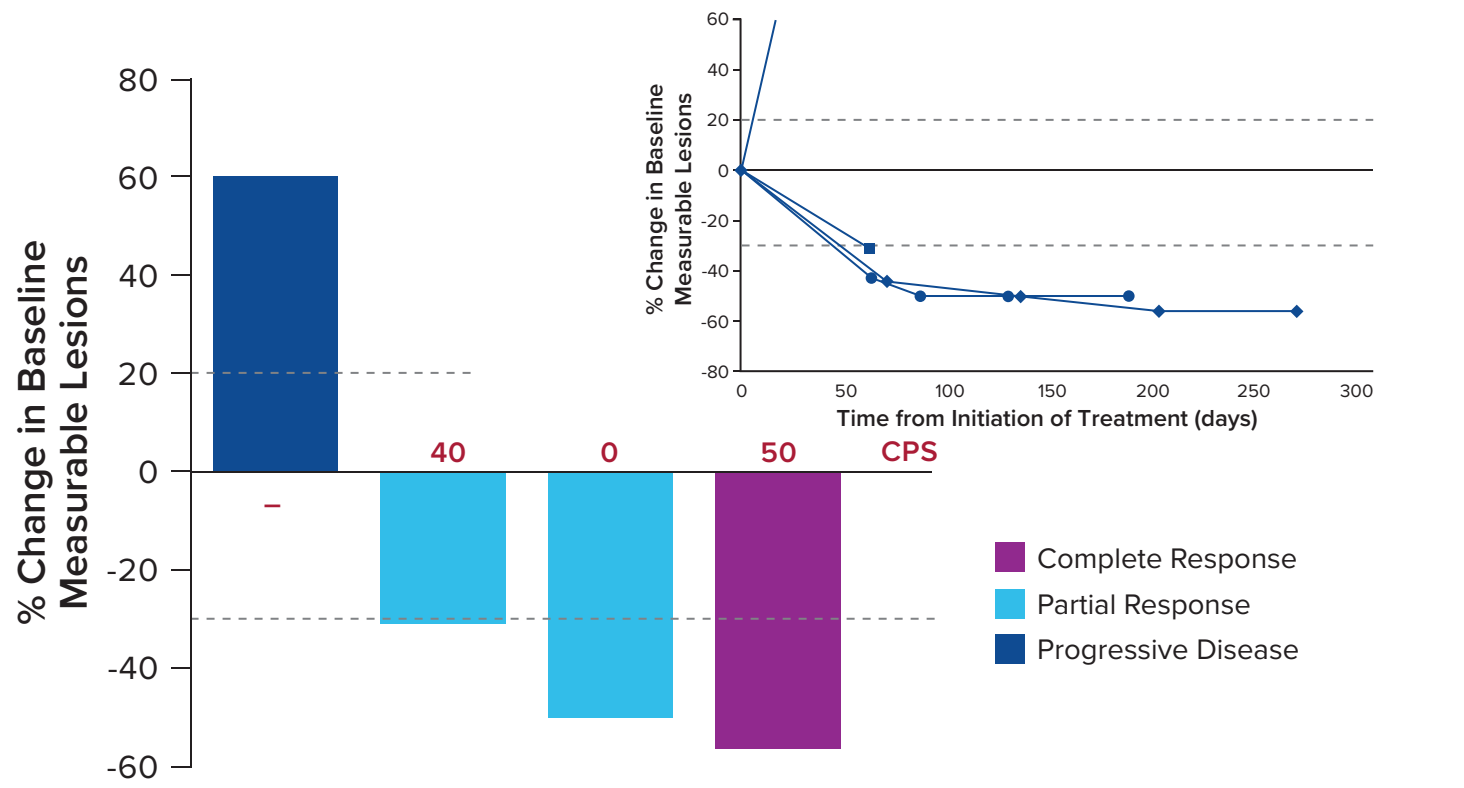
% Change in Baseline Measurable Lesions
–
40
0
50
CPS
Complete Response
Partial Response
Progressive Disease
Time from Initiation of Treatment (days)

Figure 7. Best Overall and Duration of Response in Patients While Receiving ALX148 + Pembrolizumab + 5FU + Platinum

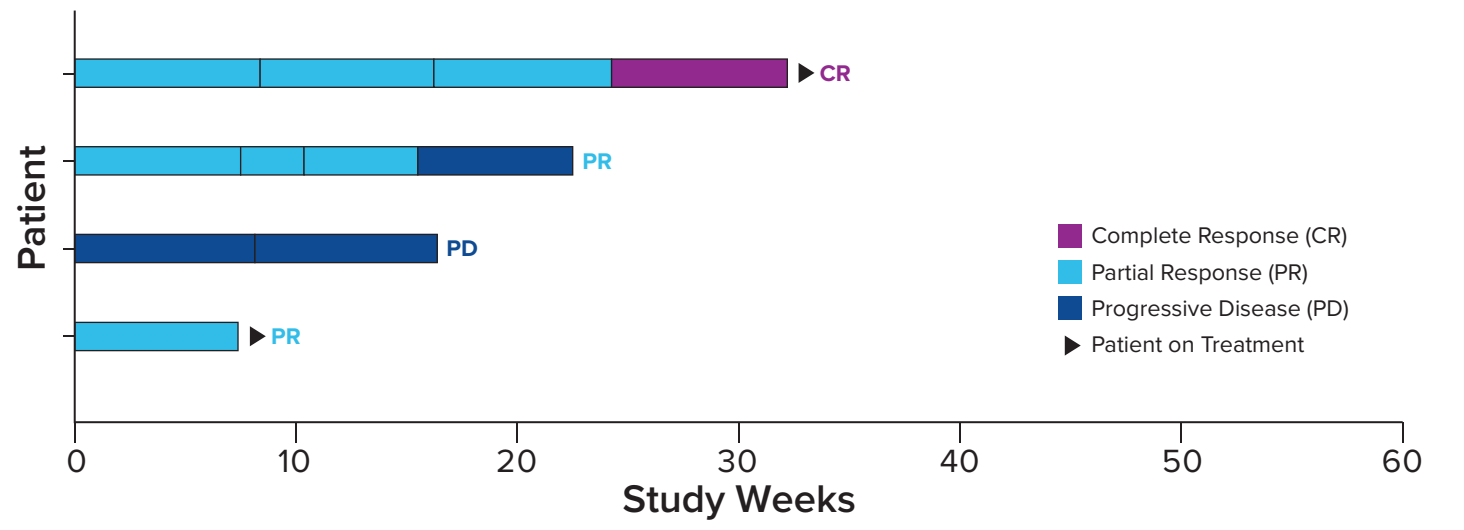
1L HNSCC
Patient
CR
PR
PD
PR
Study Weeks
Complete Response (CR)
Partial Response (PR)
Progressive Disease (PD)
Patient on Treatment

Figure 8. CD47 Target Occupancy in Chemotherapy Combination Cohorts

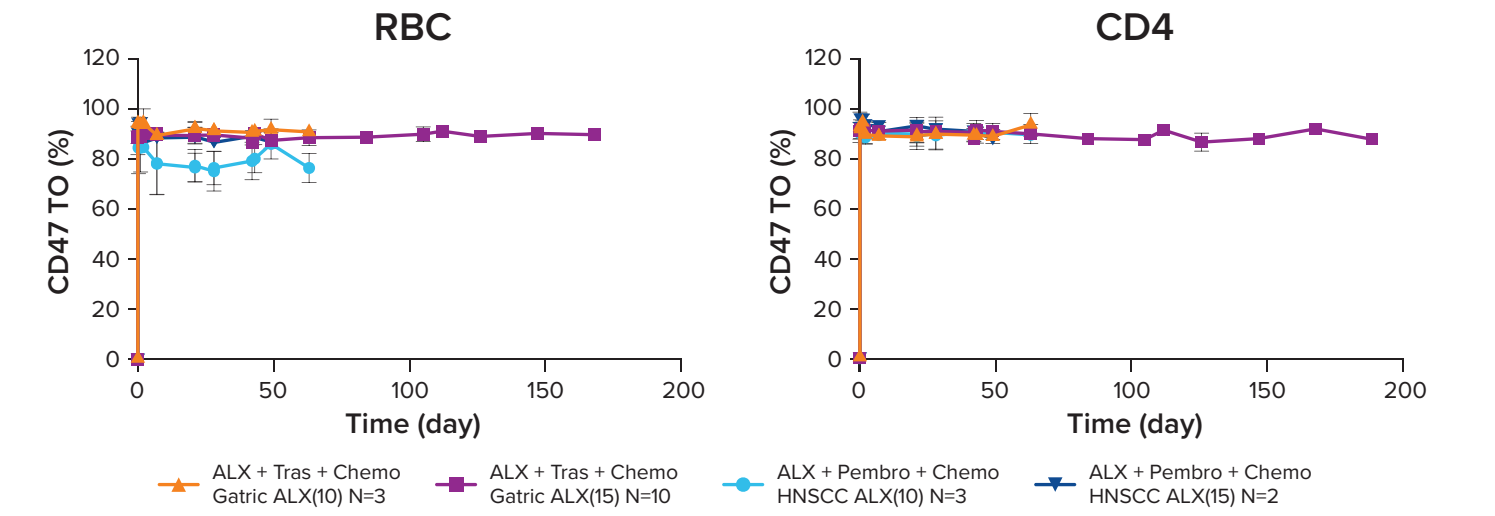
RBC
CD4
CD47 TO (%)
Time (day)
ALX + Tras + Chemo Gatric ALX(10) N=3
ALX + Tras + Chemo Gatric ALX(15) N=10
ALX + Pembro + Chemo HNSCC ALX(10) N=3
ALX + Pembro + Chemo HNSCC ALX(15) N=2

- Near complete CD47 target occupancy is maintained throughout ALX148 dosing interval when combined with chemotherapy-containing regimens.

Figure 9. Pre-Treatment IHC Results from HNSCC Patient 1 (PD-L1 positive) and Patient 2 (PD-L1 negative)

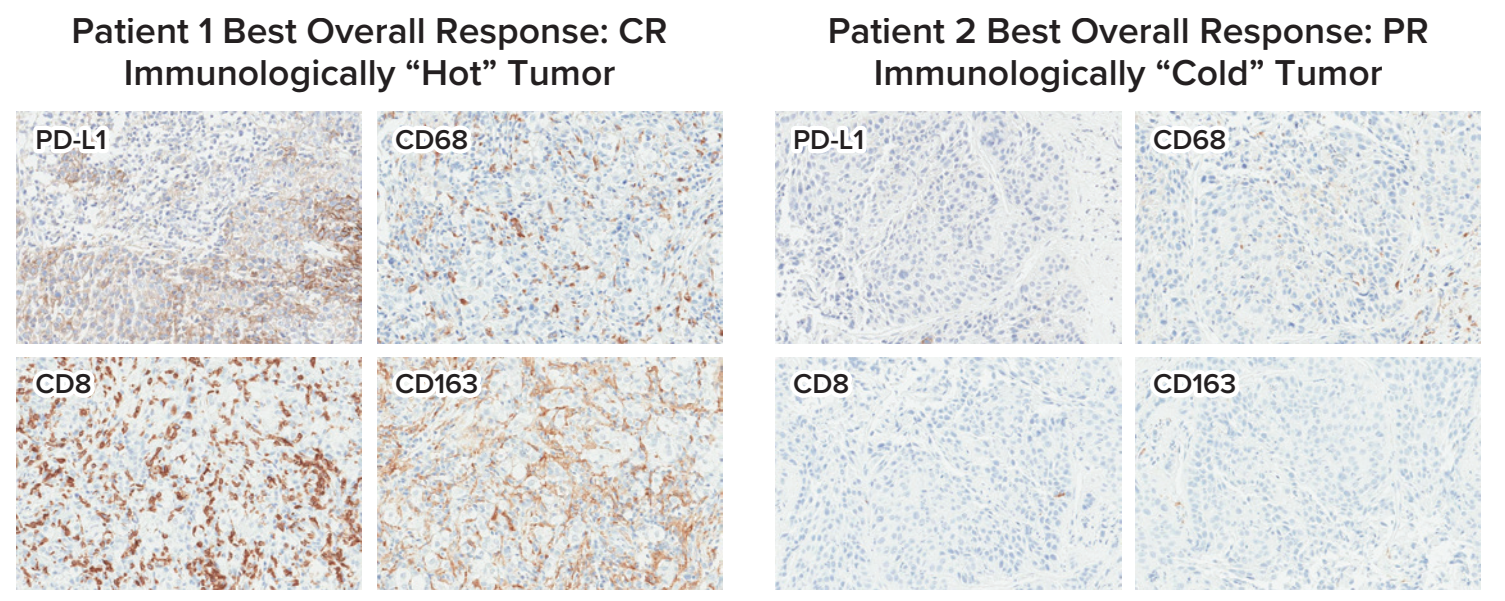
Patient 1 Best Overall Response: CR
Immunologically "Hot" Tumor
Patient 2 Best Overall Response: PR
Immunologically "Cold" Tumor
PD-L1
CD68
CD8
CD163

- **Patient 1:** HNSCC (CPS 50) characterized as immunologically "hot" with a high density of infiltrating T lymphocytes (CD8) and tumor associated macrophages and myeloid cells (CD68 and CD163).
- **Patient 2:** HNSCC (CPS 0) characterized as immunologically "cold" where immune cells are excluded from the tumor core while present at lower density in the peri-tumoral regions.

Conclusions

ALX148, a high affinity CD47 myeloid checkpoint inhibitor with an inactive Fcγ domain and favorable toxicity profile demonstrates objective response in combination with trastuzumab, pembrolizumab and multi-agent chemotherapies in patients with GC and HNSCC.

- Preliminary data suggests that ALX148 can be safely combined with the multi-agent chemotherapy regimens studied with no maximum tolerated dose reached. The maximum administered dose of ALX148 in combination was 15 mg/kg QW.
- ALX148 demonstrates promising initial ORR of 64% in patients with ≥2L HER2 positive GC in combination with trastuzumab and ramucirumab + paclitaxel that compares favorably with the historical data.
- ALX148 demonstrates initial promising anti-cancer activity including complete and partial objective responses in combination with pembrolizumab + 5FU + platinum in patients who have not received prior treatment for their advanced HNSCC, and whose tumor may be immunologically hot or cold.
- Updated data from patients with ≥2L HER2 positive GC receiving ALX148 + trastuzumab suggests promising clinical activity after their tumors have progressed upon prior trastuzumab therapy.
- Updated data from patients who have never been treated with a CPI for their ≥2L HNSCC and who received ALX148 +pembrolizumab suggests clinical activity beyond that expected from pembrolizumab monotherapy.
- Preliminary pharmacokinetics and pharmacodynamic analysis demonstrates no impact of the combination partners upon ALX148 exposure levels with full CD47 receptor occupancy.

Patients in combination cohorts continue to be followed (NCT03013218).

References

1. Weiskopf, K., *Eur J Cancer*. 2017 May;76:100-109.
2. Kauder, S.E. et al., *PLoS ONE*. 2018 August;13(8): e0201832.
3. Lakhani, N. *Journal of Clinical Oncology* 2018 36:15_suppl, 3068-3068.
4. Chow et al. *Journal of Clinical Oncology* 2020 38:15_suppl, 3056-3056.

Acknowledgments

- We would like to thank all of the participating patients and their families as well as site research staff.
- **Contact email:** info@alxoncology.com
- Presented at the Society for Immunotherapy of Cancer (SITC) Annual Meeting November 11-14, 2020 Abstract #404.

*Dr. L Chow currently at University of Texas at Austin, TX.