Pharmacodynamic Biomarker Characterization of ALX148, a CD47 Blocker, in Combination with Established Anticancer Antibodies in Patients with Advanced Malignancy

Hong I Wan[1], Laura QM Chow[2], Justin Gainor[3], Nehal Lakhani[4], Hyun Chung[5], Keun-Wook Lee[6], Jeeyun Lee[7], Patricia LoRusso[8], Yung-Jue Bang[9], Stephen Hodi[10], Rafael Santana-Davila[11], Wells Messersmith[12], Philip Fanning[1], Pierre Squifflet[1], Feng Jin[1], Tracy Kuo[1], Sangeetha Bollini[1], Jaume Pons[1], Sophia Randolph[1]

[1]ALX Oncology, Burlingame, CA, USA, and Dublin, Ireland; [2]Livestrong Cancer Institutes, Austin, TX, USA (current); [3]Massachusetts General Hospital Cancer Center, Boston, MA, USA; [4]START Midwest, Grand Rapids, MI, USA; [5]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Korea; [6]Seoul National University Bundang Hospital, Seongnam, Korea; [7]Samsung Medical Center, Seoul, Korea; [8]Yale Cancer Center, New Haven, CT, USA; [9]Seoul National University Hospital, Seoul, Korea; [10]Dana Farber Cancer Center, Boston, MA, USA; [11]Seattle Cancer Care Alliance, Seattle, WA, USA [12]University of Colorado Cancer Center, Aurora, CO, USA.

Background

- CD47-SIRPα signaling is a myeloid checkpoint mechanism that signals the macrophage to ignore the cell on which CD47 is expressed. Tumors upregulate CD47 to evade the immune response.
- ALX148 is a fusion protein with a high affinity CD47 blocker linked to an inactive human immunoglobulin Fc region (Figure 1). It blocks the CD47-mediated 'don't eat me' signal and enhances anti-tumor immunity.[1]
- AT148001, a first-in-human phase 1 study, evaluates ALX148 administered as a single agent (Part 1) and in combination with established anti-cancer antibodies (Part 2).[2-4]

Figure 1. ALX148: A Unique High Affinity SIRPα Fusion Protein[1]

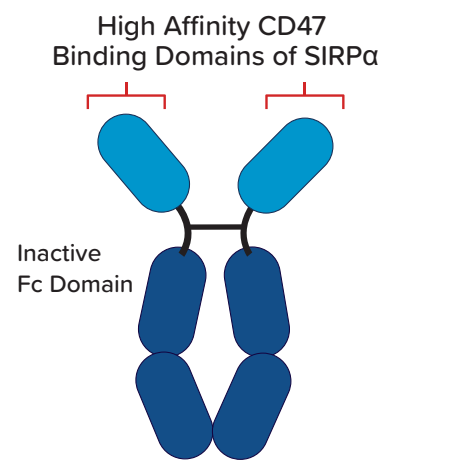
High Affinity CD47
Binding Domains of SIRPα
Inactive
Fc Domain

- Selectively binds CD47 to block its interaction with SIRPα with picomolar binding affinity
- Fc domain mutated to eliminate Fcγ receptor binding
- Molecular weight half the size of typical antibody
- Fc domain enables antibody-like PK

Figure 2. ALX148 Bridges Innate and Adaptive Immunity[1]

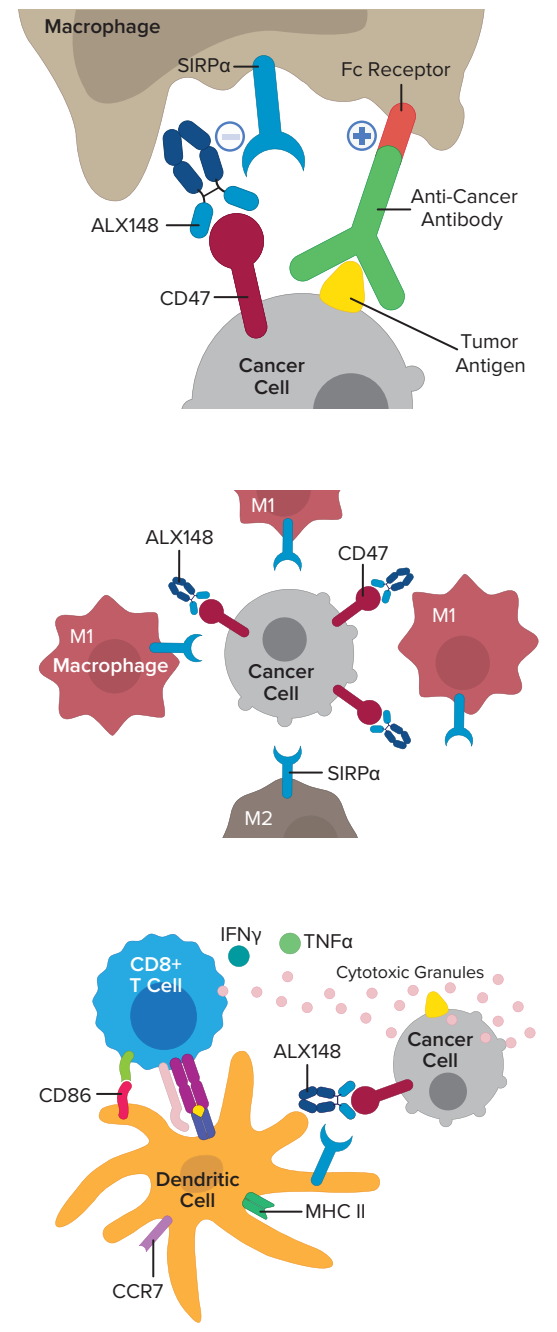
Macrophage
SIRPα
Fc Receptor
ALX148
Anti-Cancer Antibody
CD47
Cancer Cell
Tumor Antigen
M1
CD47
M1 Macrophage
M2
IFNγ
TNFα
Cytotoxic Granules
CD8+ T Cell
CD86
Dendritic Cell
MHC II
CCR7

1. **ALX148 Designed to Safely Maximize Anti-Cancer Antibody Activity**
2. **ALX148 Increases Ratio of Inflammatory M1 to Suppressive M2 in Tumor Microenvironment**
3. **ALX148 Activates Dendritic Cells and Enhances Cross-Priming of T Cells**

References

1. Kauder et al., ALX148 blocks CD47 and enhances innate and adaptive antitumor immunity with a favorable safety profile. *PLOS ONE*. 2018 13(8): e0201832.
2. Lakhani et al., A phase 1 study of ALX148, a CD47 blocker, alone and in combination with established anticancer antibodies in patients with advanced malignancy and non-Hodgkin lymphoma. *Journal of Clinical Oncology* 2018 36:15_suppl, 3068-3068.
3. Lakhani et al., A phase 1 study of ALX148: CD47 blockade in combination with anticancer antibodies to bridge innate and adaptive immune responses for advanced malignancy. *Journal for ImmunoTherapy of Cancer* 2018 6 (Suppl 1):114. Abstract 335.
4. Chow et al., A phase I study of ALX148, a CD47 blocker, in combination with established anticancer antibodies in patients with advanced malignancy. *Journal of Clinical Oncology* 2019 37:15_suppl, 2514-2514.

Acknowledgments

- We would like to thank all of the participating patients and their families as well as site research staff.
- **Contact email:** info@alxoncology.com
- Presented at the Society for Immunotherapy of Cancer (SITC) 34th Annual Meeting, National Harbor, MD, USA, Nov. 6-10, 2019

Methods

AT148001 Study Design

- **Part 1 (single agent):** No MTD reached, maximum administered dose 30 mg/kg QoW[2]
- **Part 2 (combination):** ALX148 (10 mg/kg QW) combined with standard regimens of pembrolizumab (200 mg IV Q3W) or trastuzumab (8 mg/kg IV → 6 mg/kg Q3W)

Table 1. ALX148 Combination Solid Tumor Cohorts

Combination Safety Confirmation		Combination Dose Expansion
ALX148 (10 mg/kg QW) + trastuzumab: Advanced malignancies (N=10)	➡	**ALX148 + trastuzumab: HER2 positive Gastric/GEJ (N=20)** *progressed on prior fluoropyrimidine/trastuzumab*
		ALX148 + pembrolizumab: HNSCC (N=20) *progressed on prior platinum*
ALX148 (10 mg/kg QW) + pembrolizumab: Advanced malignancies (N=12)		**ALX148 + pembrolizumab: NSCLC (N=20)** *progressed on prior CPI and/or PD-L1<50%*

- Patients with HER2-positive advanced malignancy (n=30), including gastric/GEJ cancers progressed on or after trastuzumab therapy, received ALX148 + trastuzumab. Patients with advanced malignancy (n=52,) including HNSCC progressed on platinum-based chemotherapy, as well as NSCLC [progressed on prior checkpoint inhibitor (CPI) or tumor proportion score (TPS) <50%] received ALX148 + pembrolizumab.
- Archival tumor tissue, on-treatment biopsies and whole blood samples were collected. CD8, CD68, CD163, PD-L1 and HER2 expression in tumor tissue were measured by immunohistochemistry (IHC) assays. Percent positive values for CD8, CD68 and CD163 were derived by image analysis. PD-L1 (Clone 22C3) TPS and combined positive score (CPS) were obtained by central pathologist review. HER2 levels were determined using HercepTest™.
- RNA expressions from paired tumor biopsies were assessed using NanoString IO360 Panel. Cell type abundance and pathway profiling analyses using pre-defined gene signatures were performed using NanoString nSolver Analysis Software.
- Peripheral CD47 Target Occupancy (TO) and immunophenotyping were measured by flow cytometry.

Results

Patient Baseline Characteristics

- 82 patients with advanced solid tumor malignancies have been enrolled into Part 2 combination cohorts (Table 2).

Table 2. Baseline Characteristics

	Pembrolizumab N=52	Trastuzumab N=30
Median Age, Years (range)	61 (32-81)	58 (45-79)
Sex, n		
F	23	9
M	29	21
Race, n		
White	34	13
Black	3	1
Native American	1	–
Asian	11	14
Other	3	2
ECOG PS, n		
0	16	8
1	36	22
Primary Disease, n		
NSCLC	26	–
HNSCC	20	–
Gastric/GEJ/Esophageal	–	25
Breast	–	2
Colorectal	2	–
Ovarian	2	1
Pancreatic	–	1
Peritoneal	1	–
Appendiceal	1	–
Urothelial	–	1

Note: Data Cutoff 18Apr2019.

Clinical Results

- ALX148 in combination with trastuzumab and pembrolizumab was well tolerated with no maximum tolerated dose reached. Treatment related adverse events (TRAE) were mostly of low grade and frequency and have been previously reported.[2-4]

Figure 3. ALX148 + Trastuzumab Clinical Activity in HER2 positive Gastric/GEJ Cancer Expansion Cohort

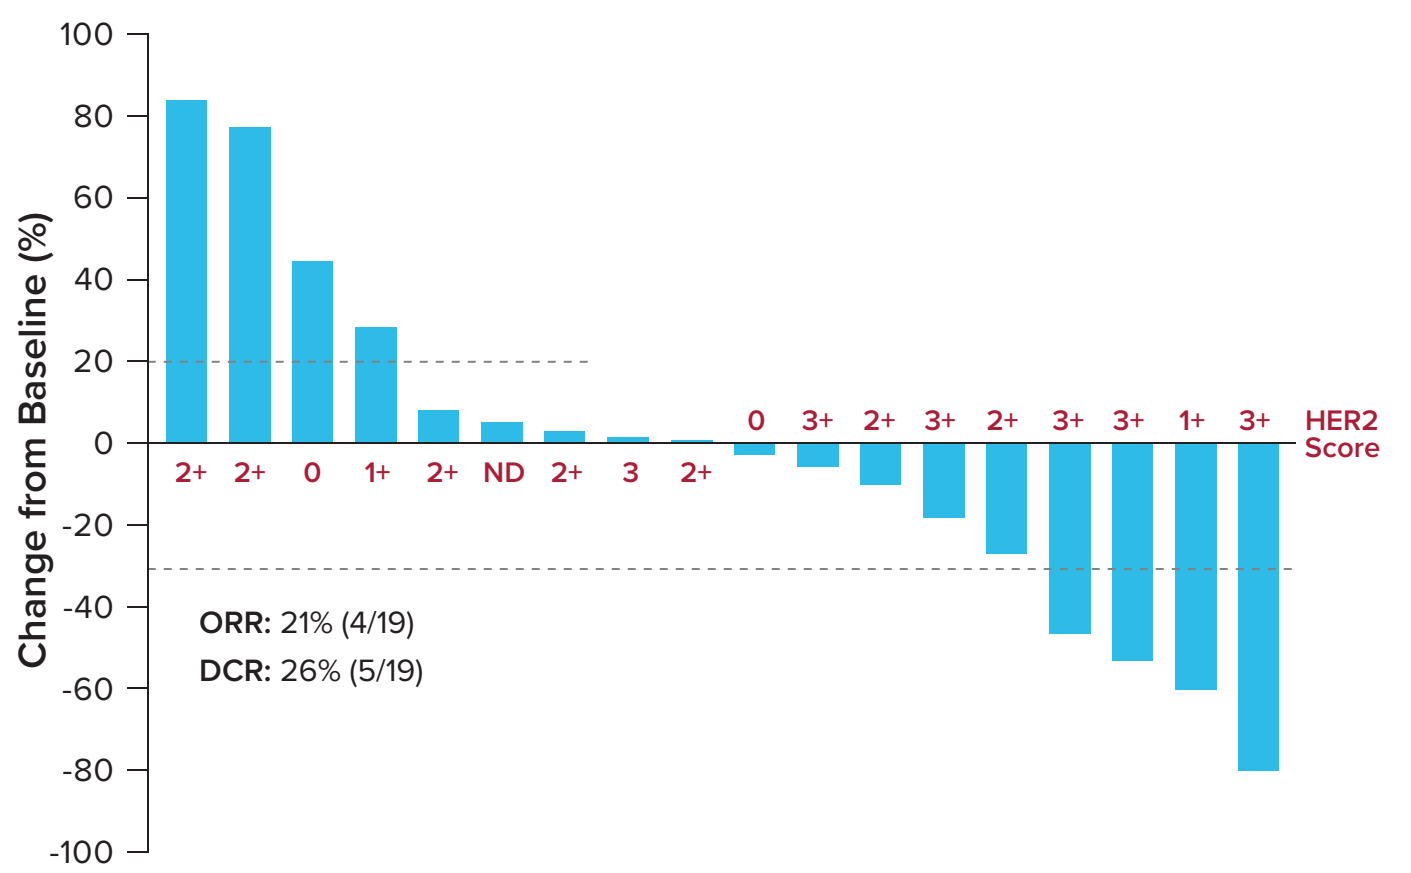
Change from Baseline (%)
2+ 2+ 0 1+ 2+ ND 2+ 3 2+
0 3+ 2+ 3+ 2+ 3+ 3+ 1+ 3+
HER2 Score
ORR: 21% (4/19)
DCR: 26% (5/19)

Note: Data Cutoff 23Sept2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment. One patient with clinical progression not included in plots; **ND** = Not Done; HER2 Score retrospectively assessed using archival tissue by a central IHC lab.

Figure 4. ALX148 + Pembrolizumab Clinical Activity in Advanced HNSCC Expansion Cohorts

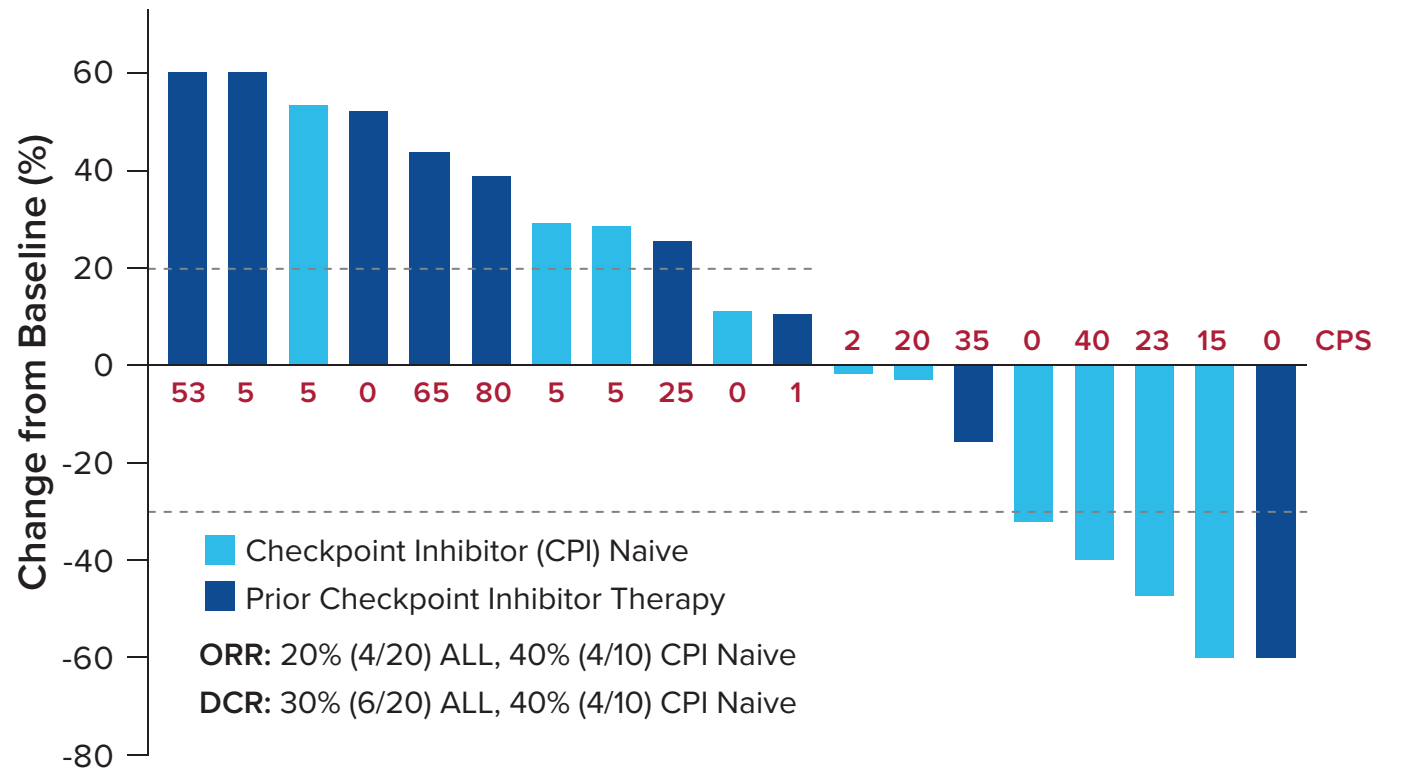
Change from Baseline (%)
53 5 5 0 65 80 5 5 25 0 1
2 20 35 0 40 23 15 0
CPS
Checkpoint Inhibitor (CPI) Naive
Prior Checkpoint Inhibitor Therapy
ORR: 20% (4/20) ALL, 40% (4/10) CPI Naive
DCR: 30% (6/20) ALL, 40% (4/10) CPI Naive

Notes: Data Cutoff 23Sept2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment; One patient with progressive disease not included in plots.

Figure 5. ALX148 + Pembrolizumab Clinical Activity in Advanced NSCLC Expansion Cohort

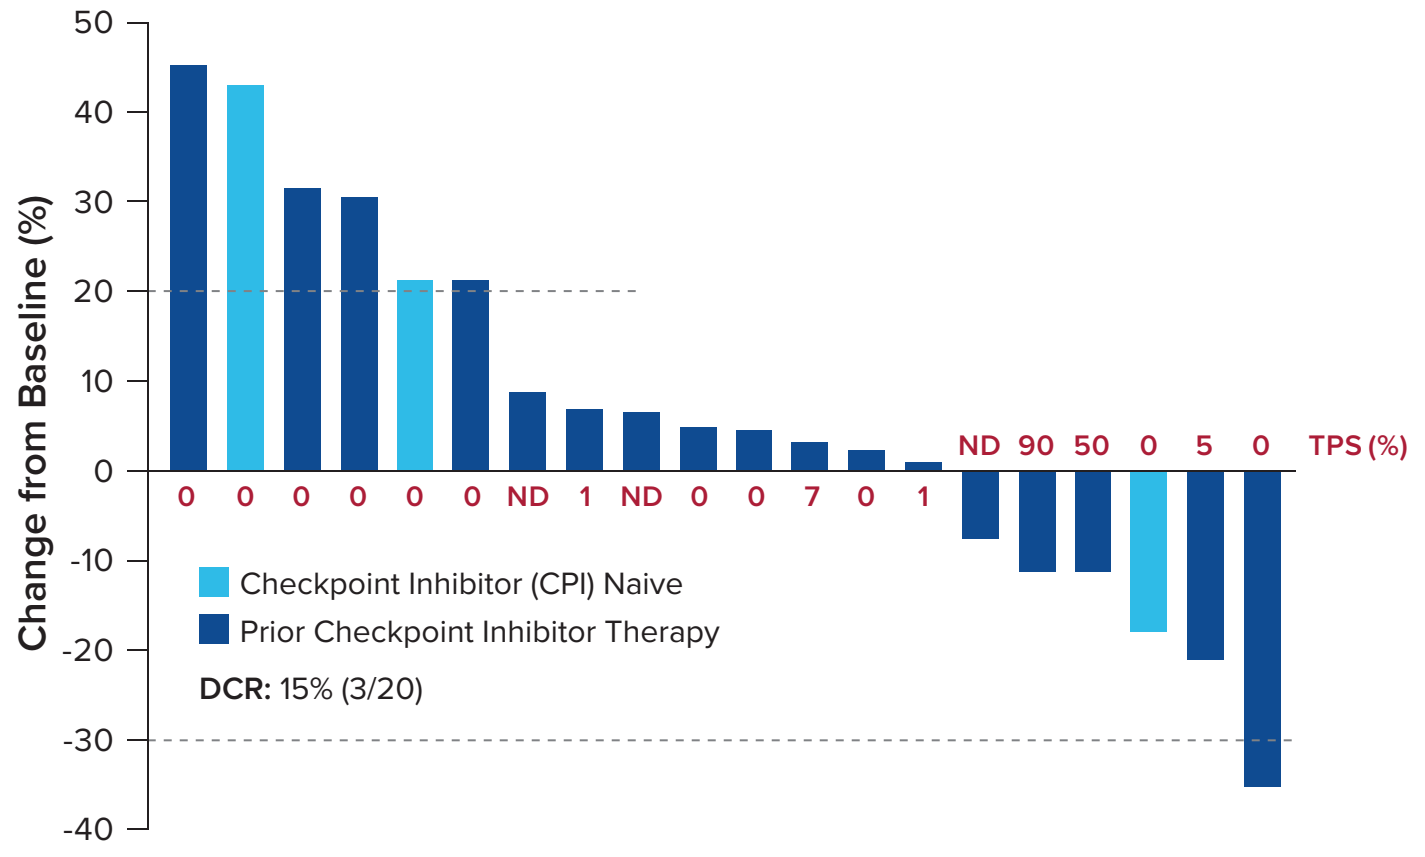
Change from Baseline (%)
0 0 0 0 0 0 ND 1 ND 0 0 7 0 1
ND 90 50 0 5 0
TPS (%)
Checkpoint Inhibitor (CPI) Naive
Prior Checkpoint Inhibitor Therapy
DCR: 15% (3/20)

Note: Data Cutoff 23Sept2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment.

Pharmacodynamic Results

Figure 6. Complete CD47 Target Occupancy by ALX148

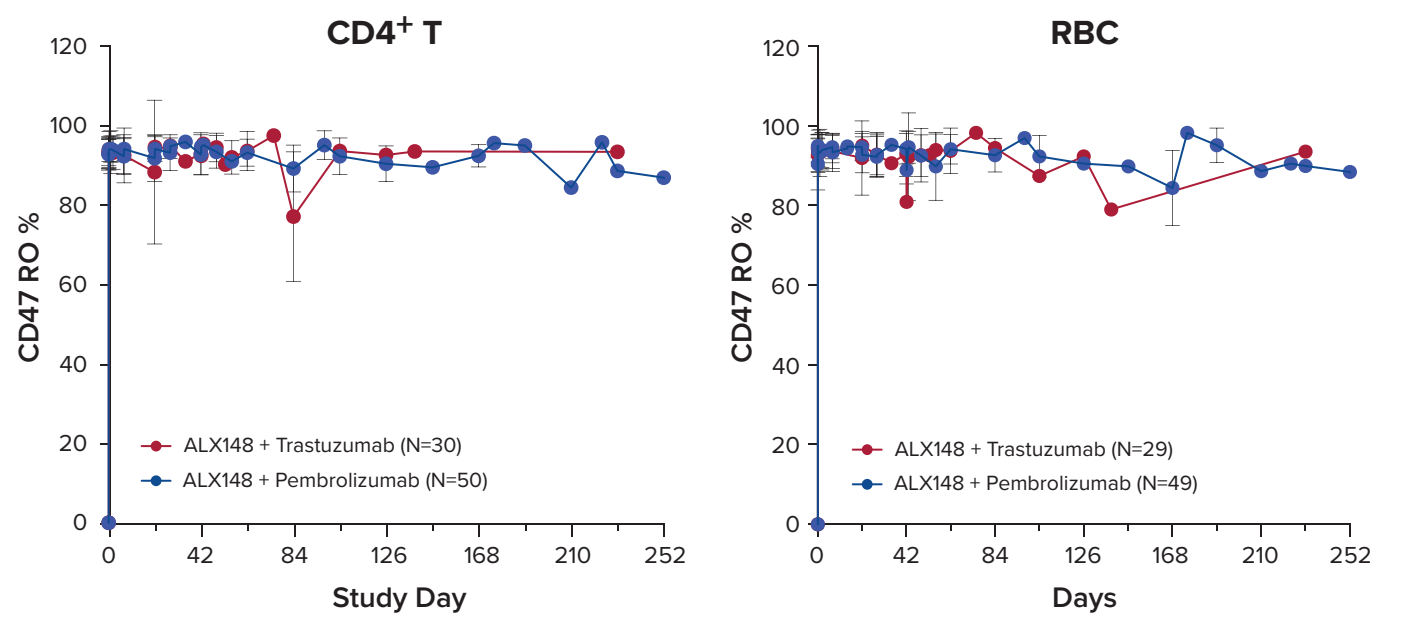
CD4+ T
RBC
CD47 RO %
ALX148 + Trastuzumab (N=30)
ALX148 + Pembrolizumab (N=50)
ALX148 + Trastuzumab (N=29)
ALX148 + Pembrolizumab (N=49)
Study Day
Days

Figure 7. No Changes of Circulating Immune Cell Profiles Following ALX148 Combinations

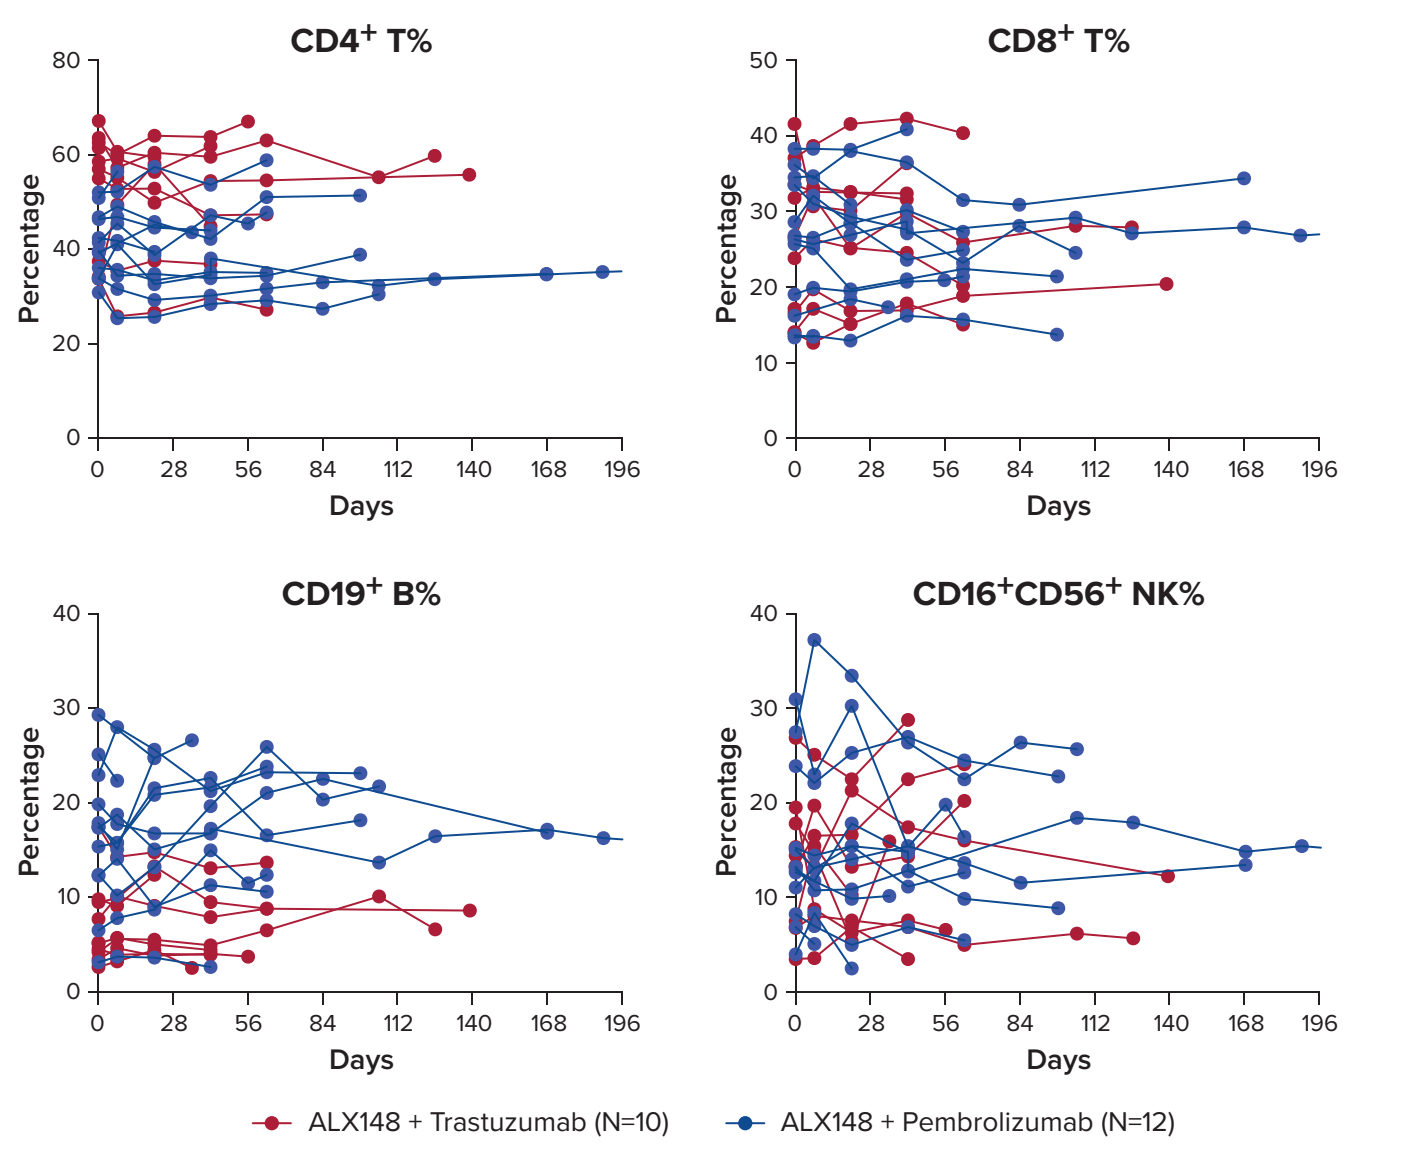
CD4+ T%
CD8+ T%
CD19+ B%
CD16+CD56+ NK%
Percentage
Days
ALX148 + Trastuzumab (N=10)
ALX148 + Pembrolizumab (N=12)

Figure 8. Increased Infiltrating Macrophages or Lymphocytes in Intratumoral Areas as Assessed by IHC Following ALX148 Combinations

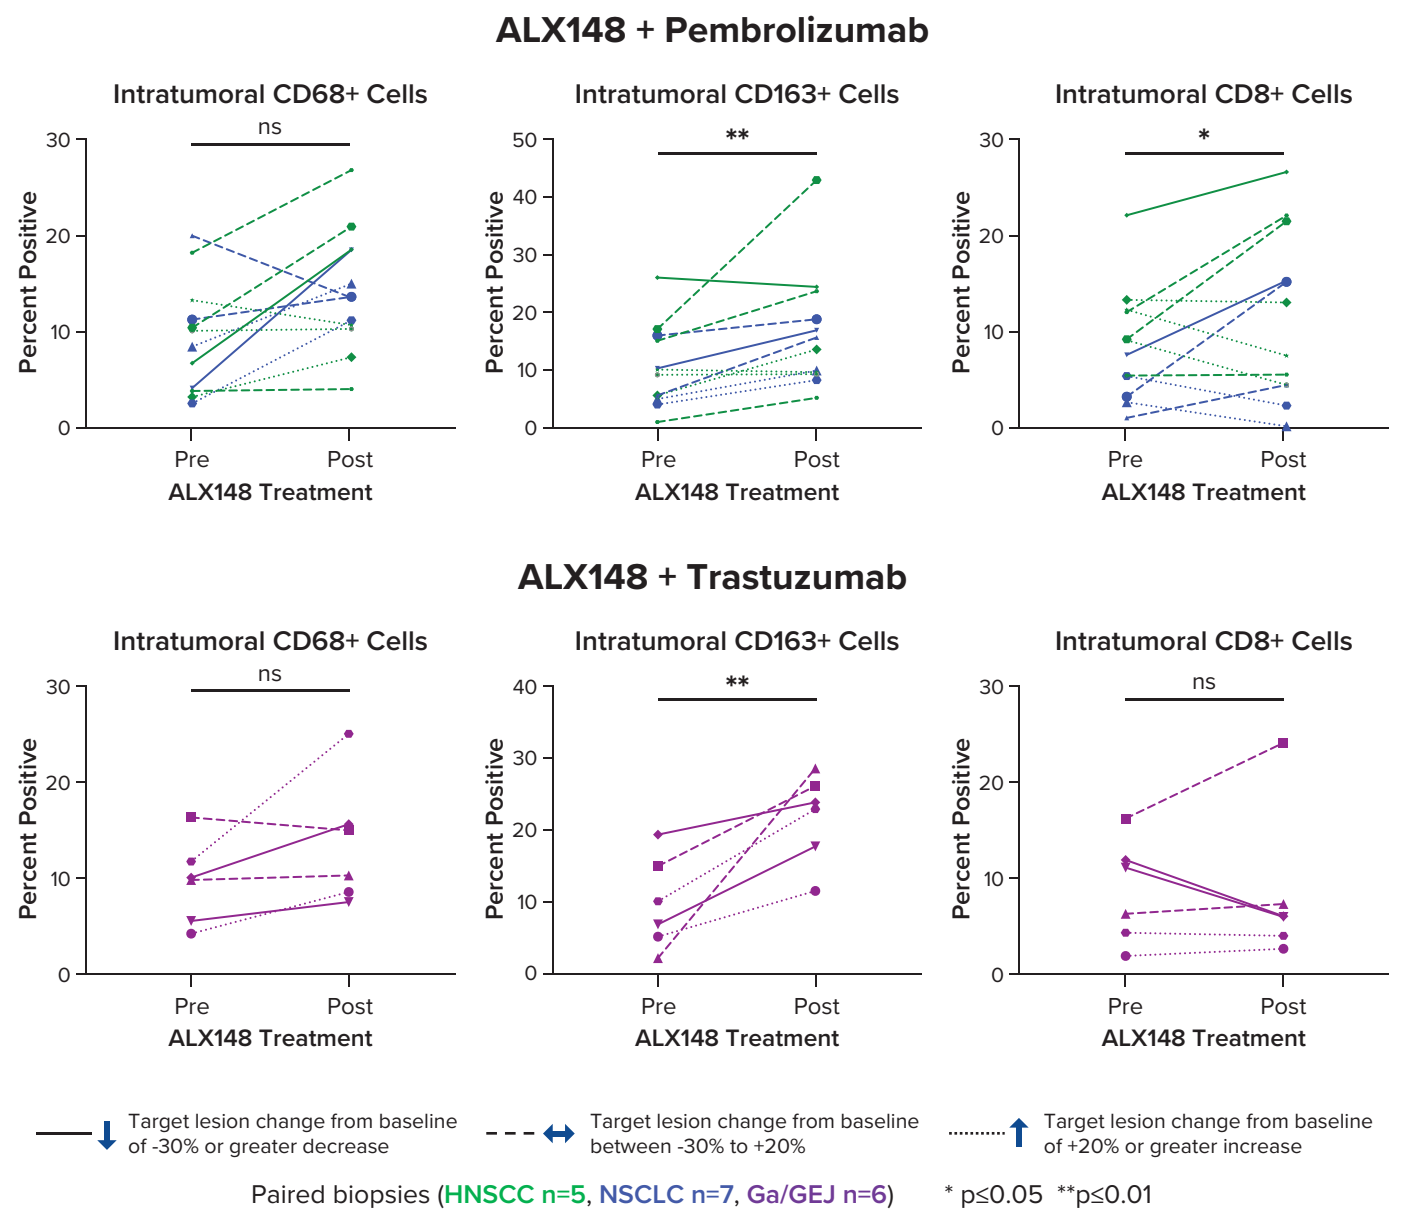
ALX148 + Pembrolizumab
Intratumoral CD68+ Cells
Intratumoral CD163+ Cells
Intratumoral CD8+ Cells
ALX148 + Trastuzumab
Percent Positive
ALX148 Treatment
Target lesion change from baseline of -30% or greater decrease
Target lesion change from baseline between -30% to +20%
Target lesion change from baseline of +20% or greater increase
Paired biopsies (HNSCC n=5, NSCLC n=7, Ga/GEJ n=6)
* p≤0.05 **p≤0.01

Figure 9. Effects of ALX148 Combinations on Immune Cell Subsets in Tumor Biopsies As Assessed by RNA Expression

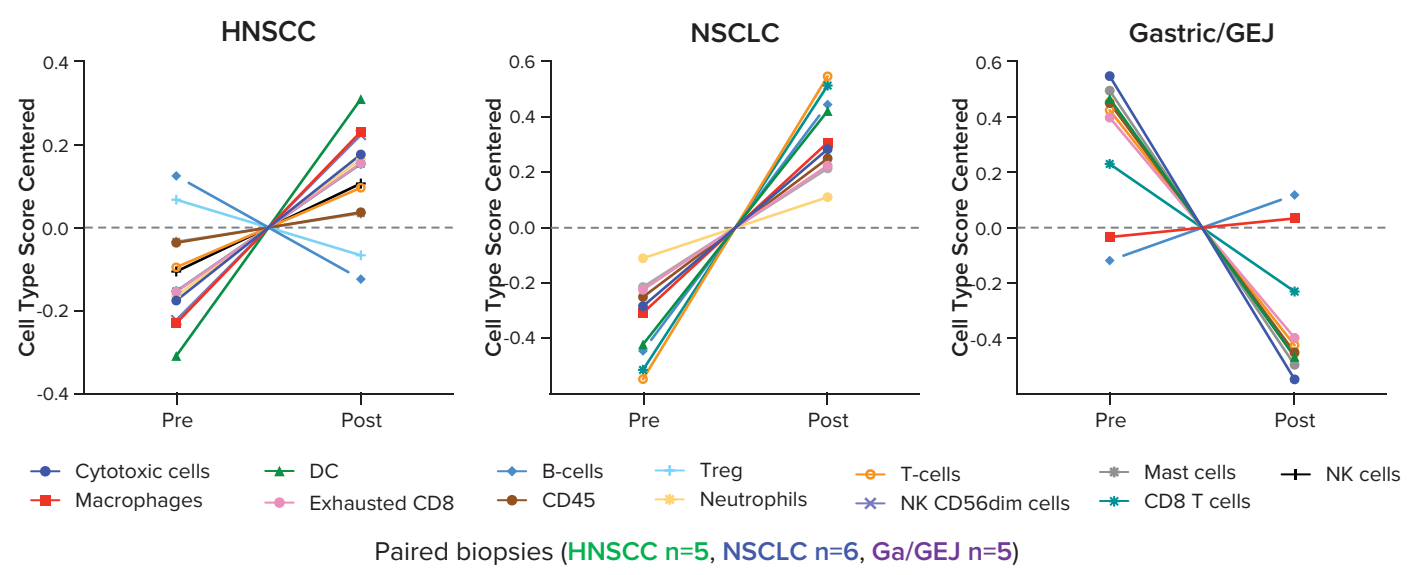
HNSCC
NSCLC
Gastric/GEJ
Cell Type Score Centered
Cytotoxic cells
Macrophages
DC
Exhausted CD8
B-cells
CD45
Treg
Neutrophils
T-cells
NK CD56dim cells
Mast cells
CD8 T cells
NK cells
Paired biopsies (HNSCC n=5, NSCLC n=6, Ga/GEJ n=5)

Figure 10. Effects of ALX148 Combinations on Immune Pathways in Tumor Biopsies As Assessed by RNA Expression

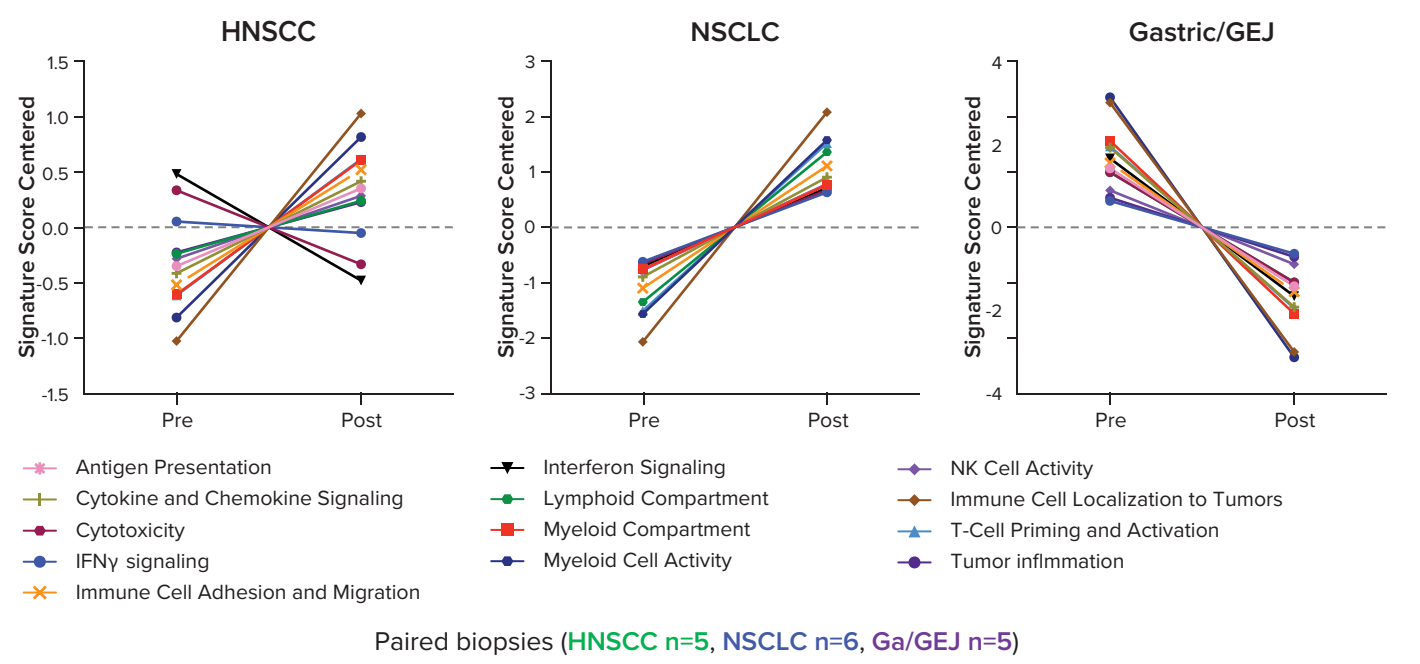
HNSCC
NSCLC
Gastric/GEJ
Signature Score Centered
Antigen Presentation
Cytokine and Chemokine Signaling
Cytotoxicity
IFNγ signaling
Immune Cell Adhesion and Migration
Interferon Signaling
Lymphoid Compartment
Myeloid Compartment
Myeloid Cell Activity
NK Cell Activity
Immune Cell Localization to Tumors
T-Cell Priming and Activation
Tumor inflmmation
Paired biopsies (HNSCC n=5, NSCLC n=6, Ga/GEJ n=5)

Figure 11. IHC and RNA Expression Results from Patient 1 HNSCC PD-L1 (+)

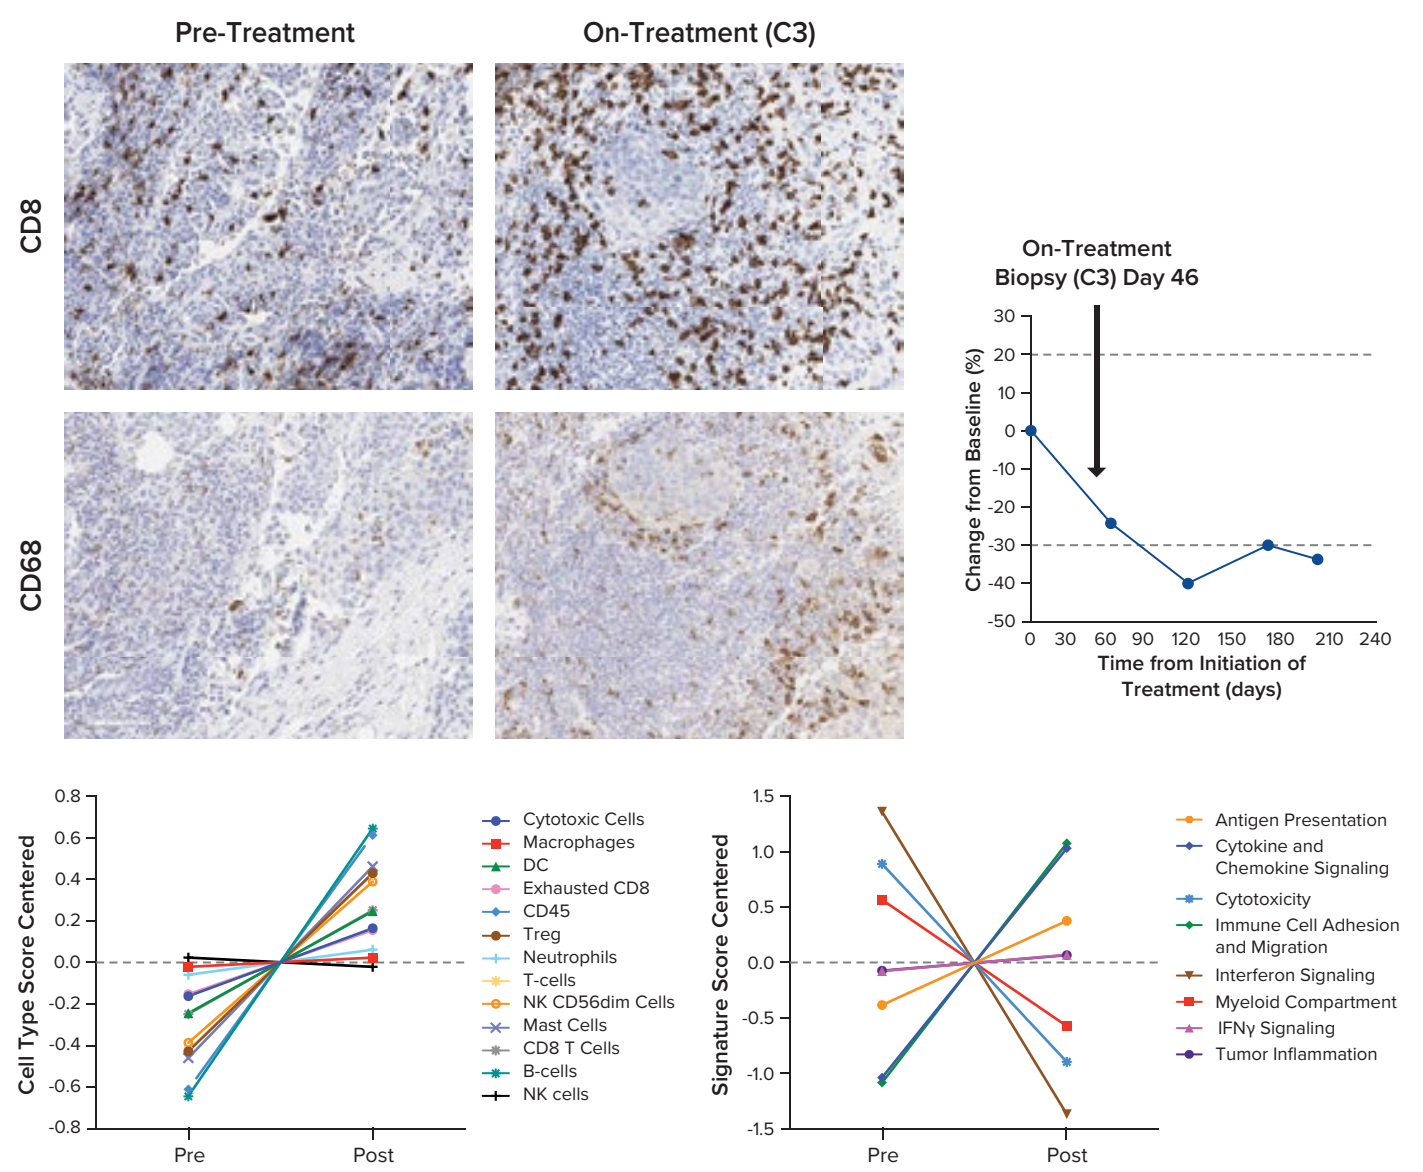
Pre-Treatment
On-Treatment (C3)
CD8
CD68
On-Treatment Biopsy (C3) Day 46
Change from Baseline (%)
Time from Initiation of Treatment (days)
Cell Type Score Centered
Signature Score Centered

Figure 12. IHC and RNA Expression Results from Patient 2 NSCLC PD-L1 (-)

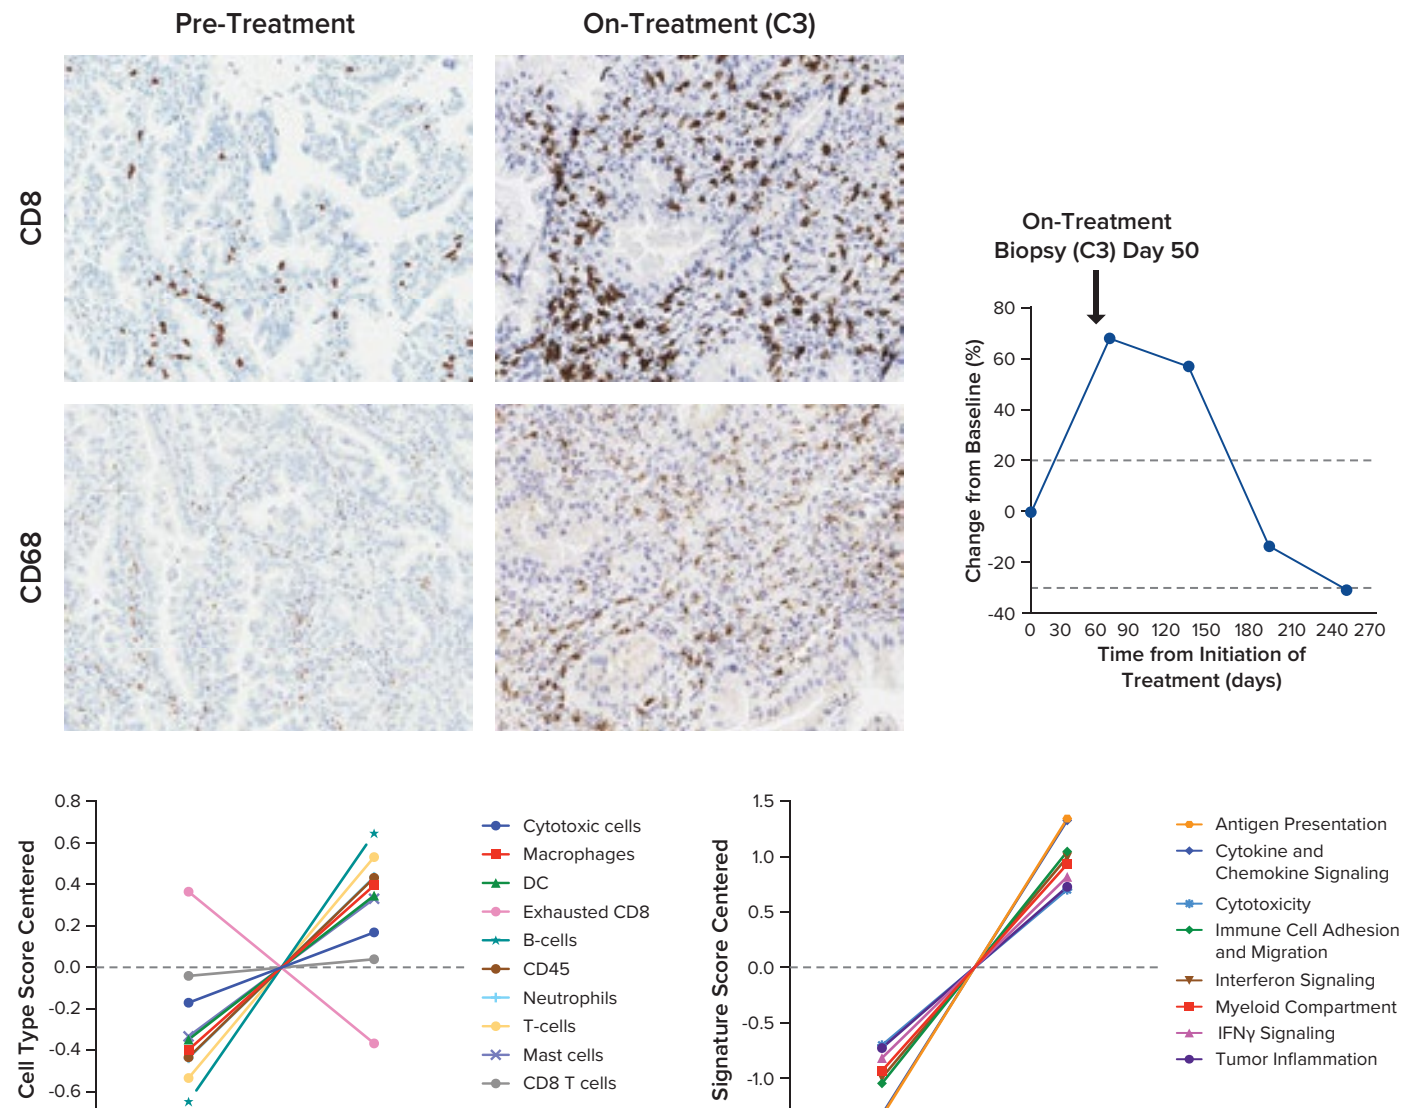
Pre-Treatment
On-Treatment (C3)
CD8
CD68
On-Treatment Biopsy (C3) Day 50
Change from Baseline (%)
Time from Initiation of Treatment (days)
Cell Type Score Centered
Signature Score Centered

Figure 13. IHC and RNA Expression Results from Patient 3 Gastric Cancer HER2 (3+)

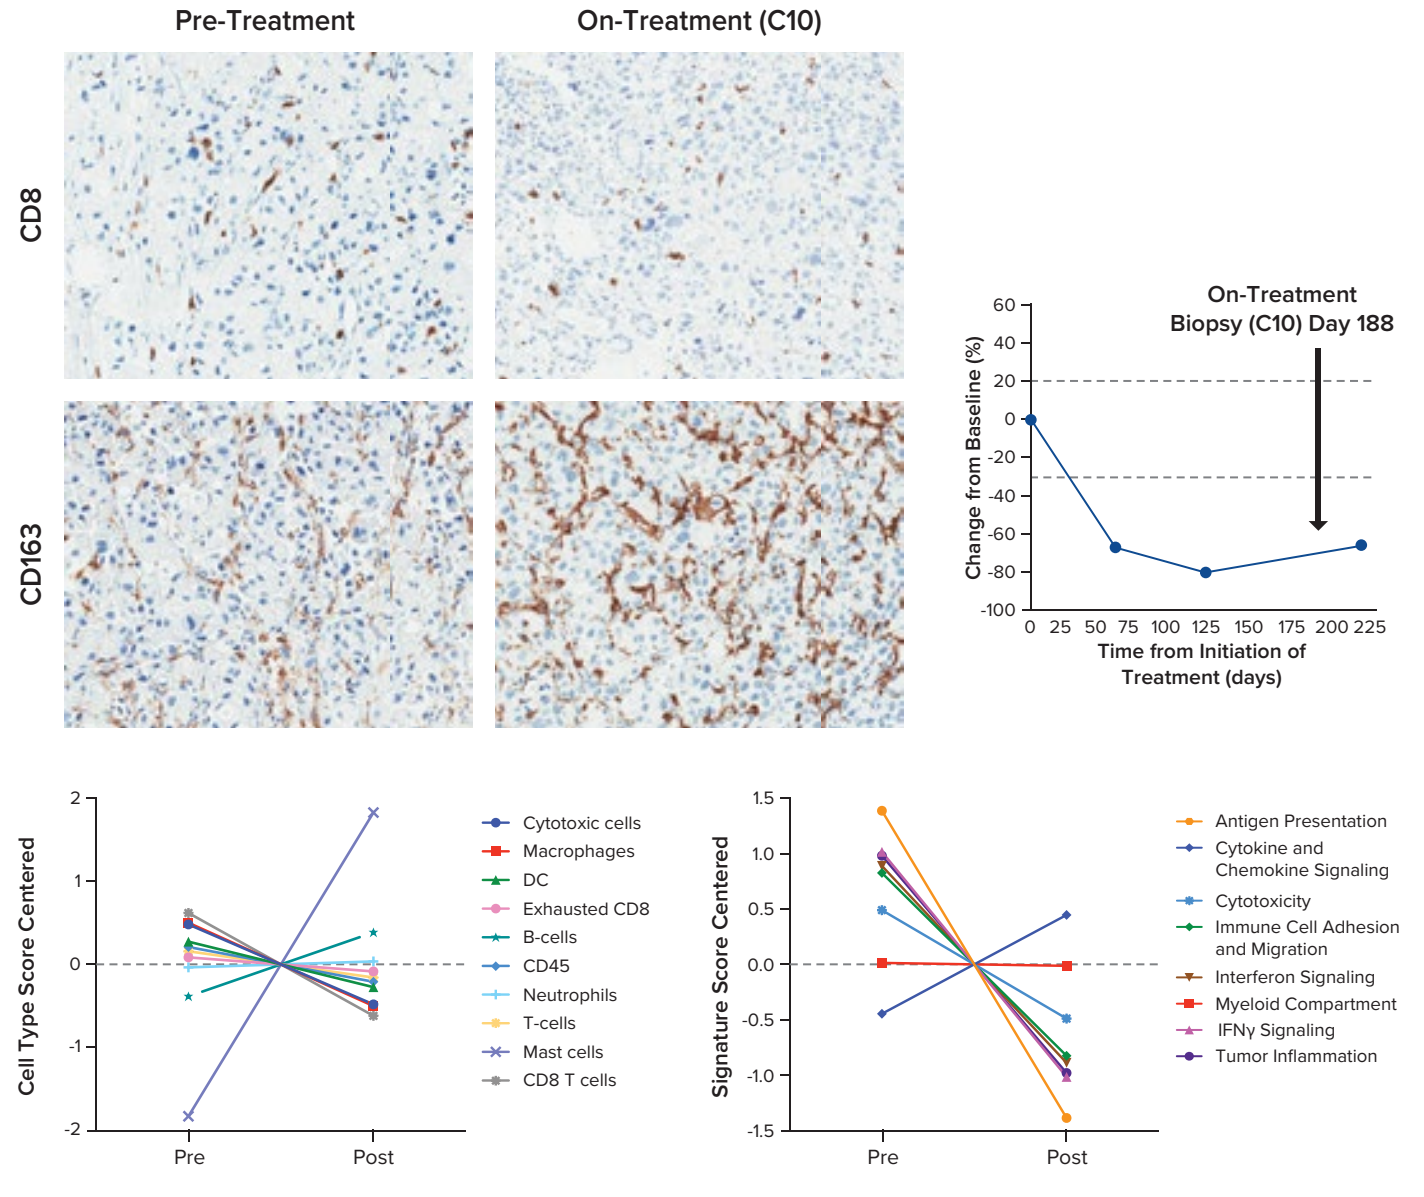
Pre-Treatment
On-Treatment (C10)
CD8
CD163
On-Treatment Biopsy (C10) Day 188
Change from Baseline (%)
Time from Initiation of Treatment (days)
Cell Type Score Centered
Signature Score Centered

Note: Mast cells signature for this sample was determined based on a partial gene list.

Conclusions

Intended for combination, ALX148 is designed to block the CD47-SIRPa myeloid checkpoint pathway, safely maximizing innate and adaptive immune response to cancer.

- ALX148 in combination with standard regimens of trastuzumab or pembrolizumab is well tolerated with a favorable hematologic safety profile.[2-4]
- ALX148 demonstrates emerging anti-cancer activity that compares favorably with anticipated activity of trastuzumab alone in patients with advanced gastric/GEJ cancer that has progressed on prior HER2-targeted therapies; ALX148 also demonstrates emerging anti-cancer activity that compares favorably with single agent pembrolizumab in patients with HNSCC that has progressed on prior systemic therapy.
- ALX148 at 10 mg/kg QW achieves complete CD47 target occupancy with no impact on circulating immune cells in combination with trastuzumab and pembrolizumab.
- Assessment of tumor infiltrating immune cells and molecular signatures in paired tumor biopsies reveals differences in pharmacodynamic changes following different ALX148 combinations, providing insights to ALX148's mechanism as a myeloid checkpoint inhibitor. Further studies are needed to confirm the relationship between pharmacodynamic changes and clinical responses.
 - ALX148 in combination with pembrolizumab showed increased immune infiltration of both innate and adaptive immune cells as well as increased inflammation signatures.
 - ALX148 in combination with trastuzumab showed increased macrophage infiltration without increased inflammation signatures.

Patients in combination cohorts continue to be followed (NCT03013218).