A Phase 1 Study of ALX148, a CD47 Blocker, in Combination with Established Anticancer Antibodies in Patients with Advanced Malignancy

Laura QM Chow[1], Justin Gainor[2], Nehal Lakhani[3], Hyun Chung[4], Keun-Wook Lee[5], Jeeyun Lee[6], Patricia LoRusso[7], Yung-Jue Bang[8], Stephen Hodi[9], Philip Fanning[10], Pierre Squifflet[11], Yonggang Zhao[10], Feng Jin[10], Hong Wan[10], Jaume Pons[10], Sophia Randolph[10], Wells Messersmith[12]

[1]University of Washington, Seattle, WA, USA; [2]Massachusetts General Hospital Cancer Center, Boston, MA, USA; [3]START Midwest, Grand Rapids, MI, USA; [4]Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Korea; [5]Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, Korea; [6]Samsung Medical Center, Seoul, Korea; [7]Yale Cancer Center, New Haven, CT, USA; [8]Seoul National University Hospital, Seoul, Korea; [9]Dana Farber Cancer Center, Boston, MA, USA; [10]ALX Oncology, Burlingame, CA, USA, and Dublin, Ireland; [11]International Drug Development Institute, Brussels, Belgium; [12]University of Colorado Cancer Center, Aurora, CO, USA.

Background

- CD47-SIRPα signaling is a myeloid checkpoint mechanism that signals the macrophage to ignore the cell on which CD47 is expressed.[1]
- Tumors upregulate CD47 to evade the immune response.
- ALX148 is a fusion protein with a high affinity CD47 blocker linked to an inactive human immunoglobulin Fc region (Figure 1). It blocks the CD47-mediated 'don't eat me' signal and enhances anti-tumor immunity.[2]
- AT148001, a first-in-human phase 1 study, evaluates ALX148 administered as a single agent (Part 1) and in combination with established anticancer antibodies (Part 2).

Figure 1. ALX148: A Unique High Affinity SIRPα Fusion Protein

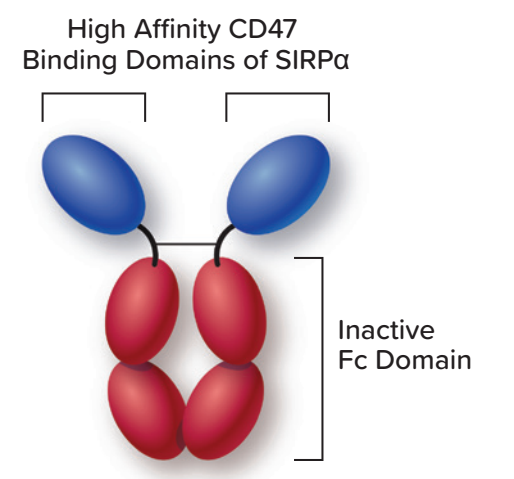
High Affinity CD47
Binding Domains of SIRPα
Inactive
Fc Domain

- Selectively binds CD47 to block its interaction with SIRPα with picomolar binding affinity.
- Fc domain is modified to eliminate binding to Fc gamma receptors to minimize toxicity.
- Molecular weight is half the size of typical antibody allowing twice the molar concentration to be delivered to tumor.
- Antibody-like pharmacokinetics.

Figure 2. ALX148 Bridges Innate and Adaptive Immunity[2]

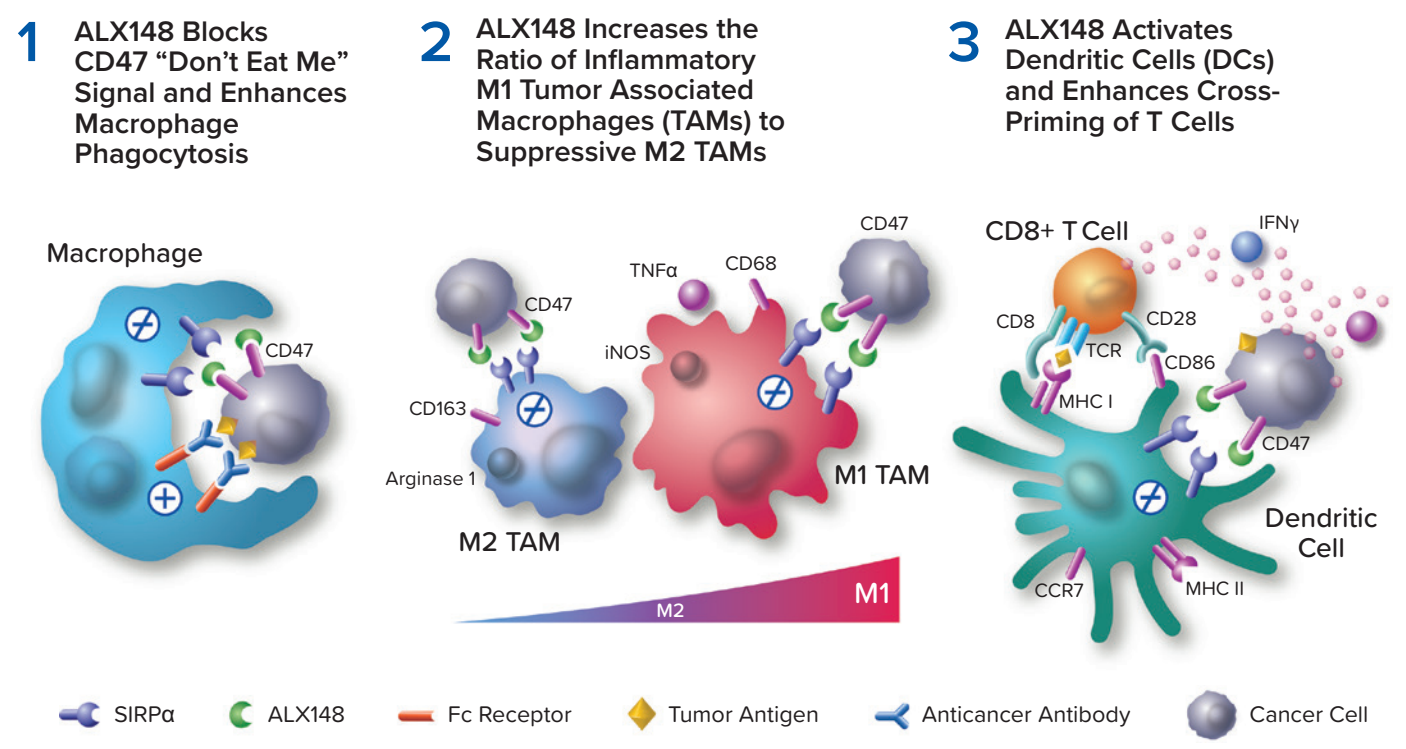
1 ALX148 Blocks CD47 "Don't Eat Me" Signal and Enhances Macrophage Phagocytosis
2 ALX148 Increases the Ratio of Inflammatory M1 Tumor Associated Macrophages (TAMs) to Suppressive M2 TAMs
3 ALX148 Activates Dendritic Cells (DCs) and Enhances Cross-Priming of T Cells
Macrophage
CD47
CD163
Arginase 1
M2 TAM
TNFα
CD68
iNOS
M1 TAM
M2
M1
CD8+ T Cell
IFNγ
CD8
CD28
TCR
CD86
MHC I
CCR7
MHC II
Dendritic Cell
SIRPα
ALX148
Fc Receptor
Tumor Antigen
Anticancer Antibody
Cancer Cell

Methods

AT148001 Study Design

- **Part 1 (single agent):** No MTD reached, maximum administered dose 30 mg/kg QOW.[3]
- **Part 2 (combination):** ALX148 (10 mg/kg QW) combined with standard regimens of trastuzumab (8 mg/kg IV → 6 mg/kg Q3W) or pembrolizumab (200 mg IV Q3W). Details below.

Table 1. ALX148 Combination Solid Tumor Cohorts

Combination Dose Confirmation		Combination Dose Expansion
ALX148 + trastuzumab Advanced Malignancies (N=10)	→	**ALX148 + trastuzumab: HER2 positive Gastric/GEJ (N=20)** *progressed on prior fluoropyrimidine and trastuzumab*
ALX148 + pembrolizumab Advanced Malignancies (N=12)		**ALX148 + pembrolizumab: HNSCC (N=20)** *progressed on prior platinum*
		ALX148 + pembrolizumab: NSCLC (N=20) *progressed on prior CPI and/or PD-L1<50%*

- **Primary study objective:** Characterize ALX148 safety profile as a single agent (Part 1) and in combination with established anticancer antibodies (Part 2).[3]
- ALX148 serum concentrations were analyzed using a validated ligand binding ELISA.
- CD47 target occupancy (TO) in peripheral blood T lymphocytes and erythrocytes was measured by flow cytometry.
- CD8, CD68, CD163 and PD-L1 on tumor tissue measured by immunohistochemistry (IHC) assays. Percent positive values for CD8, CD68 and CD163 were obtained by image analysis. PD-L1 (Clone 22C3) tumor proportion score (TPS) and combined positive score (CPS) were obtained by pathologist review.
- Here we report initial results from the fully enrolled ALX148 plus trastuzumab or pembrolizumab solid tumor combination cohorts as of April 18, 2019.

Results

Patient Baseline Characteristics

- 82 patients with advanced solid tumor malignancies have been enrolled into Part 2 combination cohorts (Table 2).

Table 2. Baseline Characteristics

	Trastuzumab N=30	Pembrolizumab N=52
Primary Disease, n		
Gastric/GEJ/Esophageal	25	–
HNSCC	–	20
NSCLC	–	26
Breast	2	–
Colorectal	–	2
Ovarian	1	2
Pancreatic	1	–
Peritoneal	–	1
Appendiceal	–	1
Urothelial	1	–
Median Age		
Years (range)	58 (45-79)	61 (32-81)
Sex, n		
F	9	23
M	21	29
Race, n		
White	13	34
Asian	14	11
Other	3	7
ECOG PS, n		
0	8	16
1	22	36

Safety

- ALX148 in combination with trastuzumab or pembrolizumab was well tolerated, and most treatment related adverse events (TRAE) were of low grade and frequency.
- The most common TRAE in combination with trastuzumab was Fatigue (26.7%), and with pembrolizumab was AST increased (15.4%). TRAEs ≥Grade 3 severity were of low frequency (Tables 3 and 4).
- One treatment related serious adverse event of febrile neutropenia in combination with trastuzumab was reported. Three treatment related serious adverse events in combination with pembrolizumab (1 autoimmune hemolytic anemia, 1 febrile neutropenia, and 1 neutropenia) were reported.

Treatment Related Adverse Events*

Table 3. ALX148 + Trastuzumab (N=30)

Adverse Event	Total n (%)	≥Grade 3
Fatigue	8 (26.7)	–
Platelets Decreased	4 (13.3)	2 (6.7)
Decreased Appetite	3 (10)	–
Pyrexia	3 (10)	–
Anemia	2 (6.7)	–
Nausea	2 (6.7)	–
Neutropenia	2 (6.7)	2 (6.7)

Table 4. ALX148 + Pembrolizumab (N=52)

Adverse Event	Total n (%)	≥Grade 3
AST Increased	8 (15.4)	–
ALT Increased	7 (13 .5)	1 (1.9)
Fatigue	6 (11.5)	–
Pruritus	5 (9.6)	–
Anemia	4 (7.7)	1 (1.9)
Infusion Reaction	4 (7.7)	–
Platelets Decreased	4 (7.7)	2 (3.8)
Alkaline Phosphatase Increased	3 (5.7)	–
Arthralgia	3 (5.8)	–
Pyrexia	3 (5.8)	–
WBC Decreased	3 (5.8)	–
Decreased Appetite	2 (3.8)	–
Myalgia	2 (3.8)	–
Nausea	2 (3.8)	–
Neutropenia	2 (3.8)	1 (1.9)
Rash	2 (3.8)	–

* Data cutoff April 18, 2019; Events occurring in ≥2 patients.

Response

ALX148 Combination Clinical Activity

- **Dose Confirmation Cohorts (N=22)**
 - **ALX148 + trastuzumab (n=10):** 8 evaluable patients – 3SD (2 Breast, 1 GEJ)
 - **ALX148 + pembrolizumab (n=12):** 10 evaluable patients – 1PR confirmed (NSCLC; CPI refractory); 3SD (1 Appendiceal, 2 NSCLC)
- **Dose Expansion Cohorts (N=60)**
 - **ALX148 + trastuzumab, HER2 positive Gastric/GEJ (n=20):** 18 evaluable patients – 4PR (confirmed), 5SD
 - **ALX148 + pembrolizumab, HNSCC (n=20):** 19 evaluable patients – 3PR (2 confirmed, 1 unconfirmed), 6SD
 - **ALX148 + pembrolizumab, NSCLC (n=20):** 18 evaluable patients – 8SD

Figure 3. ALX148 + Trastuzumab Clinical Activity in HER2 positive Gastric/GEJ Cancer Expansion Cohort

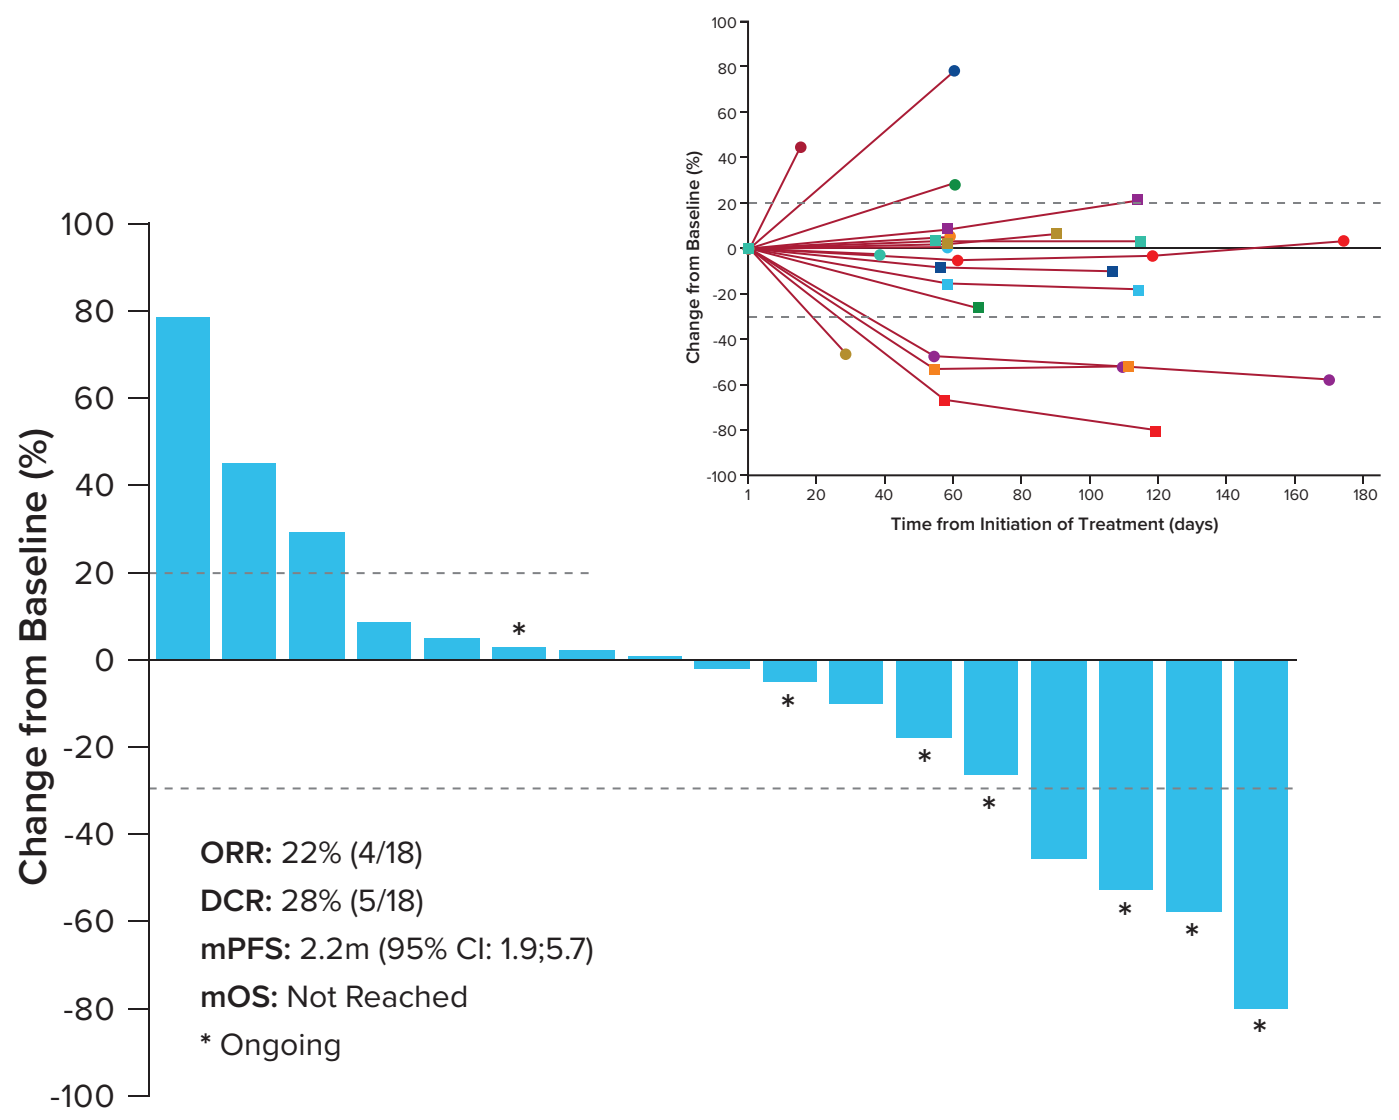
Change from Baseline (%)
ORR: 22% (4/18)
DCR: 28% (5/18)
mPFS: 2.2m (95% CI: 1.9;5.7)
mOS: Not Reached
* Ongoing
Time from Initiation of Treatment (days)

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment.

Figure 4. Duration of Treatment in Patients with Gastric/GEJ Cancer

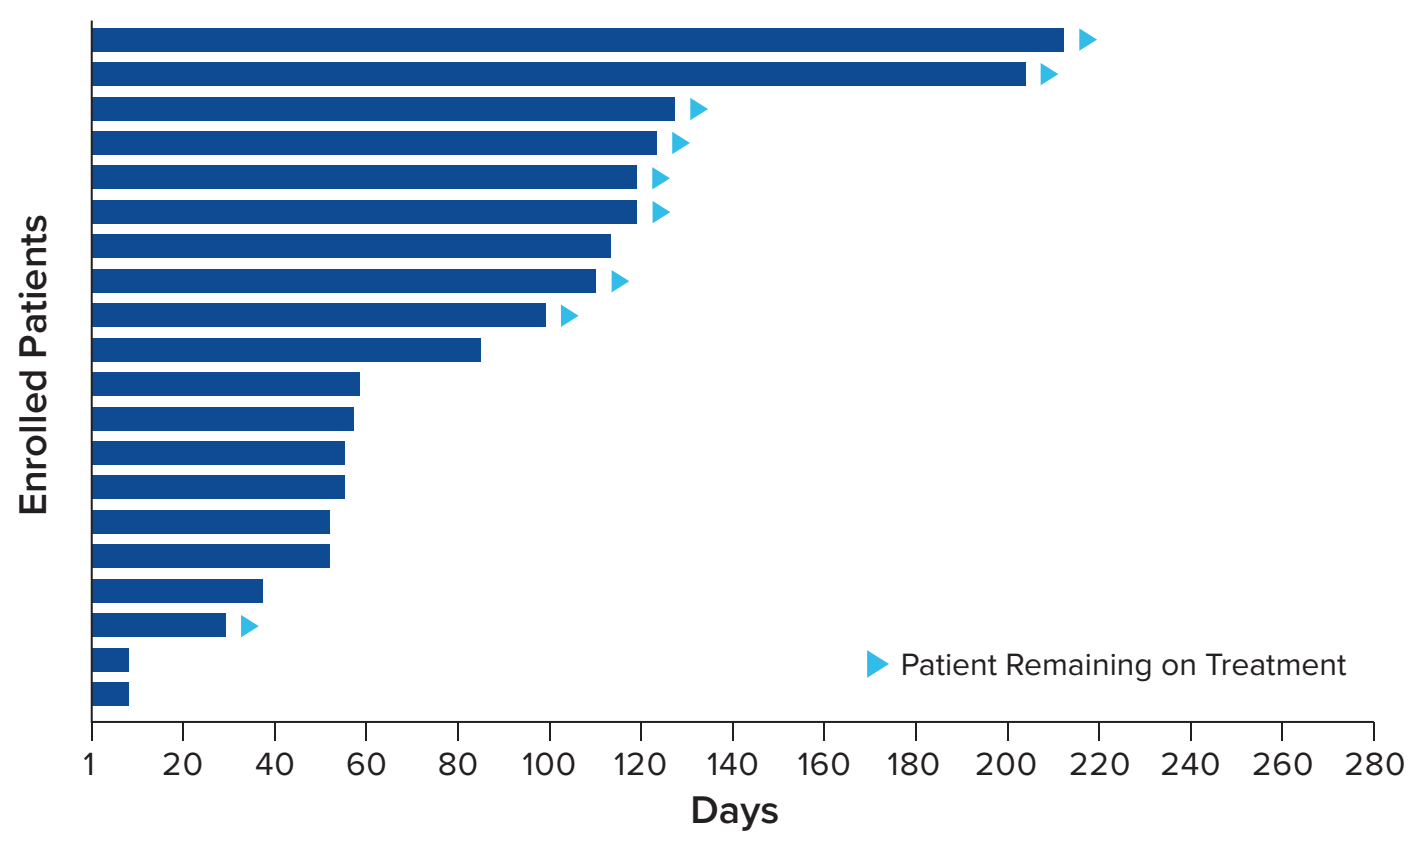
Enrolled Patients
Patient Remaining on Treatment
Days

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase.

Figure 5. ALX148 + Pembrolizumab Clinical Activity in HNSCC Expansion Cohort

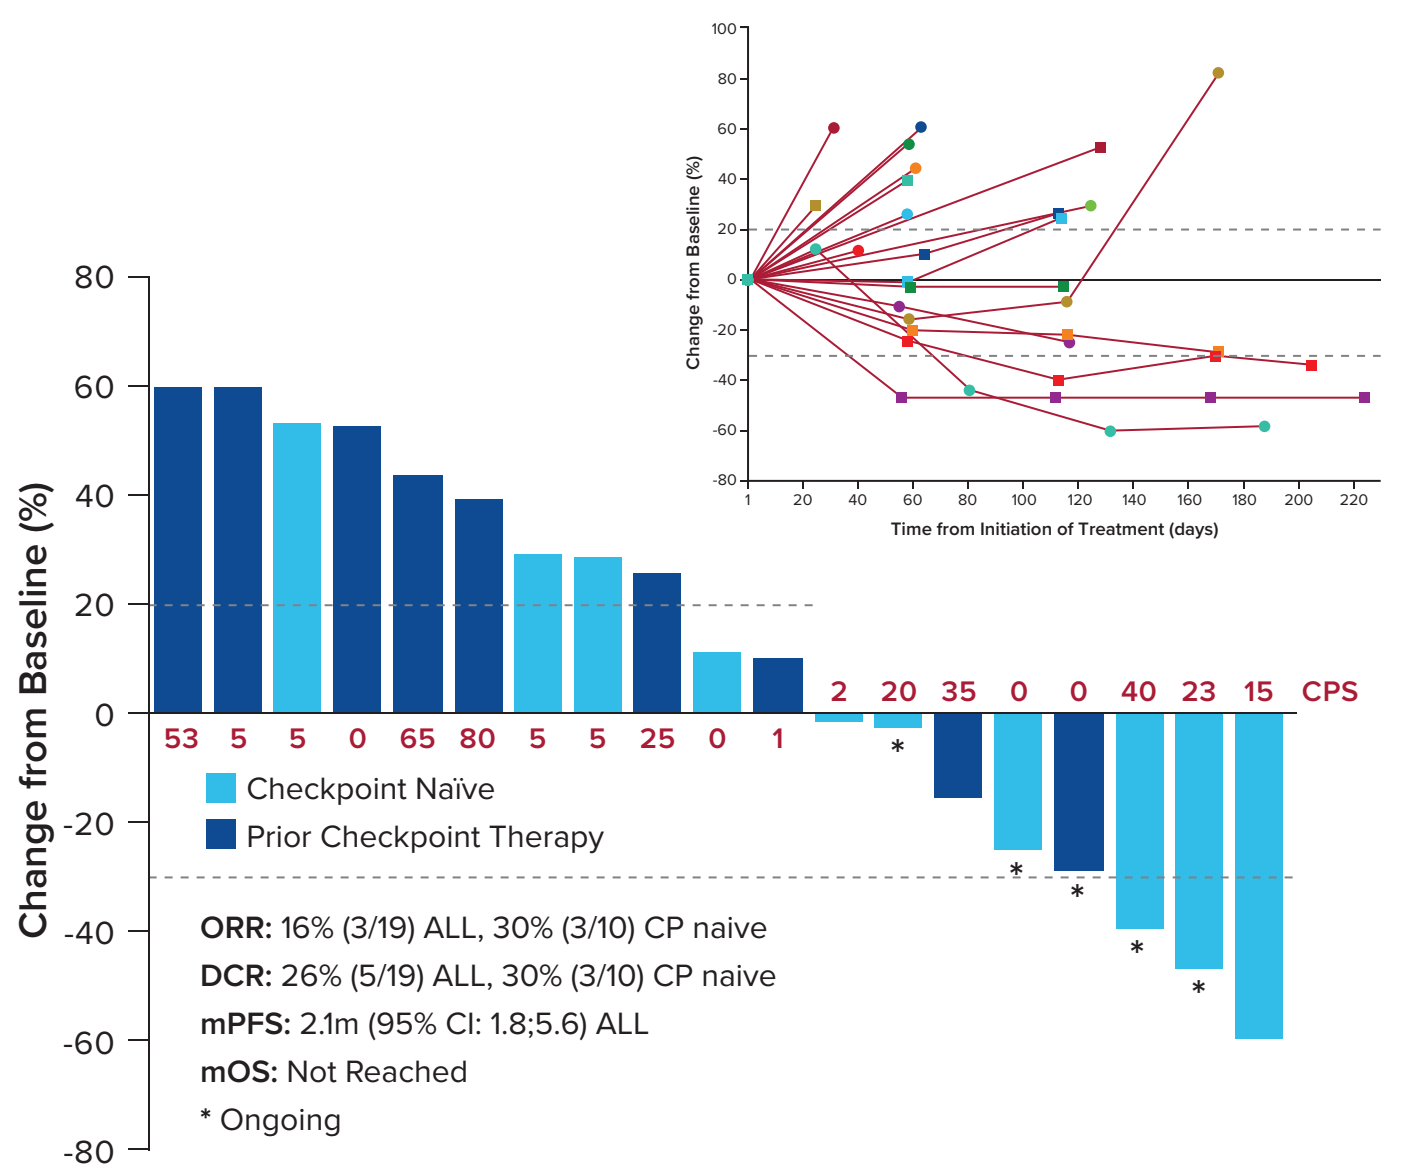
Change from Baseline (%)
53 5 5 0 65 80 5 5 25 0 1 2 20 35 0 0 40 23 15 CPS
Checkpoint Naïve
Prior Checkpoint Therapy
ORR: 16% (3/19) ALL, 30% (3/10) CP naive
DCR: 26% (5/19) ALL, 30% (3/10) CP naive
mPFS: 2.1m (95% CI: 1.8;5.6) ALL
mOS: Not Reached
* Ongoing
Time from Initiation of Treatment (days)

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment.

Figure 6. Duration of Treatment in Patients with HNSCC

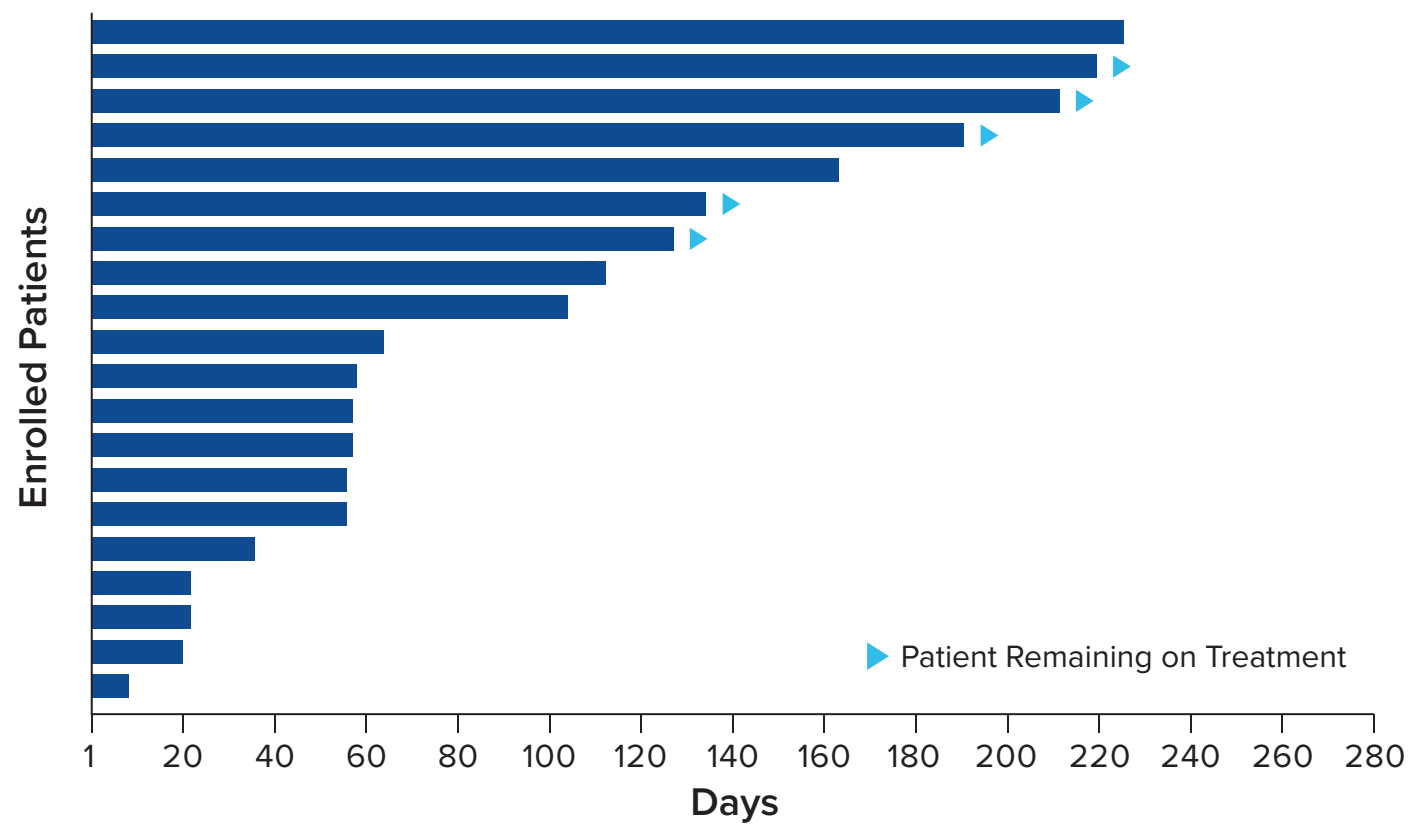
Enrolled Patients
Patient Remaining on Treatment
Days

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase.

Figure 7. ALX148 + Pembrolizumab Clinical Activity in NSCLC Expansion Cohort

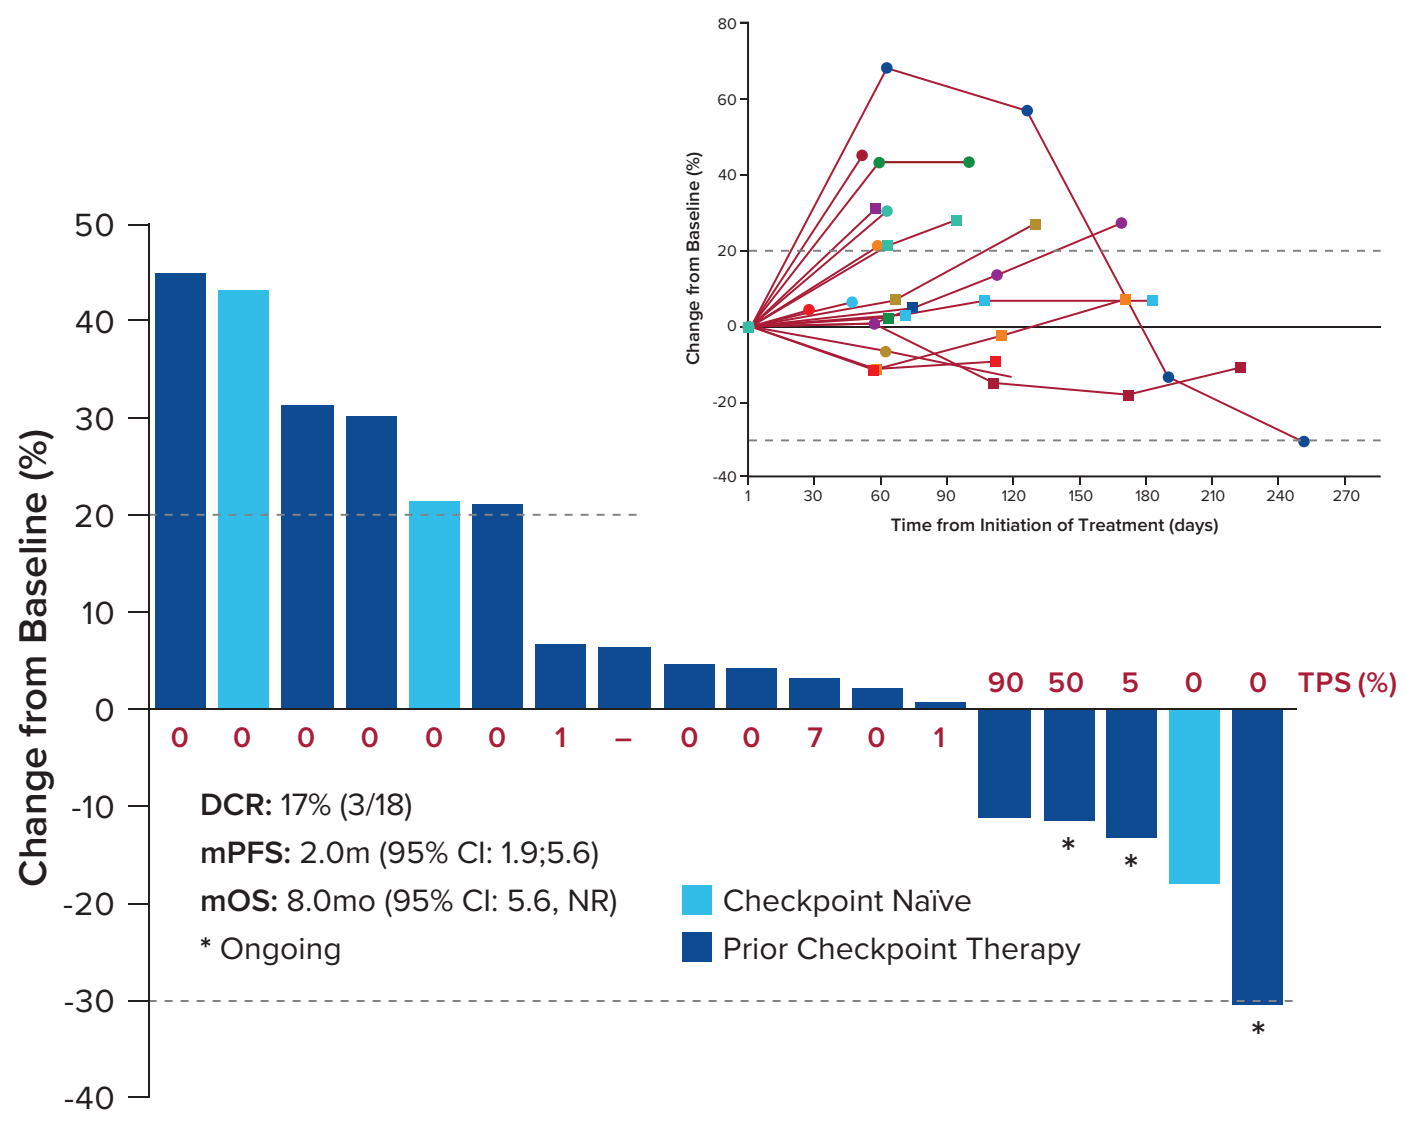
Change from Baseline (%)
0 0 0 0 0 0 1 – 0 0 7 0 1 90 50 5 0 0 TPS (%)
DCR: 17% (3/18)
mPFS: 2.0m (95% CI: 1.9;5.6)
mOS: 8.0mo (95% CI: 5.6, NR)
* Ongoing
Checkpoint Naïve
Prior Checkpoint Therapy
Time from Initiation of Treatment (days)

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase, had a baseline assessment, and at least one post-baseline disease assessment.

Figure 8. Duration of Treatment in Patients with NSCLC Cancer

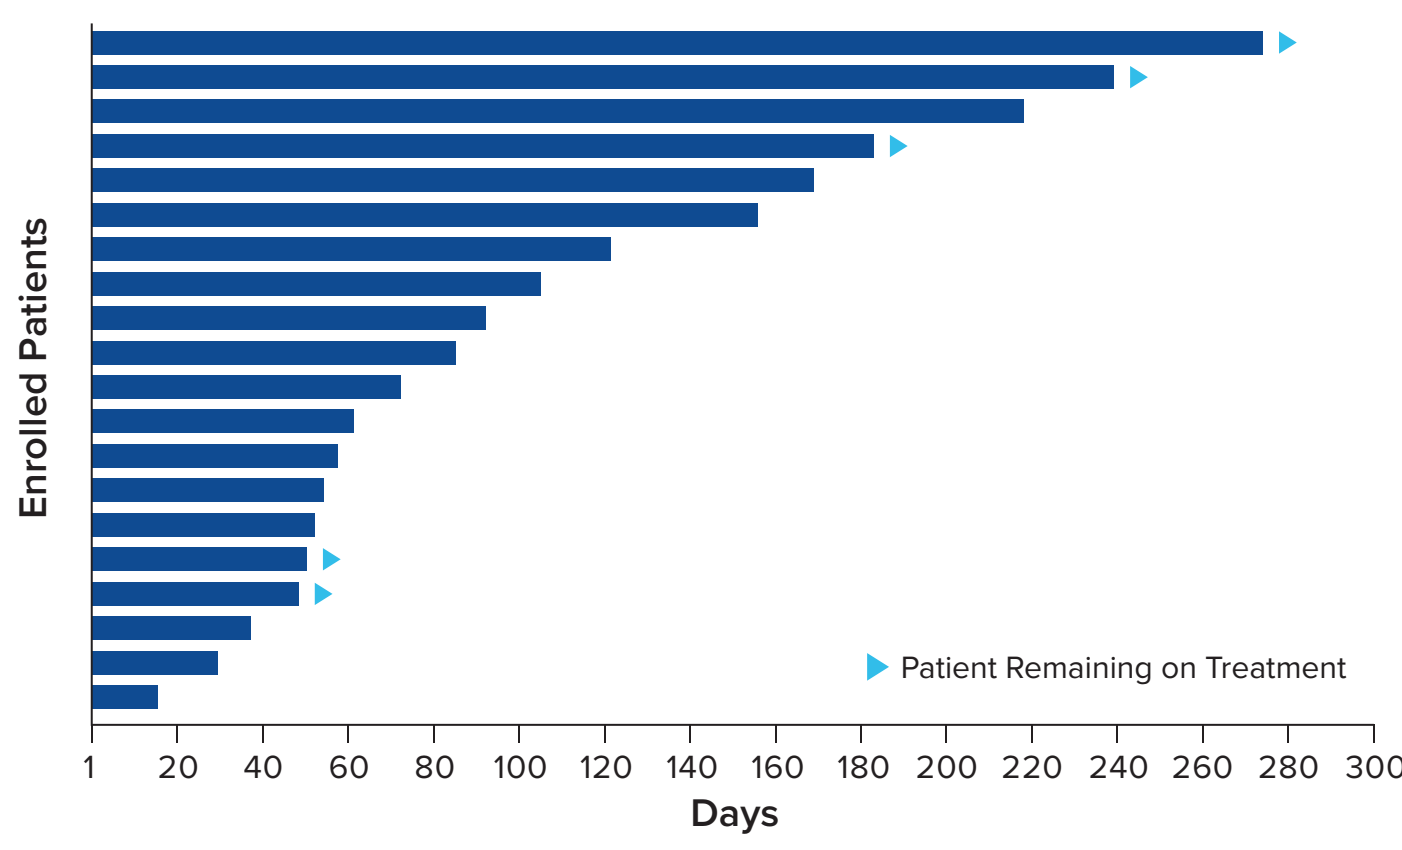
Enrolled Patients
Patient Remaining on Treatment
Days

Data cutoff April 18, 2019. Patients who received at least one dose of ALX148 in the expansion phase.

Figure 9. ALX148 PK and CD47 Target Occupancy from Combination Cohorts

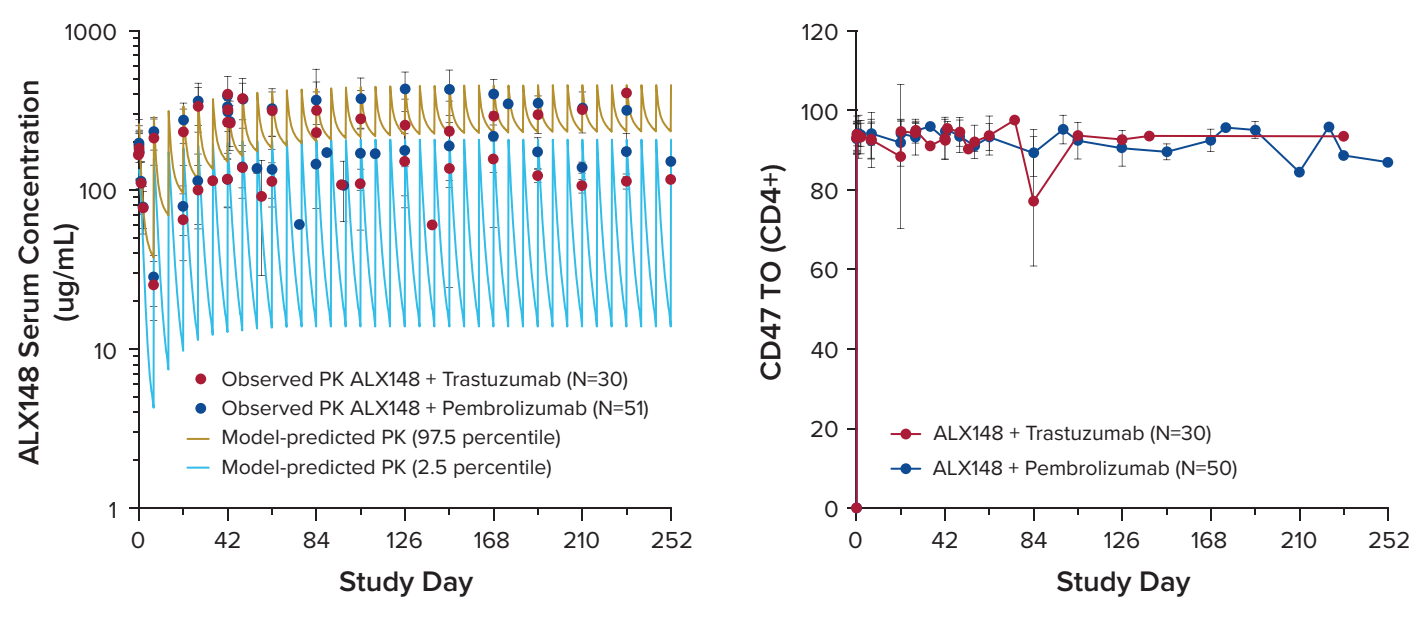
ALX148 Serum Concentration (ug/mL)
Observed PK ALX148 + Trastuzumab (N=30)
Observed PK ALX148 + Pembrolizumab (N=51)
Model-predicted PK (97.5 percentile)
Model-predicted PK (2.5 percentile)
Study Day
CD47 TO (CD4+)
ALX148 + Trastuzumab (N=30)
ALX148 + Pembrolizumab (N=50)
Study Day

ALX148 PK time points included intensive sampling in cycle 1 and 3, pre-dose and end-of-infusion only in every cycle thereafter.

- ALX148 PK observations from combination cohorts are within predicted 95% intervals based on established population PK model.[4]
- Steady-state half-life of ALX148 (10 mg/kg QW) is predicted to be ~16 days.
- Near complete CD47 target occupancy is maintained throughout ALX148 dosing interval.

Increased Tumor Associated Macrophages and Infiltrating Lymphocytes Seen After Treatment with ALX148 Combinations

Figure 10. IHC Results from Paired Biopsies (4 NSCLC, 6 HNSCC, 1 Gastric and 1 Esophageal Ca)

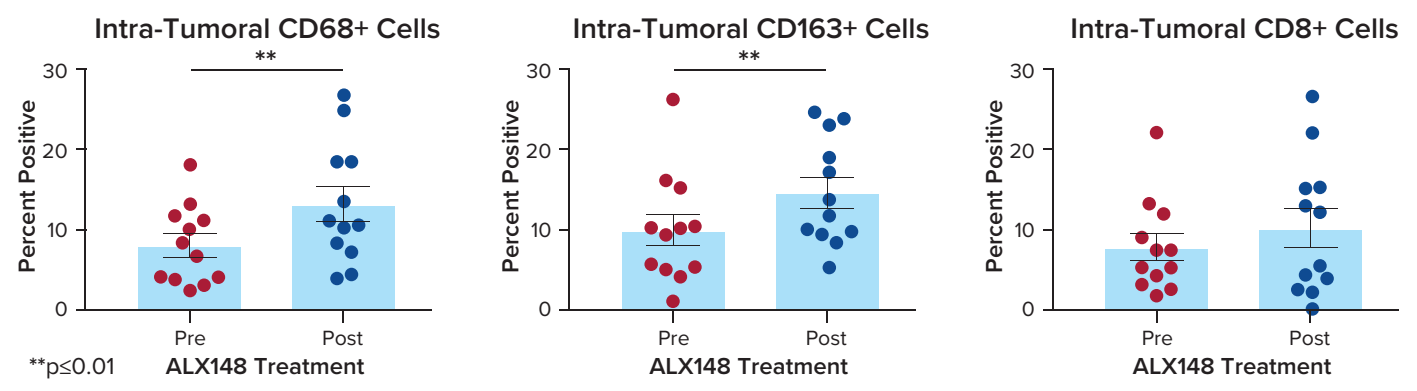
Intra-Tumoral CD68+ Cells
Intra-Tumoral CD163+ Cells
Intra-Tumoral CD8+ Cells
Percent Positive
Pre
Post
ALX148 Treatment
**p≤0.01

Figure 11. Increased CD68+ Macrophages and CD8+ T Cells Seen in Intra-Tumoral and Peri-Tumoral Areas Following Treatment

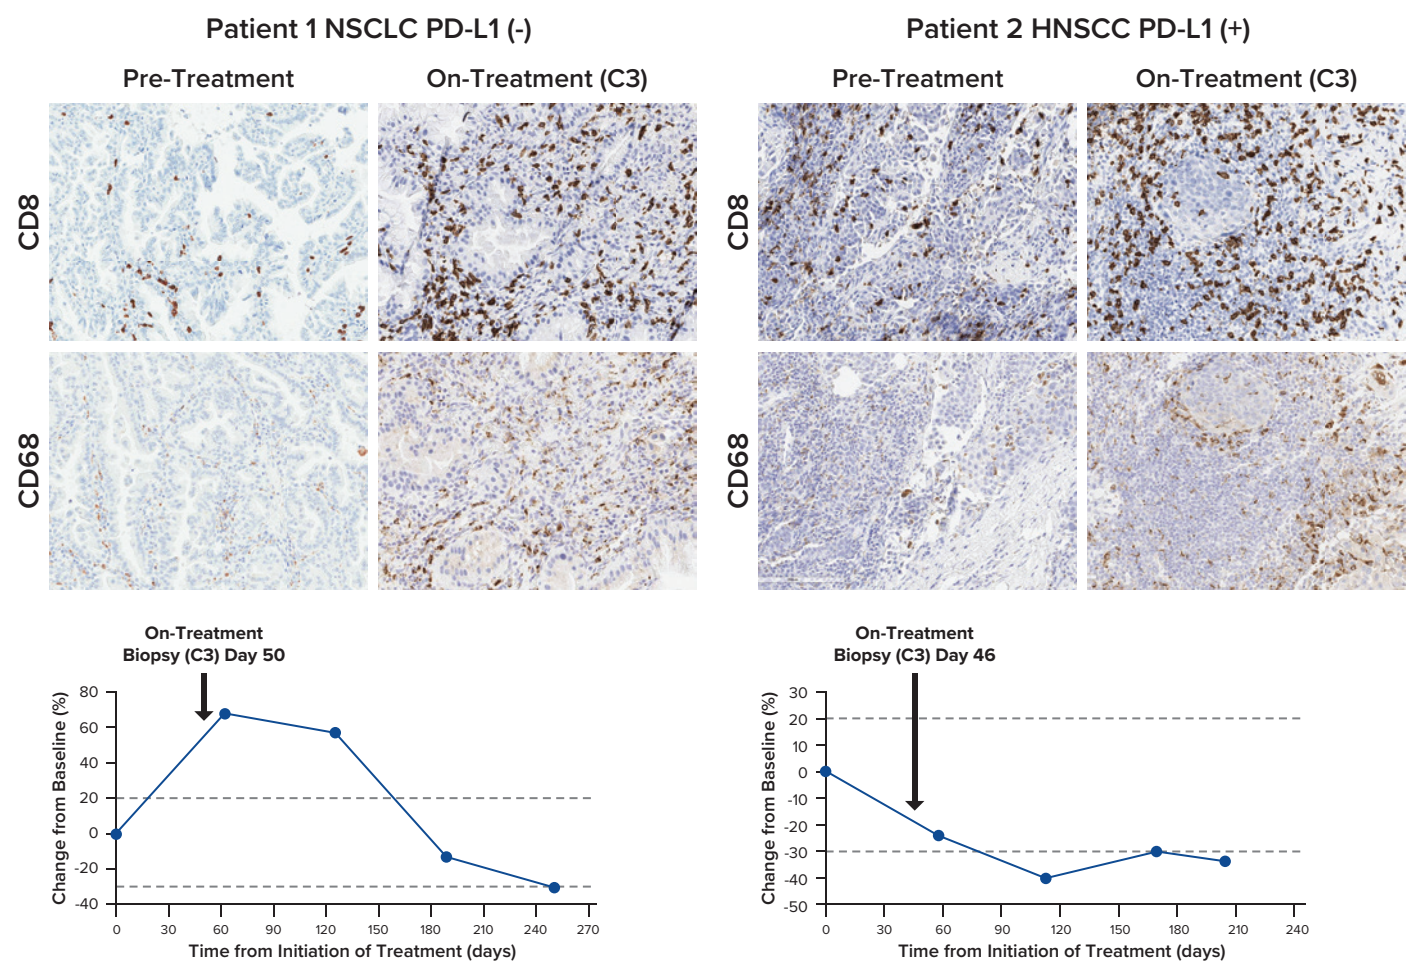
Patient 1 NSCLC PD-L1 (-)
Patient 2 HNSCC PD-L1 (+)
Pre-Treatment
On-Treatment (C3)
CD8
CD68
On-Treatment Biopsy (C3) Day 50
On-Treatment Biopsy (C3) Day 46
Change from Baseline (%)
Time from Initiation of Treatment (days)

Conclusions

Intended for combination with anti-cancer antibodies, ALX148 is designed to avoid the dose-limiting toxicities associated with other CD47-targeted approaches while maximizing the innate and adaptive immune response to cancer.

- ALX148 (10 mg/kg QW; molar equivalent to 20 mg/kg of an antibody) in combination with standard regimens of trastuzumab or pembrolizumab is well tolerated with a favorable hematologic safety profile.
- ALX148 demonstrates emerging anti-cancer activity in combination with trastuzumab in patients with HER2 positive Gastric/GEJ tumors that have progressed on prior HER2 targeted therapies.
- ALX148 demonstrates emerging anti-cancer activity in combination with pembrolizumab in patients with:
 - ≥2L HNSCC that compares favorably to the pembrolizumab single agent experience.
 - NSCLC including tumors resistant/refractory to prior checkpoint inhibitor therapy.
- ALX148 demonstrates antibody-like PK and complete CD47 target occupancy in combination with trastuzumab or pembrolizumab.
- Preliminary data from paired tumor biopsies suggests increased intra-tumoral macrophages and CD8+ T cells following ALX148 treatment.

Patients in combination cohorts continue to be followed (NCT03013218).

References

1. Weiskopf, K., *Eur J Cancer*. 2017 May;76:100-109.
2. Kauder, S.E. et al., *PLoS ONE*. 2018 August;13(8): e0201832.
3. Lakhani N. et al, #P335, SITC 2018.
4. Jin F. et al., #P340, SITC 2018.

Acknowledgments

- We would like to thank all of the participating patients and their families as well as site research staff.
- **Contact email:** info@alxoncology.com
- Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, IL, USA, May 31, 2019 - June 4, 2019.