Discovery of Monoclonal Antibodies Targeting Myeloid Checkpoint SIRPα To Enhance Anti-Tumor Immunity

Sim J[1], Sangalang E[1], Sockolosky J[1], Chen A[1], Harrabi O[1], Doyle L[1], Kauder S[1], Harriman B[3], Izquierdo S[3], Carter J[2], Wibowo AS[2], Kuo T[1],Wan H[1], Pons J[1]

[1]ALX Oncology; [2]Helix Biostructures; [3]Ligand Pharmaceuticals

Background & Objective

Signal-regulatory protein α (SIRPα) is an innate immune checkpoint receptor expressed primarily on myeloid cells and neurons[1]. SIRPα suppresses innate immunity upon interaction with its ligand, CD47. Targeting the CD47-SIRPα pathway represents a novel therapeutic approach to enhance anti-cancer immunity. Ongoing clinical trials to inhibit this pathway through targeting CD47 have shown promising results in reducing tumor burden[2]. Unlike CD47 which is expressed ubiquitously, SIRPα expression is more restricted. Therefore, targeting SIRPα may result in differential safety and efficacy profiles versus that of CD47-targeted therapies.

Objectives of our study:

- Discover anti-SIRPα antibodies that display anti-tumor activities, bind prevalent alleles of SIRPα (v1, v2) in global patient population, cross-reactive to mouse and monkey SIRPα for therapeutic translation.
- Identify diverse anti-SIRPα antibodies as useful tools to interrogate biology of CD47-SIRPα pathway.

Methods

- *Chicken immunization and gel encapsulated microenvironment (GEM) screen*. Wildtype and human antibody transgenic chickens were used for immunization[3]. GEM screen consists of a single-antibody secreting B-cell encapsulated in a droplet containing up to 3 different fluorescent reporter beads each coated with a target antigen of interest. B-cell clones that produce antibodies that bind to the immobilized targets were detected with a secondary antibody against chicken IgY, visualized via fluorescent microscopy, isolated, and sequenced.
- *Cloning and expression of scFv and antibodies*. V-genes were amplified from selected B-cell clones and subcloned as scFv-Fc for expression in HEK293FS. Select sc-Fv-IgG1 clones were reformatted as full IgG antibody and expressed in HEK293FS.
- *Surface Plasmon Resonance (SPR)*. Single concentration (100 nM) off-rate screen was carried out to profile binding to various SIRPα (human v1, human v2, mouse NOD/BL6/BALBc, cynomolgus), human SIRPβ and human SIRPγ. Epitope binning was carried out using a classical sandwich approach. For K_D determination, SIRP analytes were injected in a "one-shot" kinetic mode and flowed over immobilized anti-SIRPα antibodies.
- *Crystallization of Fab:SIRPα complexes*. The antibodies were generated as Fabs with a His-tag at C-terminus of truncated hinge domain. Crystallization used sparse matrix screening kits from Qiagen.
- *Phagocytosis*. *In vitro* monocyte-derived macrophages were polarized using M-CSF as described[4].

Results

Chicken immunization and GEM screen

- Based on low homology between chicken SIRPα and human, mouse & cynomolgus SIRPα, chickens were used for immunization.

```
                1                                                          60
Human v1    (1) EEELQVIQPDKSVLVAAGETATLRCTATSLIPVGPIQWFRGAGPGRELIYNQKEGHFPRV
Human v2    (1) EEELQVIQPDKSVSVAAGESAILHCTVTSLIPVGPIQWFRGAGPARELIYNQKEGHFPRV
    Cyno    (1) EEELQVIQPEKSVSVAAGESATLNCTATSLIPVGPIQWFRGVGPGRELIYHQKEGHFPRV
   Mouse    (1) -KELKVTQPEKSVSVAAGDSTVLNCTLTSLLPVGPIKWYRGVGQSRLLIYSFTGEHFPRV
 Chicken    (1) -DFKLQQPQSSVVVIKGDTLTLNCTASGSGPIGAVKWVKGWGSDNQTVYEHKGSFPRVM
                61                                                        120
Human v1   (61) TTVSDLTKRNNMDFSIRIGNITPADAGTYYCVKFRKGSP-DDVEFKSGAGTELSVRAKPS
Human v2   (61) TTVSESTKRENMDFSISISNITPADAGTYYCVKFRKGS--PDTEFKSGAGTELSVRAKPS
    Cyno   (61) TPVSDPTKRNNMDFSIRISNITPADAGTYYCVKFRKGS--PDVELKSGAGTELSVRAKPS
   Mouse   (60) TNVSDATKRNNMDFSIRISNVTPEDAGTYYCVKFQKGPSEPDTEIQSGGGTEVYVLAKPS
 Chicken   (59) RAVPDPTN----DFTIRISNVSLEDAGTYYCVKLRKGIV-DDVVFTRGGGTEVSVHA---
```

- Immunization was carried out by a multi-allele (IgV domains of hSIRPα v1 and v2) and multi-species regimen to generate pan-allelic and -mammalian reactive antibodies.
- Eight immunized chickens produced high titer antibodies against SIRPα v1, v2 and mouse SIRPα. Various antigen-coated GEM bead combinations were used that facilitated discovery of anti-SIRPα with unique profiles. Over 200 anti-SIRPα clones were identified and cloned as scFv-Fc for SPR characterization.

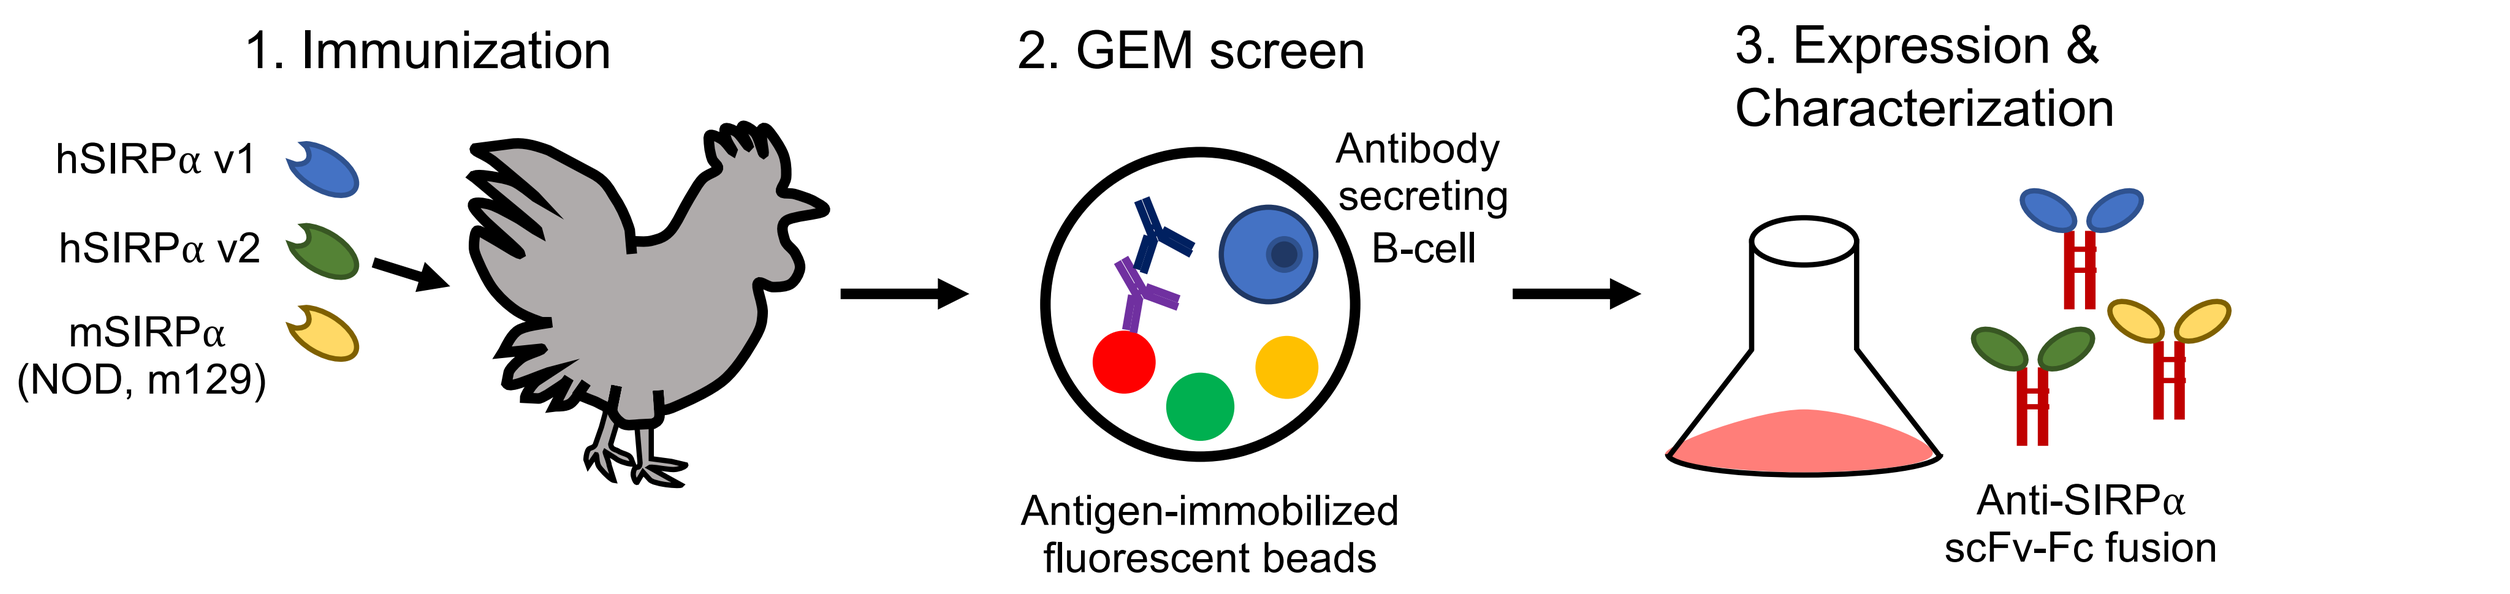
1. Immunization
hSIRPα v1
hSIRPα v2
mSIRPα
(NOD, m129)
2. GEM screen
Antibody secreting B-cell
Antigen-immobilized fluorescent beads
3. Expression & Characterization
Anti-SIRPα scFv-Fc fusion

Chicken[a]	Immunization Scheme				GEM Screen			No. of scFv-Fc Screened
	Initial	Boost 1	Boost 2	Boost 3	Bead 1	Bead 2	Bead 3	
WT 21288	V1	V2	V1	V2	V1	V2	NOD	34
					V2	NOD	Complex[b]	
WT 21292	V1	V2	m129	V1	V1	V2	m129	9
					V2	m129	Complex	
SynVH 22260	V1	V2	V1	V2	V1	V2	NOD	12
					V2	NOD	Complex	
SynVH 22280	V1	V2	m129	V1	V1	V2	M129	60
					V2	m129	Complex	
					V2	BALBc	Complex	
					V2	Complex	-	
Omni 22843	V1	NOD	V2	NOD	V1	V2	SIRPγ	50
					V1	V2	Complex	
					V2	BALBc	Complex	
					V2	Complex	-	
Omni 23504	V1	NOD	V2	NOD	V1	V2	Complex	4
					V1	V2	SIRPγ	
Omni 23941	V1	NOD	V2	NOD	V1	V2	Complex	10
					V1	V2	SIRPγ	
Omni 23975	V1	NOD	V2	NOD	V1	V2	Complex	21
					V1	V2	SIRPγ	

a. SynVH chickens contain humanized V_H immunoglobulin (Ig) repertoires paired with natural chicken light chain repertoire; OmniChickens® contain humanized V_H and V_L Ig repertoires; b. Complex = high affinity CV1 SIRPα variant and IgS domain of CD47 is pre-complexed before GEM screen

Anti-SIRPα antibodies with diverse binding profiles, SIRP reactivity & broad epitope coverage

- Majority of anti-SIRPα antibodies bind both human SIRPα v1 and v2 and cross-react with cynomolgus and mouse SIRPα (m129, NOD, BL6); a subset has specificity for hSIRPα v1 (clones 1, 123, 179, and 194), hSIRPα v2 (clone 110); a small number bind SIRPα, SIRPγ, not SIRPβ (clones 1, 9, 93, 106); SIRPα, SIRPβ1, not SIRPγ (clones 173, 174) and one specific for SIRPα with limited binding to SIRPβ1 and SIRPγ (clone 179).

Antibody	Bin	Source	V1	v2	cyno	m129	NOD	BL6	SIRPγ	SIRPβ1	CD47 Blocking
119	1	H	4.15E-04	1.48E-04	2.34E-04	NB	NB	NB	3.95E-04	4.28E-04	block
120	1	H	4.04E-04	1.49E-04	2.36E-04	NB	NB	NB	3.93E-04	4.25E-04	block
121	1	H	4.79E-04	1.12E-04	3.02E-04	NB	NB	NB	4.82E-04	5.64E-04	block
122	1	H	4.98E-04	2.26E-04	2.66E-04	NB	NB	NB	3.61E-04	3.52E-04	block
135	1	H	6.58E-04	8.78E-05	4.73E-04	NB	NB	NB	1.46E-03	5.21E-04	block
16	1	S	6.22E-05	1.11E-04	1.32E-04	1.76E-04	1.28E-03	2.35E-03	8.17E-05	1.13E-04	block
17	1	S	1.24E-04	1.97E-04	1.93E-04	2.34E-04	6.12E-04	1.13E-03	1.02E-04	1.25E-04	block
21	1	S	1.80E-04	2.07E-04	2.33E-04	2.81E-04	2.64E-03	8.06E-04	1.84E-04	1.90E-04	block
22	1	S	1.21E-04	1.46E-04	1.77E-04	1.53E-04	5.83E-04	7.36E-04	1.08E-04	1.38E-04	block
23	1	S	8.35E-05	1.35E-04	1.60E-04	1.34E-04	8.23E-04	1.32E-03	1.04E-04	1.14E-04	block
24	1	S	2.57E-04	3.17E-04	3.82E-04	4.02E-04	3.86E-03	1.26E-03	1.97E-04	1.94E-04	block
25	1	S	1.12E-04	2.09E-04	2.19E-04	1.33E-04	7.79E-04	2.90E-04	1.41E-04	1.71E-04	block
26	1	S	5.81E-05	1.11E-04	9.67E-04	1.08E-03	7.64E-04	NT	8.35E-05	1.29E-04	block
27	1	S	5.81E-05	9.97E-05	1.38E-04	1.36E-04	1.30E-03	1.84E-03	9.46E-05	8.97E-05	block
28	1	S	2.11E-05	1.47E-05	1.15E-04	3.06E-04	6.16E-04	1.86E-03	3.27E-05	6.29E-05	block
29	1	S	9.61E-05	1.78E-04	3.87E-04	1.14E-03	NT	1.86E-03	2.16E-04	2.79E-04	block
30	1	S	1.32E-04	1.88E-04	3.35E-04	4.76E-04	NT	2.44E-03	1.93E-04	2.37E-04	block
55	1	S	4.18E-05	7.16E-05	9.63E-05	1.43E-04	1.19E-03	1.97E-03	4.69E-05	6.48E-05	block
56	1	S	2.28E-04	2.86E-04	2.95E-04	4.06E-04	1.06E-03	4.29E-03	2.15E-04	3.62E-04	block
59	1	S	4.98E-05	8.63E-05	1.06E-04	2.09E-04	1.35E-03	2.39E-03	4.28E-05	1.07E-04	block
60	1	S	1.40E-04	1.86E-04	1.92E-04	2.21E-04	7.26E-04	1.50E-03	1.00E-04	1.53E-04	block
65	1	S	1.45E-04	1.72E-04	1.88E-04	1.59E-04	6.07E-04	7.86E-04	1.34E-04	1.54E-04	block
66	1	S	1.18E-04	2.06E-04	2.50E-04	1.27E-04	4.90E-04	1.02E-03	1.26E-04	1.38E-04	block
69	1	S	2.05E-04	2.67E-04	2.55E-04	2.46E-04	7.09E-04	7.65E-04	1.99E-04	2.29E-04	block
70	1	S	2.25E-04	2.90E-04	2.96E-04	2.70E-04	6.72E-04	9.87E-04	2.22E-04	2.25E-04	block
71	1	S	3.35E-04	4.03E-04	3.64E-04	4.17E-04	1.12E-03	3.41E-03	3.16E-04	3.88E-04	block
73	1	S	1.90E-05	7.49E-05	1.27E-04	1.64E-04	1.48E-03	3.73E-04	7.30E-05	1.08E-04	block
74	1	S	1.61E-04	2.78E-04	3.37E-04	4.12E-04	NT	1.37E-03	2.25E-04	2.66E-04	block
76	1	S	1.40E-04	2.09E-04	2.77E-04	1.79E-04	5.76E-04	9.98E-04	1.77E-04	1.87E-04	block
201	1	S	3.46E-05	4.27E-05	6.36E-04	8.18E-04	2.15E-03	1.11E-03	1.85E-04	4.10E-03	block
202	1	S	3.75E-05	5.40E-05	1.13E-04	4.58E-05	5.09E-04	6.30E-04	4.44E-05	2.05E-03	block
206	1	S	3.50E-05	3.60E-05	6.24E-04	9.01E-04	2.23E-03	1.57E-03	1.92E-04	4.37E-03	block
1	1	W	2.20E-03	NB	NB	0.00212	7.97E-03	1.68E-03	2.07E-03	NB	block
14	1	W	5.42E-05	7.64E-05	6.49E-04	3.73E-02	4.98E-03	1.81E-02	1.26E-04	6.01E-05	block
115	2	H	7.86E-06	1.95E-05	2.22E-05	NB	NB	NB	3.34E-05	3.69E-05	Kick-off
116	2	H	2.84E-05	5.07E-05	7.01E-05	NB	NB	NB	8.25E-05	3.09E-05	Kick-off
117	2	H	1.20E-05	3.40E-05	1.83E-05	NB	NB	NB	4.30E-06	3.54E-05	kick-off
118	2	H	9.17E-06	4.12E-05	6.97E-05	NB	NB	NB	1.13E-05	1.31E-05	Kick-off
132	2	H	3.34E-05	2.06E-05	4.28E-05	NB	NB	NB	1.05E-05	1.01E-05	Kick-off
191	2	H	6.67E-05	3.53E-05	5.54E-05	NB	NB	NB	NT	1.01E-05	Kick-off
198	2	H	3.67E-05	4.95E-05	8.34E-05	NB	NB	NB	NT	4.72E-05	kick-off
136	3	H	8.45E-04	1.61E-03	1.79E-03	4.59E-04	3.86E-04	4.78E-03	LB	3.60E-03	Non-block
137	3	H	1.65E-03	1.88E-03	2.04E-03	5.15E-04	4.43E-04	4.63E-03	LB	3.76E-03	Non-block
175	3	H	8.39E-04	1.83E-03	1.77E-03	6.05E-04	4.17E-04	3.20E-03	NT	2.99E-03	Non-block
177	3	H	3.32E-03	5.37E-03	4.47E-03	1.04E-03	7.36E-04	NT	NT	NT	Non-block
178	3	H	2.23E-03	3.17E-03	3.44E-03	1.12E-03	7.62E-04	NT	NT	4.73E-03	Non-block
180	3	H	1.60E-03	2.27E-03	2.50E-03	8.06E-04	5.76E-04	4.66E-03	NT	3.75E-03	Non-block
184	3	H	2.53E-03	3.91E-03	3.90E-03	8.21E-04	6.38E-04	4.22E-03	NT	NT	Non-block
185	3	H	1.22E-03	2.05E-03	1.78E-03	7.17E-04	5.73E-04	5.14E-03	NT	3.85E-03	Non-block
189	3	H	1.06E-03	2.81E-03	3.31E-03	8.37E-04	5.04E-04	3.90E-03	NT	2.78E-03	Non-block
190	3	H	1.56E-03	1.99E-03	2.17E-03	7.73E-04	5.63E-04	NT	NT	2.56E-03	Non-block
3	4	W	4.10E-04	2.76E-04	2.18E-03	6.55E-04	1.19E-03	1.97E-03	1.62E-04	1.92E-03	Non-block
173	4	H	1.12E-03	5.04E-03	LB	NB	NB	NB	NB	LB	Non-block
174	4	H	1.43E-03	3.99E-03	LB	NB	NB	NB	NB	LB	Non-block
209	4	S	6.59E-04	4.01E-03	LB	NT	NB	NB	LB	6.54E-05	Non-block
213	4	S	LB	6.85E-04	2.60E-03	NT	NB	NB	LB	LB	Non-block
214	4	S	LB	6.77E-04	3.51E-03	NT	NB	NB	LB	LB	Non-block
123	5	H	1.55E-03	NB	1.57E-03	NB	NB	NB	1.43E-03	LB	Non-block
149	5	H	4.15E-03	5.44E-04	LB	NB	NB	NB	LB	3.31E-04	Non-block
161	5	H	3.37E-03	3.48E-04	LB	NB	NB	NB	LB	3.21E-03	Non-block
162	5	H	6.84E-04	LB	LB	NB	NB	NB	4.66E-03	1.98E-03	Non-block
163	5	H	5.35E-04	6.64E-03	LB	NB	NB	NB	3.15E-03	1.70E-03	Non-block
164	5	H	5.95E-04	9.89E-03	LB	NB	NB	NB	4.26E-03	1.67E-03	Non-block
194	5	H	1.17E-03	NB	1.31E-03	NT	NB	NB	NT	LB	Non-block
218	5	S	7.06E-05	5.47E-05	2.49E-05	NT	NB	NB	2.12E-05	NT	Non-block
45	6	W	7.24E-05	1.44E-05	NB	NB	NB	NB	LB	LB	Non-block
9	NT	W	1.29E-04	1.40E-04	3.38E-04	NB	NB	NT	1.03E-04	NB	Block
93	NT	W	9.15E-05	1.09E-04	4.19E-04	NB	NB	NT	1.09E-04	NB	Block
106	NT	S	9.15E-04	6.14E-04	NB	NB	NB	NT	7.26E-04	NB	Block
110	NT	S	NB	2.40E-04	NB	NB	NB	NT	LB	LB	Block
179	NT	H	1.74E-03	NB	NB	NB	NB	NB	NB	NB	NT

Binding specificity (K_{off}, 1/s)

Binding
LB/NB (low/no binding)
NT (not tested)

LB = K_{off} 5-10x10^{-3}
NB = K_{off} > 1x10^{-2}
W=wt, H=OmniChicken, S=SynVH chicken

- Binding of antibodies to various SIRP proteins was determined by SPR using single concentration (100nM) off-rate screen. Binding was fitted using a 1:1 Langmuir and K_{off} (1/s) values calculated.
- The panel of SIRP proteins generated for SPR screens are the IgV domain from human SIRPα v1 (NP_542970.1), human SIRPα v2 (CAA71403.1), cynomolgus SIRPα (EHH65484.1), mouse 129 SIRPα (162330193), NOD/C57BL/6 SIRPα sequences are designed as described in reference[5], human SIRPγ (NP_061026.2), human SIRPβ1 (NP_006056.2). All the proteins are expressed and produced in HEK293FS.

- Epitope binning showed the antibodies can be divided into 6 distinct bins: blockers (bin 1), kick-offs (bin2), and non-blockers that subdivide into 4 bins (bin 3, 4, 5, and 6).

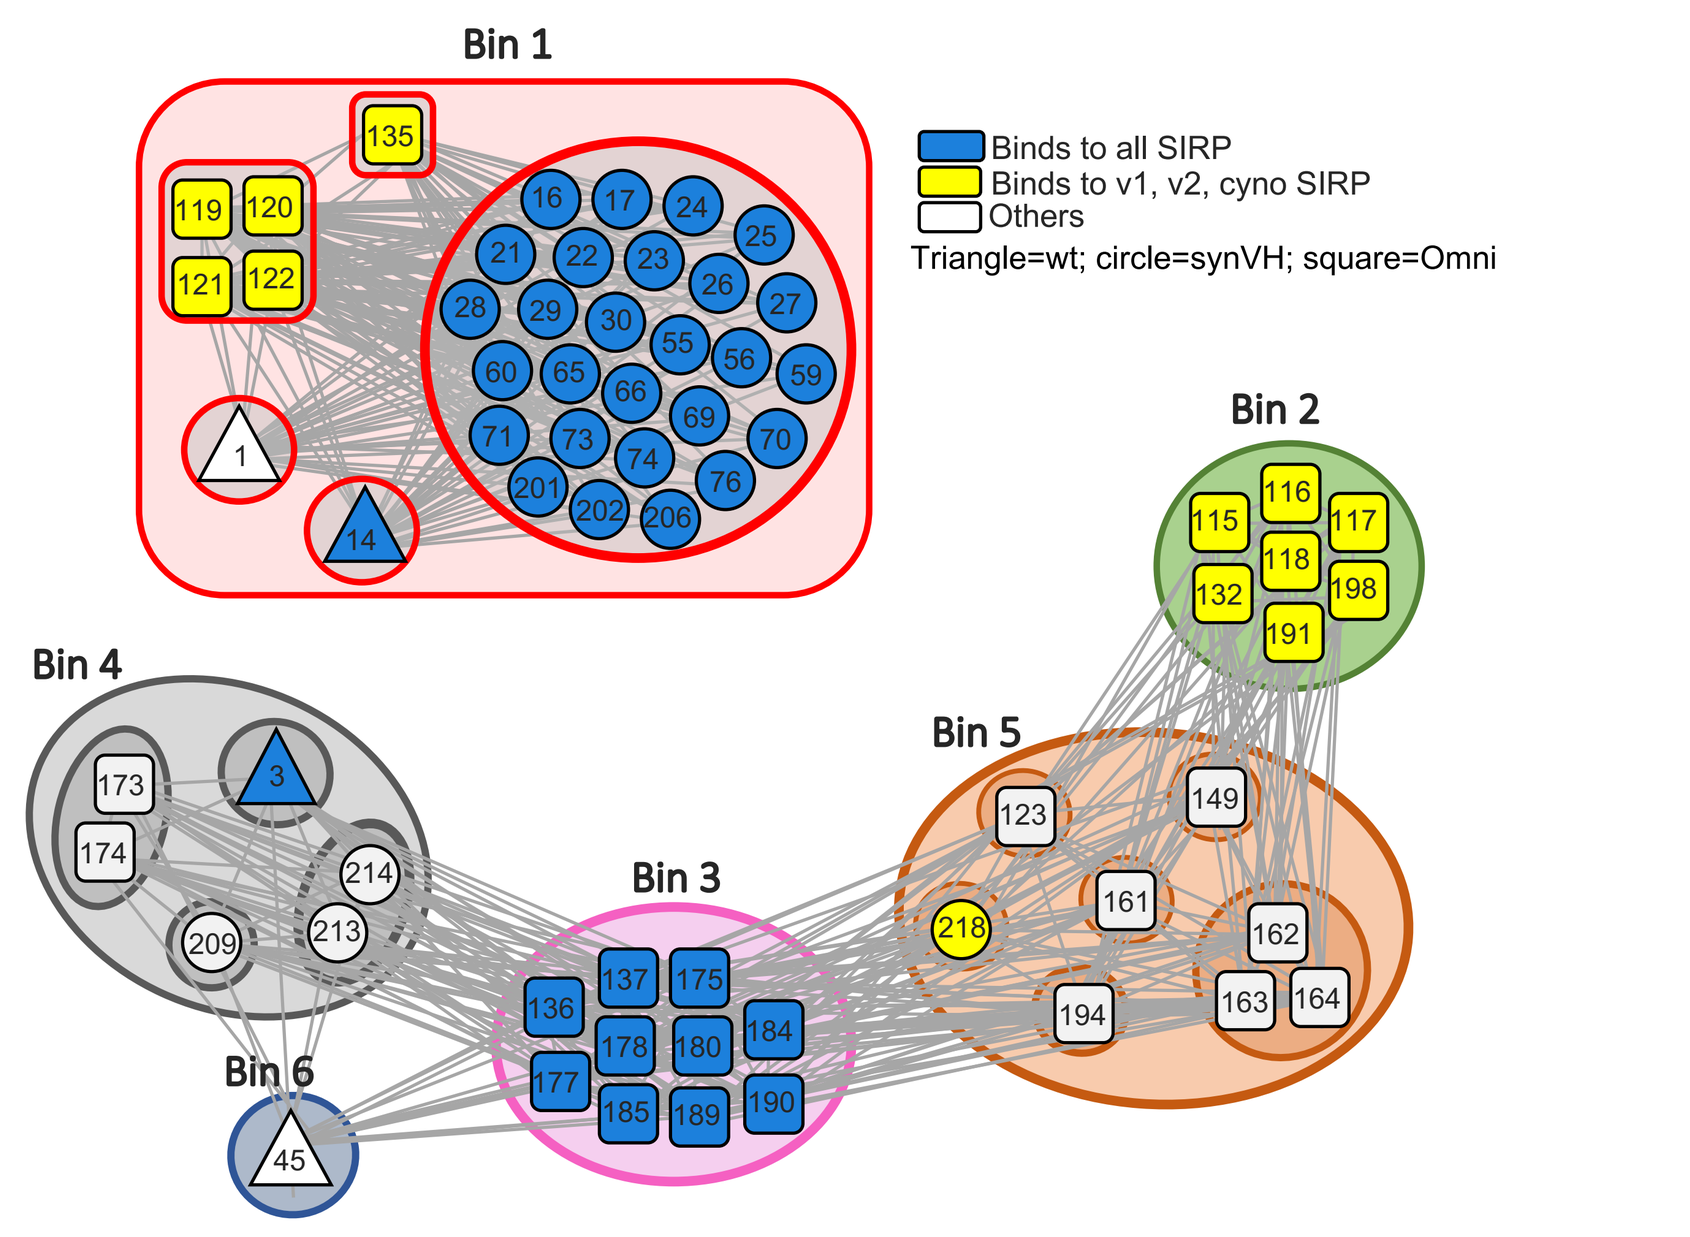
Bin 1
135
119 120
121 122
16 17 24 25 21 22 23 26 27 28 29 30 55 56 59 60 65 66 69 70 71 73 74 76 201 202 206
1
14
Binds to all SIRP
Binds to v1, v2, cyno SIRP
Others
Triangle=wt; circle=synVH; square=Omni
Bin 2
115 116 117 118 132 198 191
Bin 4
173 3 174 214 209 213
Bin 3
136 137 175 177 178 180 184 185 189 190
Bin 5
123 149 218 161 162 194 163 164
Bin 6
45

- Binning experiments were conducted by immobilizing each anti-SIRPα antibody (ligand) followed by capture of the recombinant SIRPα antigen and the pairwise interrogation of an in solution antibody (analyte) binding to the antibody-captured SIRPα antigen.
- Epitope binning was performed again by reversing the mAb ligand/analyte orientation to validate the data set.

- Anti-SIRPα antibodies were categorized into three types based on their inhibition of CD47-SIRPα interaction – blocking, non-blocking or kick-off antibodies.

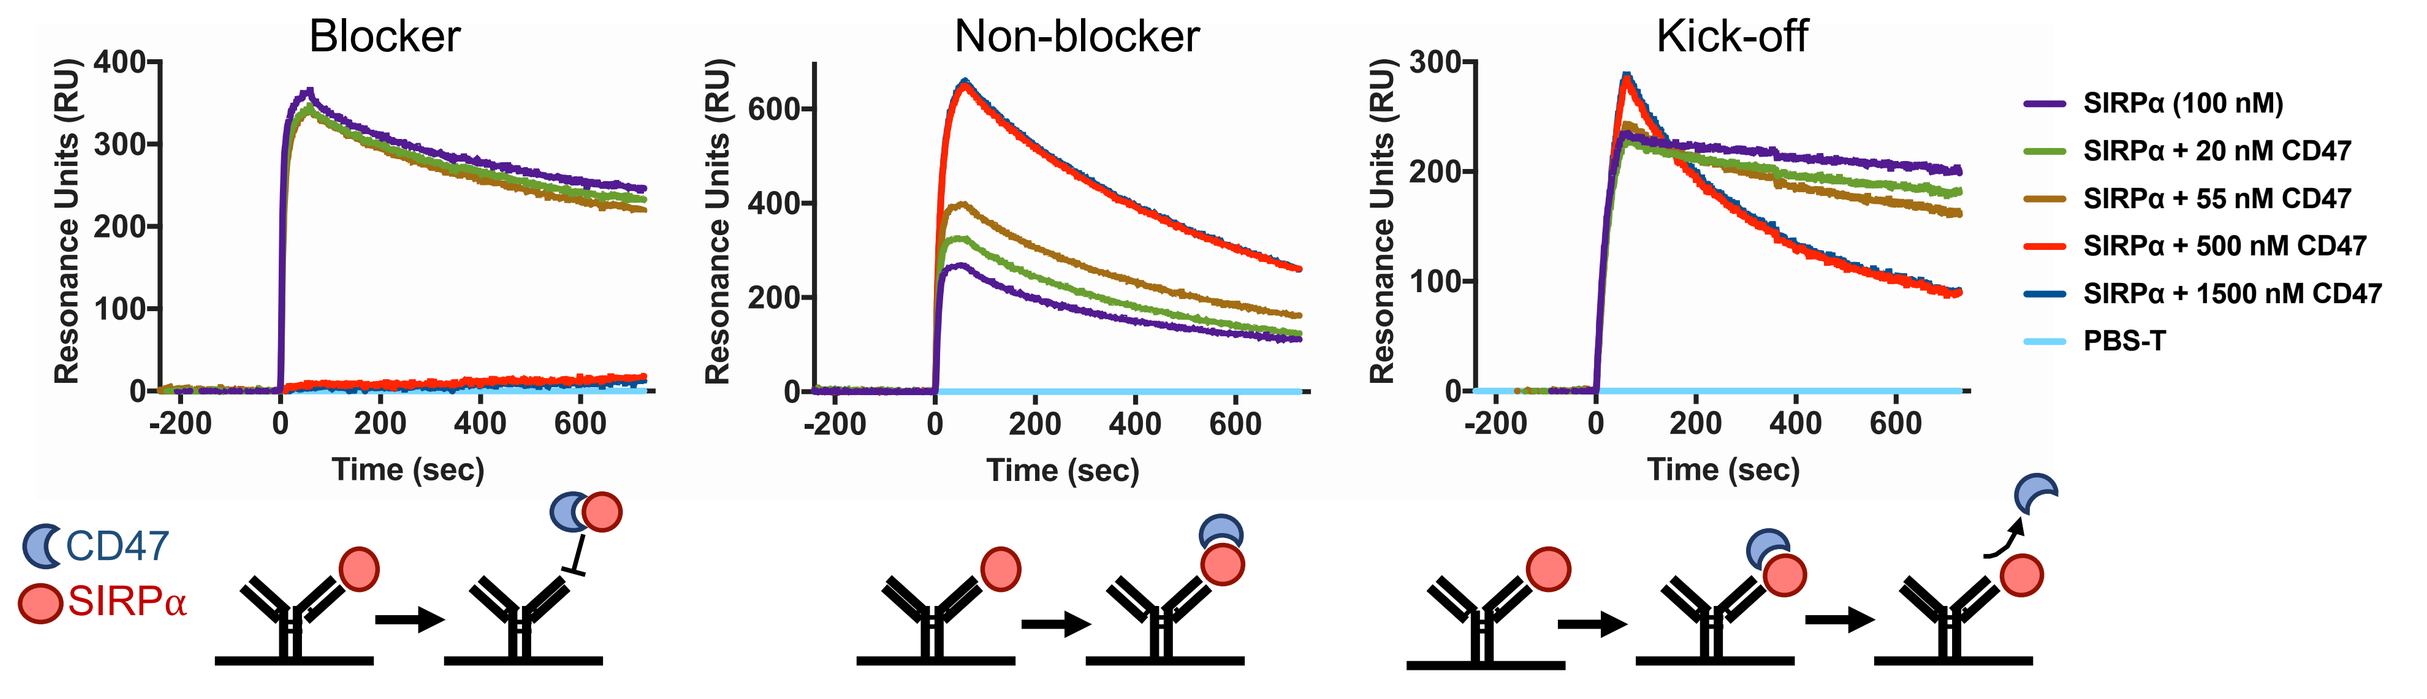
Blocker
Non-blocker
Kick-off
Resonance Units (RU)
Time (sec)
SIRPα (100 nM)
SIRPα + 20 nM CD47
SIRPα + 55 nM CD47
SIRPα + 500 nM CD47
SIRPα + 1500 nM CD47
PBS-T
CD47
SIRPα

- Anti-SIRPα antibodies were first captured on a chip. Increasing concentrations of CD47 (0 to 1500 nM) were added to 100 nM of high affinity SIRPα prior to injection
- At saturating concentrations of CD47 (>100 nM, antibodies that bind overlapping epitopes on SIRPα (blocking) fail to bind the SIRPα/CD47 complex, whereas antibodies that bind non-competitive epitopes (non-blocking) form a classical sandwich with the SIRPα/CD47 complex. A third type, kick-off antibodies, form a transient complex between antibody:SIRPα:CD47 prior to displacement of CD47 from antibody-bound SIRPα

Anti-SIRPα antibodies bind with high affinity to SIRPα

- A diverse panel of antibodies bind to human SIRPα v1 and v2 with high affinity ranging from low nanomolar to picomolar (K_D, M).
- A select number of clones (21, 119, 135, 136) also bind similarly to human SIRPα alleles and cynomolgus SIRPα; in addition, clones 21 and 136 also cross-react with mouse SIRPα alleles which will facilitate preclinical mouse studies.

Bin	Antibody	Source	V1	V2	Cyno	NOD	BL6	BALB/c	SIRPγ	SIRPβ1	CD47 Blocking
1	119	H	1.83E-10	6.82E-11	1.12E-10	NB	NB	NB	2.67E-10	3.42E-10	Block
1	135	H	1.51E-10	2.90E-11	9.69E-11	NB	NB	NB	5.39E-10	1.10E-10	Block
1	21	S	<1.00E-12	<1.00E-12	3.61E-12	5.47E-10	4.60E-10	1.05E-09	<1.00E-12	<1.00E-12	Block
2	115	H	4,26E-10	1.86E-09	2.41E-09	NB	NB	NB	B	B	Kick-off
3	136	H	4.58E-10	1.63E-09	2.15E-09	5.54E-10	1.27E-08	3.50E-10	2.39E-08	4.35E-09	Non-block
4	3	W	1.62E-10	7.67E-11	2.29E-09	1.63E-09	3.65E-09	1.16E-09	8.36E-08	1.63E-09	Non-Block
4	173	H	9.37E-10	9.28E-09	4.46E-08	NB	NB	NB	NB	B	Non-Block
4	209	S	1.71E-10	5.01E-09	3.90E-08	NB	NB	NB	8.99E-09	B	Non-Block
4	213	S	6.05E-09	1.69E-09	4.49E-08	NB	NB	NB	2.02E-09	1.71E-08	Non-Block
5	123	H	6.05E-10	NB	1.62E-09	NB	NB	NB	7.47E-10	7.07E-08	Non-Block
5	149	H	8.73E-10	2.38E-10	7.64E-09	NB	NB	NB	1.89E-09	2.06E-10	Non-Block
5	161	H	1.03E-09	1.27E-10	6.35E-09	NB	NB	NB	2.84E-09	2.63E-09	Non-Block
5	162	H	4.50E-10	1.57E-08	1.26E-08	NB	NB	NB	3.97E-09	3.00E-09	Non-Block
5	194	H	4.97E-10	NB	9.11E-10	NB	NB	NB	9.47E-10	5.36E-08	Non-Block
5	218	S	1.23E-10	2.76E-10	5.99E-11	NB	NB	NB	6.36E-11	B	Non-Block
6	45	W	6.63E-11	1.34E-10	NB	NB	NB	NB	2.71E-08	1.06E-08	Non-Block

- All antibodies are screened as full IgG1 except 173, 209, 213, 123, 149, 161, 162, 194, 218 are screened as scFv-Fc
- NB = no binding; B = binding confirmed and exact K_D value was not determined
- W=wt, H=OmniChicken, S=SynVH chickens

Crystal structures of anti-SIRPα Fab fragments in complex with SIRPα

- High resolution structures (1.83-2.68 A) of 5 Fabs in complex with IgV domain of SIRPα were elucidated. The antibodies are blocker 119 (bin1), kick-off 115 (bin2), non-blockers 136 (bin3), 3 (bin4) and 218 (bin5)

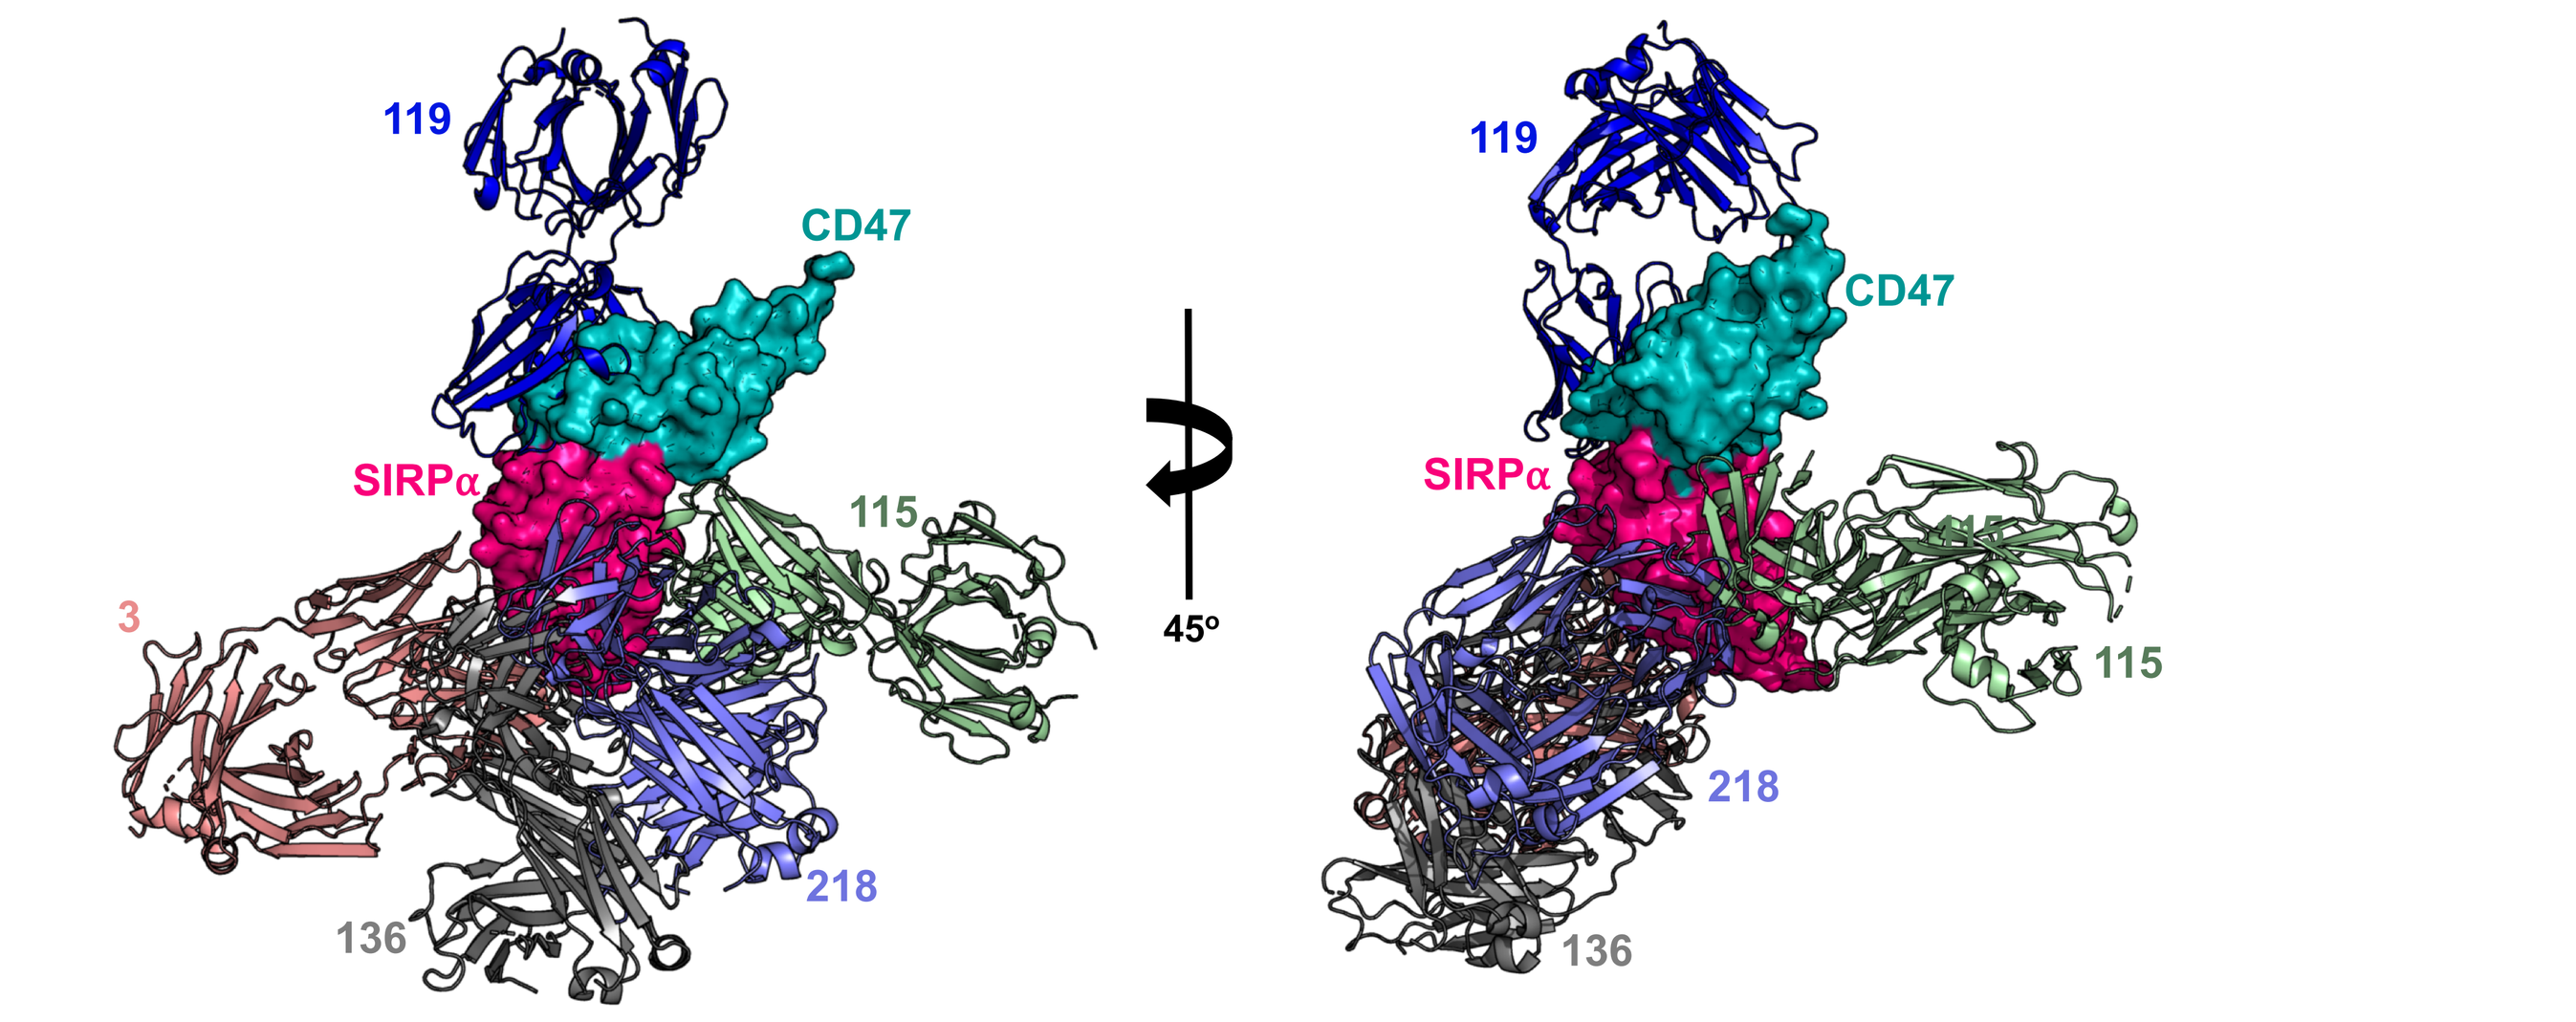
119
CD47
SIRPα
115
3
218
136
45°

- Binding epitopes of the 5 Fabs to SIRPα and their respective overlapping binding sites with other Fabs are in agreement with the epitope binning results. The binding regions are colored accordingly in the space filled and Venn diagrams shown below.

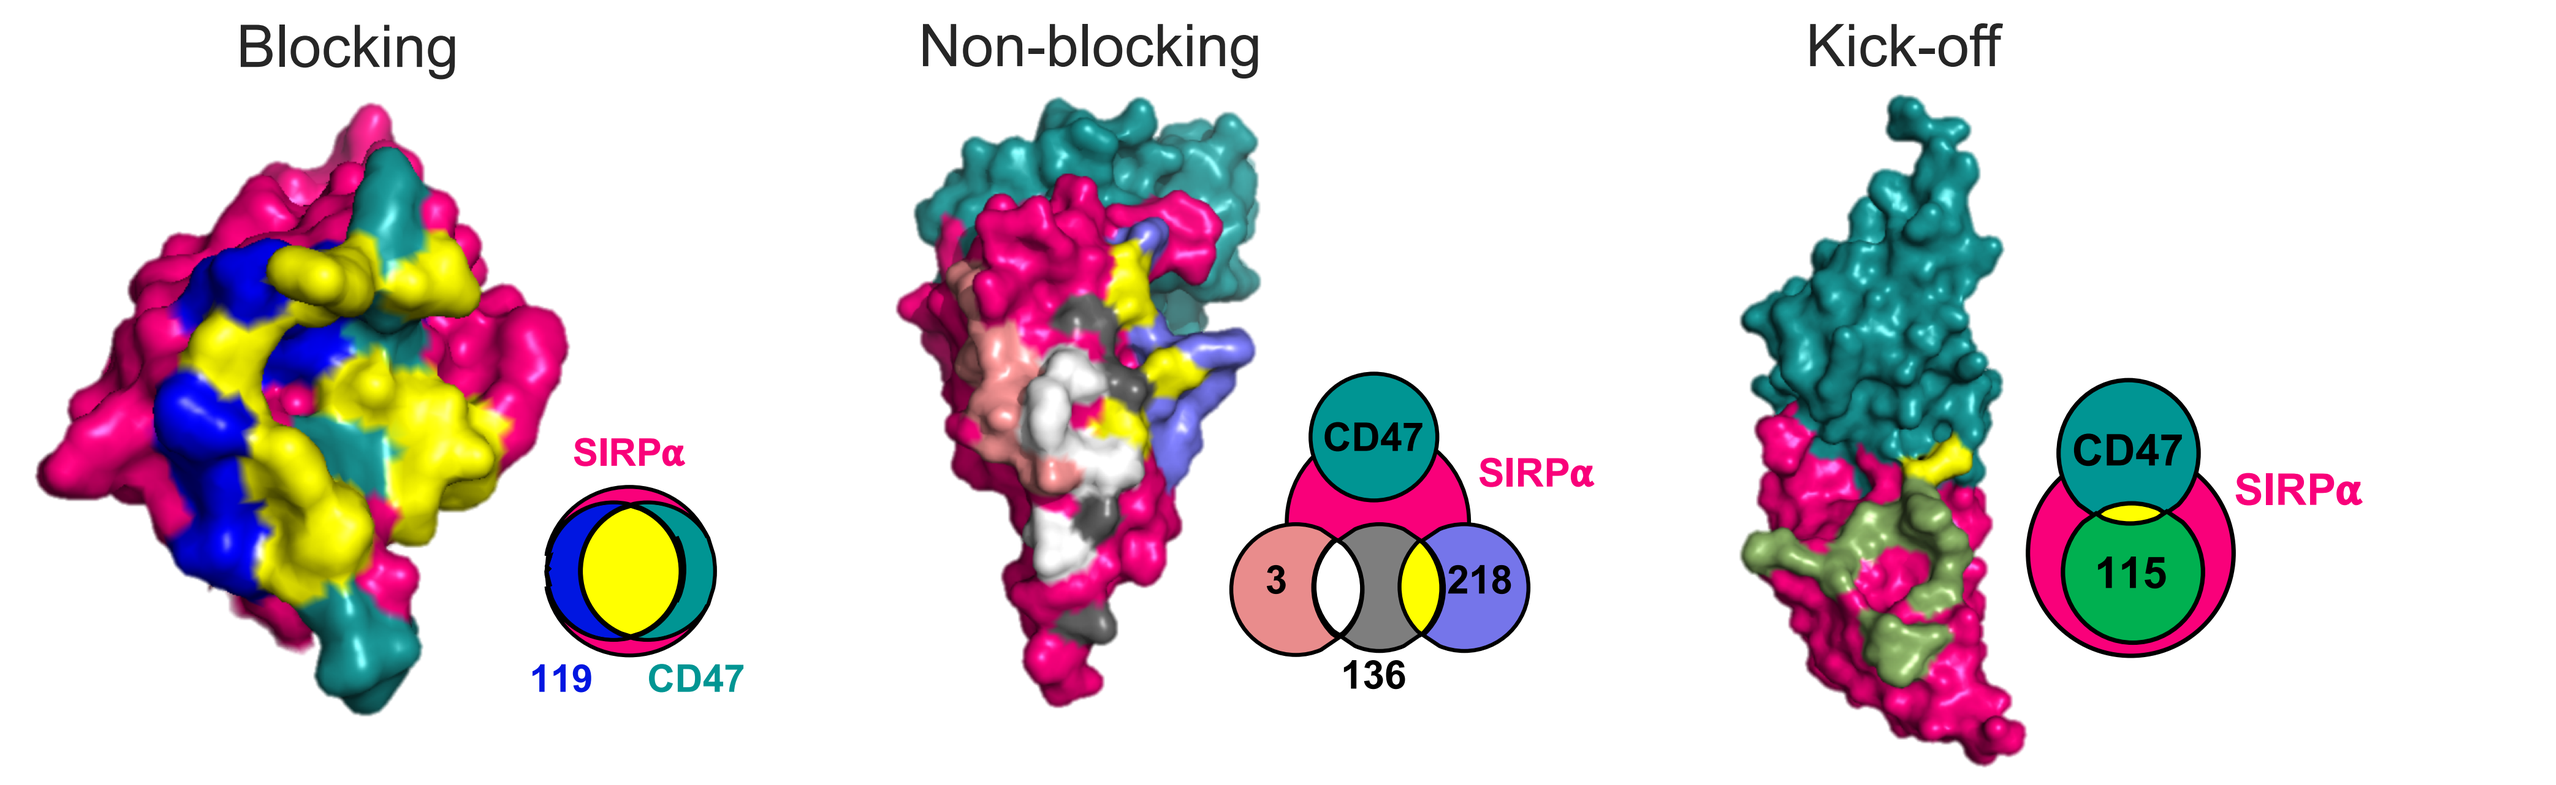
Blocking
Non-blocking
Kick-off
SIRPα
119
CD47
3
218
136
115

Anti-SIRPα antibodies can potentiate macrophage phagocytosis and enhance anti-tumor activity in vivo

- Blockers and non-blockers anti-SIRPα antibodies potentiated macrophage phagocytosis of DLD-1 tumor cells in combination with cetuximab

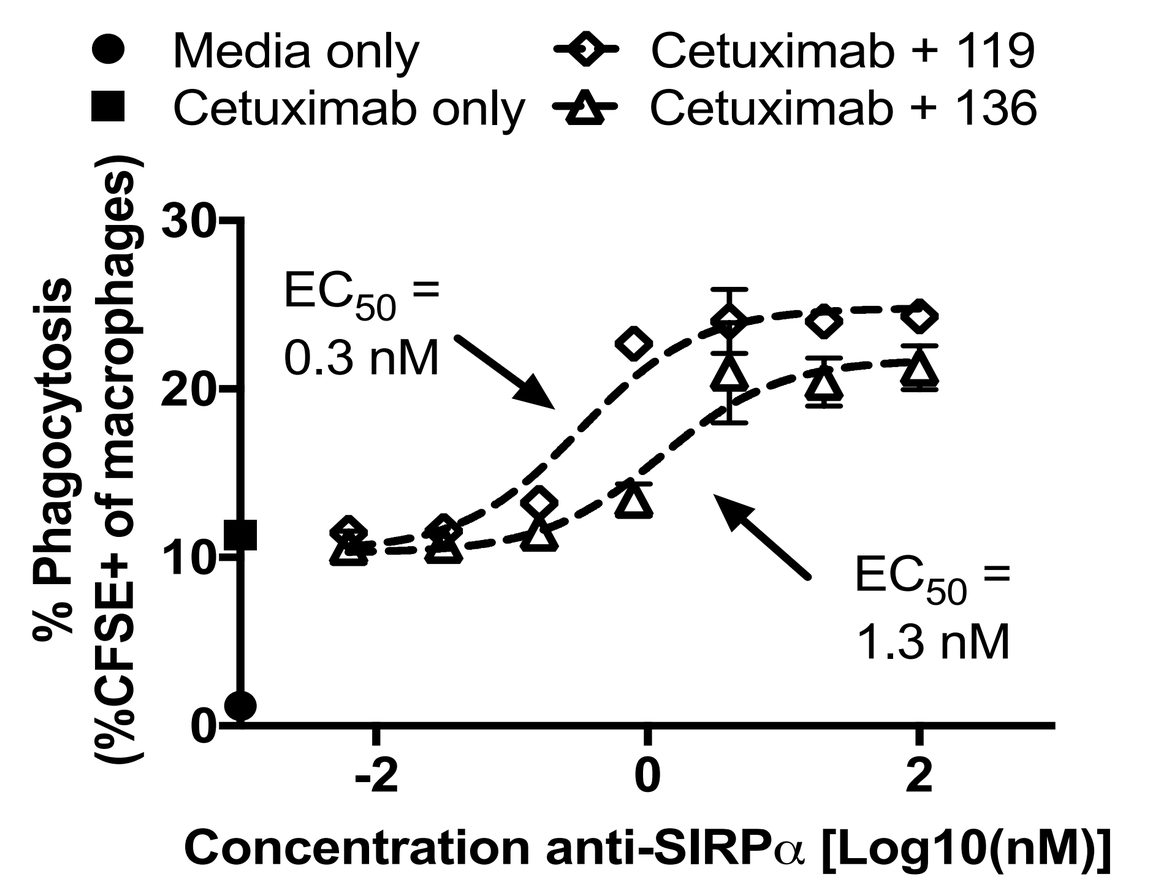
Media only
Cetuximab only
Cetuximab + 119
Cetuximab + 136
% Phagocytosis (%CFSE+ of macrophages)
EC50 = 0.3 nM
EC50 = 1.3 nM
Concentration anti-SIRPα [Log10(nM)]

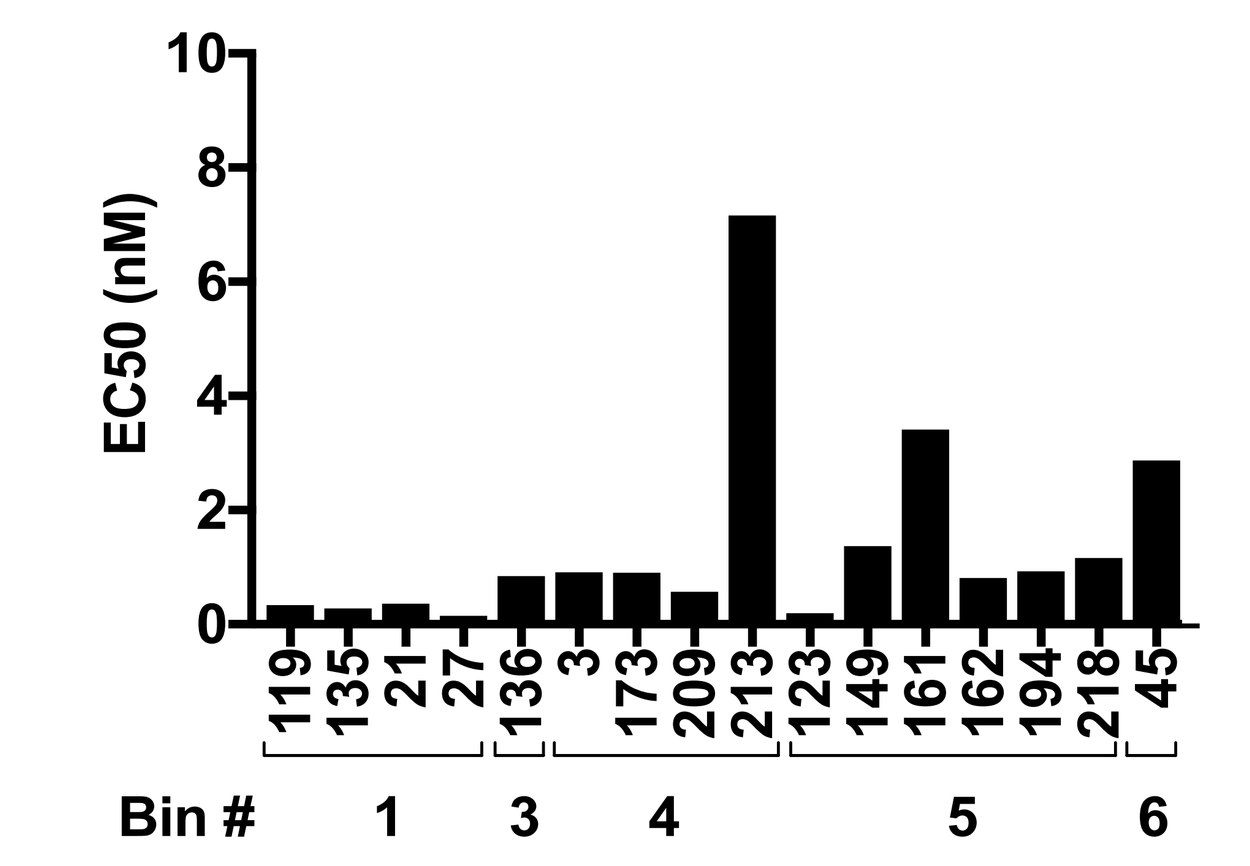
EC50 (nM)
119 135 21 27 136 3 173 209 213 123 149 161 162 194 218 45
Bin #
1
3
4
5
6

- Allele specific anti-SIRPα antibody (v1) fails to enhance phagocytosis by v1/v2 and v2/v2 macrophages. Hence, pan-allele binding anti-SIRPα antibodies are critical for clinical development to target diverse patient populations (A).
- Blocker anti-SIRPα in combination with PD-1 results in tumor inhibition in MC38 syngeneic model (B).

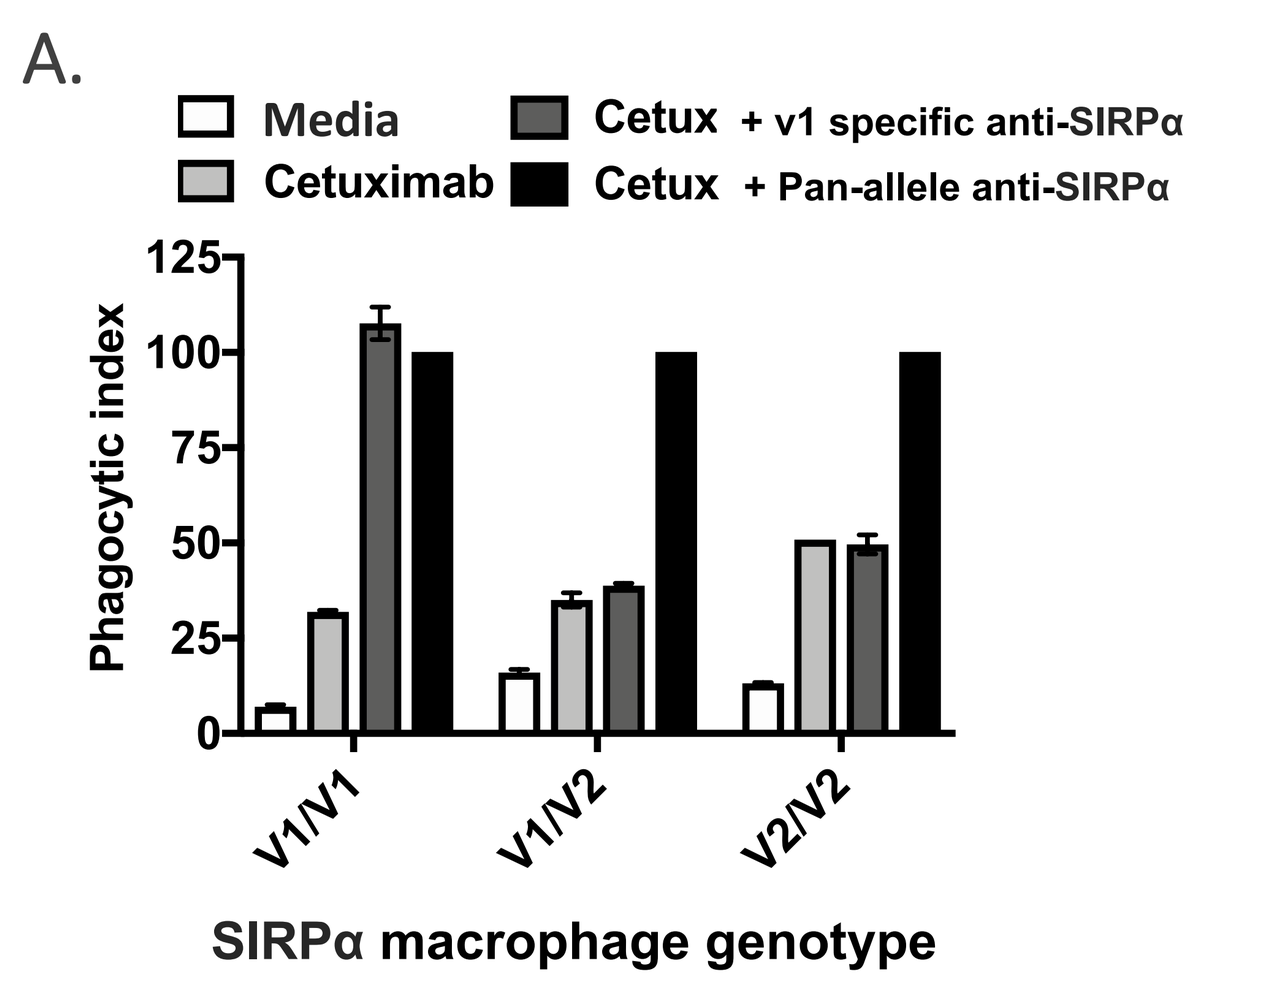
A.
Media
Cetuximab
Cetux + v1 specific anti-SIRPα
Cetux + Pan-allele anti-SIRPα
Phagocytic index
V1/V1
V1/V2
V2/V2
SIRPα macrophage genotype

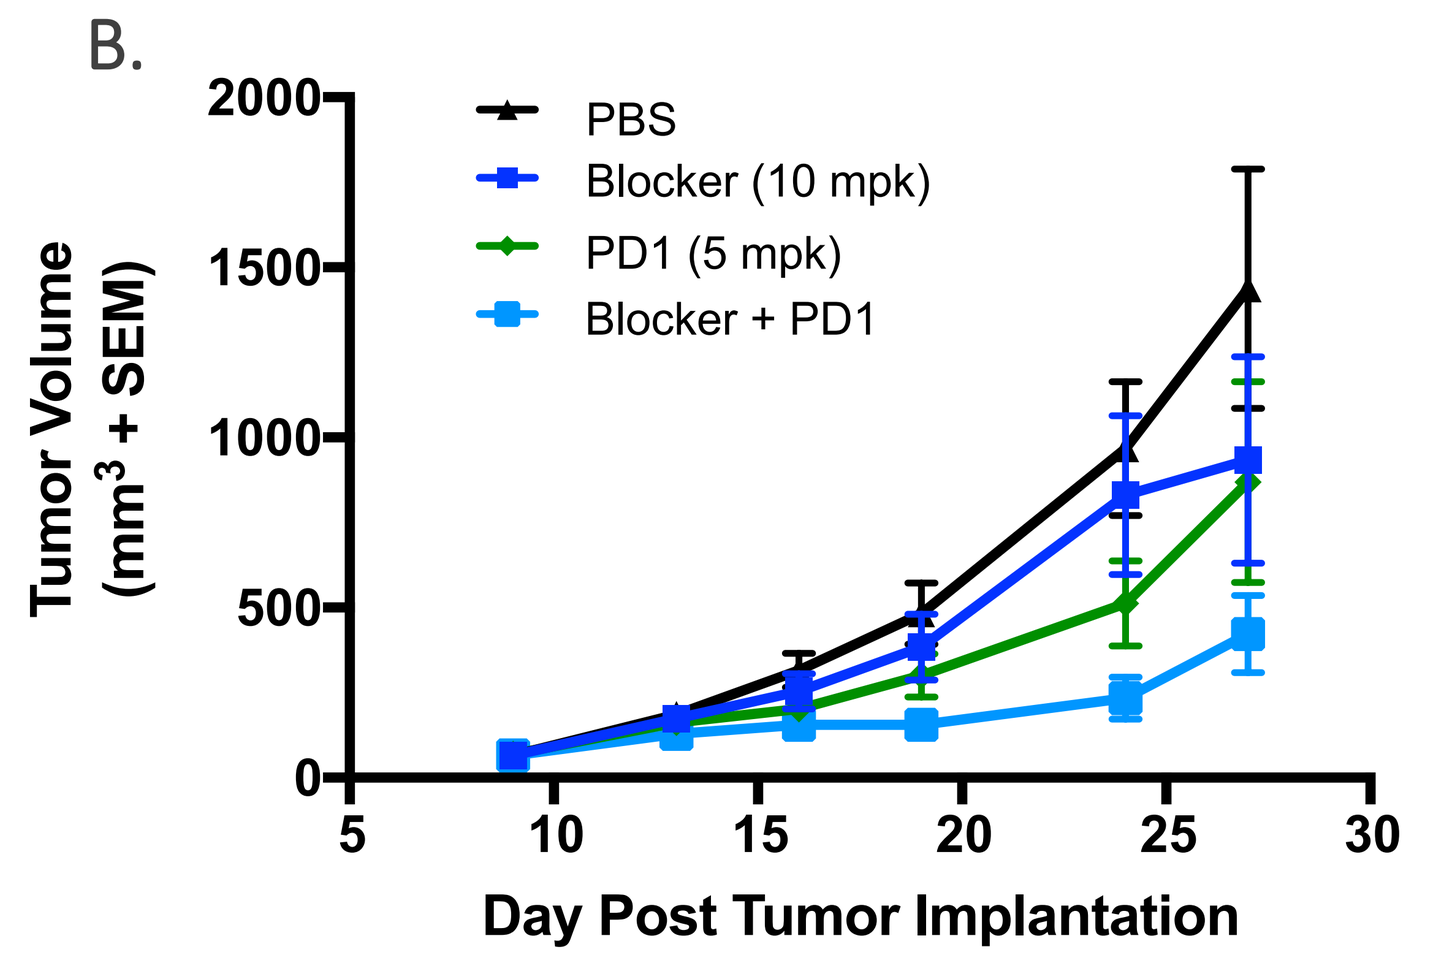
B.
PBS
Blocker (10 mpk)
PD1 (5 mpk)
Blocker + PD1
Tumor Volume (mm3 + SEM)
Day Post Tumor Implantation

Concluding remarks

- A panel of anti-SIRPα antibodies with diverse binding profiles, SIRP reactivity and broad epitope coverage were identified and will be useful tools for interrogation of the SIRP biology and CD47-SIRPα checkpoint
- Selected anti-SIRPα antibodies display high affinity binding to both human SIRPα v1 and v2, species cross-reactivity, and anti-tumor activities. These anti-SIRPα antibodies are attractive for further development to evaluate the therapeutic potential of targeting SIRPα in cancer patients.

References:

1. Barclay AN, Van den Berg TK (2014) *Annu Rev Immunol* 32:25–50.
2. Lakhani N, et al. (2018) *SITC*, p Abstract nr P335.
3. Ching KH, et al. (2018) *MAbs* 10(1):71–80.
4. Kauder SE, et al. (2018) *PLoS One* 13(8):e0201832.
5. Iwamoto C, et al. (2014) *Exp Hematol* 42(3):163–171.e1.